FLOODLIGHT: Remote Self-Monitoring is Accepted by Patients and Provides Meaningful, Continuous Sensor-Based Outcomes Consistent With and Augmenting Conventional In-Clinic Measures

X Montalban,[1,2] P Mulero,[2] L Midaglia,[2] J Graves,[3] SL Hauser,[3] L Julian,[4] M Baker,[5] J Schadrack,[5] C Gossens,[5] A Scotland,[5] F Lipsmeier,[5] G Pointeau,[5] C Bernasconi,[5] S Belachew,[5] M Lindemann[5,6]

[1]Division of Neurology, University of Toronto, Toronto, ON, Canada; [2]Multiple Sclerosis Centre of Catalonia, Vall d'Hebron University Hospital, Barcelona, Spain; [3]University of California, San Francisco, San Francisco, CA, USA; [4]Genentech, Inc., South San Francisco, CA, USA; [5]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [6]Baden-Wuerttemberg Cooperative State University, Loerrach, Germany

BACKGROUND

- Conventional assessment of the clinical course of multiple sclerosis (MS) relies on relapse-associated and periodic 6- to 12-monthly follow-up clinical visits
- Smartphone-based, high-yield active and passive monitoring assessments may enhance clinical disease assessments,[1] compared with conventional methods for detecting subtle progressive clinical changes that may predict disease activity and long-term disease progression[2–6]
- Primary objectives of the FLOODLIGHT study (NCT02952911):
 - — Adherence to smartphone- and smartwatch-based assessments
 - — Feedback from patients and healthy controls (HCs) on the smartphone and smartwatch schedule of assessments and the impact on their daily activities, using a satisfaction questionnaire
- Other objectives are to determine the association between the FLOODLIGHT Test Battery and conventional MS clinical outcomes, and establish whether the FLOODLIGHT Test Battery can differentiate between participants with and without MS

OBJECTIVE

- To report interim analysis of adherence, results from a patient satisfaction questionnaire (PSQ) and correlations between smartphone-based and in-clinic tests from FLOODLIGHT

METHODS

Study Design

- The FLOODLIGHT study is a prospective pilot study to assess the feasibility of remote patient monitoring (RPM) using smartphones and smartwatches in patients with MS and HCs (**Table 1**)
- Eligible patients and HCs were enrolled in the study and will be assessed clinically at baseline, Week 12 and Week 24
- At the enrollment visit, participants were provided with a remote monitoring solution that includes:
 - — A preconfigured smartphone and smartwatch containing application software that prompts the user to perform various assessments, referred to as "active tests" (**Figure 1**)
 - — The application also passively records sensor data, referred to as "passive monitoring"

For smartphone assessment schedule (Figure 1) please scan here

Table 1. Demographics and characteristics for study participants at baseline

Parameter	Healthy controls (N=25)	Patients with MS (N=76)
Age, years[a]	34.92 ± 9.35	39.55 ± 7.85
Female, %	28.00	69.74
MS diagnosis (PPMS, SPMS, RRMS), %	NA	3.95, 5.26, 90.79
Time since MS symptom onset, years[a]	NA	12.04 ± 6.91
EDSS[a]	NA	2.42 ± 1.35
9HPT, seconds[a]	19.18 ± 1.86	22.31 ± 4.07
T25FW, seconds[a]	5.02 ± 0.95	6.04 ± 2.09
BBS[a]	56.00 ± 0	52.54 ± 5.72

Data cut-off: March 23, 2018.
[a]Values are stated as mean ± SD.
9HPT, 9-Hole Peg Test; BBS, Berg Balance Scale; EDSS, Expanded Disability Status Scale; MS, multiple sclerosis; NA, not applicable; PPMS, primary progressive multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis; SPMS, secondary progressive multiple sclerosis; T25FW, Timed 25-Foot Walk.

RESULTS

Adherence to Active Tests and Passive Monitoring

- Only patients with MS who completed the 24-week study were included in this adherence data analysis (n=53) to account for pending data retrieval at the interim analysis point
- Adherence to active tests was measured as proportion of study weeks with at least 3 days of completed testing (study co-primary endpoint):
 - — All active tests: 77.36%
 - — All active tests except Two-Minute Walk Test (2MWT): 81.45%
 - — 2MWT: 79.11%
- Adherence to smartphone-based passive monitoring was 83.25%, measured as proportion of study weeks with at least 3 days of at least 4 hours/day (study co-primary endpoint)
- The Kruskal–Wallis test was used to compare groups with up to five categories
 - — The association between continuous variables was assessed using Spearman's rank correlation
- No strong associations between FLOODLIGHT RPM adherence and in-clinic assessments (EDSS, FSMC, MSIS-29, PHQ-9, T25FW, 9HPT, SDMT)[a] and characteristics (age, sex, height, weight, body mass index) were found
- Weak association was found for gender with respect to active test adherence (p=0.010; mean: male=68.46%, female=82.34%) and smartwatch adherence (p=0.020; mean: male=76.43%, female=88.41%)

[a]9HPT, 9-Hole Peg Test; EDSS, Expanded Disability Status Scale; FSMC, Fatigue Scale for Motor and Cognition Function; MSIS, Multiple Sclerosis Impact Scale; PHQ, Patient Health Questionnaire; SDMT, Symbol Digit Modalities Test; T25FW, Timed 25-Foot Walk.

Satisfaction Questionnaire: Satisfaction Was Good to Excellent and Remained Stable

- The satisfaction questionnaire was assessing patients' and HCs' experience regarding smartphone and smartwatch use and its impact on their daily activities at the Week 12 visit and at study termination/early discontinuation visit
- A score was developed that sums the individual answers to all questions rescaled to 0–100 from their original Likert scale
- Satisfaction amongst patients with MS that completed the study (n=61) was good to excellent
 - — The average satisfaction score at the Week 12 visit was 74.62 out of a possible 100, and remained stable at Week 24 with 74.63 out of 100 (Wilcoxon signed-rank test p=0.41)

Performance Measures From FLOODLIGHT Active Tests at Baseline

- The performances of a set of heuristic single features derived from FLOODLIGHT active tests were assessed at baseline[a]
 - — Differences in FLOODLIGHT smartphone-based measures between HCs and patients with MS was tested using the Mann–Whitney U test
 - — Agreement between FLOODLIGHT smartphone-based measures and conventional clinical assessments was studied using Spearman's rank correlation
- FLOODLIGHT smartphone-based measures were averaged over the first week to account for missing data
- All FLOODLIGHT smartphone-based measures (except Static Balance Test [SBT]) showed significant differences between patients with MS and HCs (**Table 2A**) and/or significant agreement with the conventional corresponding clinical outcome measure (**Table 2B**)
- The non-significant results observed when exploring the Sway from the SBT vs the Berg Balance Scale (BBS) may be due to the near-complete absence of clinically apparent balance impairment in the population of patients with MS and the ceiling effect of the BBS in this setting (BBS = 52.54 ± 5.72 for patients with MS for a maximal score of 56 on this scale)

[a]Baseline: average measures over the first week.

Table 2. Examples of active test performances at baseline[a] across FLOODLIGHT Test Battery

A: Domain	A: FLOODLIGHT smartphone test	A: Feature	Healthy controls Mean	Healthy controls SD	Healthy controls N	Patients with MS Mean	Patients with MS SD	Patients with MS N	HCs vs patients with MS[b] p value	B: MS clinical outcome measure	Correlation with MS clinical outcome measure[c] R^2	Correlation p value	Correlation N
Cognition	SDMT	Correct responses	49.79	5.76	17	42.64	9.66	61	**0.003**	**SDMT**	0.625	**<0.001**	61
Hand & arm	Pinching Test	Time between two consecutive pinches (seconds)	0.41	0.04	17	0.55	0.12	64	**<0.001**	**9HPT**	0.477	**<0.001**	62
	Draw a Shape Test	Overall mean celerity	0.30	0.07	17	0.25	0.07	63	**0.021**		−0.177	0.17	61
Gait & posture	SBT	Sway[d]	10.97	12.51	17	11.91	11.30	64	0.35	**BBS**	−0.089	0.49	61
	5UTT	Turn speed (degree/second)	84.17	9.99	17	79.35	18.15	64	0.14	**T25FW**	−0.391	**0.002**	63
	2MWT	Step power	4.81	2.57	16	3.65	2.16	63	0.10		−0.421	**<0.001**	62

[a]Baseline: average measures over the first week; [b]Mann–Whitney U test; [c]Spearman's rank correlation; [d]Sway is the total length of accelerometer trajectory.
2MWT, Two-Minute Walk Test; 5UTT, Five U-Turn Test; 9HPT, 9-Hole Peg Test; BBS, Berg Balance Scale; HCs, healthy controls; MS, multiple sclerosis; N, number of observations; SBT, Static Balance Test; SD, standard deviation; SDMT, Symbol Digit Modalities Test; T25FW, Timed 25-Foot Walk.

Cognition: Smartphone-Based Symbol Digit Modalities Test

- The number of correct responses from the FLOODLIGHT smartphone-based Symbol Digit Modalities Test (SDMT)[7] was expectedly lower in patients with MS vs HCs (**Table 2A**) and correlated well with the in-clinic–performed oral SDMT score measured at baseline[a] in patients with MS (**Table 2B, Figure 2A**)
- The number of correct responses from the smartphone-based SDMT also showed significant association with the Multiple Sclerosis Impact Scale (MSIS)-29 Psychological Score at baseline[a] (**Figure 2B**)

[a]Baseline: average measures over the first week.

Figure 2. Correct responses of smartphone-based SDMT correlate with oral SDMT and patients' perceived mental/psychological status measured with the MSIS-29 at baseline[a]

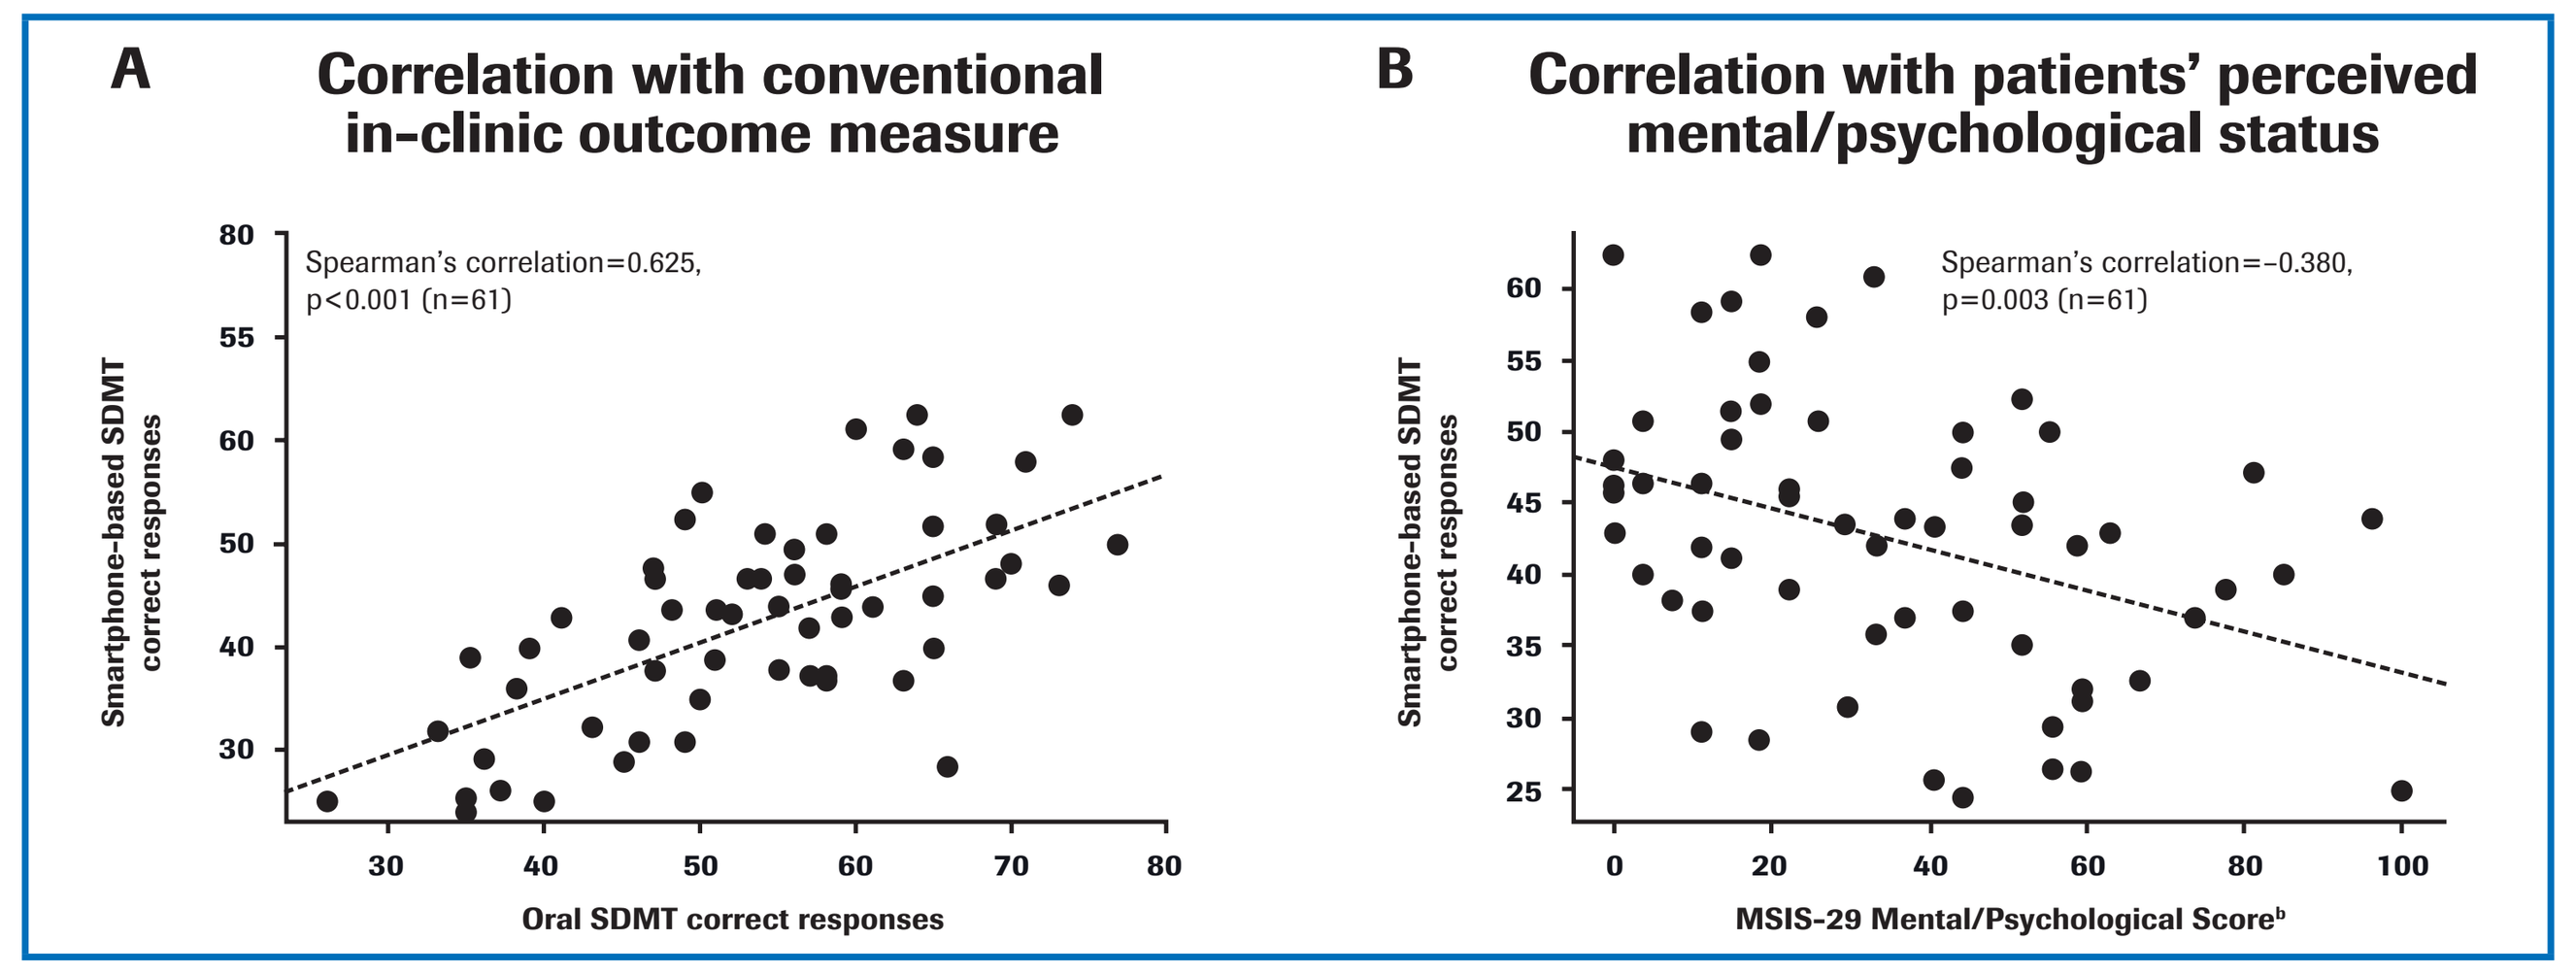
A
Correlation with conventional in-clinic outcome measure
Spearman's correlation=0.625, p<0.001 (n=61)
Smartphone-based SDMT correct responses
Oral SDMT correct responses
B
Correlation with patients' perceived mental/psychological status
Spearman's correlation=−0.380, p=0.003 (n=61)
MSIS-29 Mental/Psychological Score[b]

[a]Baseline: average measures over the first week; [b]Items 21 to 29 of the MSIS-29.
MSIS, Multiple Sclerosis Impact Scale; SDMT, Symbol Digit Modalities Test.

Hand Motor Function: Pinching Test

- In the Pinching Test, subjects were asked to pinch as many tomatoes as possible in 30 seconds
- Baseline performance on the Pinching Test measured by the time between two consecutive pinch attempts revealed differences between HCs and patients with MS (**Table 2A, Figure 4B**), and significant correlation with 9-Hole Peg Test (9HPT) times in patients with MS (**Table 2B, Figure 3A**) as well as with patients' perceived hand/arm/upper limb function[a] (**Figure 3B**)
- Significant group differences between HCs and patients with MS were also observed on 9HPT times (**Figure 4A**) measured at baseline[b]
- Grouping patients with MS into presumably normal and impaired hand/arm function based on an upper limit of normal range defined as the average 9HPT time for HCs plus two standard deviations[b] (23.91 seconds),[8] we observed that patients with MS with normal hand/arm function were discriminated from HCs by a significantly longer mean time between two consecutive pinch attempts (**Figure 4C**)

[a]Items 2, 6 and 15 of the MSIS-29; [b]Baseline: average measures over the first week.

Figure 3. Mean time between two consecutive pinch attempts correlates with average 9HPT and patients' perceived hand/arm/upper limb function

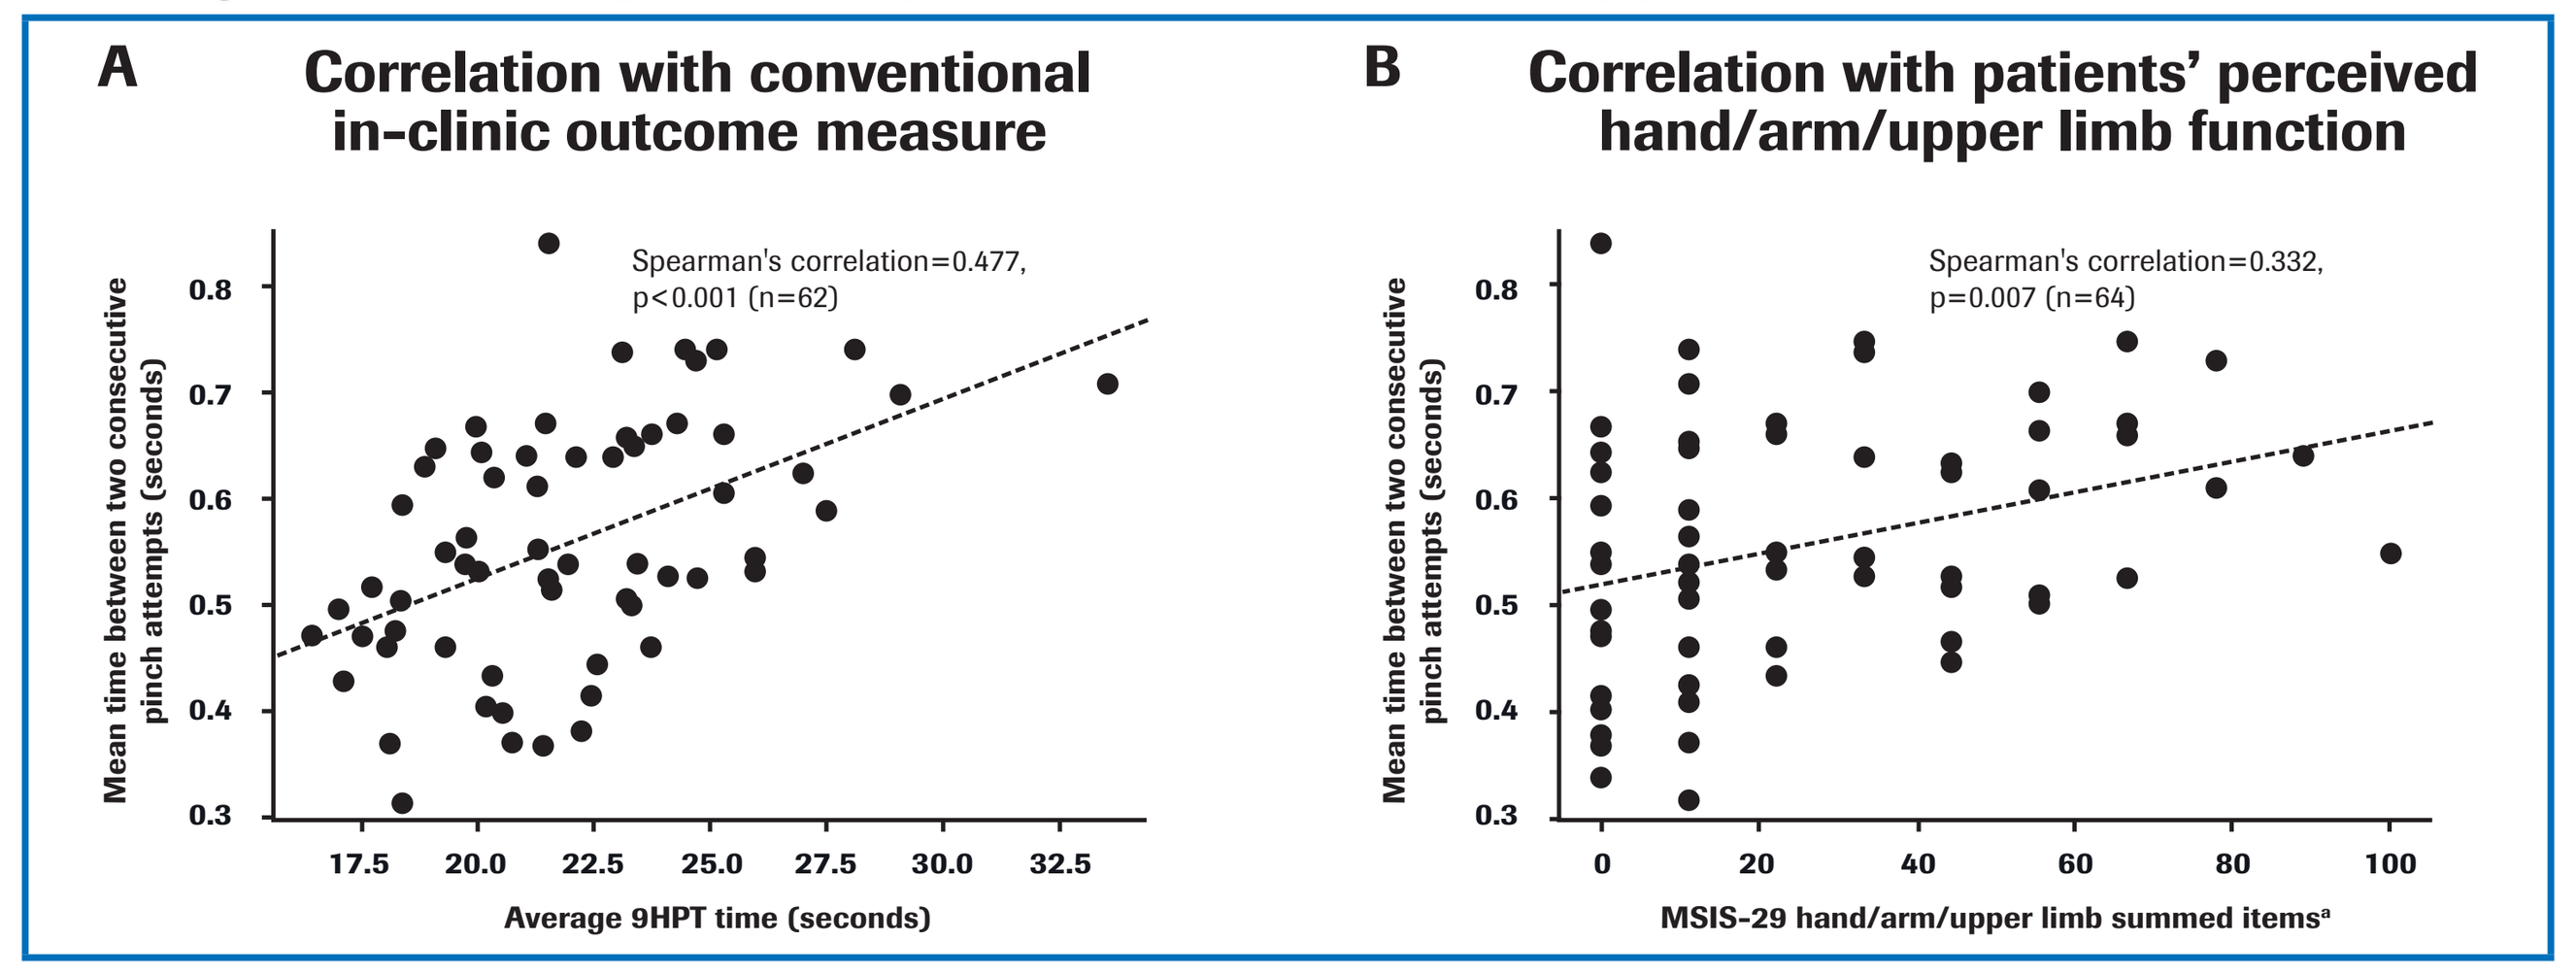
A
Correlation with conventional in-clinic outcome measure
Spearman's correlation=0.477, p<0.001 (n=62)
Mean time between two consecutive pinch attempts (seconds)
Average 9HPT time (seconds)
B
Correlation with patients' perceived hand/arm/upper limb function
Spearman's correlation=0.332, p=0.007 (n=64)
MSIS-29 hand/arm/upper limb summed items[a]

[a]Items 2, 6 and 15 of the MSIS-29 summarized and rescaled to [0, 100].
9HPT, 9-Hole Peg Test; MSIS, Multiple Sclerosis Impact Scale.

Figure 4. A sensitive assessment of Hand Motor Function through the Pinching Test

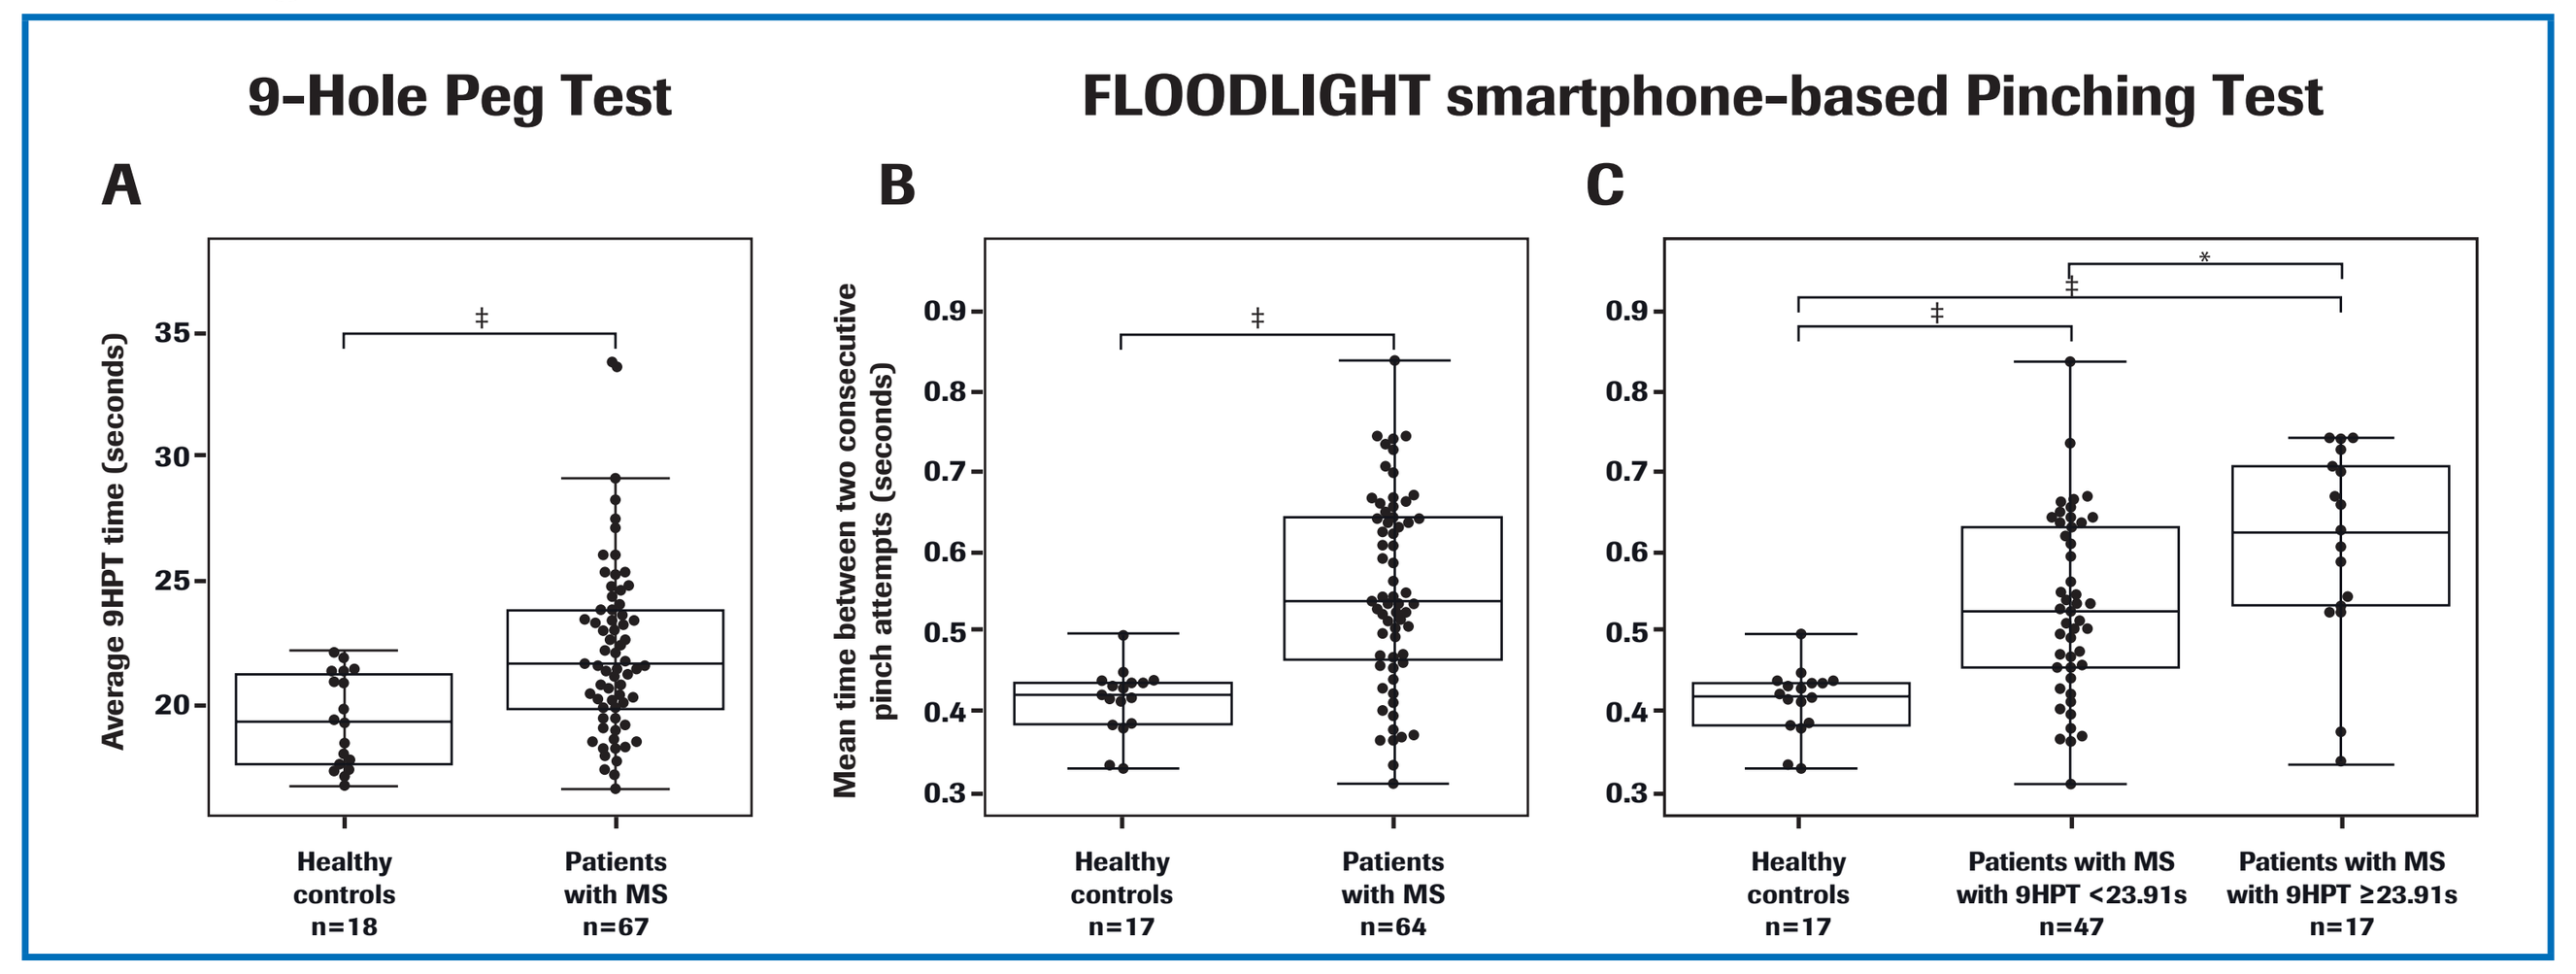
9-Hole Peg Test
FLOODLIGHT smartphone-based Pinching Test
A
Average 9HPT time (seconds)
Healthy controls n=18
Patients with MS n=67
B
Mean time between two consecutive pinch attempts (seconds)
Healthy controls n=17
Patients with MS n=64
C
Healthy controls n=17
Patients with MS with 9HPT <23.91s n=47
Patients with MS with 9HPT ≥23.91s n=17

*p<0.05; ‡p<0.001.
9HPT, 9-Hole Peg Test; MS, multiple sclerosis; s, seconds.

Hand Motor Function: Draw a Shape Test

- In the Draw a Shape Test, subjects were instructed to hold the mobile device in the untested hand and draw six prewritten alternating shapes of increasing complexity (linear, rectangular, circular, figure 8-shape and spiral) on a touchscreen of the mobile device with the second finger of the tested hand "as fast and as accurately as possible" within 30 seconds, and passing through all indicated waypoints
- Baseline[a] performance on the Draw a Shape Test measured by the overall mean celerity (derived from the accuracy to connect all waypoints relative to the time to complete the test) revealed differences between HCs and patients with MS (**Table 2A**)
 - — In contrast, the overall mean celerity in the Draw a Shape Test was not significantly correlated with 9HPT time in patients with MS (**Table 2B**), suggesting that this heuristic feature of the Draw a Shape Test may measure a different aspect of hand/finger motor function compared with 9HPT

[a]Baseline: average measures over the first week.

Gait & Posture: Five-U-Turn Test

- The aim of the Five-U-Turn Test (5UTT) is to assess gait and balance difficulties and unusual patterns in performing U-turns while walking a short distance at a comfortable pace
- A turn detection algorithm[9,10] was used to identify U-turns and calculate metrics around U-turn behavior
- Similarly to Timed 25-Foot Walk (T25FW) Test (data not shown), the baseline turn speed performance on 5UTT was not significantly different between HCs and patients with MS (**Table 2A**), which is likely to be related to the low level of ambulation impairment in this study population
- Turn speed correlated well with the in-clinic–performed T25FW times for patients with MS measured at baseline[a] (**Table 2B, Figure 5A**)
- Turn speed and the patient's perceived lower limb and ambulation function[b] showed significant association at baseline[a] (**Figure 5B**)

[a]Baseline: average measures over the first week; [b]Items 4 and 5 of the MSIS-29 rescaled to [0, 100].

Figure 5. U-turn speed measured with 5UTT correlates with T25FW and the patients' perceived lower limb and ambulation function

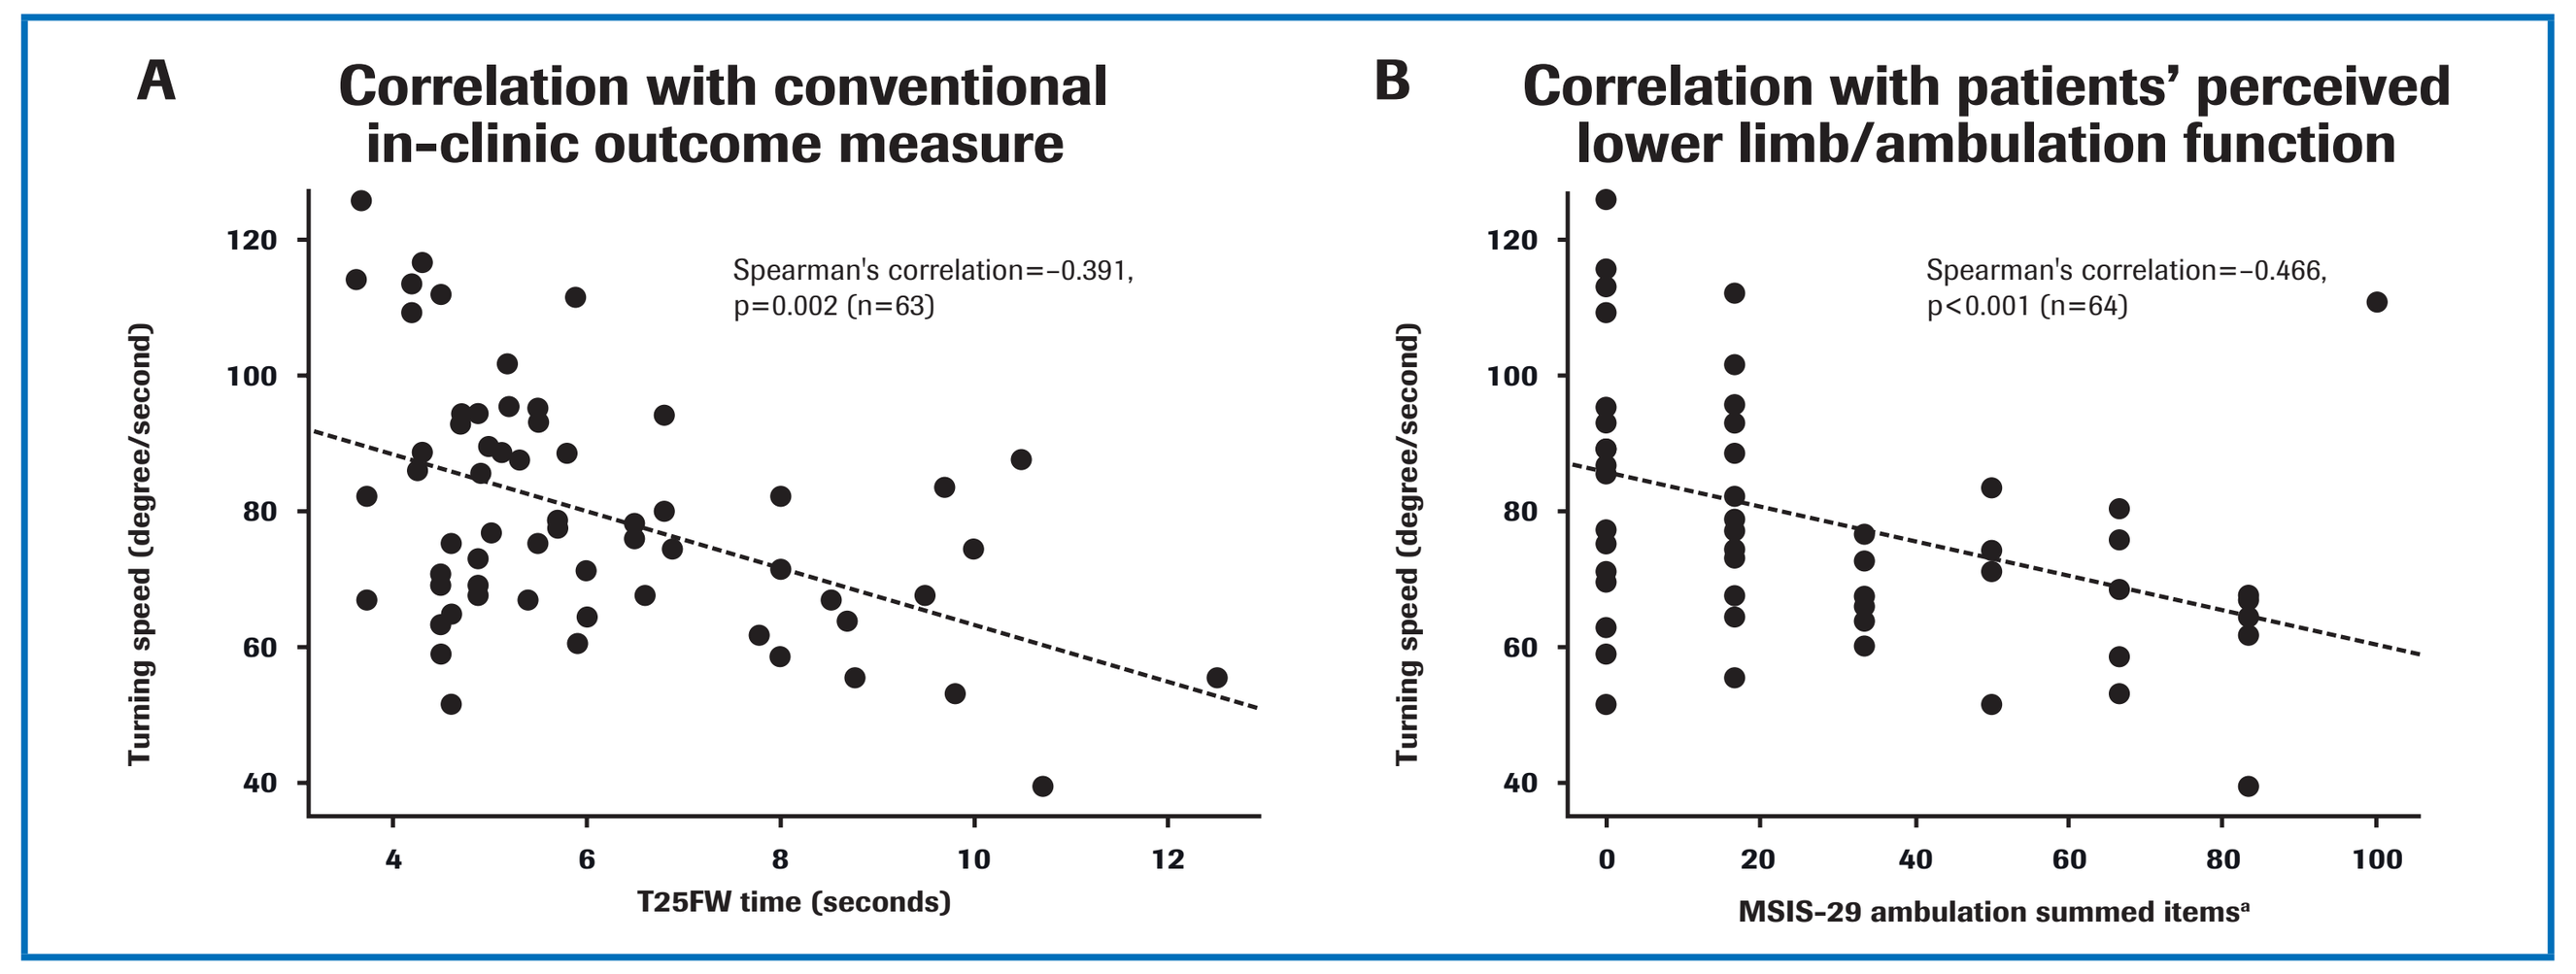
A
Correlation with conventional in-clinic outcome measure
Spearman's correlation=−0.391, p=0.002 (n=63)
Turning speed (degree/second)
T25FW time (seconds)
B
Correlation with patients' perceived lower limb/ambulation function
Spearman's correlation=−0.466, p<0.001 (n=64)
MSIS-29 ambulation summed items[a]

[a]Items 4 and 5 of the MSIS-29 summarized and rescaled to [0, 100].
5UTT, Five-U-Turn Test; MSIS, Multiple Sclerosis Impact Scale; T25FW, Timed 25-Foot Walk.

Gait & Posture: Two-Minute Walk Test

- The aim of the 2MWT is to assess ambulation speed, fatigability or abnormal gait feature patterns in a more demanding walking distance than T25FW
- Baseline[a] performance on the 2MWT measured by step power revealed differences between HCs and patients with MS (**Table 2A**) and significantly correlated with T25FW time (**Table 2B**)
 - — Step power was defined as the integral of variance in accelerometer radius for step spans detected using a step detection algorithm[11]

[a]Baseline: average measures over the first week.

CONCLUSIONS

- Preliminary data from the FLOODLIGHT study suggest that patients are highly engaged and satisfied with smartphone-based self-assessments
- FLOODLIGHT outcomes correlate with conventional in-clinic outcome measures of MS disability and with patients' perception of their mental/psychological status and hand/arm/ambulation function
- Smartphone-based FLOODLIGHT outcomes may represent a promising avenue to enable precise continuous assessment of MS disease in clinical trials and real-world practice settings

ACKNOWLEDGMENTS

We would like to thank all patients, their families, and the investigators who participated in these trials.
This research was sponsored by F. Hoffmann-La Roche Ltd, Basel, Switzerland. Writing and editorial assistance for this presentation was provided by Articulate Science, UK, and funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

REFERENCES

1. Bove R, *et al. Neurol Neuroimmunol Neuroinflamm* 2015;2:e162.
2. Roosendaal SD, *et al. Mult Scler* 2011;17:1098–1106.
3. Eshaghi A, *et al. Neuroimage* 2014;86:257–264.
4. Filippi M, *et al. Curr Opin Neurol* 2014;27:290–299.
5. Steenwijk MD, *et al. Hum Brain Mapp* 2015;36:1796–1807.
6. Khaleeli Z, *et al. Neuroimage* 2007;37:253–261.
7. Smith A. Symbol Digits Modalities Test: Manual. Los Angeles: Western Psychological Services 2007.
8. Wang YC, *et al. J Hand Ther* 2015;28:53–60.
9. El-Gohary M, *et al. Sensors* 2013;14:356–369.
10. Benedict RH, *et al. Mult Scler* 2016;22:1874–1882.
11. Lee HH, *et al. Sensors* 2015;15:27230–27250.

DISCLOSURES

X Montalban has received speaker honoraria and travel expense reimbursement for participation in scientific meetings, been a steering committee member of clinical trials or served on advisory boards of clinical trials for Actelion, Almirall, Bayer, Biogen, F. Hoffmann-La Roche Ltd, Genzyme, Merck, Novartis, Octapharma, Receptos, Sanofi, Teva and Trophos. P Mulero has nothing to disclose. L Midaglia has nothing to disclose. J Graves has received grants or research support from Biogen, Genentech, Inc., and S3 Group and has received compensation for a nonbranded resident and fellow education seminar supported by Biogen. SL Hauser serves on the board of trustees for Neurona and on scientific advisory boards for Annexon, Bionure and Symbiotix and has received travel reimbursement and writing assistance from F. Hoffmann-La Roche Ltd for CD20-related meetings and presentations. L Julian is an employee of Genentech, Inc., and a shareholder of F. Hoffmann-La Roche Ltd. M Baker is an employee and shareholder of F. Hoffmann-La Roche Ltd. J Schadrack is an employee and shareholder of F. Hoffmann-La Roche Ltd. C Gossens is an employee and shareholder of F. Hoffmann-La Roche Ltd. A Scotland is an employee of F. Hoffmann-La Roche Ltd. F Lipsmeier is an employee of F. Hoffmann-La Roche Ltd. G Pointeau is an employee of F. Hoffmann-La Roche Ltd. C Bernasconi is a contractor for F. Hoffmann-La Roche Ltd. S Belachew is an employee and shareholder of F. Hoffmann-La Roche Ltd. Michael Lindemann has nothing to disclose.

Presented at the 70th American Academy of Neurology (AAN) Annual Meeting; April 21–27, 2018; Los Angeles, CA, USA

Poster 382