Preferences, Needs and Opportunity Areas for US Multiple Sclerosis Caregivers

S Shankar,[1] B Adelman,[2] A Sorathia[2]

[1]Genentech, Inc., South San Francisco, CA, USA; [2]Karate Health, Washington, DC, USA

INTRODUCTION

- Multiple sclerosis (MS) is a chronic, demyelinating disease affecting an estimated 2.2 million patients worldwide[1]
- Specific symptoms vary widely across patients; however, in general, patients tend to accumulate considerable disability over time, which impacts their ability to work, participate in recreational activities, and engage in social interactions[2]
- Caregivers are an indispensable piece of the support system for patients living with MS at home, and their lives are also heavily impacted physically and emotionally[3]; moreover, it is not uncommon for caregivers to reduce or completely forgo employment, creating financial strain[4]
- Currently, there is limited research investigating how best to meet the needs of caregivers and ensure that they are optimally equipped to support patients
- Digital resources are increasingly being recognized by patients and providers alike as valuable platforms for health interactions,[5] and, with a reported 8 billion hours/year spent online in the United States alone,[5] they are an undeniably effective way of reaching patients wherever they are[6]
- Using MSCorner, a digital platform developed by Karate Health (Washington, DC, USA) that provides content and tools for the MS community, we investigated the experiences and needs of caregivers for patients with MS

OBJECTIVES

- To better understand the challenges experienced by caregivers and the relationship between caregivers, patients with MS and the clinical team
- To identify effective strategies to support caregivers, including shared decision-making and patient-centric programs

METHODS

- A series of online surveys were developed for patients with MS and caregivers in the United States, and were hosted on MSCorner (**Figure 1**)
- Patients were recruited using digital marketing campaigns including placements on social media and search engine results
- Surveys were completed on a volunteer basis, and participants were not compensated
- Participants with incomplete responses were contacted via email for complete information

Figure 1. Caregiver and patient surveys and disposition

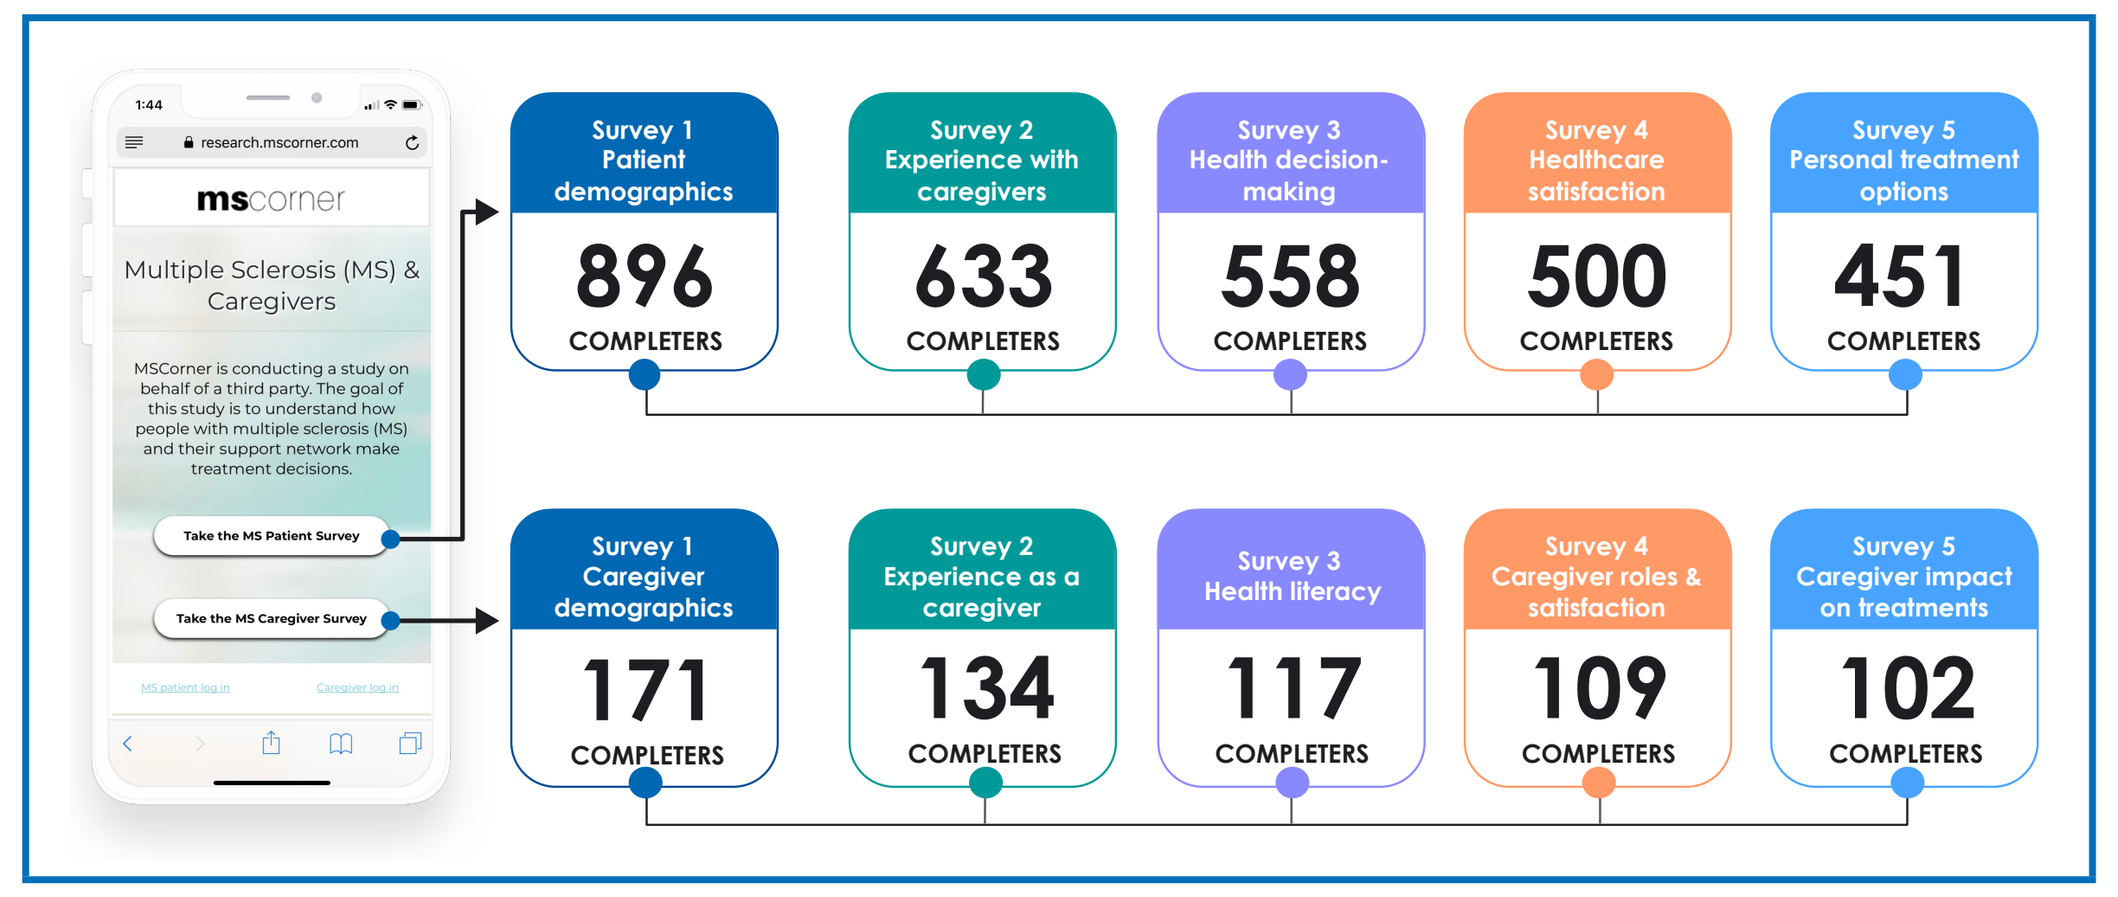

mscorner
Multiple Sclerosis (MS) & Caregivers
Take the MS Patient Survey
Take the MS Caregiver Survey
Survey 1 Patient demographics 896 COMPLETERS
Survey 2 Experience with caregivers 633 COMPLETERS
Survey 3 Health decision-making 558 COMPLETERS
Survey 4 Healthcare satisfaction 500 COMPLETERS
Survey 5 Personal treatment options 451 COMPLETERS
Survey 1 Caregiver demographics 171 COMPLETERS
Survey 2 Experience as a caregiver 134 COMPLETERS
Survey 3 Health literacy 117 COMPLETERS
Survey 4 Caregiver roles & satisfaction 109 COMPLETERS
Survey 5 Caregiver impact on treatments 102 COMPLETERS

RESULTS

Respondent Characteristics

- Respondents included 896 patients (relapsing-remitting MS, 67%; secondary progressive MS, 9%; primary progressive MS, 9%; progressive-relapsing MS, 4%; not sure, 11%) and 171 caregivers with diverse demographics from 52 states and territories (**Figure 2**)
- Caregivers primarily identified as a family member (typically a spouse), and professional caregivers were rarely used (**Figure 3**)

Figure 2. Respondent demographics

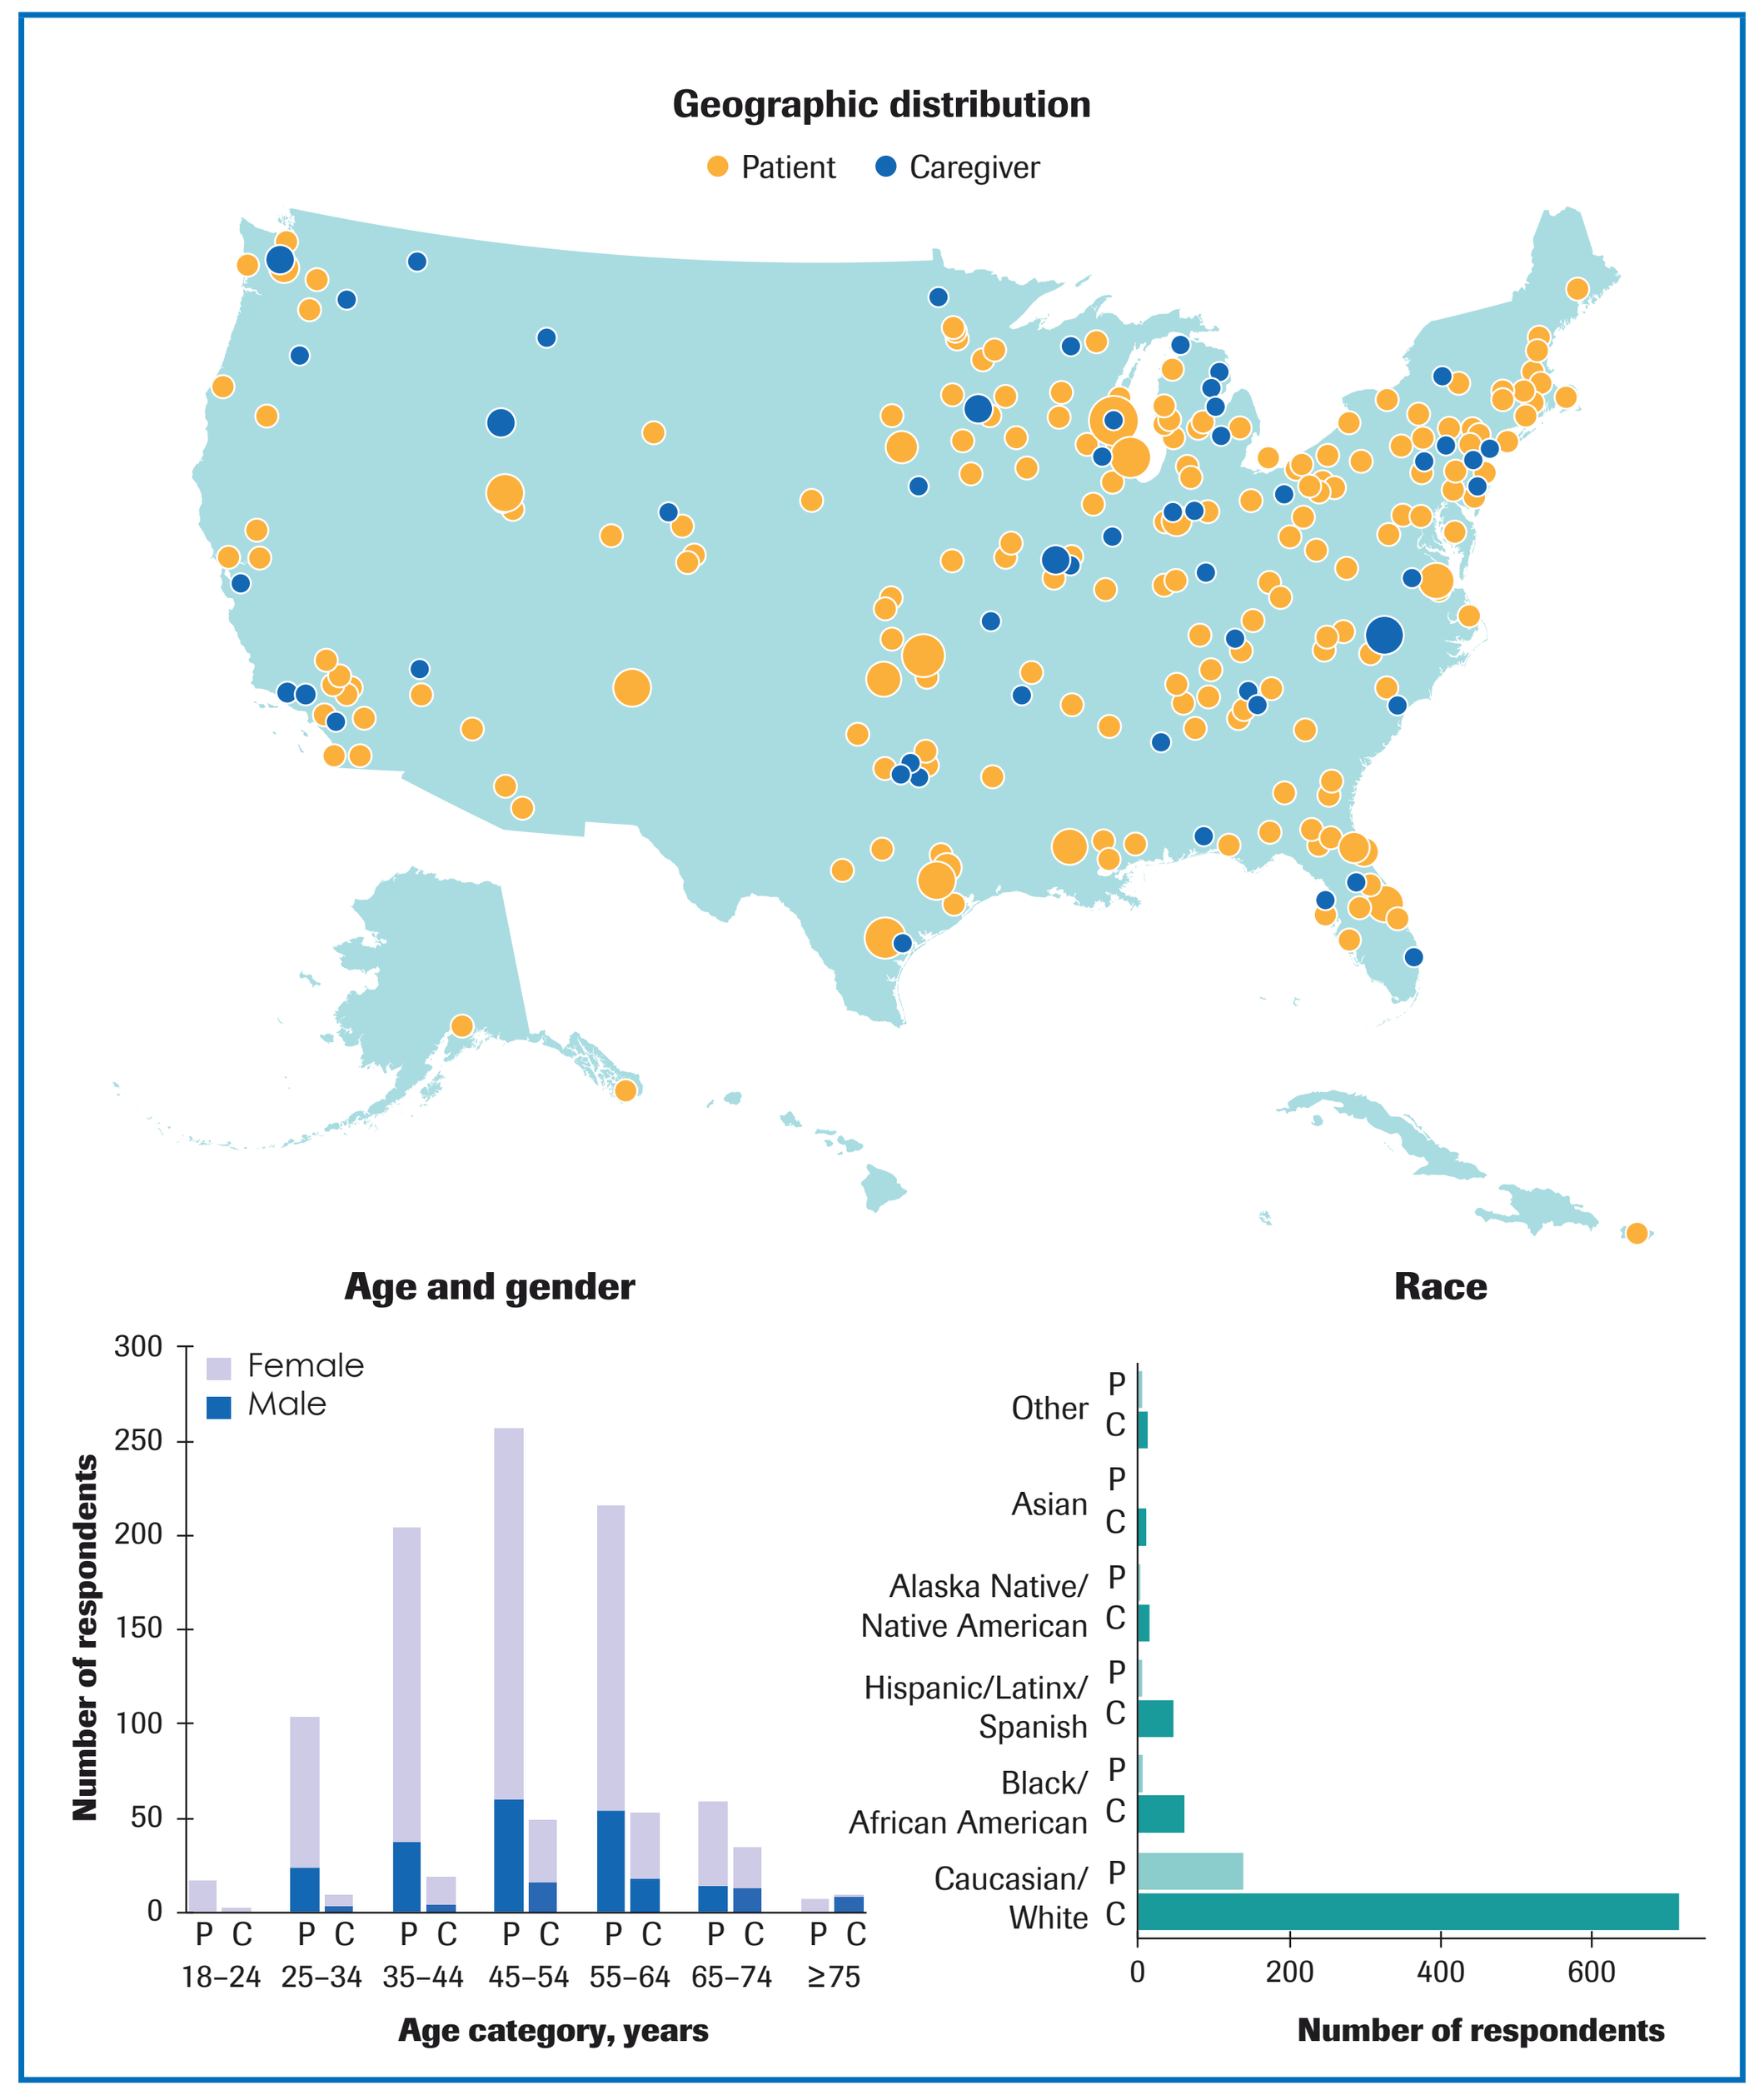

Geographic distribution
Patient
Caregiver
Age and gender
Female
Male
Number of respondents
Age category, years
18–24
25–34
35–44
45–54
55–64
65–74
≥75
Race
Other
Asian
Alaska Native/ Native American
Hispanic/Latinx/ Spanish
Black/ African American
Caucasian/ White
Number of respondents

P, patient; C, caregiver.

Figure 3. Professional caregiver utilization

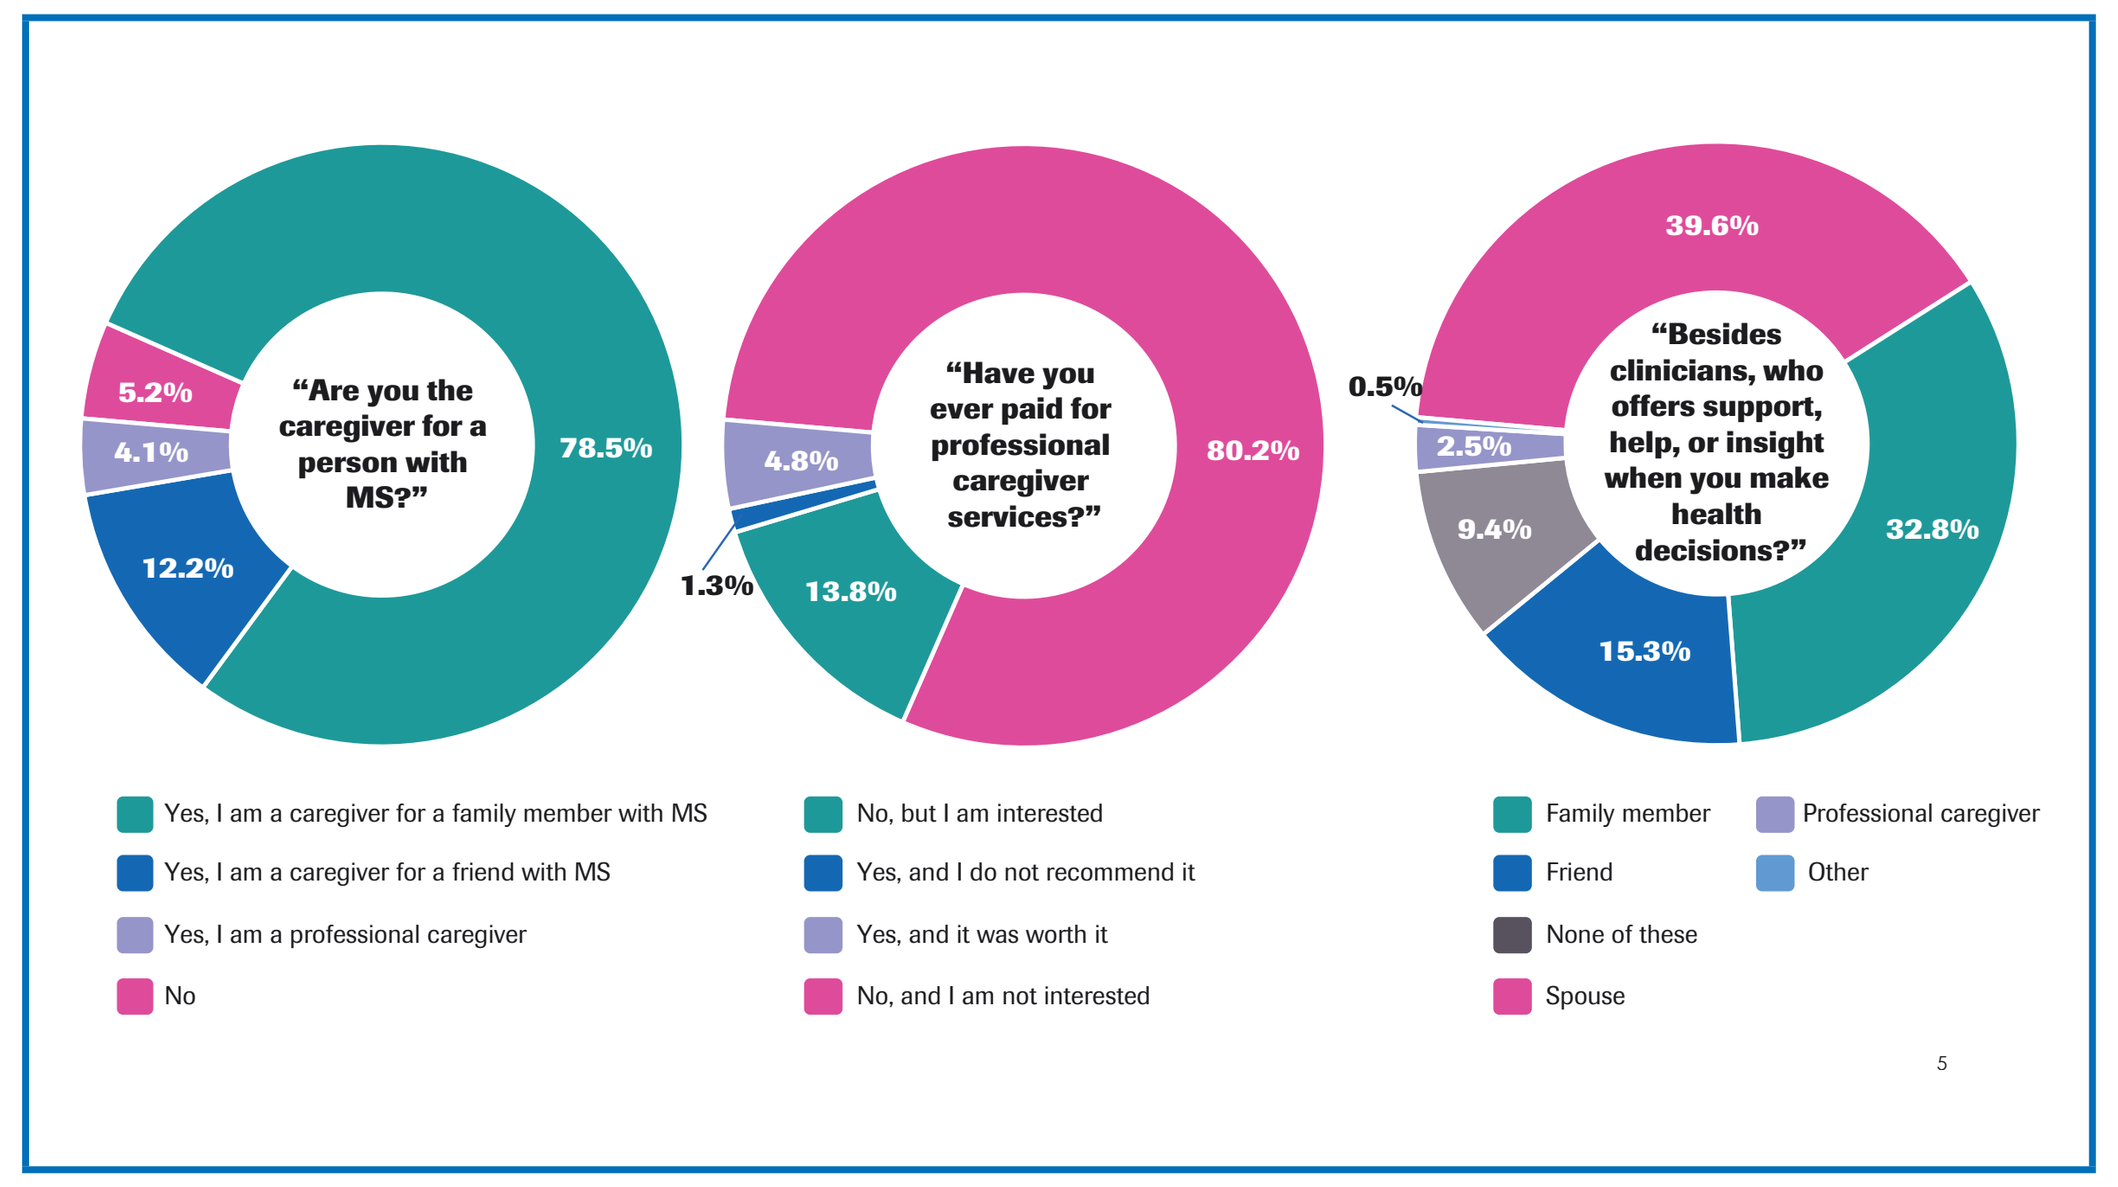

"Are you the caregiver for a person with MS?"
78.5%
12.2%
4.1%
5.2%
"Have you ever paid for professional caregiver services?"
80.2%
13.8%
1.3%
4.8%
"Besides clinicians, who offers support, help, or insight when you make health decisions?"
39.6%
32.8%
15.3%
9.4%
2.5%
0.5%
Yes, I am a caregiver for a family member with MS
Yes, I am a caregiver for a friend with MS
Yes, I am a professional caregiver
No
No, but I am interested
Yes, and I do not recommend it
Yes, and it was worth it
No, and I am not interested
Family member
Friend
None of these
Spouse
Professional caregiver
Other

Commitment of Caregiving

- Caregivers indicated that they were heavily involved in patients' day-to-day routines (**Figure 4**), supporting an average of 4.64 tasks daily
- One-third of caregivers reported giving >40 hours of care per week, for which they were largely unpaid, highlighting a considerable economic burden (**Figure 5**)

Figure 4. Domains of caregiver support

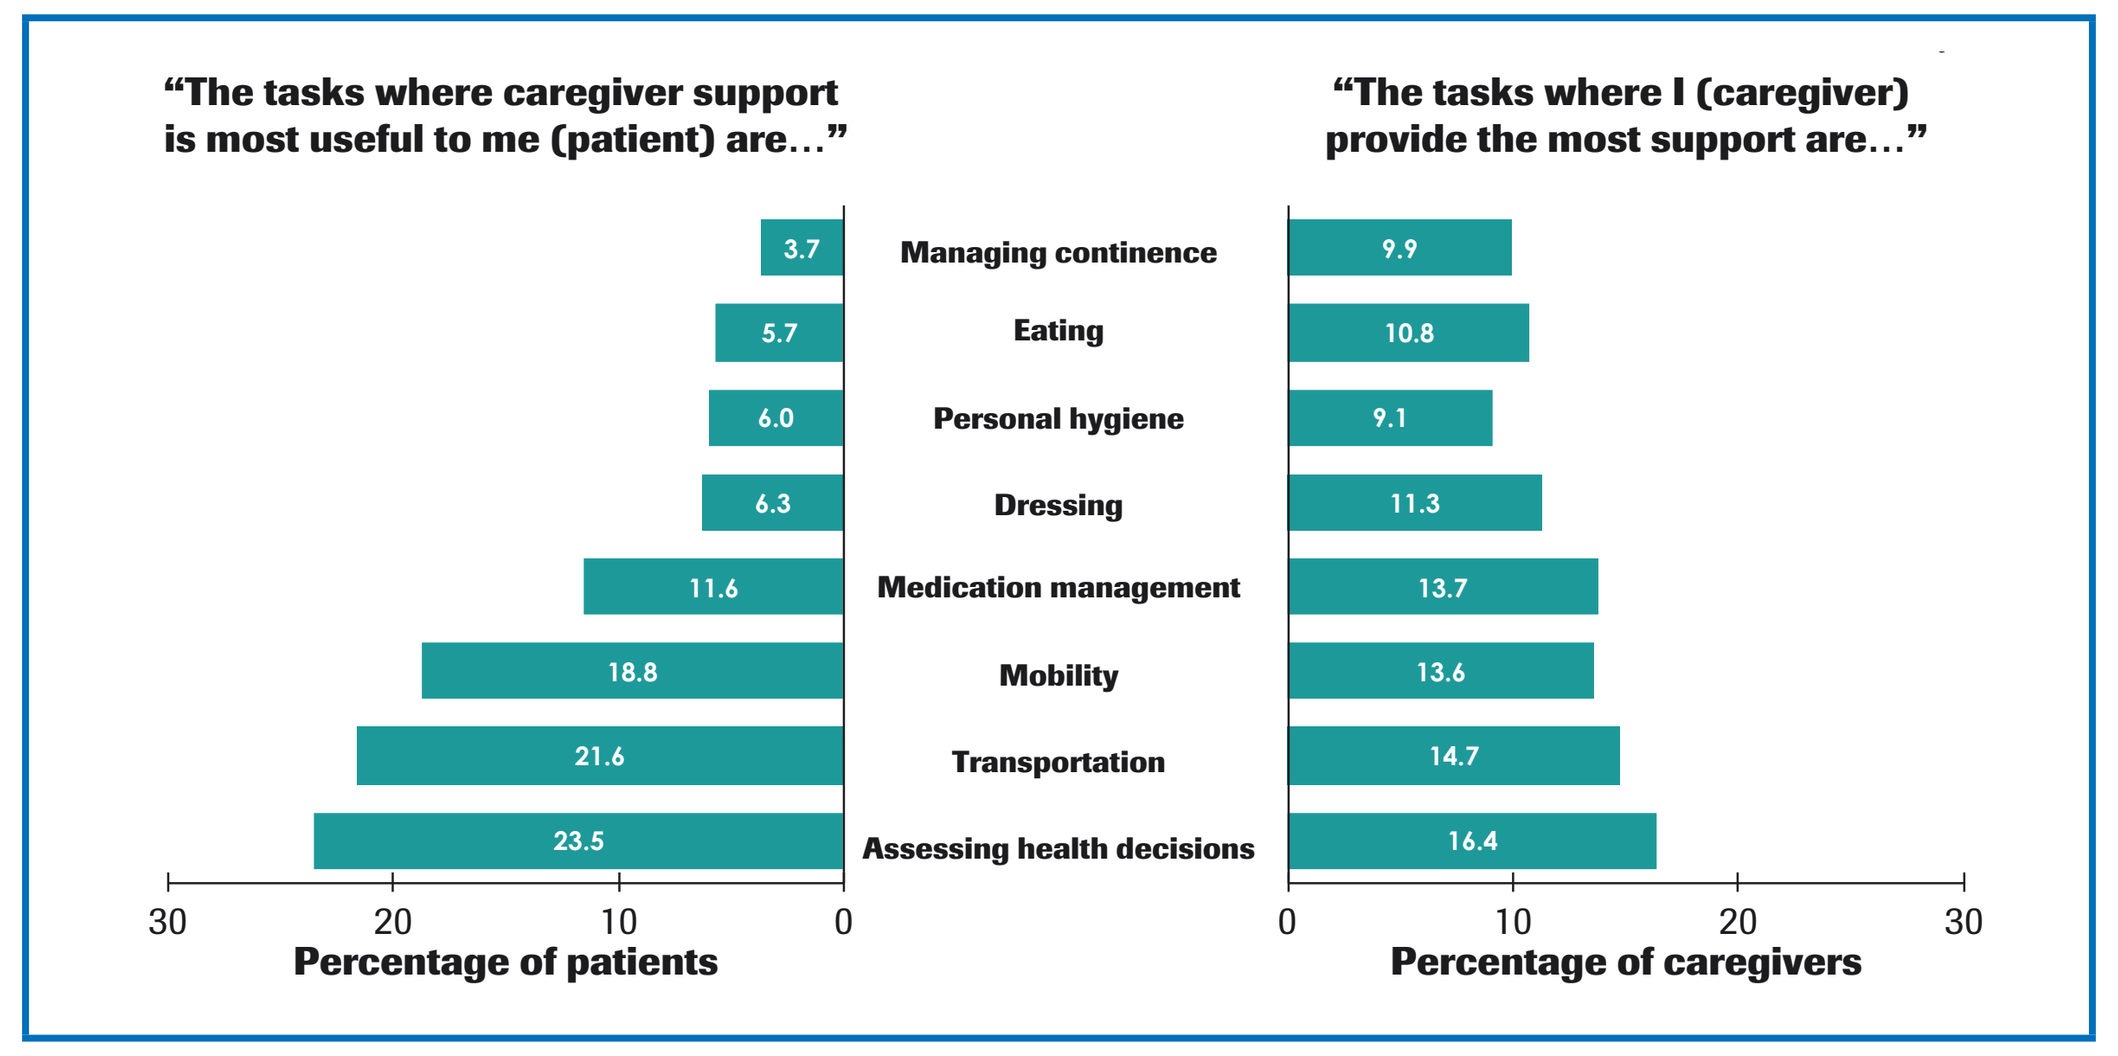

"The tasks where caregiver support is most useful to me (patient) are…"
"The tasks where I (caregiver) provide the most support are…"
Managing continence 3.7 9.9
Eating 5.7 10.8
Personal hygiene 6.0 9.1
Dressing 6.3 11.3
Medication management 11.6 13.7
Mobility 18.8 13.6
Transportation 21.6 14.7
Assessing health decisions 23.5 16.4
Percentage of patients
Percentage of caregivers

Figure 5. Caregiver time and financial investment

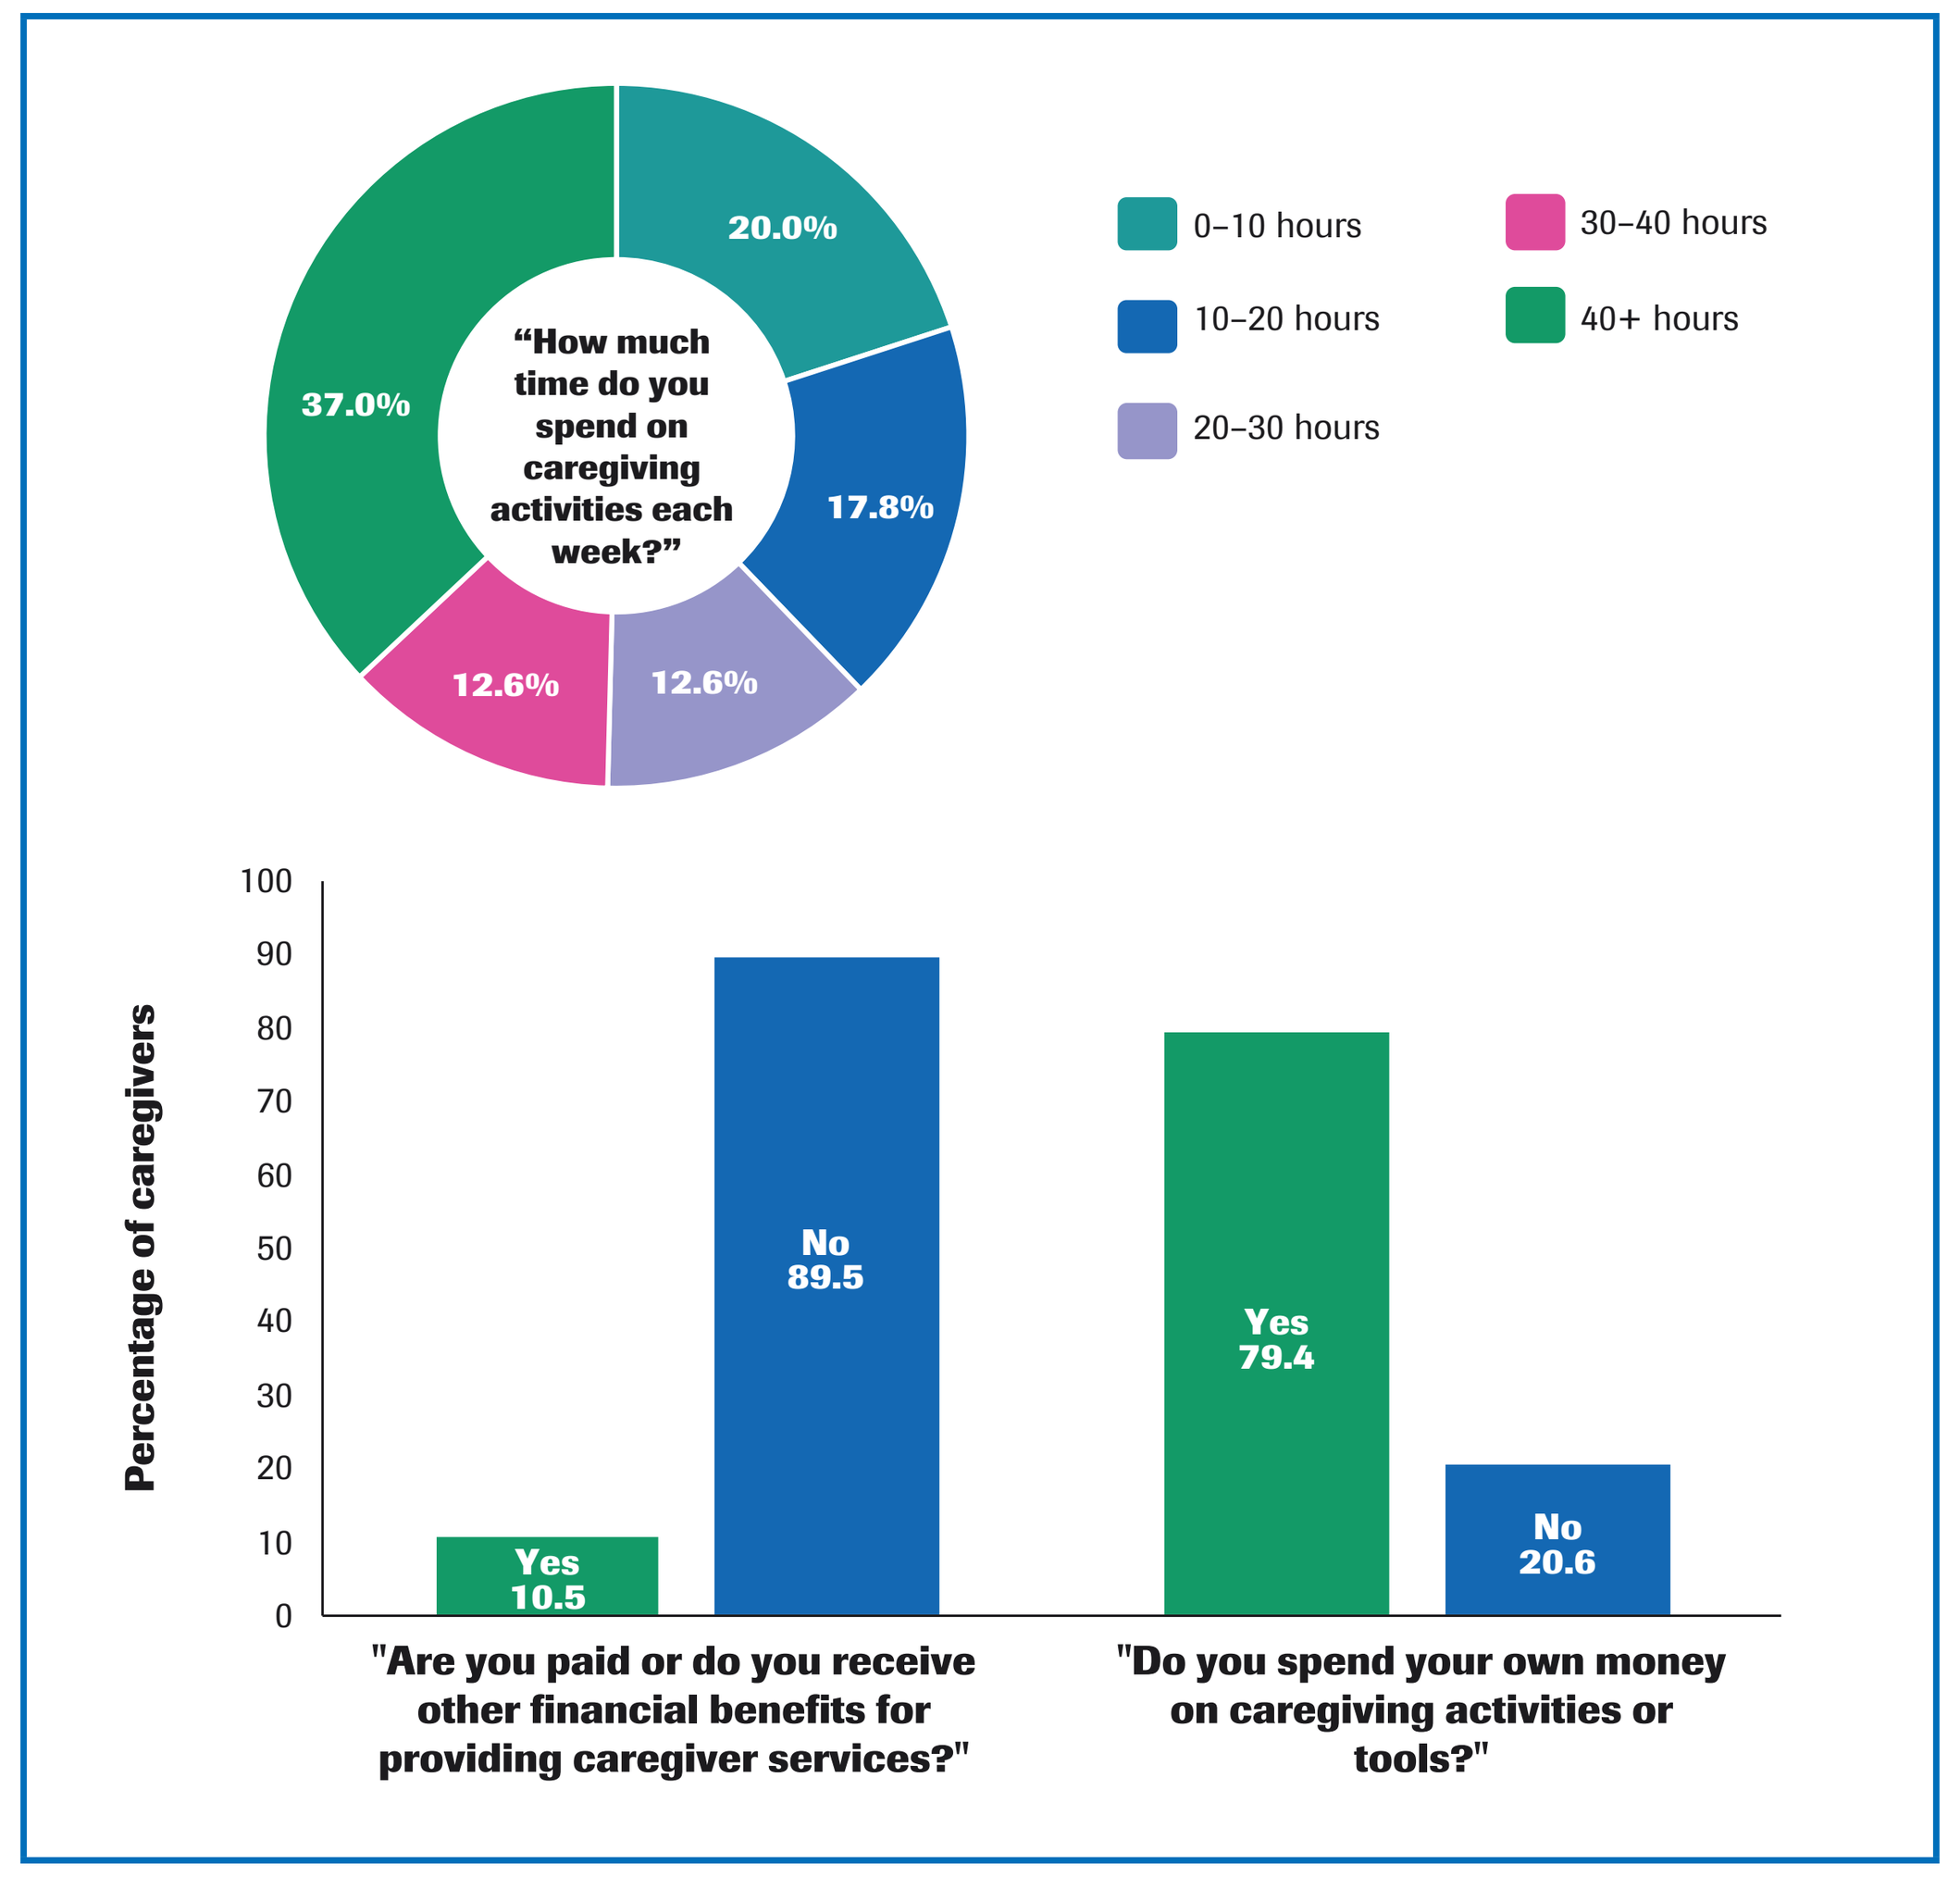

"How much time do you spend on caregiving activities each week?"
20.0%
17.8%
12.6%
12.6%
37.0%
0–10 hours
10–20 hours
20–30 hours
30–40 hours
40+ hours
Percentage of caregivers
Yes 10.5
No 89.5
Yes 79.4
No 20.6
"Are you paid or do you receive other financial benefits for providing caregiver services?"
"Do you spend your own money on caregiving activities or tools?"

Health Decision-Making

- "Assessing health-related decisions" was identified as a primary responsibility of caregivers by both patient and caregiver respondents (**Figure 4**), and 13.7% of patients ranked caregivers as being the most influential when it came to decision-making (**Figure 6**)
- Caregivers frequently sought guidance from professional healthcare providers to assist with decision-making and tended to be well-informed (**Figure 6**), with the most trusted sources of information including large health websites (e.g. WebMD) and other members of the MS care team

Figure 6. Trusted sources of support

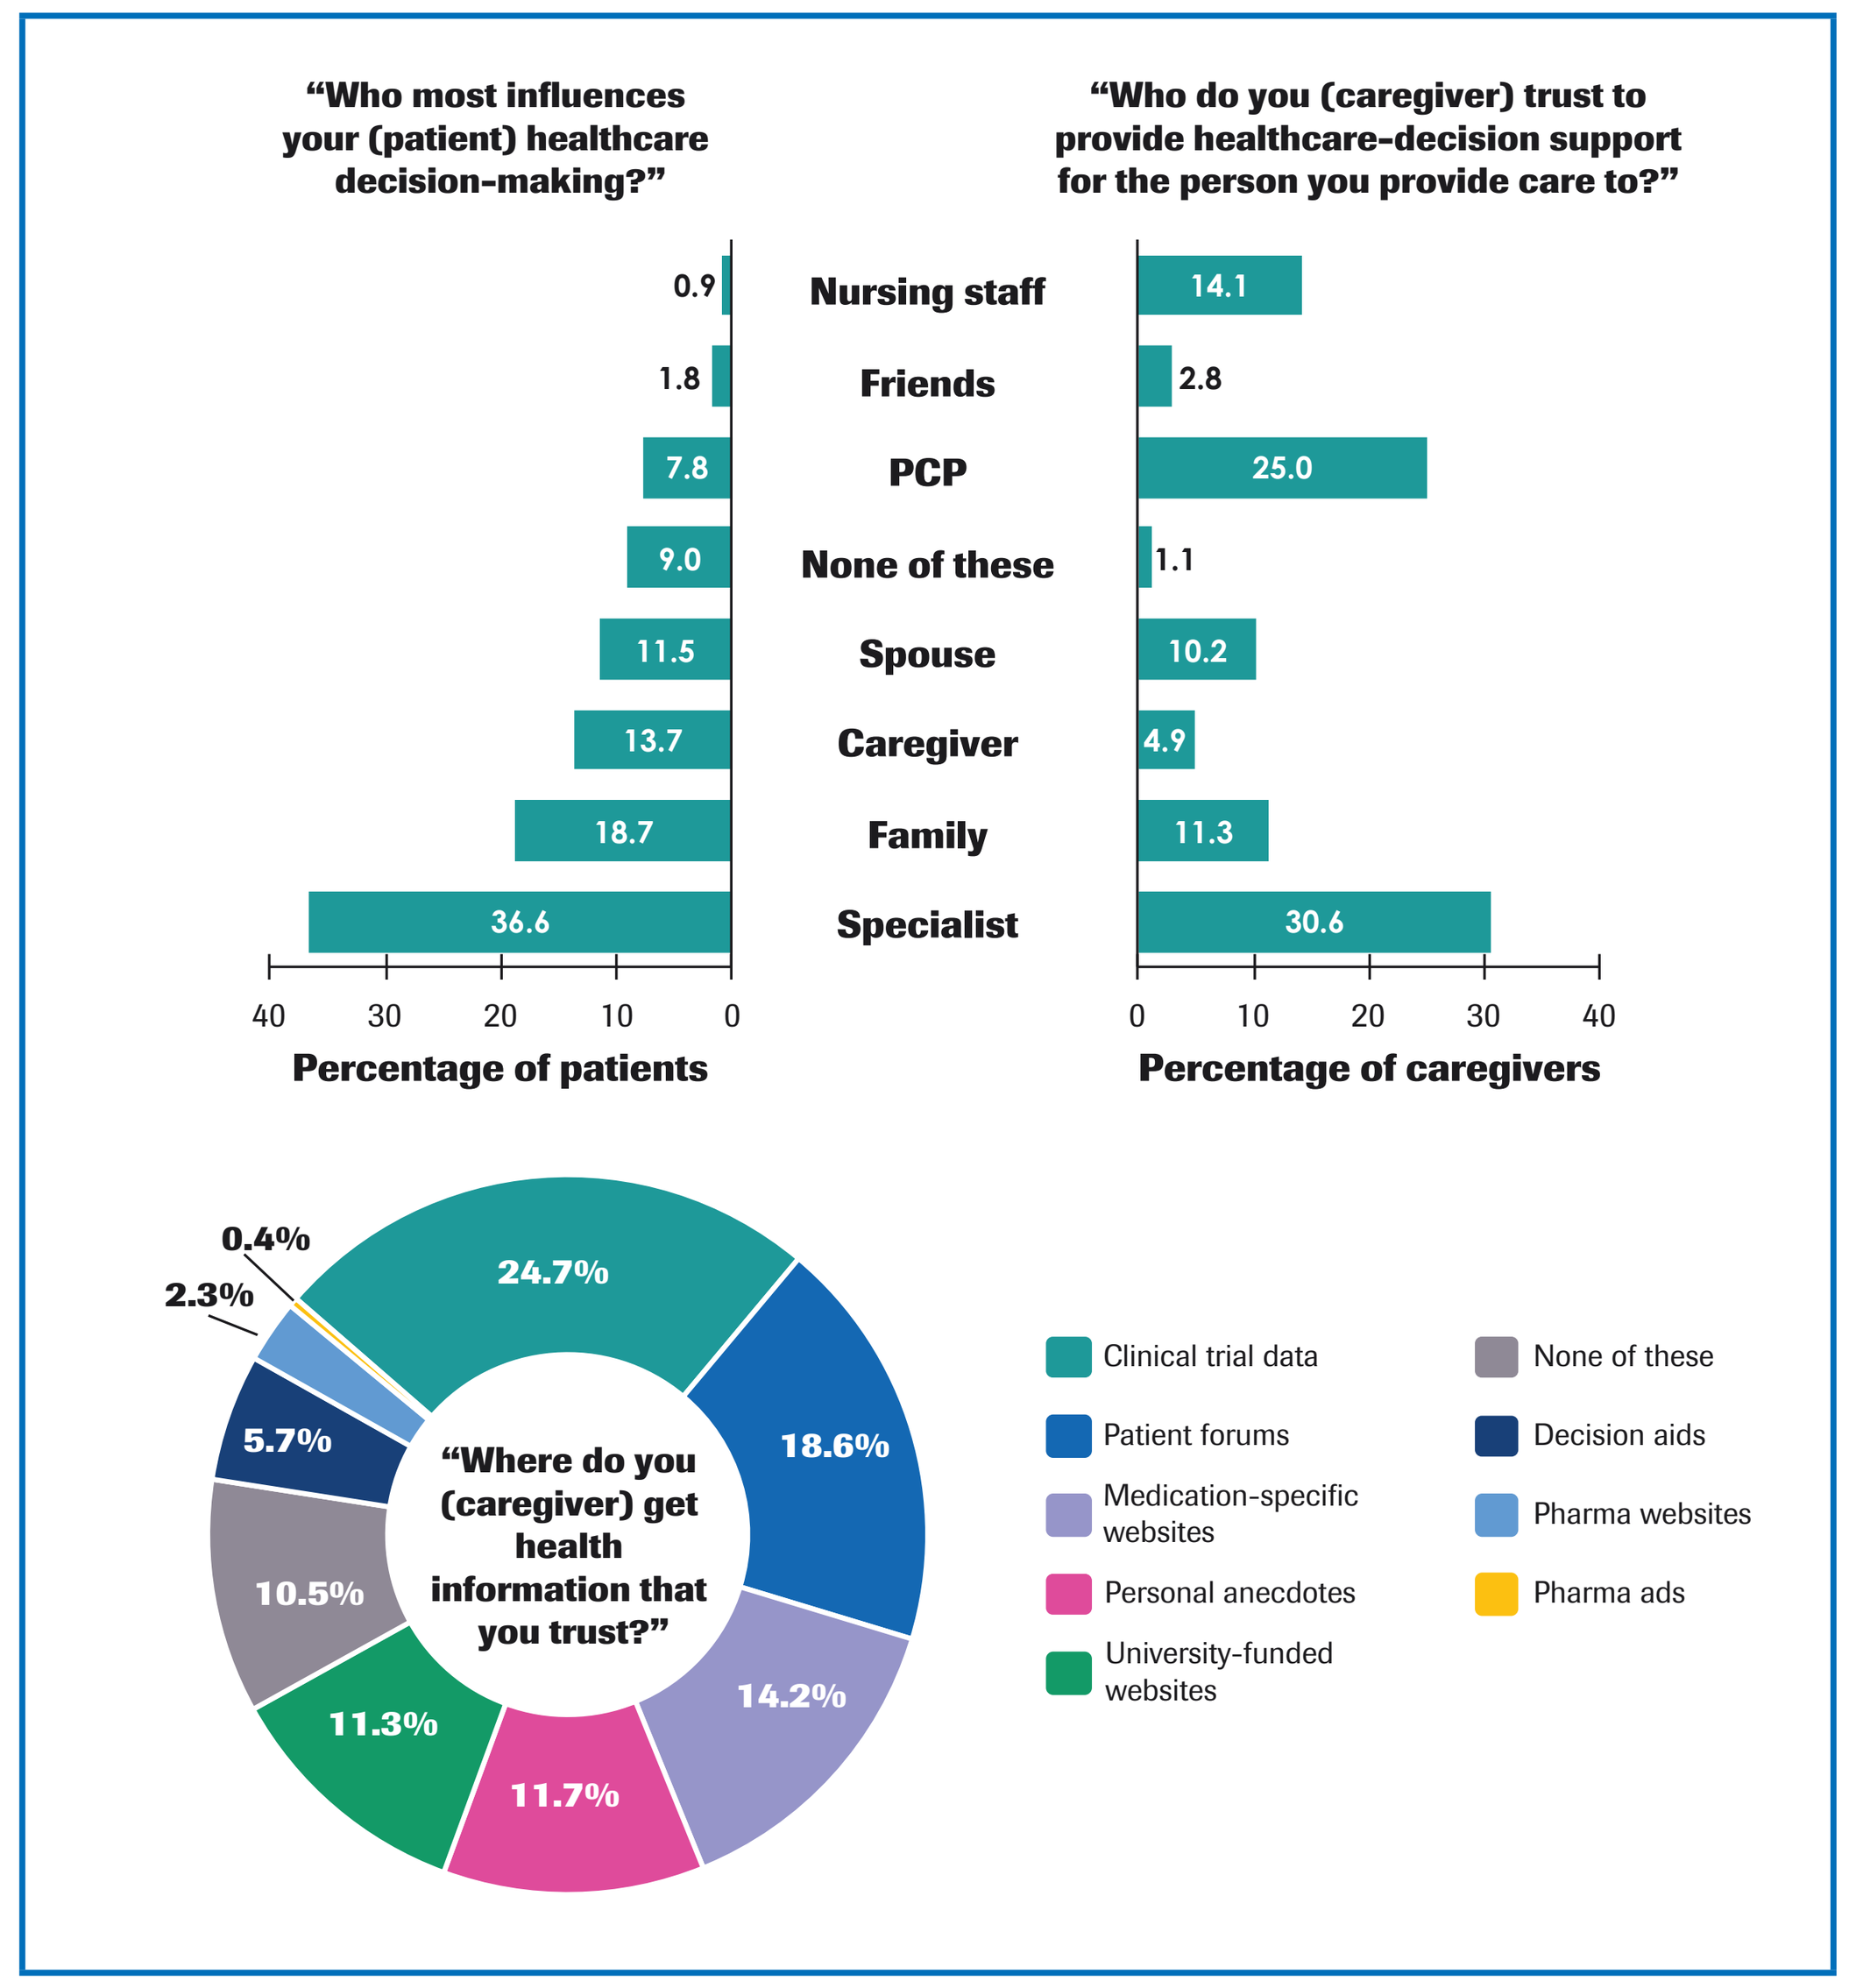

"Who most influences your (patient) healthcare decision-making?"
"Who do you (caregiver) trust to provide healthcare-decision support for the person you provide care to?"
Nursing staff 0.9 14.1
Friends 1.8 2.8
PCP 7.8 25.0
None of these 9.0 1.1
Spouse 11.5 10.2
Caregiver 13.7 4.9
Family 18.7 11.3
Specialist 36.6 30.6
Percentage of patients
Percentage of caregivers
"Where do you (caregiver) get health information that you trust?"
24.7%
18.6%
14.2%
11.7%
11.3%
10.5%
5.7%
2.3%
0.4%
Clinical trial data
Patient forums
Medication-specific websites
Personal anecdotes
University-funded websites
None of these
Decision aids
Pharma websites
Pharma ads

PCP, primary care practitioner; Pharma, pharmaceutical (company).

Caregiver Perspective and Needs

- Despite clear evidence of caregivers' significant role in the day-to-day lives of patients with MS, findings indicate a strong disconnect between caregivers and the MS care team (**Figure 7**)
 - Only 51.3% of caregivers reported feeling that they are a part of the care team and trusted, and 58.5% would recommend the MS care team to another patient with MS
 - Fewer than half (48.6%) of caregivers agreed that patient symptoms were being successfully managed with the current care plan

Figure 7. Caregiver perception of the MS team

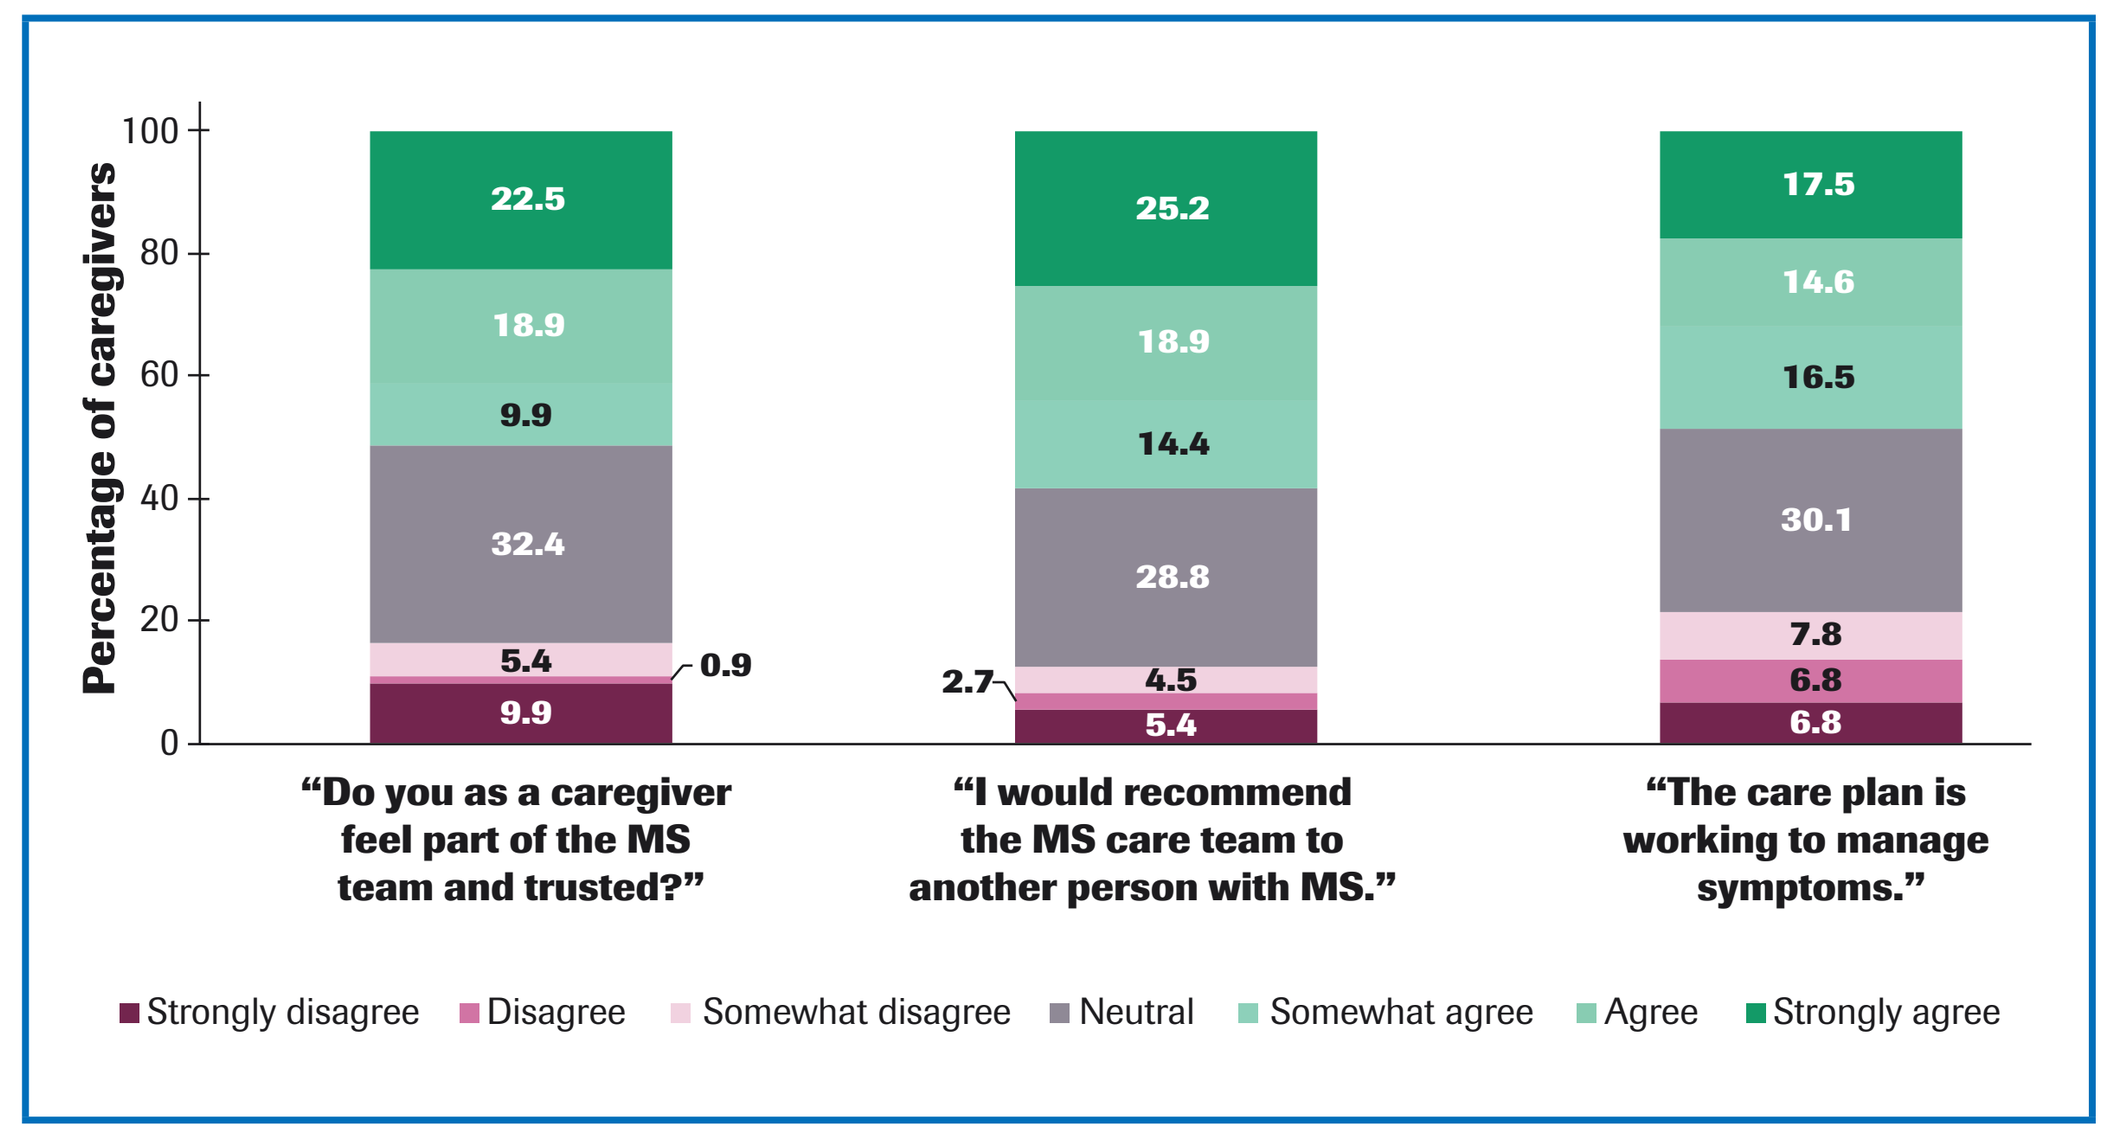

Percentage of caregivers
"Do you as a caregiver feel part of the MS team and trusted?" 9.9 0.9 5.4 32.4 9.9 18.9 22.5
"I would recommend the MS care team to another person with MS." 5.4 2.7 4.5 28.8 14.4 18.9 25.2
"The care plan is working to manage symptoms." 6.8 6.8 7.8 30.1 16.5 14.6 17.5
Strongly disagree
Disagree
Somewhat disagree
Neutral
Somewhat agree
Agree
Strongly agree

- Survey results highlight the considerable personal challenges experienced by caregivers across a number of domains that affect everyday functioning and quality of life (**Figure 8**)

Figure 8. Health impact on caregivers

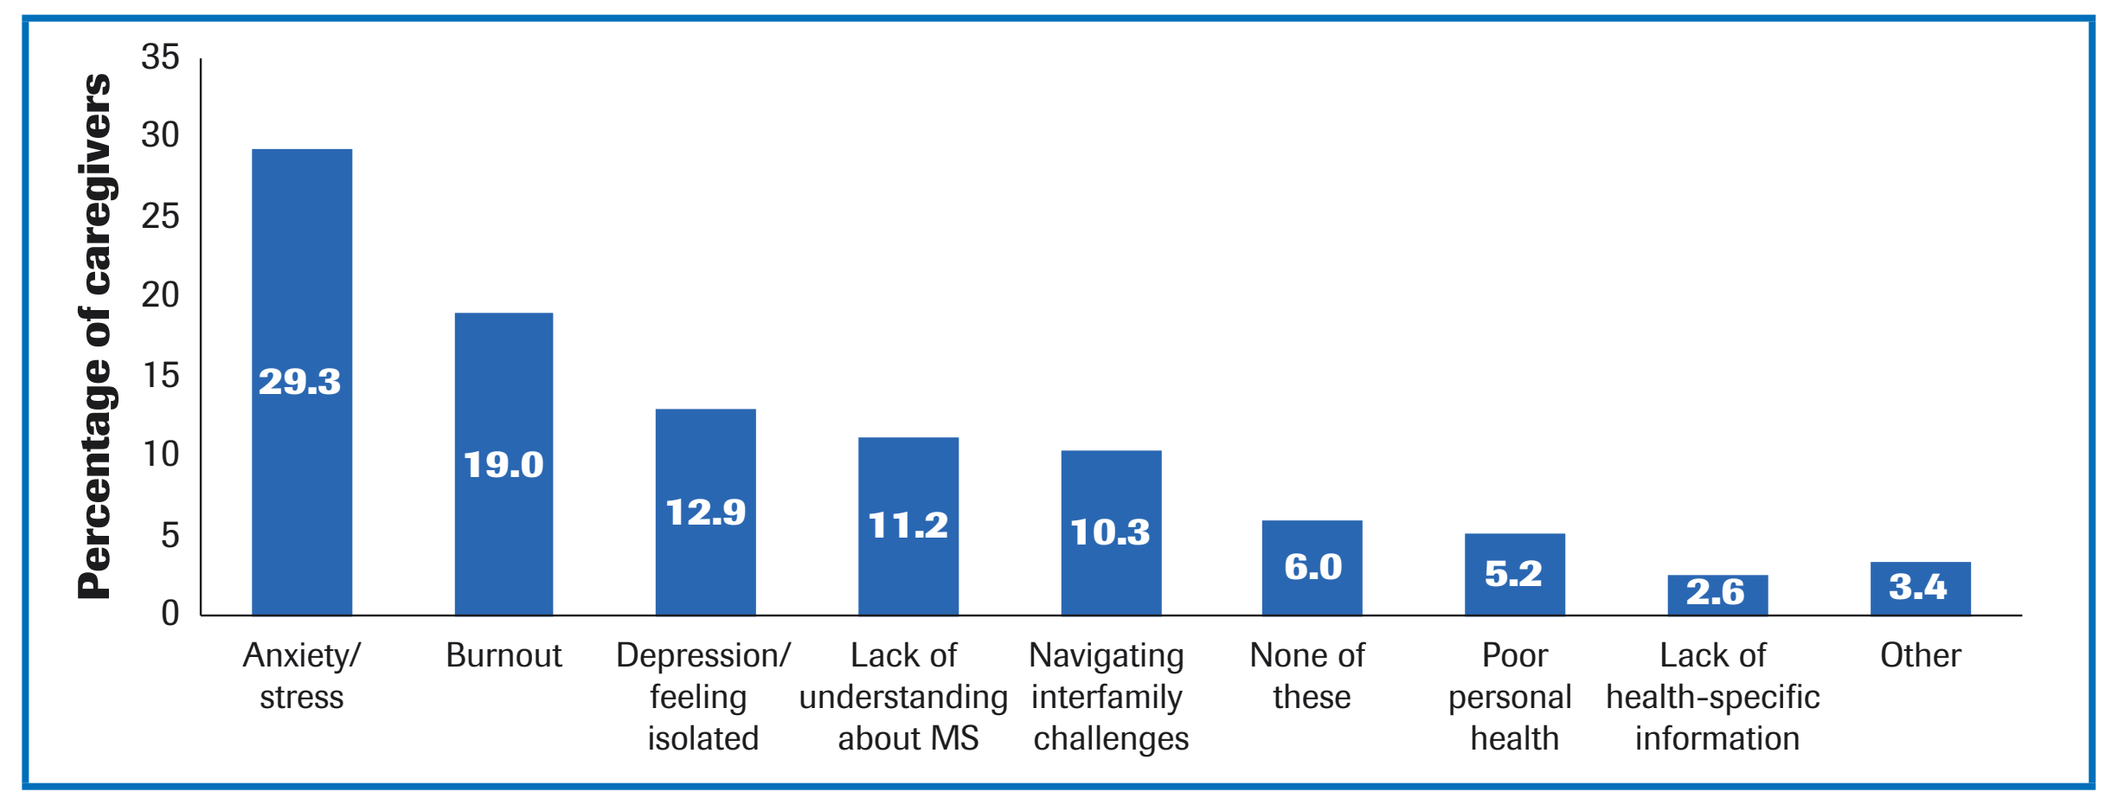

Percentage of caregivers
Anxiety/ stress 29.3
Burnout 19.0
Depression/ feeling isolated 12.9
Lack of understanding about MS 11.2
Navigating interfamily challenges 10.3
None of these 6.0
Poor personal health 5.2
Lack of health-specific information 2.6
Other 3.4

DISCUSSION AND RECOMMENDATIONS

- Despite their significant role in providing care to patients with MS, caregivers do not feel fully involved in care decisions or planning
- Evidence demonstrates the considerable impact of caregiving responsibilities on the health and overall well-being of the caregivers themselves, which ultimately will have a negative impact on patients
- Overall, findings suggest substantial value in creating caregiver-oriented content and resources that can be shared by trusted clinical providers and nonindustry organizations
- Some considerations for caregiver-oriented materials include the following:
 - Digital content has the potential to reach a wide audience and can be easily tailored to meet a specific purpose
 - Physician-oriented resources on caregiving may help to facilitate better integration of the caregiver into the care team and decision-making process

CONCLUSIONS

- There is currently a lack of resources supporting the caregivers of patients with MS
- This shortcoming can be addressed by developing caregiver-oriented content with critical input from clinical providers and thought leaders in the MS field, that can better prepare these individuals to support loved ones with MS

DISCLOSURES

S Shankar is an employee of Genentech, Inc.

B Adelman is a co-owner of Karate Health.

A Sorathia is a co-owner of Karate Health.

ACKNOWLEDGEMENTS

We would like to thank all patients and caregivers who participated in this survey. This research was conducted by Karate Health, Washington, DC, USA, and funded by Genentech, Inc., South San Francisco, CA, USA. Medical writing and editorial assistance for this presentation were provided by Health Interactions, USA, and funded by Genentech, Inc.

REFERENCES

1. GBD 2016 Multiple Sclerosis Collaborators. *Lancet Neurol* 2019;18:269–285.
2. Fillipi M, et al. *Nat Rev Dis Primers* 2018;4:43.
3. Petrikis P, et al. *J Clin Neurol* 2019;15:77–83.
4. Buchanan RJ, et al. *Int J MS Care* 2013;14:203–210.
5. Carroll CL, Ramachandran P. *Chest* 2014;145:896–902.
6. Gordon NP, Hornbrook MC. *BMC Health Serv Res* 2018;18:220.

Presented at the 2019 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC); May 28–June 1, 2019; Seattle, WA, USA

Poster FAM02