PB0325

Exposure-response modeling of emicizumab▼ for the prophylaxis of bleeding in haemophilia A patients with and without inhibitors against factor VIII

Fredrik Jonsson[1], Christophe Schmitt[2], Claire Petry[2], Francois Mercier[2], Nicolas Frey[2], Sylvie Retout[2]

[1]*qPharmetra, Cary, NC, USA;* [2]*F. Hoffmann-La Roche, Basel, Switzerland*

BACKGROUND

- Emicizumab is a bispecific humanised monoclonal antibody that binds activated factor IX (FIXa) and FX to restore the function of missing activated FVIII in persons with haemophilia A (PwHA).[1]
- Emicizumab demonstrated efficacy in bleed prevention when administered subcutaneously once weekly (QW) at 1.5 mg/kg, every two weeks (Q2W) at 3 mg/kg, and every four weeks (Q4W) at 6 mg/kg in PwHA with or without FVIII inhibitors.[1–4]
- These dosing regimens were selected based on an exposure-repeated time-to-event model[5] developed from bleeding data collected in a phase I/II study in three groups of six PwHA each who received weekly subcutaneous maintenance doses of either 0.3, 1, or 3 mg/kg.
- With this analysis, we update the exposure–response relationship for bleeding events using a much larger database including data from PwHA from phase I/II and phase III studies.
- We also investigated factors that could contribute to between-patient variability in the expected treatment response, and assess the impact of changing the dosing regimen from weekly to Q2W or Q4W on the expected treatment response.

METHODS

- A population pharmacokinetic (PK) model was developed based on data from the phase I/II study and four phase III studies.[6]
- This population PK model was used to predict typical concentration–time profiles at steady state (**Figure 1**).
- Bleeding event data were pooled from 445 PwHA with or without inhibitors participating in six clinical studies (**Table 1**).
- Those bleeding event data were analysed as count data using a pharmacokinetic/pharmacodynamic (PK/PD) model implemented in NONMEM version 7.3 (ICON Development Solutions).
- The model used herein included an E_{max} relationship for the effect of daily emicizumab concentrations on the mean daily bleed count (λ), with the maximal effect parameter fixed to 1.

Figure 1. Predicted concentration–time profiles for emicizumab under different regimens

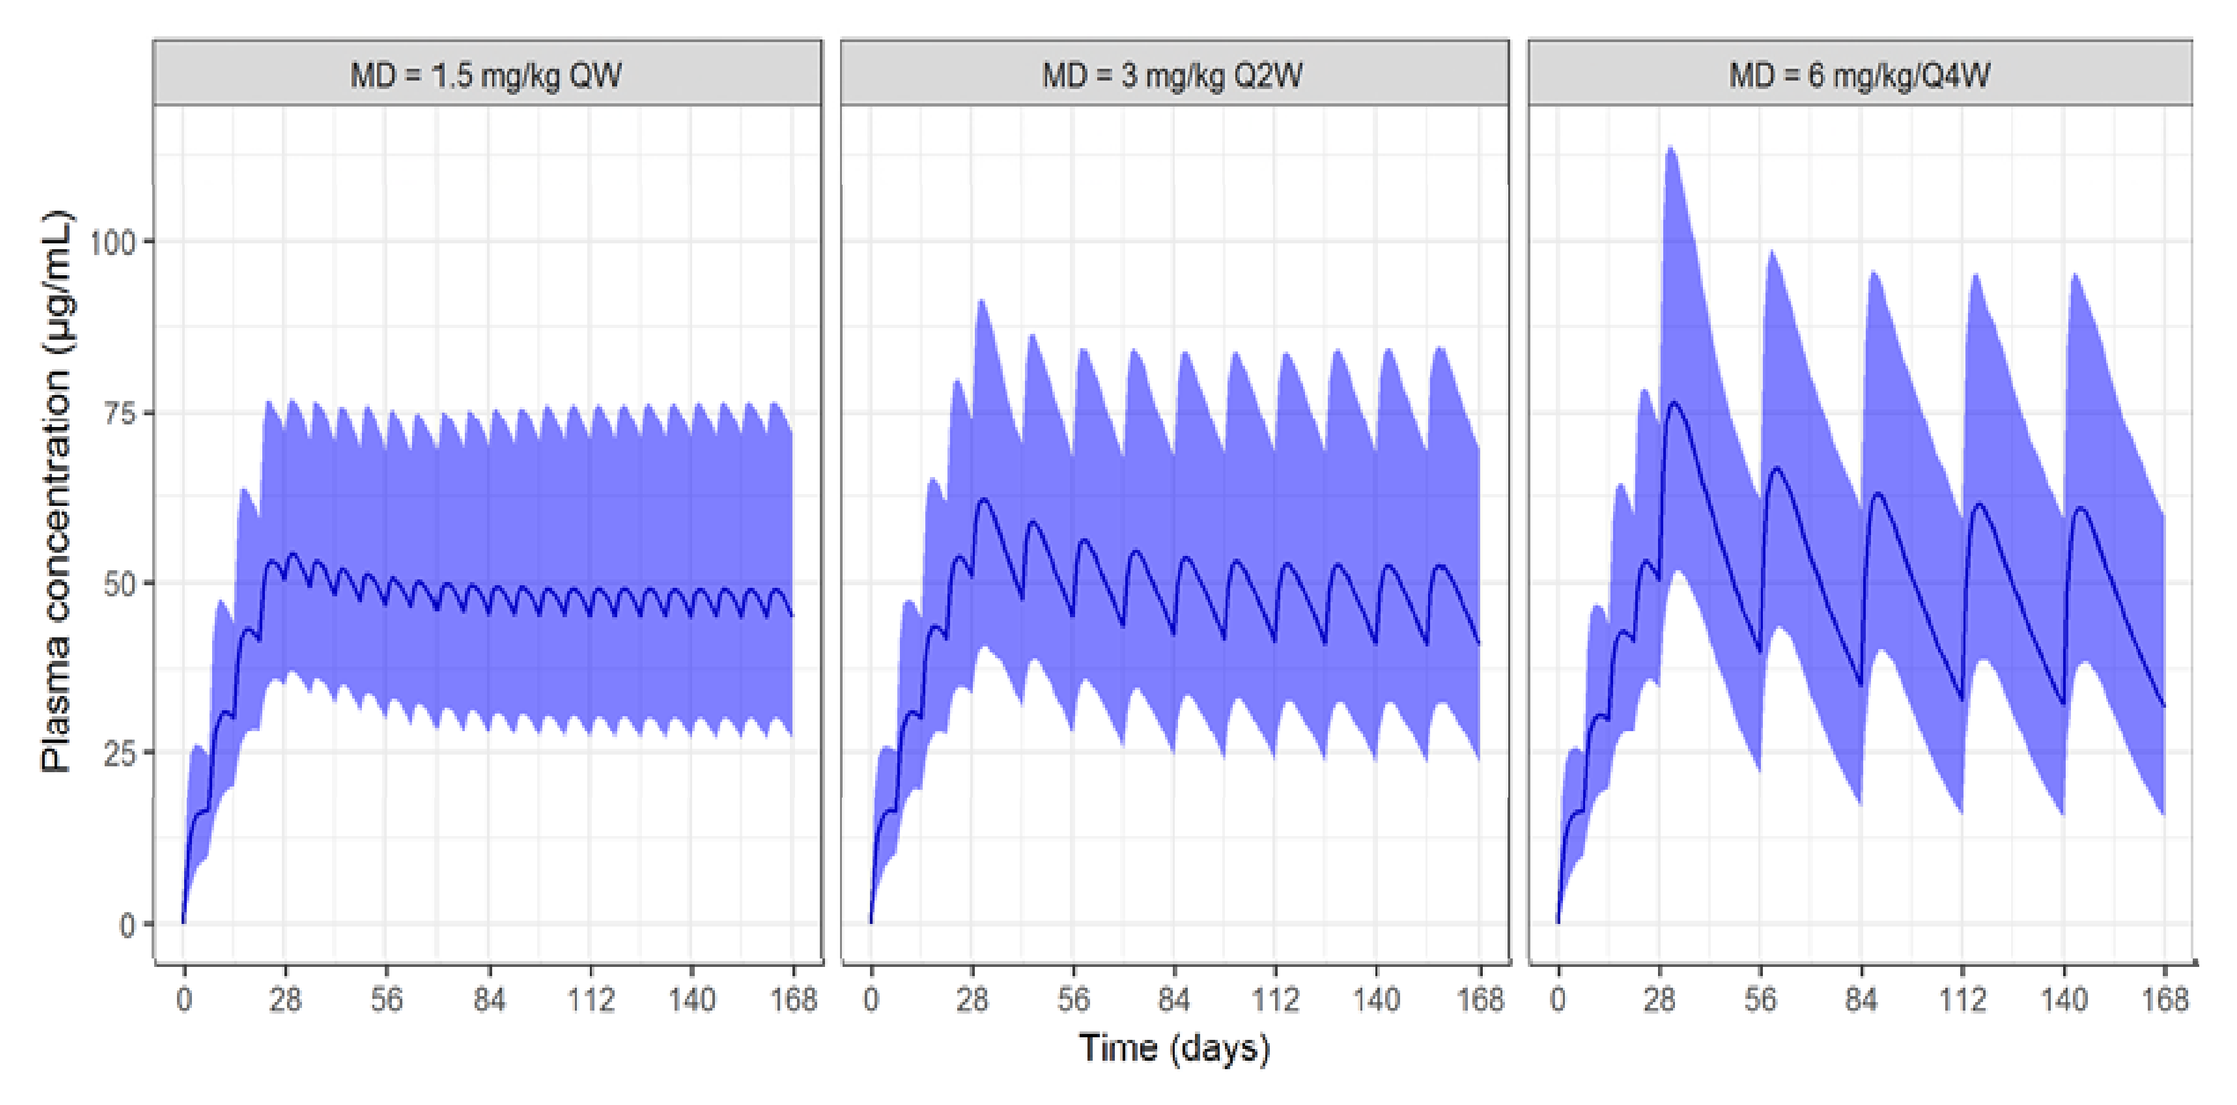
MD = 1.5 mg/kg QW
MD = 3 mg/kg Q2W
MD = 6 mg/kg/Q4W
Plasma concentration (µg/mL)
Time (days)

MD, maintenance dose. Blue curve: median of predictions; Blue shaded area: 90% prediction intervals

- The likelihood of having a bleeding event was modelled as count data, using of a constant 'baseline' bleeding count λ_{base} (mean daily bleed count with no prophylaxis) added to a time-dependent effect of emicizumab and other prophylactic treatment.
- Covariates such as concentration of FIX and FX antigens, dosing regimen or number of bleeds prior to emicizumab administration were tested on EC_{50}.

$$\lambda_{i,t} = \lambda_{base,i} \times \left(1 - \frac{\frac{C_{emi,i,t}}{EC_{50,i}} + PLX_{i,t} \times \theta_{PLX}}{1 + \frac{C_{emi,i,t}}{EC_{50,i}} + PLX_{i,t} \times \theta_{PLX}}\right)$$

$$P_{i,t}(Y = y) = \frac{\lambda_{i,t} \times (\lambda_{i,t} + y \times \delta)^{y-1} \times e^{-\lambda_{i,t} - y \times \delta}}{y!}$$

- Wherein:

$\lambda_{i,t}$	Bleeding count on day t for patient i
$\lambda_{base,i}$	Baseline λ_{base} for patient i
$C_{emi,i,t}$	Predicted plasma concentration of emicizumab on day t for patient i
$EC_{50,i}$	EC_{50} for patient i
$PLX_{i,t}$	Status of ongoing/no use of FVIII prophylactic treatment at time t for patient i (the first week of emicizumab treatment may be concomitant to the effect of FVIII prophylactic treatment)
θ_{PLX}	Effect of another prophylactic treatment on bleeding hazard
$P_(i,t)(Y=y)$	Probability of observing y bleeding events on day t for patient i
δ	Dispersion factor for the generalised Poisson distribution

- The model was subsequently used to simulate the relationship between emicizumab concentration and bleeding count over 1 year (equivalent to annualised bleeding rate [ABR]).

Table 1. Clinical studies included in this analysis

Study [Ref.no]	ACE002JP[7,8]*	HAVEN 1[1]	NIS[9]	HAVEN 2[2]*	HAVEN 3[3]	HAVEN 4[4]	Total
Emicizumab dosing regimen	Cohort 1: 1 mg/kg followed by 0.3 mg/kg QW Cohort 2: 3 mg/kg followed by 1 mg/kg QW Cohort 3: 3 mg/kg QW	3 mg/kg QW for 4 weeks followed by 1.5 mg/kg QW	NIS; no emicizumab given	3 mg/kg QW for 4 weeks followed by 1.5 mg/kg QW	Arms A and D: 3 mg/kg QW for 4 weeks followed by 1.5 mg/kg QW Arms B and C†: 3 mg/kg QW for 4 weeks followed by 3 mg/kg Q2W	Run-in cohort: 6 mg/kg QW Expansion cohort: 3 mg/kg QW for 4 weeks followed by 6 mg/kg Q4W	
N	16‡	109	60§	61	151¶	48	**445**
Age, year, median (range)	34.5 (12–58)	28 (12–75)	26 (2–75)	7.12 (1.22–15.7)	38 (13–77)	38 (14–68)	**29 (1.22–77)**
Body weight, kg, median (range)	63.4 (40.8–81.7)	70 (40.1–156)	65 (13.5–111)	22.6 (9.5–53)	76.5 (43–139)	74.2 (43.3–102)	**68.5 (9.5–156)**
FVIII inhibitors, present/absent, n	9/7	109/0	40/20	61/0	0/151	8/40	**227/218**
Patients with up-titration, n	4	5	0	0	5	0	**14**
Bleeding events per patient during non-intervention, median (range)	9.5 (0–38)	5 (0–35)	4 (1–33)	5 (1–12)	0.5 (0–36)	–	**4 (0–38)**
Bleeding events per patient during intervention, median (range)	2 (0–74)	0 (0–65)	–	0 (0–2)	0 (0–13)	0 (0–18)	**0 (0–74)**
Study non-intervention period duration, days, median (range)	168 (168–186)	145 (0–650)	90 (0–294)	0 (0–312)	168 (0–382)	–	**85 (0–650)**
Study intervention period duration, days, median (range)	1380 (248–1570)	434 (27–659)	–	202 (56–440)	206 (0–351)	168 (164–308)	**210 (0–1570)**

NIS, non-interventional study. For all studies except NIS, emicizumab was administered as a subcutaneous injection.*Two patients from ACE002JP and two from HAVEN 2 were excluded from the population PK data set; †Following an initial period of 24 weeks of no prophylaxis, participants in Arm C were eligible to switch to emicizumab prophylaxis; ‡16/18 total participants; §60/94 total participants; ¶151/152 total participants

RESULTS

- The estimated EC_{50} (3.89 µg/mL), was low compared with the expected concentration range at steady state under the intended dosing regimens, which tend to be distributed around 50 µg/mL (**Table 2 and Figure 1**).

Table 2. Parameter estimates of the final model

Parameter	Estimate	Relative standard error (%)	95% confidence interval
λ (mean daily bleed count)	0.0225	7.07	[0.0194; 0.0256]
δ (dispersion parameter of Generalised Poisson distribution)	0.0238	13.7	[0.0174; 0.0302]
Effect of prophylactic treatment among non-inhibitor patients	0.797	2.01	[0.766; 0.828]
EC_{50} (µg/mL)	3.89	3.50	[3.62; 4.16]
ω^2_λ Interindividual variability in mean daily bleed count	1.19	7.66	[1.01; 1.37]

- The predictive performance of the count data model were evaluated using a simulation approach; it adequately predicted the bleeding onset over time, before and after the start of emicizumab treatment.
 - The present analysis, while confirming previous findings from phase I/II data,[5] considerably improves the precision of the estimates.
- The simulated mean ABR in the absence of emicizumab or any other prophylactic treatment was 17.3. This decreased with increasing emicizumab concentration, with a relatively flat relationship for concentrations above approximately 30 µg/mL, where the predicted mean ABR was <1.83, corresponding to a >89.4% reduction.
 - These concentrations were achieved with all three dosing regimens (**Figure 2**).
- Previous prophylaxis with FVIII treatment had, as expected, a profound effect on bleeding rate at baseline, with a 44% (θ_{PLX} = 0.797) difference in bleeding hazard between patients with and without FVIII prophylaxis.
- Owing to the mechanism of action of emicizumab, there were no reasons to expect a different PK/PD relationship between patients with and without FVIII inhibitors. *De facto*, the model nicely described the PK/PD relationship for all patients.
- Of note, none of the covariates tested (see methods) had a significant impact on the EC_{50} of the PK/PD relationship.
 - Likewise, the frequency of dosing regimens did not impact the PK/PD relationship nor the treatment response.
- An analysis of the timing of bleeding events in HAVEN 3 (which included QW and Q2W dosing, analysed separately) found no evidence of a connection between occurrence of bleeding and timing of the last emicizumab dose.[10]

▼ This medicinal product is subject to additional monitoring. This will allow quick identification of new safety information. Healthcare professionals are asked to report any suspected adverse reactions. These should be reported to the Regulatory authorities in your country according to your national requirements.

Figure 2. Simulated exposure–response of emicizumab

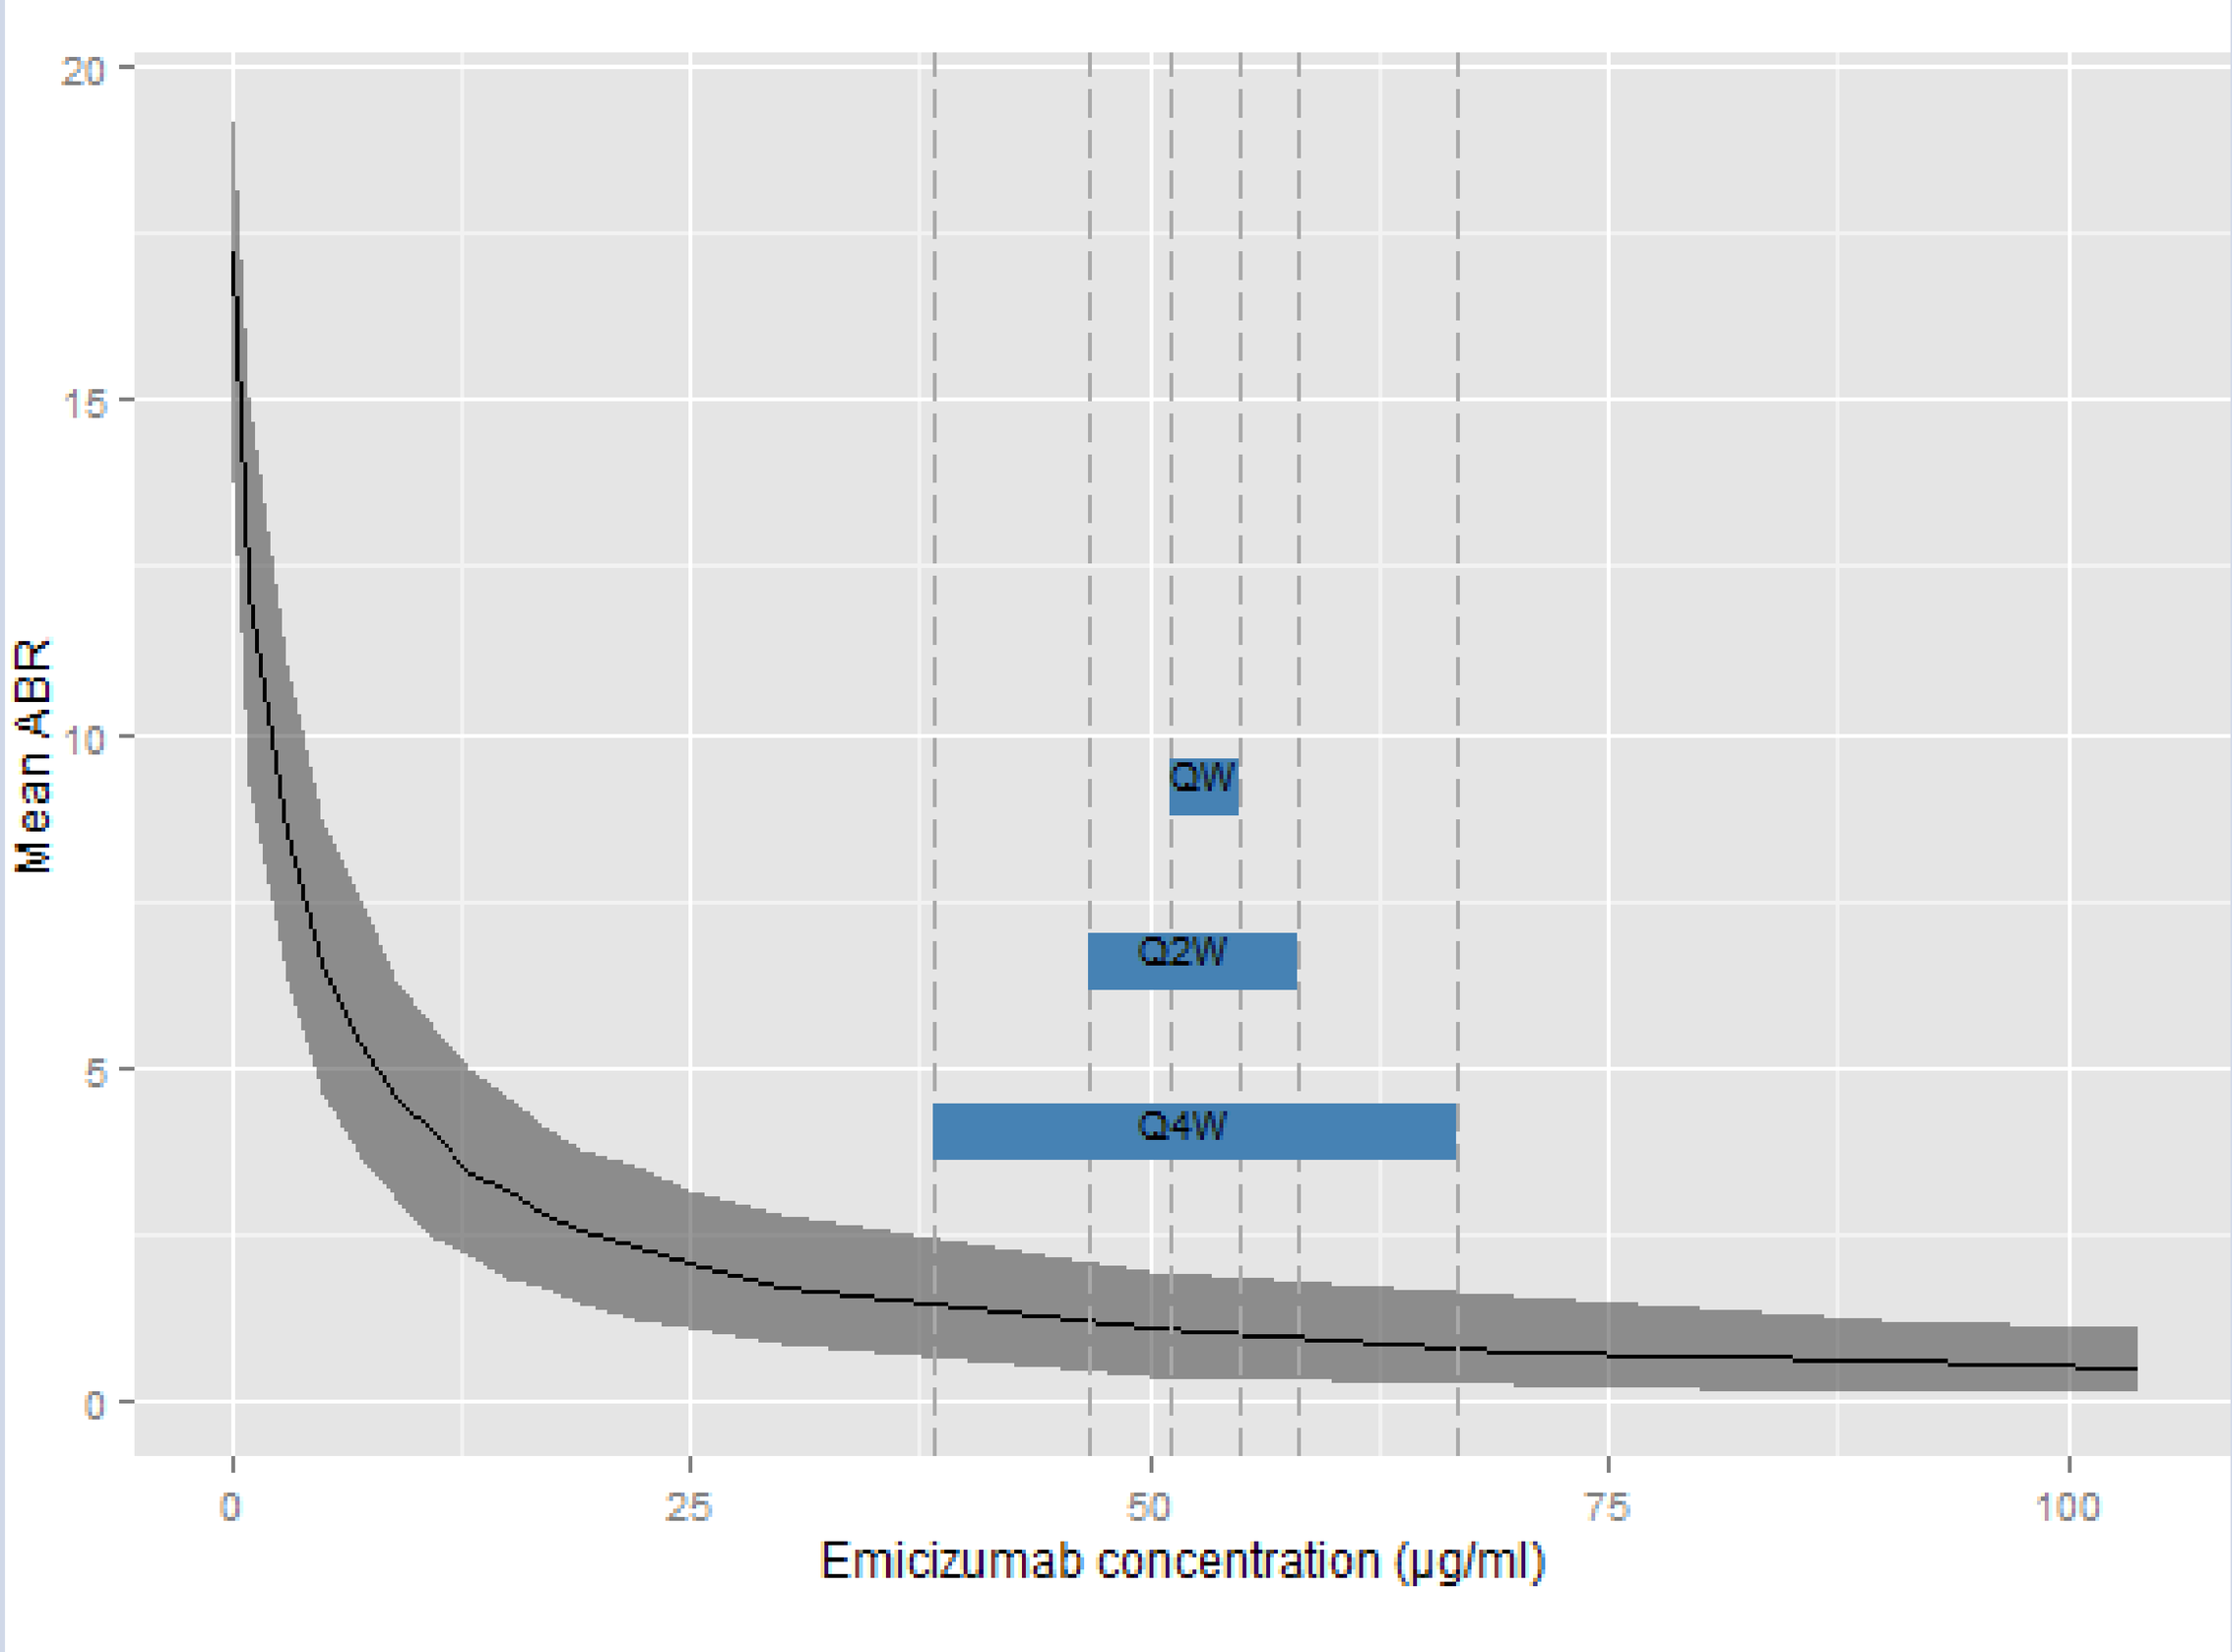
Mean ABR
QW
Q2W
Q4W
Emicizumab concentration (µg/ml)

Envelope around the simulated means indicate 2.5th and 97.5th percent boundaries of the prediction intervals. Horizontal blue bars indicate the ranges of predicted concentrations in typical patients at maintenance doses of 1.5 mg/kg QW, 3 mg/kg Q2W and 6 mg/kg Q4W

CONCLUSIONS

- The relationship between emicizumab PK and occurrence of bleeding events was adequately characterised by a count data model.
- Previous prophylactic treatment with FVIII reduced the baseline bleeding rate but did not impact the overall PK/PD relationship.
- None of the covariates tested (concentration of FIX and FX antigens, dosing regimen or number of bleeding events prior to emicizumab administration) had a significant impact on the overall PK/PD relationship.
- No factors were identified that could contribute to between-patient variability in the expected treatment response.
- Concentrations above 30 µg/mL are predicted to provide clinically meaningful control of bleeding. Effective concentrations were achieved with all three approved therapeutic dosing regimens.

ACKNOWLEDGEMENTS

Editorial assistance for this poster was provided by Rebecca A. Bachmann, PhD and was funded by F. Hoffman-La Roche Ltd. This
study was funded by F. Hoffman-La Roche Ltd and qPharmetra, LLC.

REFERENCES

1. Oldenburg J, et al. N Engl J Med 2017;377:809–18.
2. Young G, et al. Blood 2019 [submitted].
3. Malhangu J, et al. N Engl J Med 2018;379:811–22.
4. Pipe S, et al. Lancet Haematol 2019;doi: 10.1016/S2352-3026(19)30054-7.
5. Yoneyama K, et al. Clin Pharmacokinet 2018;57:1123–34.
6. Retout S, et al. Clin Pharmacokinet 2019 [submitted].
7. Shima M, et al. N Engl J Med 2016;374:2044–53.
8. Shima M, et al. Blood Adv 2017;1:1891–99.
9. Kruse-Jarres R, et al. Haemophilia 2019;25:213–20.
10. Mahlangu J, et al. ISTH 2019; poster PB0694.

PUSHED FOR TIME?

Receive a copy of this poster
http://bit.ly/2NePGpK

Request additional presentations of trials sponsored/supported by Roche at this congress
RocheMedically@gcc-global.com

Presented at the International Society on Thrombosis and Haemostasis (ISTH) Congress | Melbourne, Australia | 6–10 July 2019