Design of a prospective, longitudinal, natural history study in Huntington's disease (HD)

Gregory Hooper (1), Dylan Trundell (1), Giuseppe Palermo (2), Thomas Kremer (2), Elsbeth Frick (2), Lauren Boak (2), the Natural History Study Investigators, Rachelle Doody (2,3) and Scott Schobel (2)

(1) Roche Products Ltd., Welwyn Garden City, UK. (2) F. Hoffmann-La Roche Ltd., Basel, Switzerland. (3) Genentech Inc., South San Francisco, CA, USA.

Summary

mHTT is a protein thought to cause HD, and can be measured in collected spinal fluid.[1] This study will provide valuable information on the relationship between levels of mHTT and a wide range of assessments that measure HD-related symptoms and their impact on daily function. Spinal fluid will be collected when patients begin the study and at 3, 9 and 15 months. Progression of the disease will be followed closely over 15 months and the results are expected to aid interpretation of clinical trials.

Study Objectives

Primary	To examine the predictive value of baseline CSF mHTT levels on measures of HD progression assessed by measuring change from baseline at 3, 9 and 15 months in the following endpoints: ▪ clinical: cUHDRS, TFC, TMS, SDMT, SWR Test and IS. ▪ biomarkers of neuronal injury:[1] CSF NfL, Tau. ▪ brain atrophy (by MRI); decline in whole brain volume, caudate volume.
Secondary	To investigate the temporal profile of longitudinal changes in CSF mHTT levels within the patient, and the association of these changes with clinical measures, markers of neuronal injury and clinical outcomes from the primary endpoint at baseline, and 3, 9 and 15 months.
Exploratory	▪ To examine the association of the Roche HD app with standard HD clinical assessments, biomarkers and patient-reported outcomes. ▪ To examine the association of potential prognostic biomarkers and genetic modifiers with HD disease status and disease progression.

As no medication will be given to the patients during the study, safety assessments will consist of monitoring and recording AEs and SAEs related to study procedures.

Patient Eligibility Criteria

Select Inclusion Criteria

- Aged 25–65 years.
- Early manifest, Stage I or Stage II HD (defined as TFC of 7–13, inclusive).
- Genetically confirmed disease (CAG repeat length ≥36 in *HTT* by direct DNA testing).
- Be capable of undergoing MRI scans, lumbar puncture and blood draws.
- Stable medical, psychiatric and neurologic status for 12 weeks prior to screening and at the time of enrollment.

Select Exclusion Criteria

- Any condition, including severe chorea, that would prevent either writing or performing pen and paper, or smartphone-based tasks.
- History of attempted suicide or suicidal ideation with plan that required hospital visit and/or change in level of care within 12 months prior to screening.
- Current active psychosis, confusional state or violent behaviour.
- Treatment with an investigational drug within 30 days prior to screening or 5 half-lives of the investigational drug, whichever is longer.
- Any serious medical condition or clinically significant laboratory, vital sign, or electrocardiogram abnormalities at screening that, precludes the patient's safe participation in and completion of the study.
- Antiplatelet or anticoagulant therapy within the 14 days prior to screening.
- History of bleeding diathesis or coagulopathy.
- History of gene therapy or cell transplantation or any other experimental brain surgery.
- Presence of implanted shunt for the drainage of CSF or an implanted CNS catheter.

HD Natural History Study Design

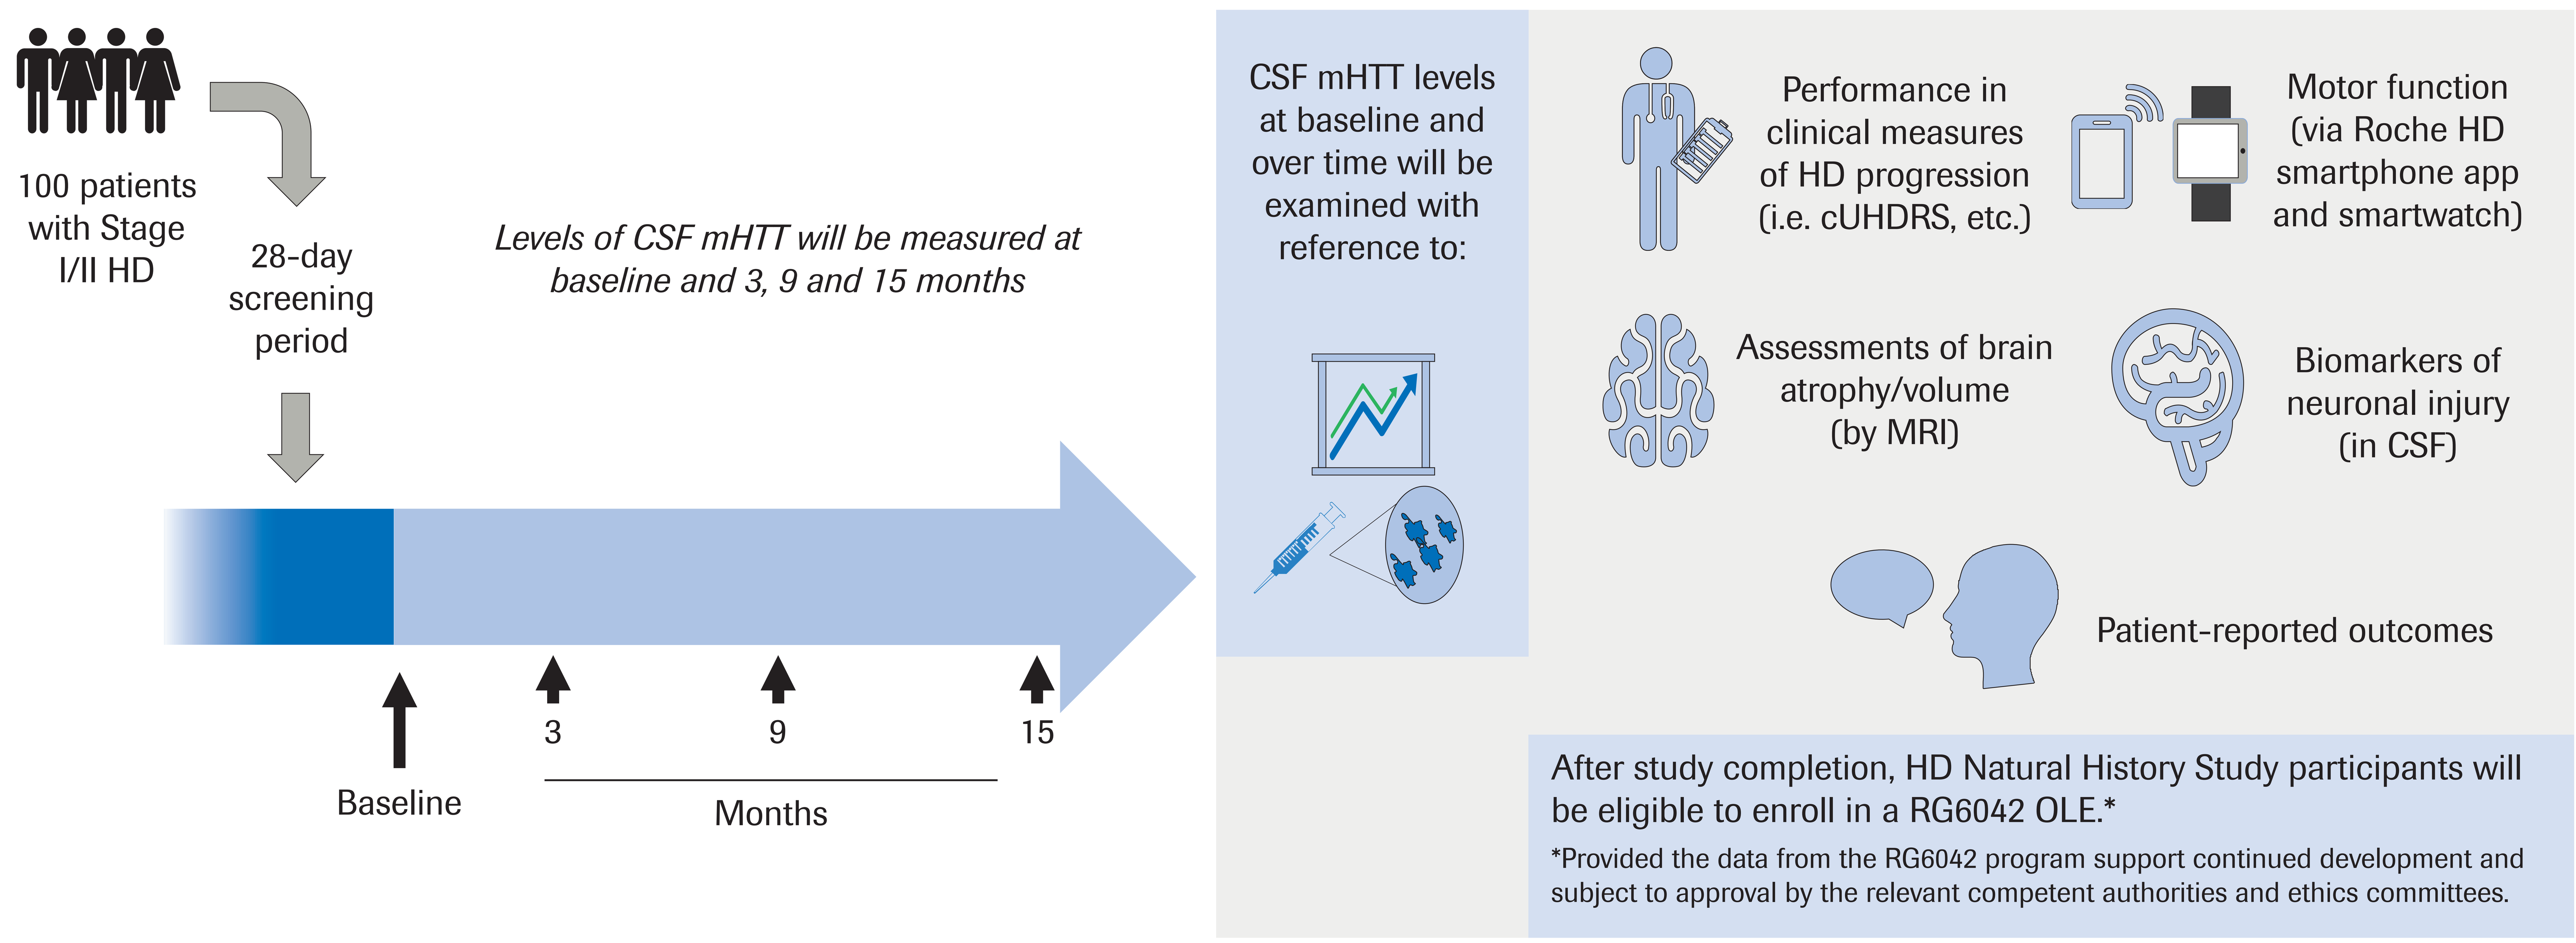
100 patients with Stage I/II HD
28-day screening period
Levels of CSF mHTT will be measured at baseline and 3, 9 and 15 months
Baseline
3
9
15
Months
CSF mHTT levels at baseline and over time will be examined with reference to:
Performance in clinical measures of HD progression (i.e. cUHDRS, etc.)
Motor function (via Roche HD smartphone app and smartwatch)
Assessments of brain atrophy/volume (by MRI)
Biomarkers of neuronal injury (in CSF)
Patient-reported outcomes
After study completion, HD Natural History Study participants will be eligible to enroll in a RG6042 OLE.*
*Provided the data from the RG6042 program support continued development and subject to approval by the relevant competent authorities and ethics committees.

Conclusions

This study will provide valuable information on the relationship between putative biomarkers, including CSF mHTT, and clinical outcomes in HD. It is expected to aid interpretation of current and future clinical trials.

Abbreviations

AE, adverse event; CNS, central nervous system; CSF, cerebrospinal fluid; cUHDRS, composite Unified Huntington's Disease Rating Scale; HD, Huntington's disease; IS, independence scale; *htt*, huntingtin gene; mHTT, mutant huntingtin protein; MRI, magnetic resonance imaging; NfL, neurofilament light chain; OLE, open-label extension; SAE, serious adverse event; SDMT, Symbol Digit Modalities Test; SWR, Stroop Word Reading; TFC, Total Functional Capacity; TMS, Total Motor Score.

Acknowledgments

We thank all the patients who participate in our studies and their families. This study is funded by F. Hoffmann La-Roche. We acknowledge the support of our colleagues, and the HD Natural History study investigators who contributed to the development of the study protocol; Sarah Tabrizi, Ed Wild, Blair Leavitt, Bernhard Landwehrmayer, Carsten Saft, Joseph Priller, Hugh Rickards, Anne Rosser, David Craufurd and Roger Barker. The authors thank Craig O'Hare, PhD, of MediTech Media UK for providing editorial support for this poster, which was funded by F. Hoffmann La-Roche in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

References

1. Wild EJ, et al. *J Clin Invest.* 2015; 125:1979-1986a.

Please scan using your QR reader application to access this poster on your mobile device.
NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details.
Alternatively this can be accessed at: https://bit.ly/2CRmCxN

Presented at HSG 2018: Unlocking HD, Houston, TX, USA; November 8–10, 2018