A study of risdiplam (RG7916) in babies with pre-symptomatic spinal muscular atrophy

E Bertini[1], JW Day[2], M Al Muhaizea[3], H Xiong[4], L Servais[5], A Prufer[6], J Buchbjerg[7]*, WY Yeung[8], K Gorni[7] and O Khwaja[7]

[1]Department of Neurosciences and Neurorehabilitation, Bambino Gesù Children's Research Hospital IRCCS, Rome, Italy; [2]Department of Neurology, Stanford University, Palo Alto, CA, USA; [3]Department of Neurosciences, King Faisal Specialist Hospital & Research Center-Riyadh, Kingdom of Saudi Arabia; [4]Department of Pediatrics, Peking University First Hospital, Beijing 100034, China; [5]Institute of Myology, Paris, France and Reference Center for Neuromuscular Disease, Centre Hospitalier Régional de La Citadelle, Liège, Belgium; [6]Federal University of Rio de Janeiro, Rio de Janeiro, Brazil; [7]Roche Pharmaceutical Research and Early Development, Roche Innovation Center Basel, Switzerland; [8]Roche Products Ltd, Welwyn Garden City, United Kingdom
*jeppe.buchbjerg@roche.com

Background

- Spinal muscular atrophy (SMA) is a severe, progressive, inherited neuromuscular disease leading to loss of motor function and reduced life expectancy.[1]
- SMA is caused by reduced levels of the survival of motor neuron (SMN) protein due to deletions and/or mutations of the *SMN1* gene.[1]
 — A second SMN gene, SMN2, only produces low levels of functional SMN protein.[2]
- Risdiplam (RG7916; RO7034067) is an investigational, orally administered, centrally and peripherally distributed small molecule that modulates *SMN2* pre-mRNA splicing to increase SMN protein levels.[3]
 — Risdiplam is currently under investigation in three clinical trials in patients with SMA[4–6]
- Most motor neuron degeneration occurs in the first months of life in babies with Type 1 SMA, thus the timing of therapeutic intervention is crucial.[7]
- In recent clinical studies babies with pre-symptomatic SMA have shown a higher and more rapid achievement of motor milestones and reduced disease severity when SMN upregulation therapy was administered early.[8]

Please scan using your QR reader application to access the graphs and data presented in this poster. NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details. Alternatively this can be accessed at: http://bit.ly/2NShnUC

Study design

- RAINBOWFISH (BN40703) is an open-label, single-arm, multicenter clinical study to investigate the efficacy, safety, PK, and PD of risdiplam in babies with genetically diagnosed SMA who are not yet presenting with symptoms.

RAINBOWFISH Birth–6 weeks old	Key inclusion criteria	• Genetic documentation of 5q *SMN1* homozygous gene deletion or mutation or compound heterozygous mutation (regardless of *SMN2* copy number). • Up to 6 weeks (42 days) of age at the time of first dose. • Adequately recovered from any acute illness at baseline. • Absence of clinical signs or symptoms prior to dosing that are strongly suggestive of SMA.
	Key exclusion criteria	• Concomitant or previous administration of an *SMN2*-targeting antisense oligonucleotide, *SMN2*-splicing modifier, or gene therapy. • Requiring invasive ventilation, tracheostomy or awake non-invasive ventilation. • Presence of significant concurrent syndromes or diseases.

Study schema

- Recruitment will be global.
- Enrolled babies will receive risdiplam orally once daily for 24 months, followed by an open-label extension phase of at least 36 months.

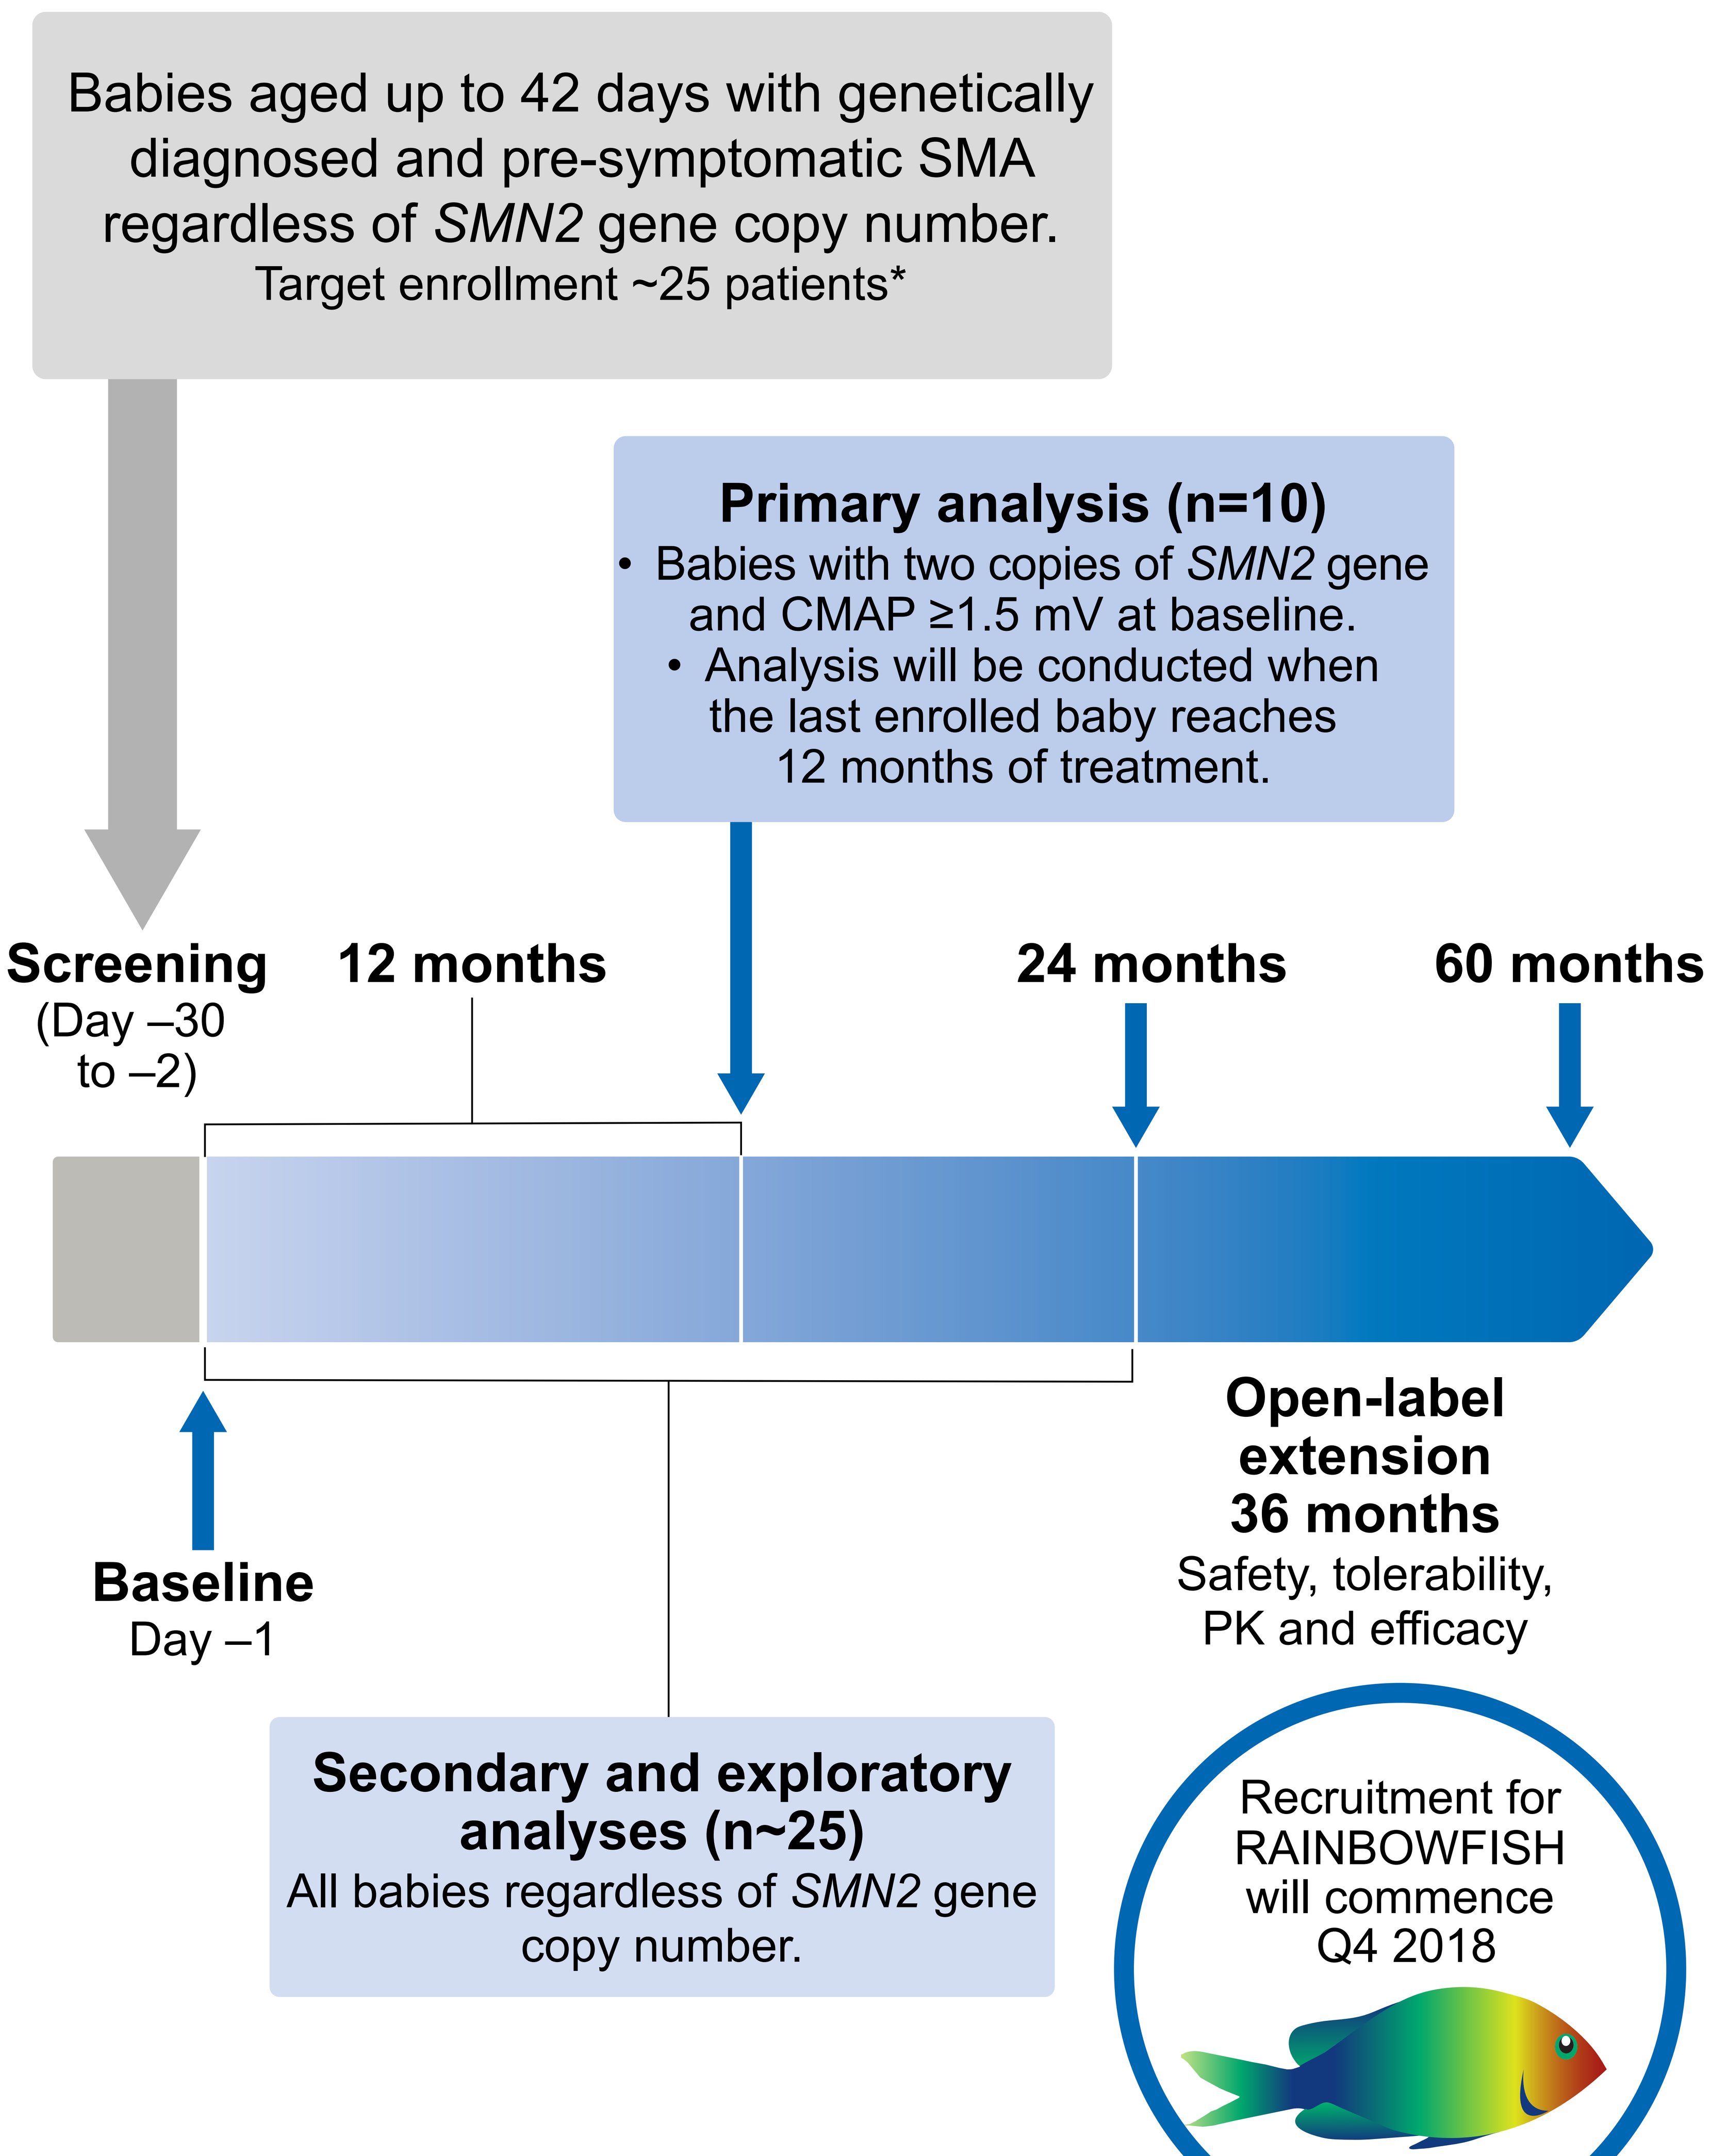
Babies aged up to 42 days with genetically diagnosed and pre-symptomatic SMA regardless of *SMN2* gene copy number.
Target enrollment ~25 patients*
Primary analysis (n=10)
• Babies with two copies of *SMN2* gene and CMAP ≥1.5 mV at baseline.
• Analysis will be conducted when the last enrolled baby reaches 12 months of treatment.
Screening (Day –30 to –2)
12 months
24 months
60 months
Baseline Day –1
Open-label extension 36 months
Safety, tolerability, PK and efficacy
Secondary and exploratory analyses (n~25)
All babies regardless of *SMN2* gene copy number.
Recruitment for RAINBOWFISH will commence Q4 2018

*Recruitment will close when 10 babies with two *SMN2* copies and a baseline of CMAP ≥1.5 mV have been enrolled.

Primary efficacy objective

To evaluate the efficacy of risdiplam in babies with two *SMN2* copies and CMAP ≥1.5 mV at baseline as determined by:

proportion of babies sitting without support for 5 seconds after 12 months on treatment as assessed by Gross Motor Scale of the BSID-III.

Secondary efficacy objectives

To evaluate the efficacy of risdiplam on:

- the development of clinical symptoms of SMA
- survival and permanent ventilation
- the achievement of motor milestones defined in the BSID-III and by the HINE
- CHOP-INTEND motor function scale
- growth measures
- nutritional status (ability to swallow and level of solid food intake)
- degree of innervation by CMAP
- PD effects (SMN mRNA and SMN protein levels in blood)
- respiratory effects by plethysmography

Additional objectives

- Health status and health-related QoL
- Biomarkers
- PK profile of risdiplam
- Safety of risdiplam
- Other developmental milestones (cognition and speech)

Acknowledgments

We would like to thank our collaborators at PTC Therapeutics and the SMA Foundation. The study was funded by F. Hoffmann-La Roche AG, Basel, Switzerland. Writing and editorial assistance was provided by Lindsey Weedon, of MediTech Media, UK, in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

Abbreviations

BSID-III, Bayley Scales of Infant and Toddler Development, Third Edition; CHOP-INTEND, Children's Hospital of Philadelphia Infant Test of Neuromuscular Disorders; CMAP, compound muscle action potential; HINE, Hammersmith Infant Neurological Examination; PD, pharmacodynamics; PK, pharmacokinetics; QoL, quality of life; SMA, spinal muscular atrophy; SMN, survival of motor neuron.

References

1. Mercuri E, et al. *Lancet Neurol.* 2012; 11:443–452;
2. Hamilton G and Gillingwater TH. *Trends Mol Med.* 2013; 19:40–50;
3. Ratni H, et al. *J Med Chem.* 2018; 61:6501–6517;
4. ClinicalTrials.gov.NCT02913482 (accessed Sep 2018)
5. ClinicalTrials.gov.NCT02908685 (accessed Sep 2018)
6. ClinicalTrials.gov.NCT03032172 (accessed Sep 2018)
7. Govoni A, et al. *Mol Neurobiol.* 2018; 55:6307–6318;
8. Hwu W, et al. *Neuromusc Disord.* 2017; 27:S212.

Presented at the 23rd International Annual Congress of the World Muscle Society, Mendoza, Argentina, October 2–6, 2018