FIREFISH: Risdiplam (RG7916) improves motor function in babies with Type 1 SMA

G Baranello[1*], L Servais[2], JW Day[3], N Deconinck[4], E Mercuri[5], A Klein[6], B Darras[7], R Masson[1], H Kletzl[8], Y Cleary[8], M El-Khairi[9], T Seabrook[8], C Czech[8], M Gerber[8], P Somugompely[8], K Gelblin[8], K Gorni[8] and O Khwaja[8]

1. Carlo Besta Neurological Research Institute Foundation, Developmental Neurology Unit, Milan, Italy; 2. Institute of Myology, Paris, France and Reference Center for Neuromuscular Disease, Centre Hospitalier Régional de La Citadelle, Liège, Belgium; 3. Department of Neurology, Stanford University, Palo Alto, CA, USA; 4. Queen Fabiola Children's University Hospital, Université Libre de Bruxelles, Brussels, Belgium; Neuromuscular Reference Center UZ Ghent; Ghent, Belgium; 5. Paediatric Neurology and Nemo Center, Catholic University and Fondazione Policlinico Gemelli, Rome, Italy; 6. University Children's Hospital Basel, Basel, Switzerland; Inselspital, Bern, Switzerland; 7. Boston Children's Hospital, Harvard Medical School, Boston, MA, USA; 8. Roche Pharmaceutical Research and Early Development, Roche Innovation Center Basel, Basel, Switzerland; 9. Roche Products Ltd, Welwyn Garden City, UK

***Giovanni.Baranello@istituto-besta.it**

Background

- Type 1 spinal muscular atrophy (SMA) is a devastating neuromuscular disease with untreated babies failing to achieve major motor milestones and typically dying before 2 years of age.[1]
- SMA is caused by reduced levels of the survival of motor neuron (SMN) protein due to deletions and/or mutations of the *SMN1* gene.[1,2]
 - A second SMN gene, *SMN2*, produces low levels of functional SMN protein.[3]
- Increasing preclinical evidence indicates that SMA is a multisystem disorder.[4]
 - Therapies that increase SMN protein levels systemically may have broader therapeutic benefit than those targeting motor neurons alone.
- Risdiplam (RG7916; RO7034067) is an orally administered, centrally and peripherally distributed small molecule that modulates *SMN2* pre-mRNA splicing to increase SMN protein levels.[5]

Study design

- FIREFISH[6] is an ongoing, multicenter, open-label, two-part, operationally seamless study.
 - Part 1: Dose-finding (followed by open-label extension) – enrollment is complete.
 - Part 2: Efficacy and safety at the dose selected in Part 1 is ongoing.
 - Open-label risdiplam treatment for 24 months.

FIREFISH Type 1 SMA 1–7 months old Two *SMN2* gene copies		**Part 1 (N=21)***	**Part 2 (N=40)†**
	Primary endpoints	• Safety, tolerability, PK and PD of risdiplam. • Dose selection for Part 2.	• Proportion of babies sitting without support for 5 seconds after 12 months on treatment as assessed by Gross Motor Scale of the BSID-III.
	Secondary endpoints		• Motor function (HINE-2, CHOP-INTEND), PD/PK, safety, time to death or permanent ventilation, RP.

*Actual number of babies enrolled. †Target recruitment. Part 1 included multiple doses.

Risdiplam has been well tolerated at all doses assessed in FIREFISH Part 1

- To date, there have been no drug-related safety findings leading to withdrawal in any patient exposed to risdiplam, and no stopping rules have been met.
- Adverse events were reflective of the underlying disease:
 - 10 babies (47.6%) had at least one serious AE; two babies experienced fatal complications.*
 - Most frequent AEs: pyrexia, diarrhea, upper respiratory tract infection, ear infection, pneumonia, constipation, vomiting, cough, upper respiratory tract inflammation (see supplementary page).
- There have been no significant ophthalmological findings to date.

No baby has required tracheostomy, reached permanent ventilation† or lost the ability to swallow

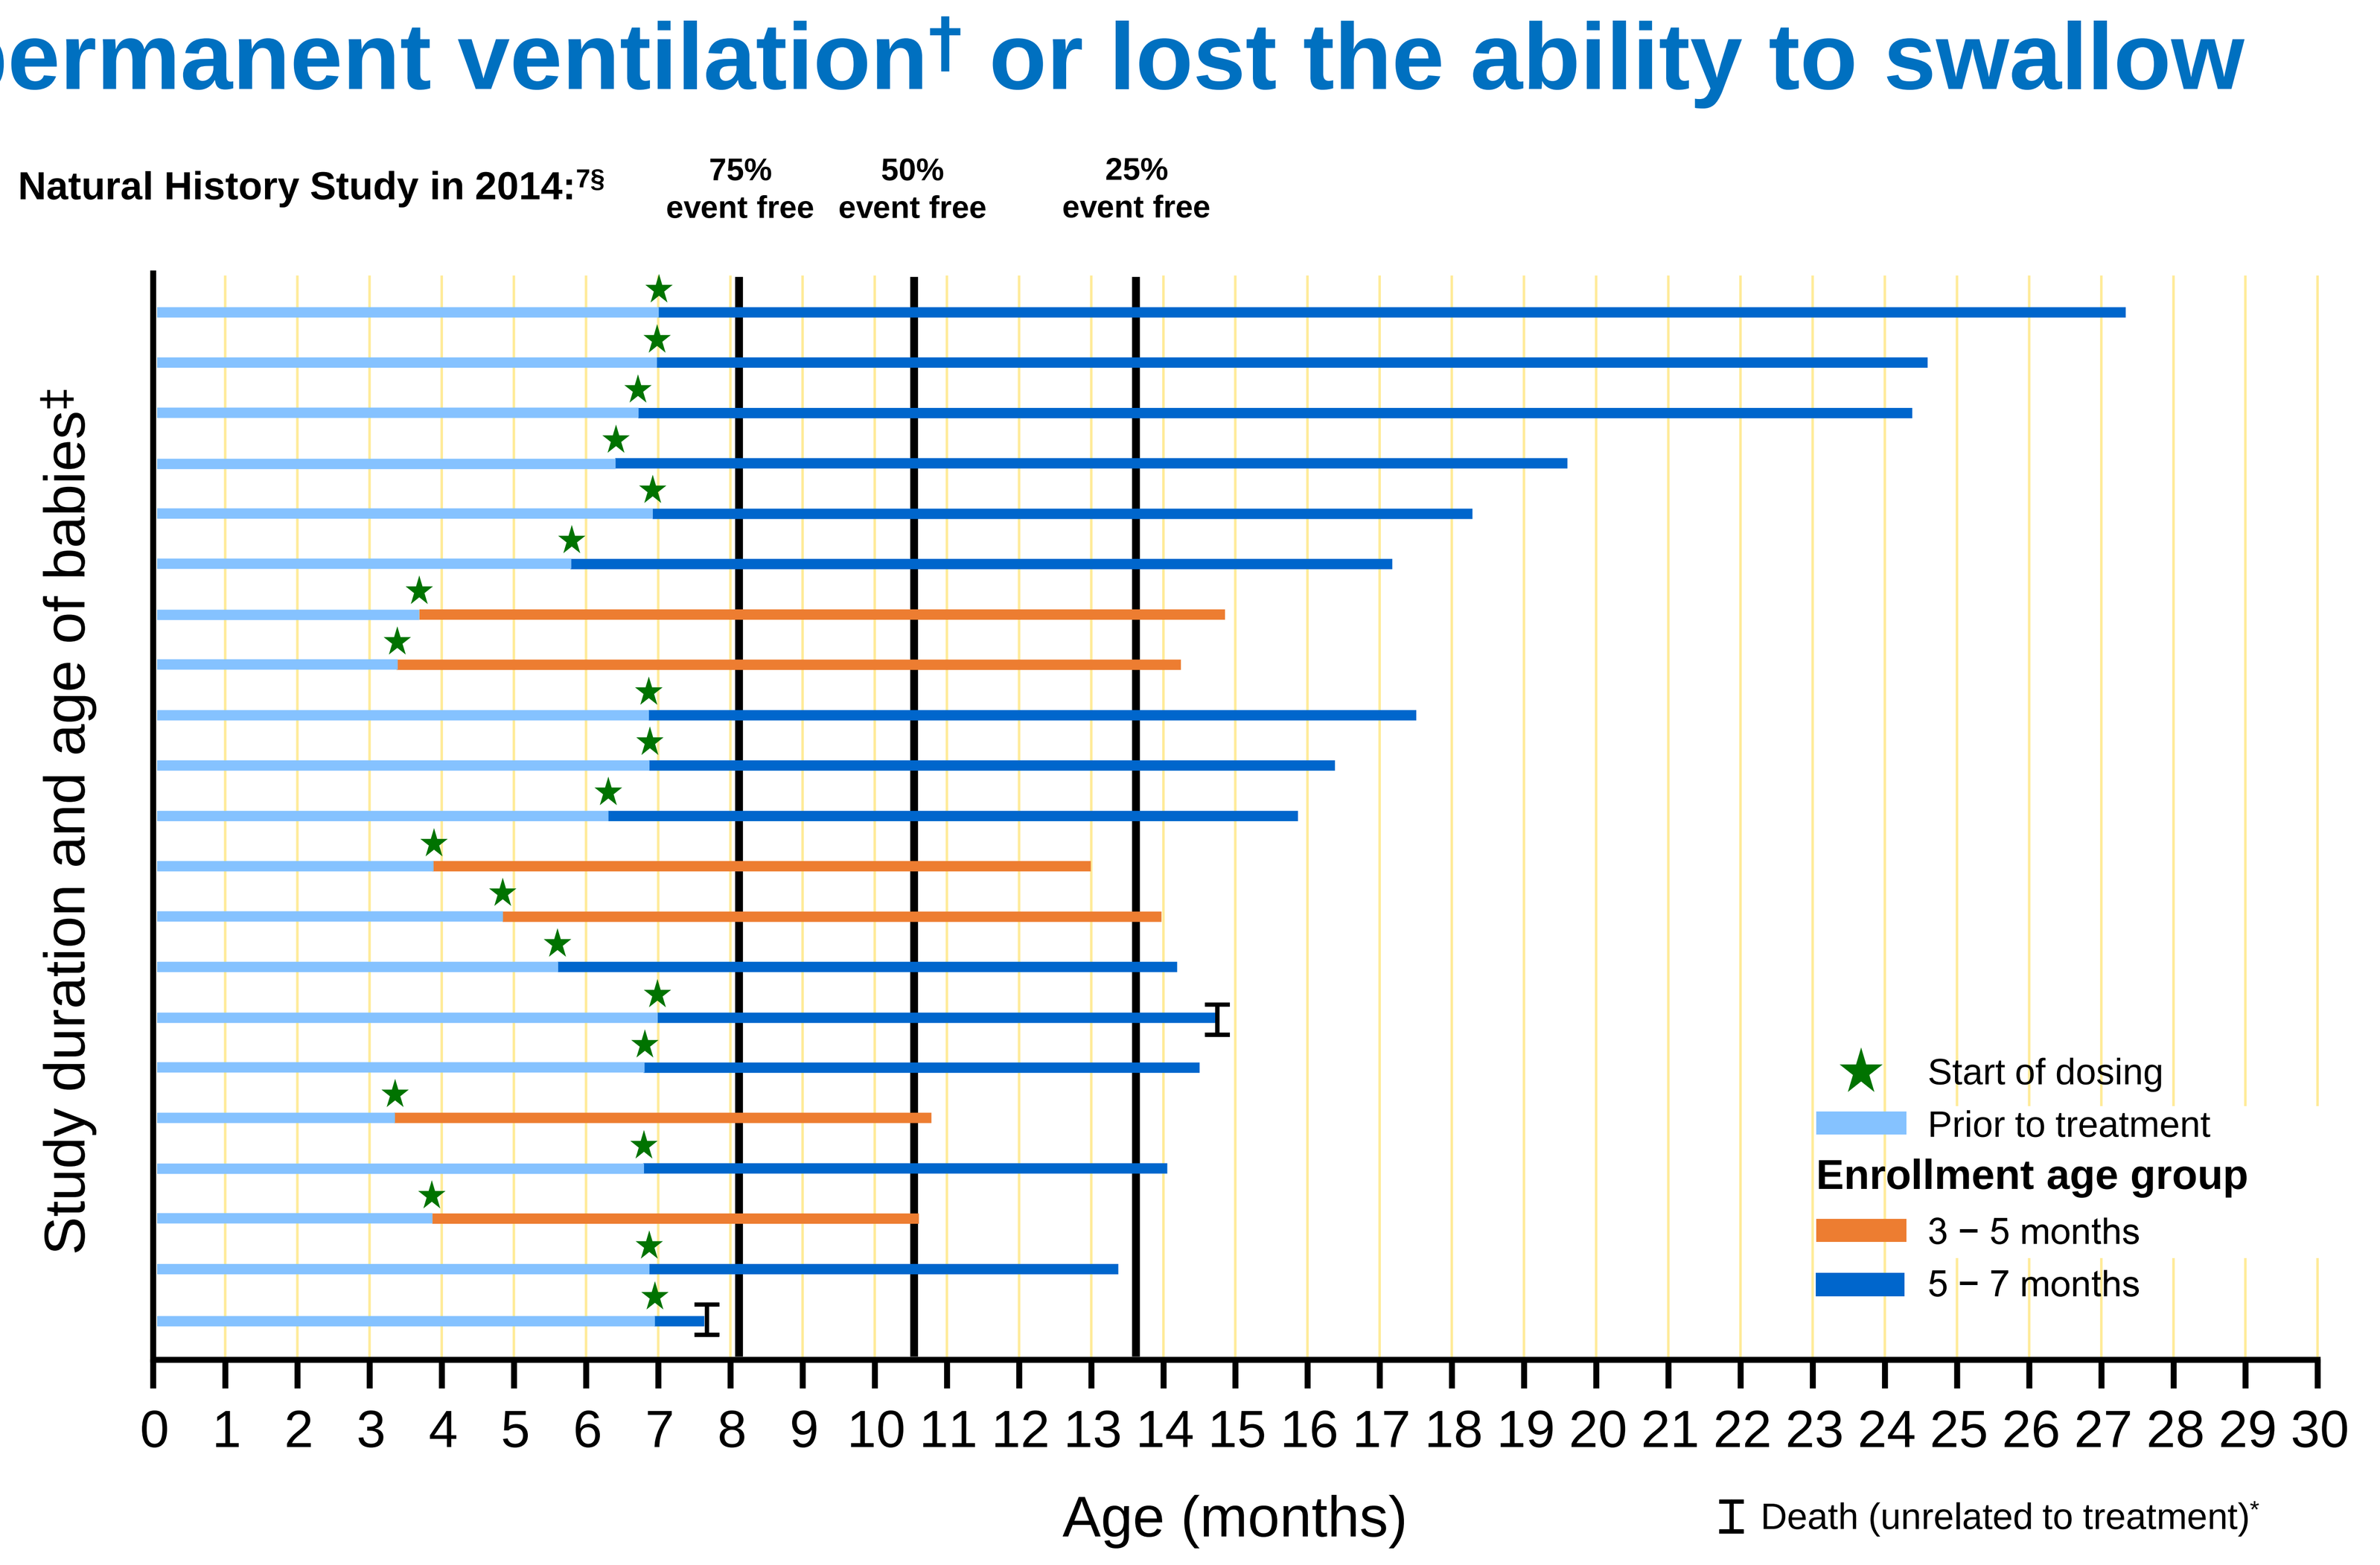
Natural History Study in 2014:7§
75% event free
50% event free
25% event free
Study duration and age of babies‡
Start of dosing
Prior to treatment
Enrollment age group
3 – 5 months
5 – 7 months
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 30
Age (months)
Death (unrelated to treatment)*

- 20/21 babies (95%) in FIREFISH were event-free at 10.5 months of age compared with 50% of babies at the same age in natural history studies[7]
- Median exposure to treatment: 9.53 months (0.7–20.3), N=21.

*Fatal events were reported in two patients: (1) respiratory tract infection viral in female infant aged 7 months at enrollment. First symptoms started on Day 4 with fatal outcome on study Day 21. The event was complicated by bilateral atelectasis. (2) Fatal cardiac arrest and respiratory failure on study Day 236 in female infant aged 7 months at enrollment on concurrent night ventilation (BIPAP for less than 16 hours per day) in the context of suspected aspiration; †permanent ventilation defined as ≥16 hours per day continuously for ≥2 weeks or continuous intubation ≥30 days; ‡Study duration is measured from the start date of the first dose to the date of the data cut off; §The median age at the combined endpoint for subjects with two *SMN2* copies was 10.5 months (IQR 8.1–13.6); event free = alive and no need for permanent ventilation (defined as ≥16 hours per day continuously for ≥2 weeks)[7]; Data cut-off: 7 September 2018.

Supplementary information

Please scan using your QR reader application to access the graphs and data presented in this poster. NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details. Alternatively this can be accessed at: http://bit.ly/2xwWSSV

Continuous improvement in median CHOP-INTEND total score from baseline in babies with Type 1 SMA

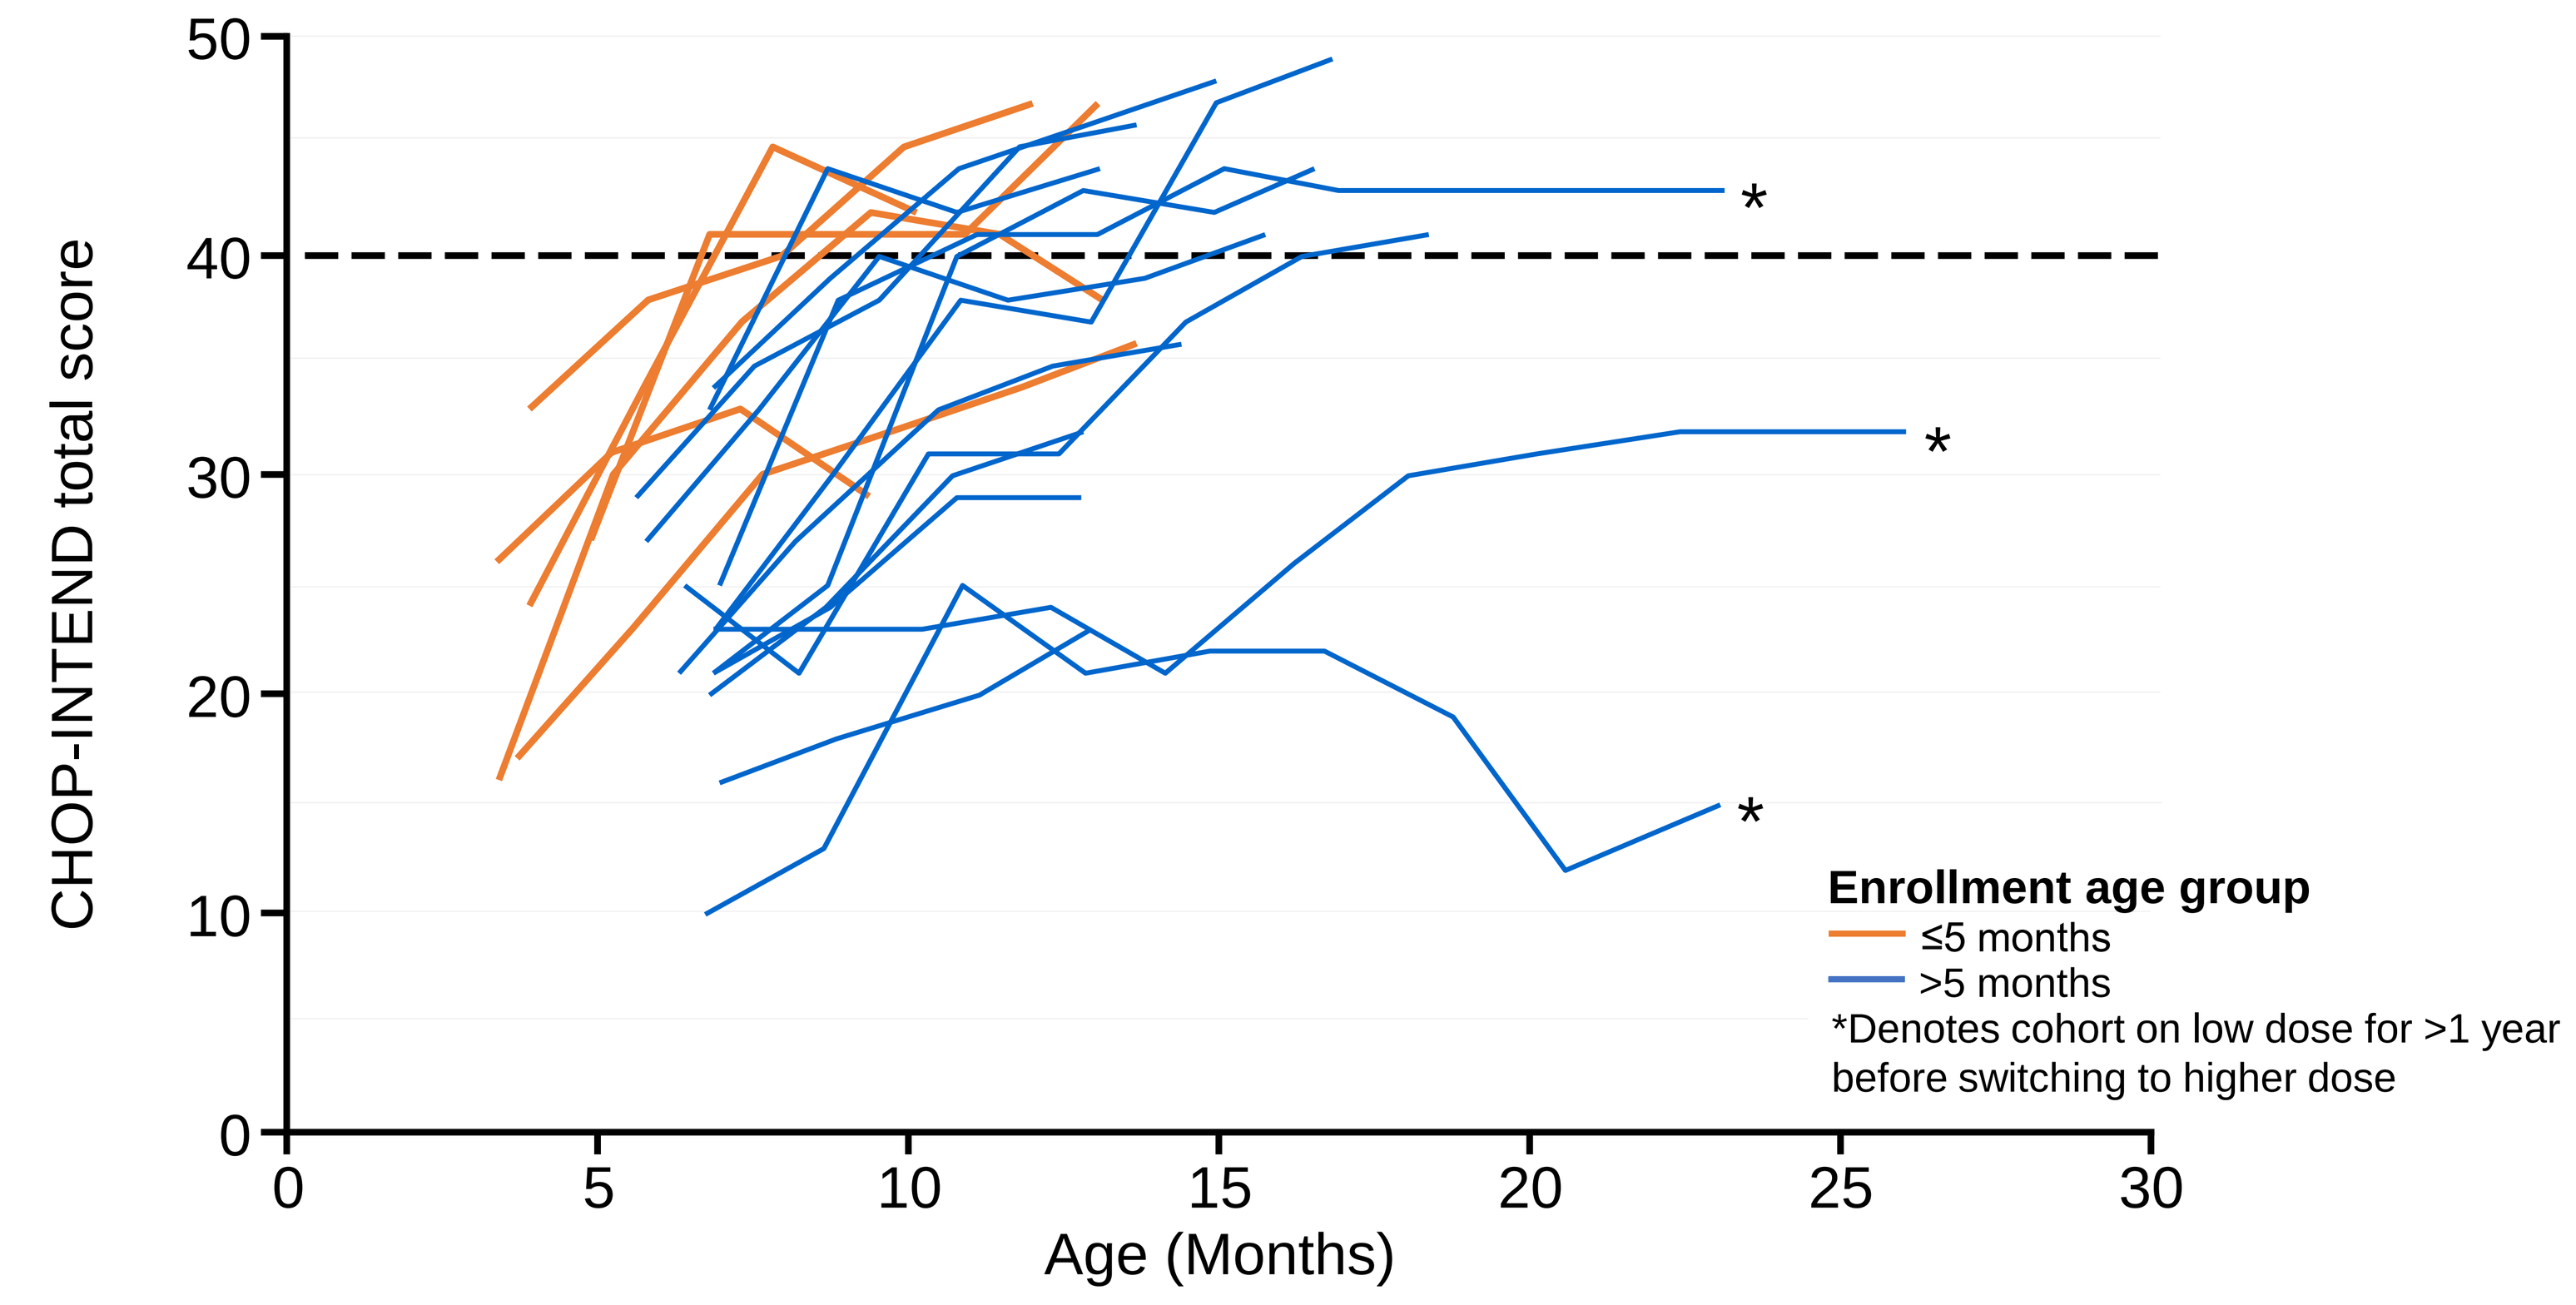
CHOP-INTEND total score
0
10
20
30
40
50
0
5
10
15
20
25
30
Age (Months)
Enrollment age group
≤5 months
>5 months
*Denotes cohort on low dose for >1 year before switching to higher dose

Each line represents an individual patient. Intent-to-treat patients from FIREFISH Part 1 (N=21). Data cut-off: 7 Sep 2018.

93% of babies had a ≥4-point improvement in CHOP-INTEND total score from baseline*

The median change from baseline in CHOP-INTEND total score was 16*

57% of babies had achieved a CHOP-INTEND total score of ≥40 points*

*At Day 245 (8 months), which is the latest timepoint at which a majority of babies have follow-up data available. N=14.

Improved motor function and milestones achieved in FIREFISH Part 1

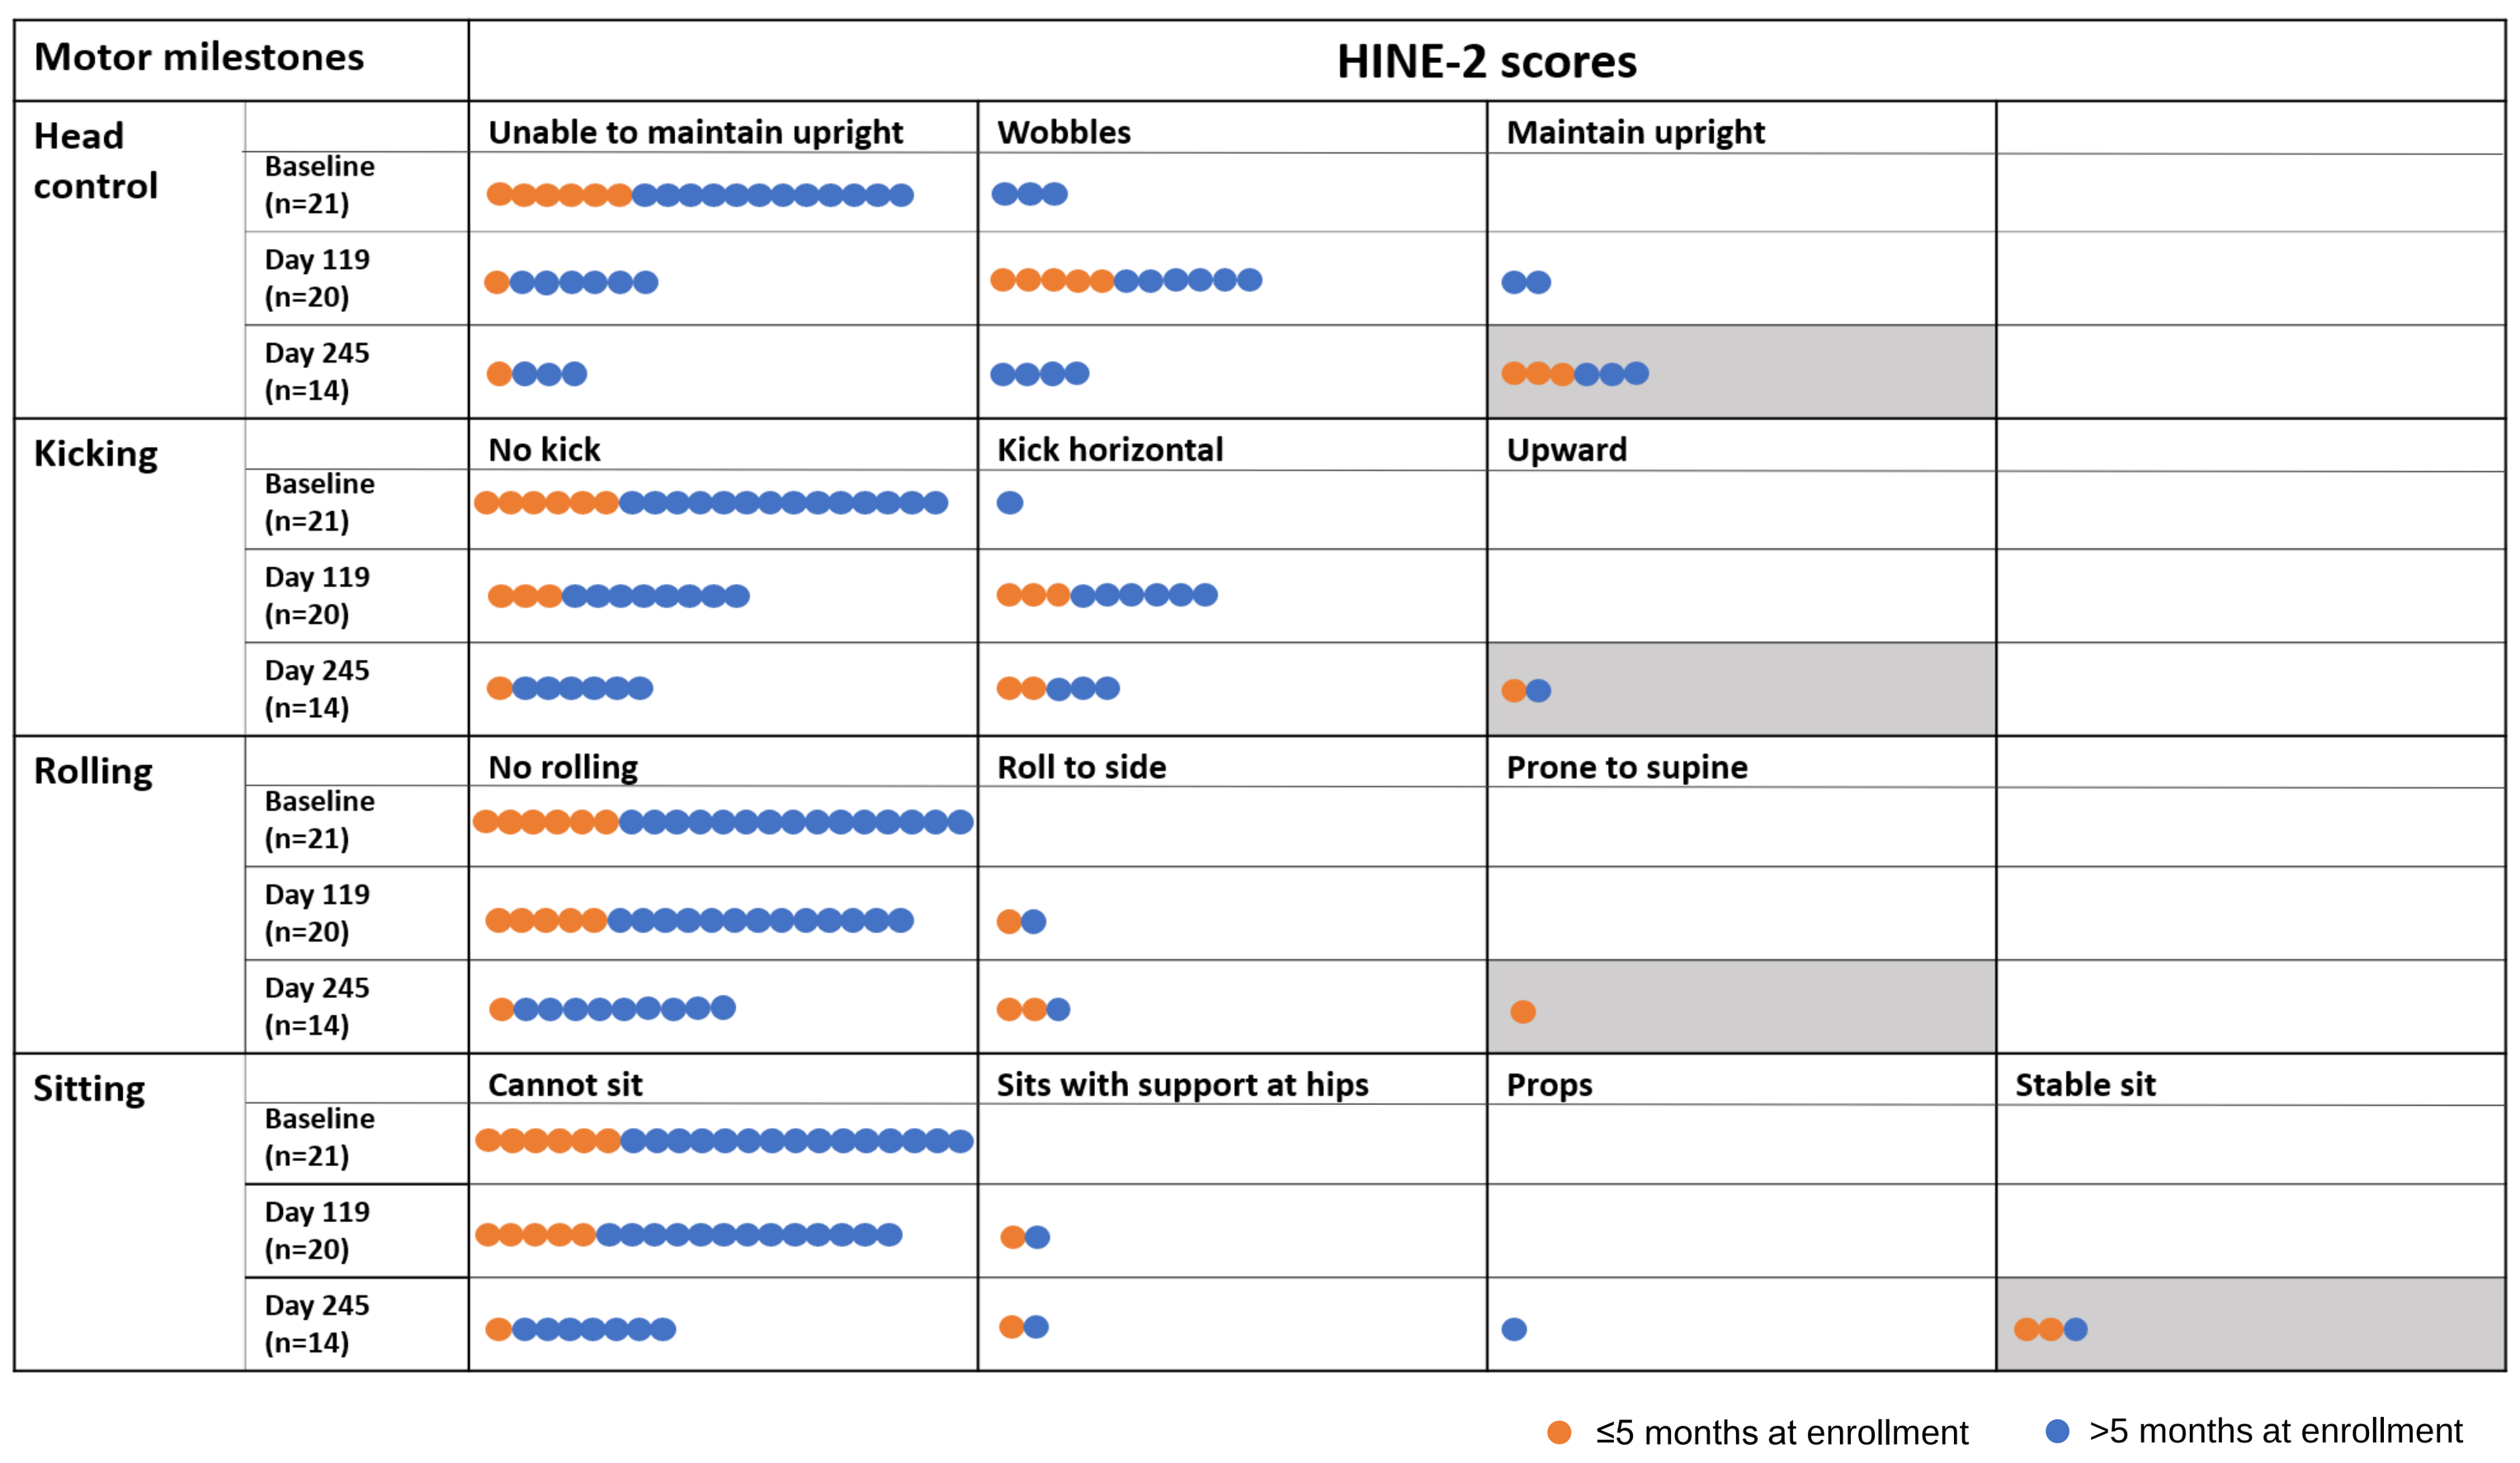
Motor milestones
HINE-2 scores
Head control
Baseline (n=21)
Day 119 (n=20)
Day 245 (n=14)
Unable to maintain upright
Wobbles
Maintain upright
Kicking
No kick
Kick horizontal
Upward
Rolling
No rolling
Roll to side
Prone to supine
Sitting
Cannot sit
Sits with support at hips
Props
Stable sit
≤5 months at enrollment
>5 months at enrollment

Each circle represents an individual baby. Intent-to-treat babies from FIREFISH Part 1 (N=21). Data cut-off: 7 September 2018.

95% (20/21) babies were event-free at 10.5 monthsof age compared with 50% of babies event-free at the same age in natural history studies[7]

No baby has required tracheostomy, reached permanent ventilation or lost the ability to swallow*

At Day 245 of treatment, 6/14 (43%) babies were able to sit (with or without support), including 3 (21%) who achieved unassisted sitting†

Global recruitment into FIREFISH Part 2 is underway and will assess the efficacy and safety of risdiplam

*Latest visit day: 546 days; †Day 245 (8 months) is the latest time point at which the majority of babies have follow-up data available.

Abbreviations

BSID-III, Bayley Scales of Infant Development, Third Edition; BIPAP, bilevel positive airway pressure; CHOP-INTEND, Children's Hospital of Philadelphia Infant Test of Neuromuscular Disorders; HINE-2, Hammersmith Infant Neurological Examination Module 2; IQR, interquartile range; PD, pharmacodynamics; PK, pharmacokinetics; PNCR, Pediatric Neuromuscular Research Network for SMA; RP, respiratory plethysmography; SMA, spinal muscular atrophy; SMN, survival of motor neuron.

Acknowledgments

We would like to thank the babies and their families for participation in this study, as well as the investigators and trial staff involved in the FIREFISH study. This study is funded by F. Hoffmann La-Roche AG. We would also like to thank our collaborators at PTC Therapeutics and the SMA Foundation. The study was funded by F. Hoffmann-La Roche AG, Basel, Switzerland. Writing and editorial assistance was provided by Lindsey Weedon, of MediTech Media, UK, in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

References

1. Mercuri E, et al. *Lancet Neurol.* 2012; 11:443–452;
2. Lefebvre S, et al. *Cell.* 1995; 80:155–165;
3. Lorson CL, et al. *PNAS.* 1999; 96:6307–6311;
4. Hamilton G and Gillingwater TH. *Trends Mol Med.* 2013; 19:40–50;
5. Ratni H, et al. *J Med Chem.* 2018. 61:6501–6517
6. Clinicaltrials.gov/ct2/show/NCT02913482 (Accessed July 2018).
7. Finkel R, et al. *Neurology.* 2014; 83:810–8;

Presented at the 23rd International Annual Congress of the World Muscle Society, Mendoza, Argentina, October 2–6, 2018

SUPPLEMENTARY INFORMATION – the content below was not in the poster presented at WMS 2018 but is available via a QR code

FIREFISH: Risdiplam (RG7916) improves motor function in babies with Type 1 SMA

G Baranello[1], L Servais[2], JW Day[3], N Deconinck[4], E Mercuri[5], A Klein[6], B Darras[7], R Masson[1], H Kletzl[8], Y Cleary[8], M El-Khairi[8], T Seabrook[8], C Czech[8], M Gerber[8], P Somugompely[8], K Gelblin[8], K Gorni[8], O Khwaja[8]

1. Carlo Besta Neurological Research Institute Foundation, Developmental Neurology Unit, Milan, Italy; 2. Institute of Myology, Paris, France and Reference Center for Neuromuscular Disease, Centre Hospitalier Régional de La Citadelle, Liège, Belgium; 3. Department of Neurology, Stanford University, Palo Alto, CA, USA; 4. Queen Fabiola Children's University Hospital, Université Libre de Bruxelles, Brussels, Belgium; Neuromuscular Reference Center UZ Ghent; Ghent, Belgium; 5. Paediatric Neurology and Nemo Center, Catholic University and Fondazione Policlinico Gemelli, Rome, Italy; 6. University Children's Hospital Basel, Basel, Switzerland; Inselspital, Bern, Switzerland; 7. Boston Children's Hospital, Harvard Medical School, Boston, MA, USA; 8. Roche Pharmaceutical Research and Early Development, Roche Innovation Center Basel, Basel, Switzerland; 9. Roche Products Ltd, Welwyn Garden City, UK

*Giovanni.Baranello@istituto-besta.it

FIREFISH Part 1: Patient baseline characteristics

	All babies (N=21)
Gender	
Female, n (%)	15 (71)
Male, n (%)	6 (29)
Age at diagnosis (months)	
Median (range)	3.0 (0.9–5.4)
Age at first dose (months)	
Median (range)	6.7 (3.4–7.0)
Weight at baseline (g)	
Median (range)	6,700 (5,150–8,855)
CHOP-Intend score	
Mean (SD)	23.6 (6.0)
HINE-2 score	
Mean (SD)	1.0 (0.7)

Data cut-off: 7 September 2018.

No patients have left the study due to drug-related adverse events

		Babies (N=21)
Total number of AEs, N Babies with at least one AE, n (%)		138 21 (100)
Most frequent AEs,* number of babies (%)	Pyrexia	11 (52.4)
	Diarrhea	6 (28.6)
	Upper respiratory tract infection	4 (19.0)
	Ear infection	3 (14.3)
	Pneumonia	3 (14.3)
	Constipation	3 (14.3)
	Vomiting	3 (14.3)
	Cough	3 (14.3)
	Upper respiratory tract inflammation	3 (14.3)
Patients with at least one serious AE, n (%)		10 (47.6)
Patients with at least one Grade 3–5 AE, n (%)		8 (38.1)
Discontinuations due to drug-related AEs, n		0

*Investigator text for AEs encoded using MedDRA version 21.0. Percentages based on N in column headings. Data cut-off: 7 September 2018.

Abbreviations

AE, adverse event; CHOP-INTEND, Children's Hospital of Philadelphia Infant Test of Neuromuscular Disorders; HINE-2, Hammersmith Infant Neurological Examination Module 2; IQR, interquartile range; SAE, serious adverse event; SD, standard deviation.

Supplementary page

(This page provides additional online content and will not be available at the poster stand. Attendees will be able to view and download this page by scanning the QR code on the main poster)

At Day 245, the median change from baseline in CHOP-INTEND total score was 16 (n=14)

Median change in CHOP-INTEND total score from baseline

	Aged ≤5 months at enrollment	Aged >5 months at enrollment	Overall
Day 56	8.0 (n=6)	4.5 (n=14)	5.5 (n=20)
Day 119	13.5 (n=6)	10.0 (n=14)	11.0 (n=20)
Day 182	14.5 (n=6)	11.5 (n=14)	12.0 (n=20)
Day 245	18.5 (n=4)	14.5 (n=10)	16.0 (n=14)

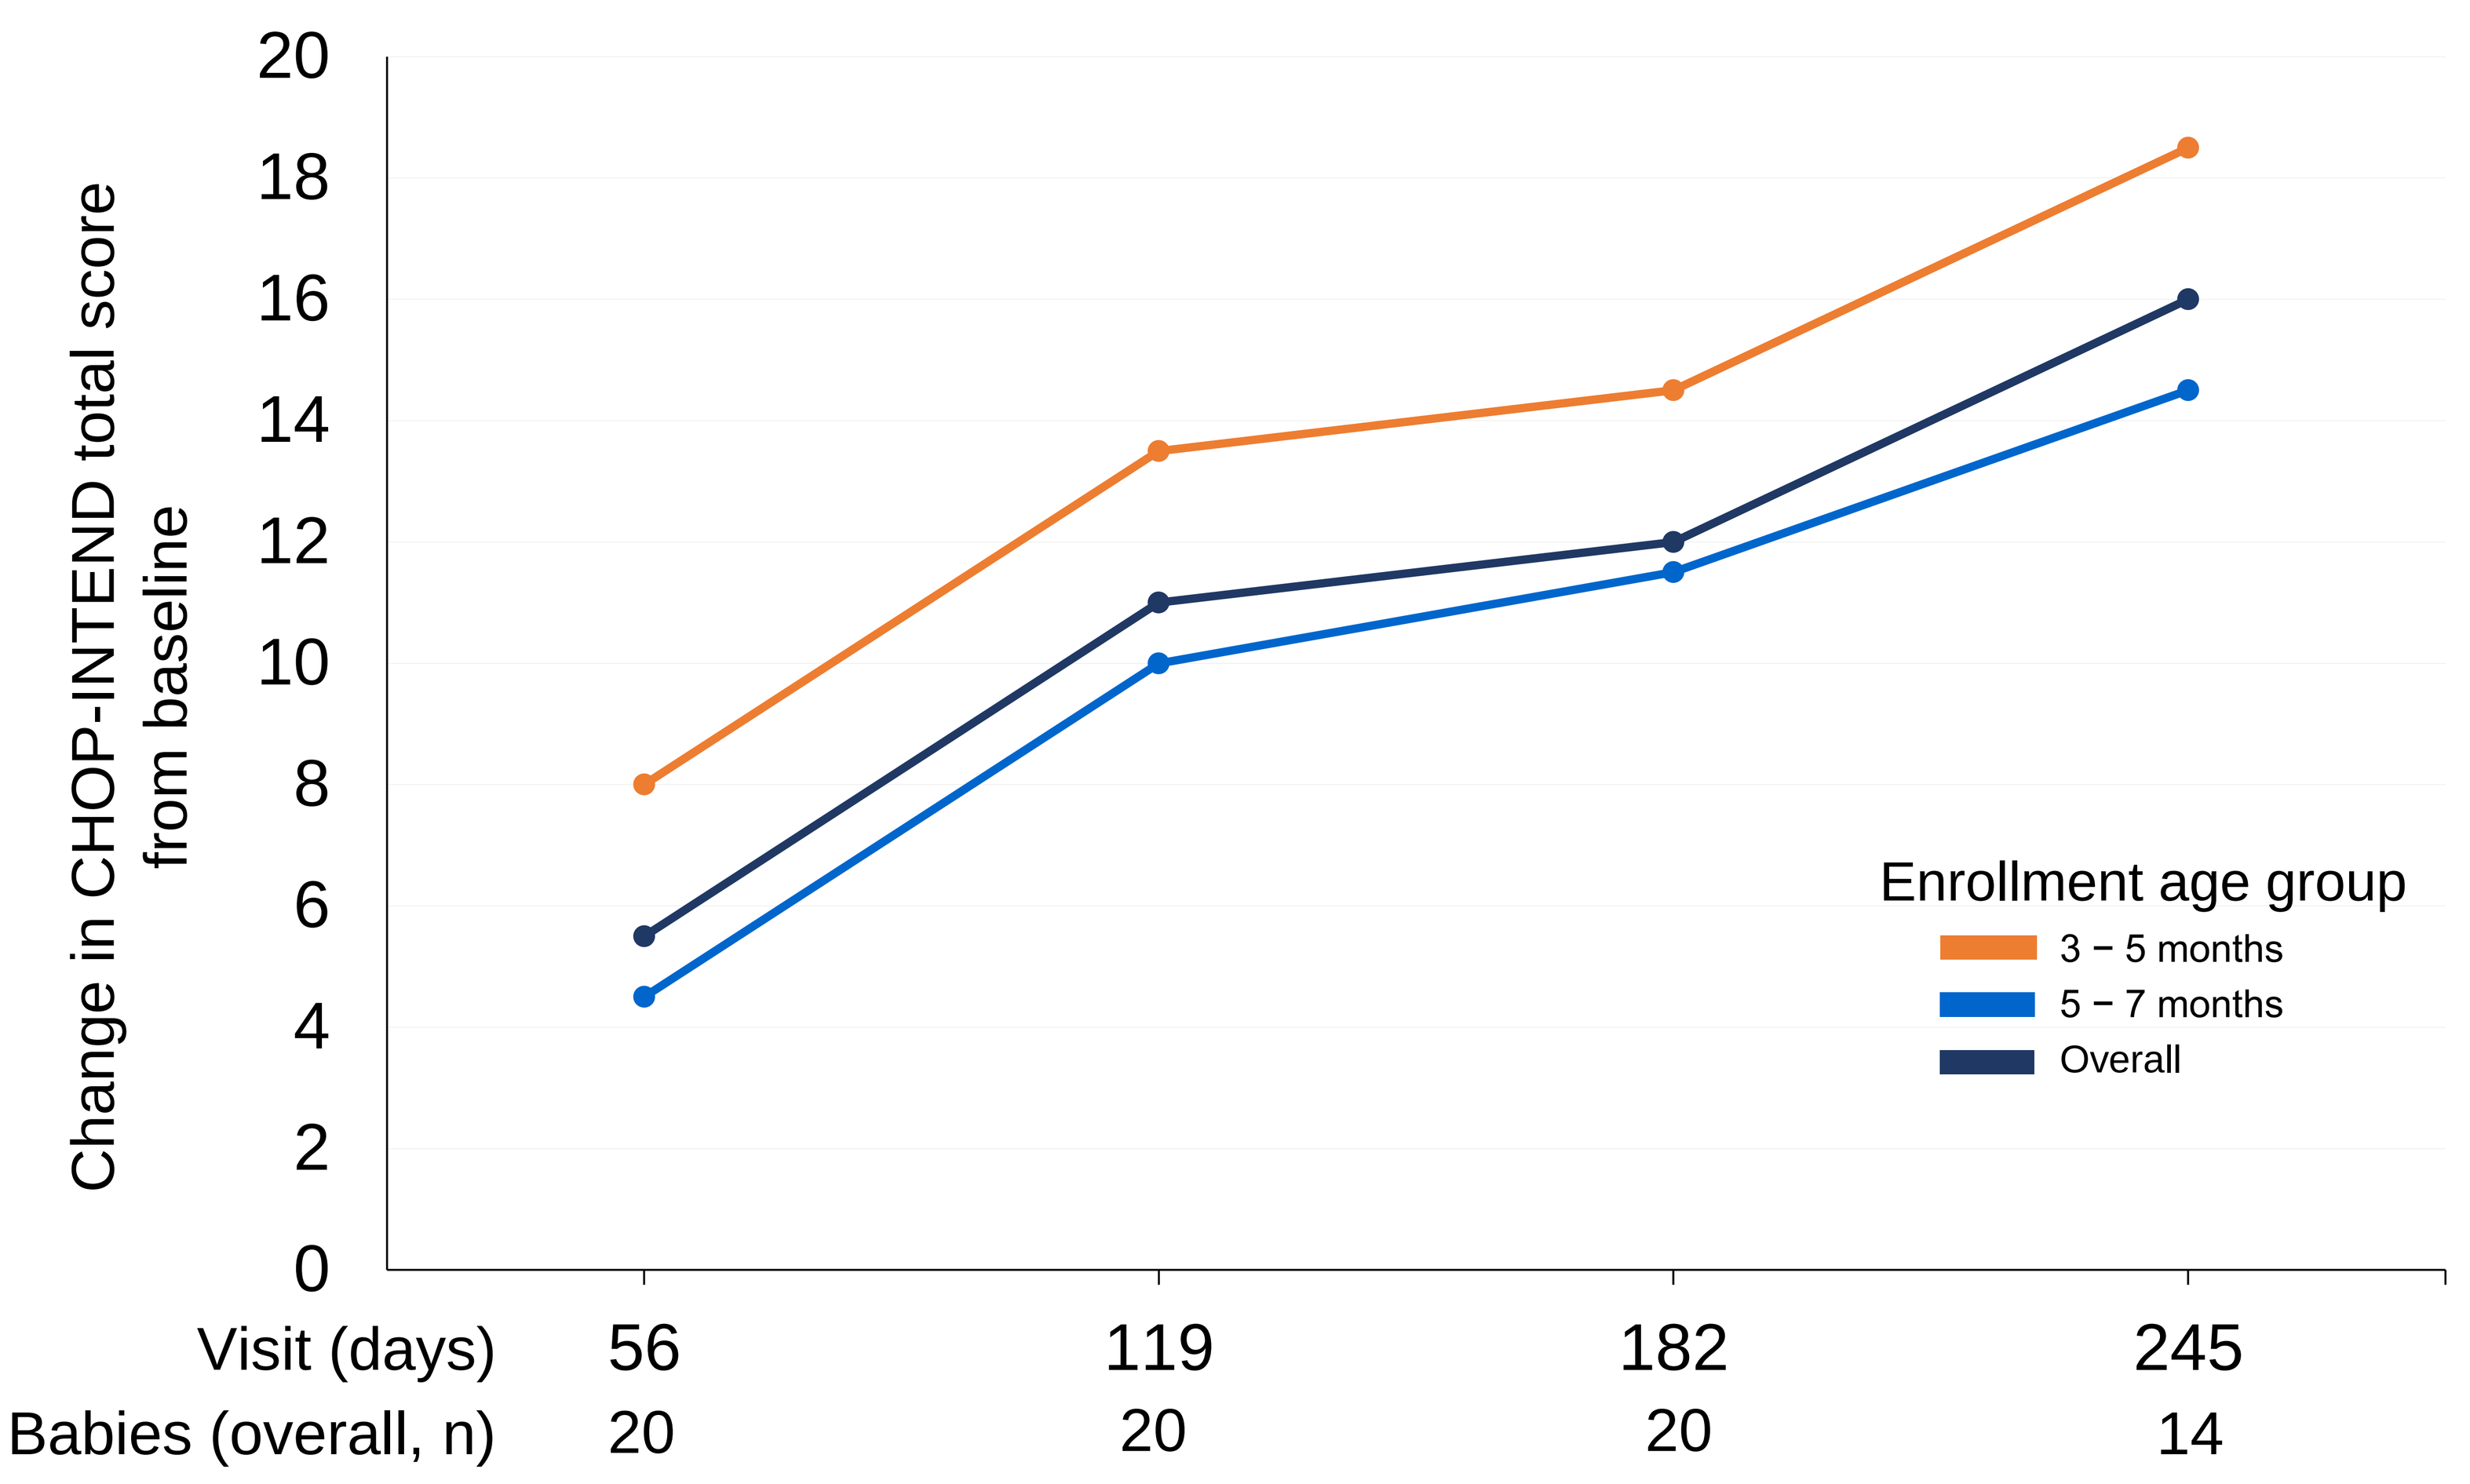

Change in CHOP-INTEND total score from baseline
0
2
4
6
8
10
12
14
16
18
20
Enrollment age group
3 – 5 months
5 – 7 months
Overall
Visit (days)
56
119
182
245
Babies (overall, n)
20
20
20
14

Intent-to-treat patients from FIREFISH Part 1 (N=21). Day 245 (8 months) is the latest time point at which the majority of babies have follow-up data available. Data cut-off: 7 September 2018.

CHOP-INTEND total score: The majority of babies showed a ≥4-point improvement from baseline

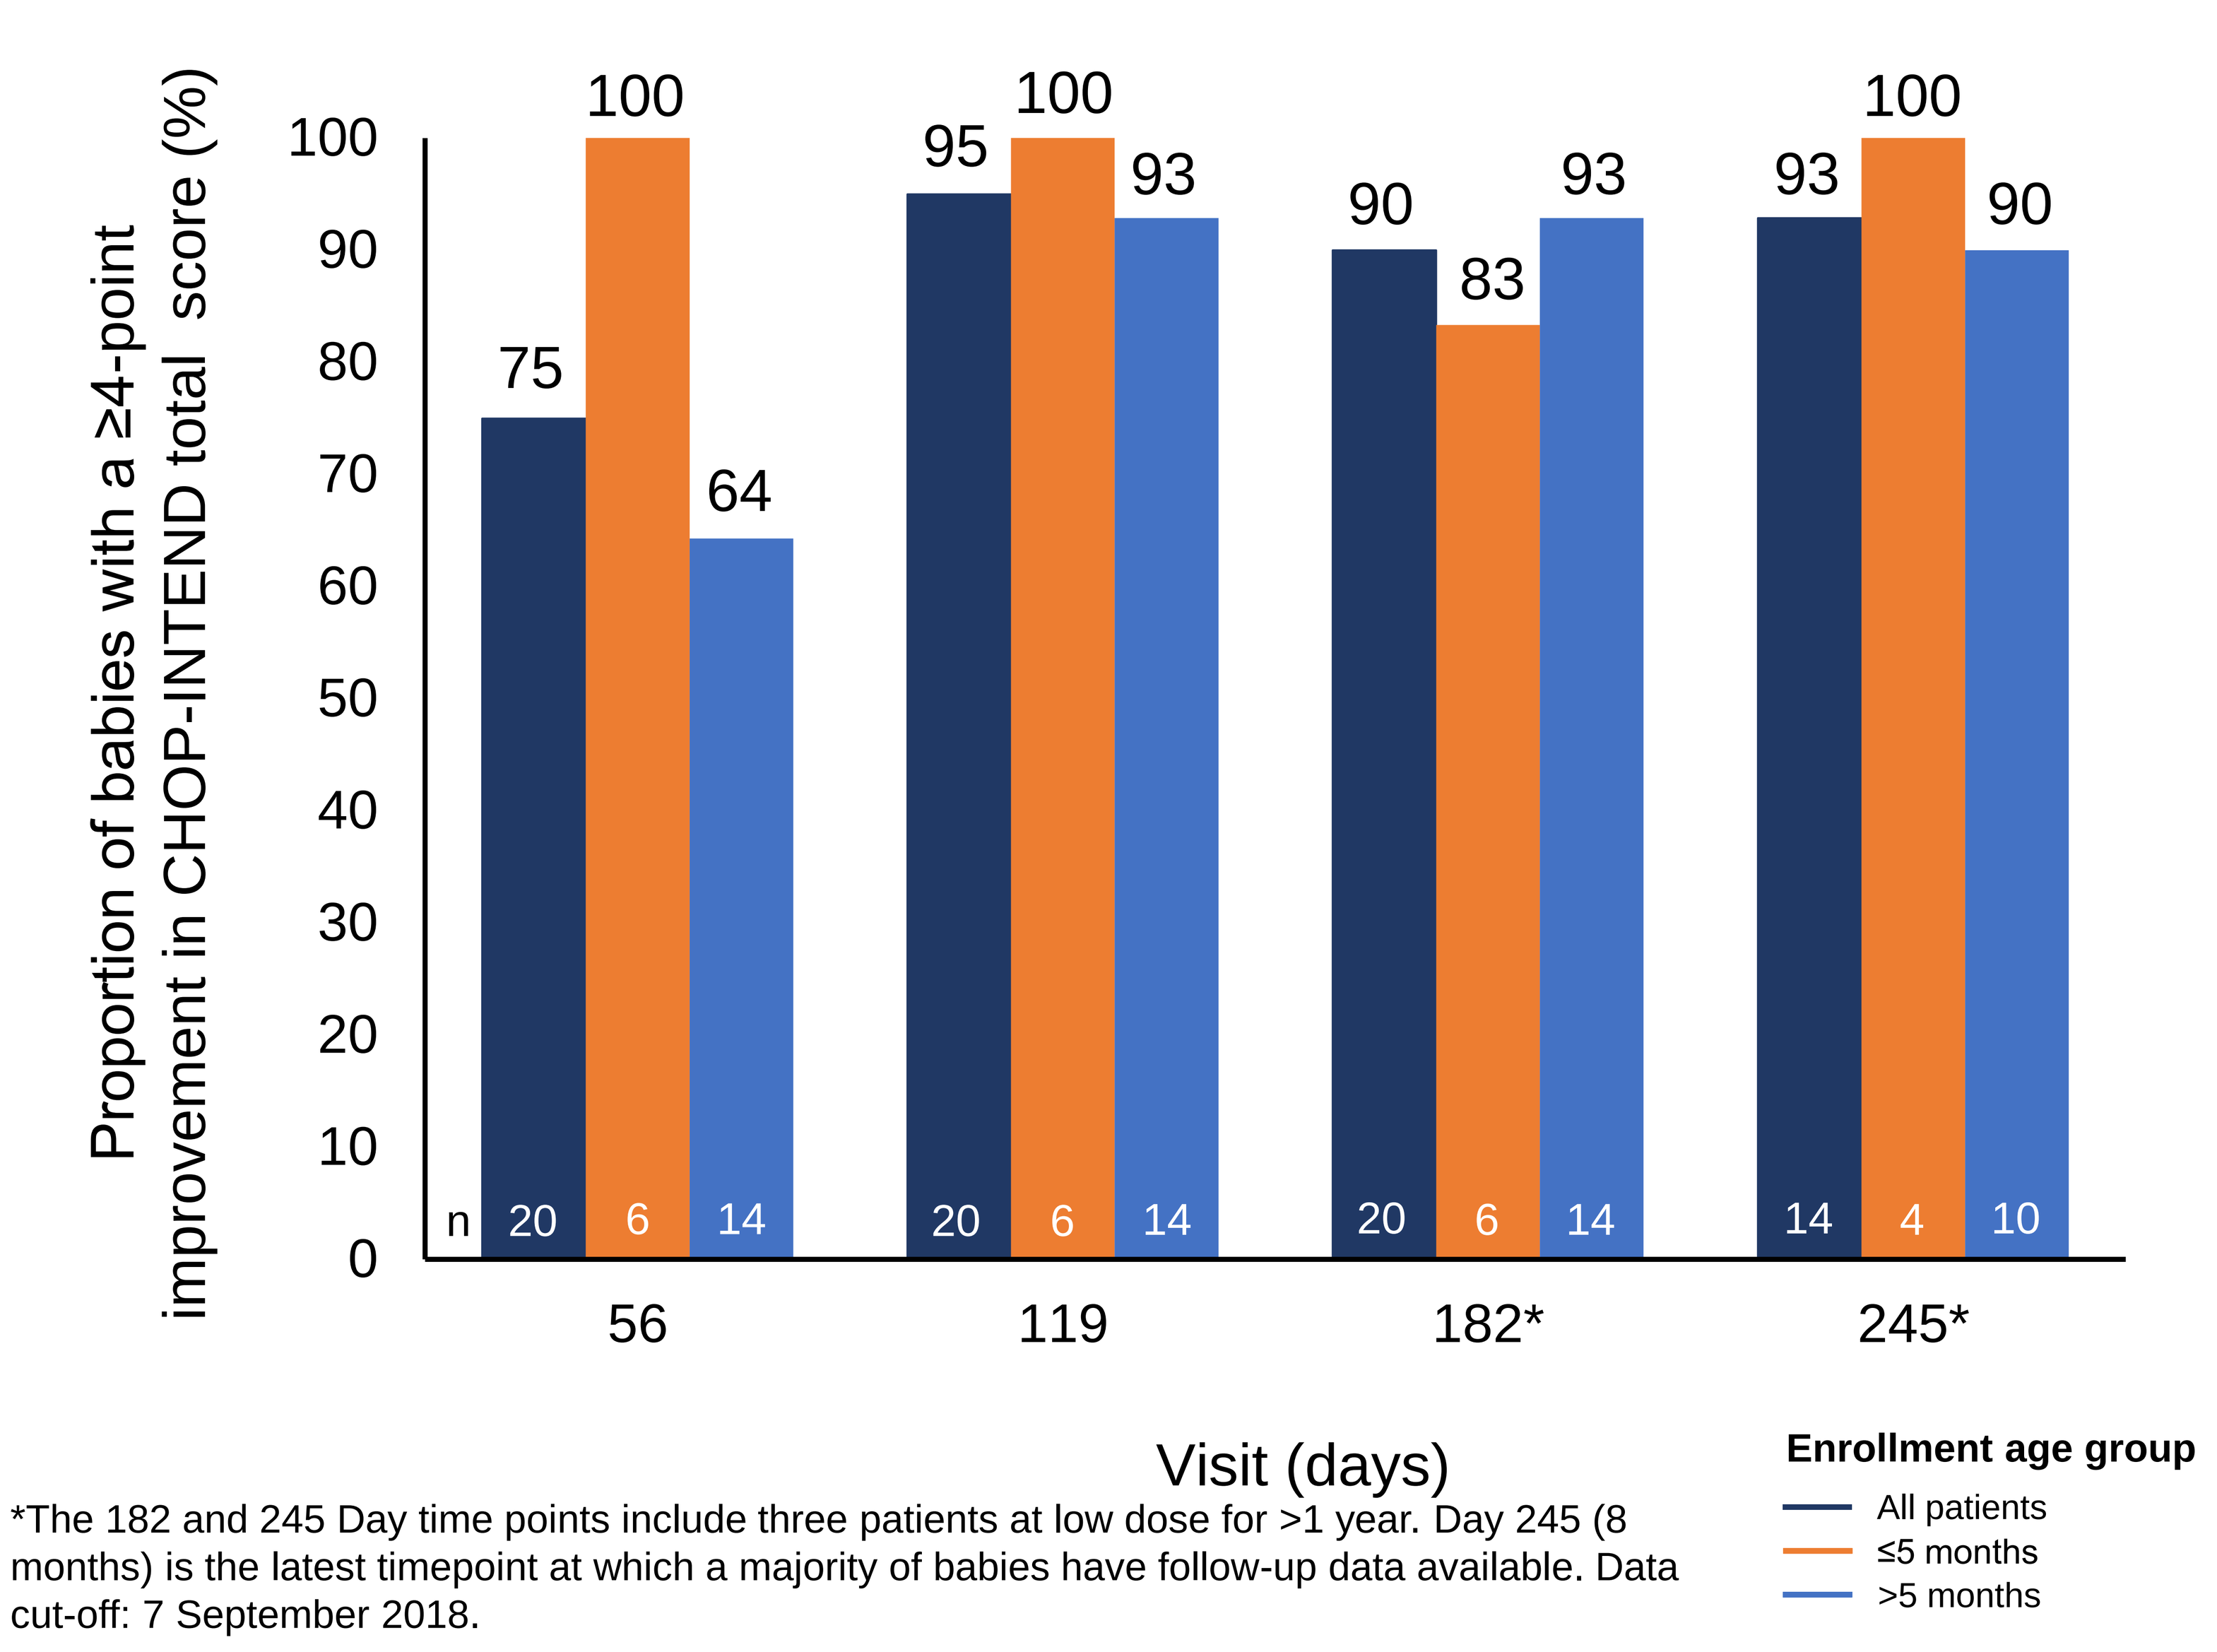

Proportion of babies with a ≥4-point improvement in CHOP-INTEND total score (%)
0
10
20
30
40
50
60
70
80
90
100
75
100
64
95
100
93
90
83
93
93
100
90
n
20
6
14
20
6
14
20
6
14
14
4
10
56
119
182*
245*
Visit (days)
Enrollment age group
All patients
≤5 months
>5 months

*The 182 and 245 Day time points include three patients at low dose for >1 year. Day 245 (8 months) is the latest timepoint at which a majority of babies have follow-up data available. Data cut-off: 7 September 2018.