PA3028

Disease Characteristics and Outcomes in a Swedish Cohort of Patients With Pulmonary Fibrosis

C. Magnus Sköld,[1,2] Karen Bartley,[3] Aaron Levine,[4] Lisen Arnheim-Dahlstrom,[4] Klaus-Uwe Kirchgaessler,[5] Christer Janson,[6] Giovanni Ferrara[1,2]

[1]Karolinska University Hospital, Stockholm, Sweden; [2]Karolinska Institutet, Stockholm, Sweden; [3]Genentech, Inc., South San Francisco, CA, USA; [4]IQVIA, Solna, Sweden; [5]F. Hoffmann-La Roche, Ltd., Basel, Switzerland; [6]Uppsala University, Uppsala, Sweden

AIMS

- Idiopathic pulmonary fibrosis (IPF) is a debilitating, progressive, fatal, fibrosing lung disease[1]
- IPF prognosis is poor, with survival rates lower than those reported for many common types of cancer[2,3]
 - Risk factors for mortality include male sex, older age ($>$ 70 years), tobacco use, reduced lung function, pronounced fibrosis at time of diagnosis and pulmonary hypertension[1]
 - IPF is furthermore associated with comorbidities, such as gastro-oesophageal reflux disease and vascular diseases[4,5]
- Here, we assessed disease characteristics and outcomes in patients with pulmonary fibrosis in Sweden

METHODS

- This retrospective, longitudinal cohort study utilised linked datasets from Swedish population-based registers from 2001–2015
- **Figure 1** describes the construction of the study cohorts
 - The pulmonary fibrosis cohort was constructed using the following criteria in order to refine the population for patients most likely to have IPF: a registration of International Statistical Classification of Diseases and Related Health Problems, 10th Revision (ICD-10) code J84.1 ('other interstitial pulmonary diseases with fibrosis') (since no specific coding for IPF is available in the ICD-10), $\geqslant$ 40 years of age and without a competing diagnosis after the initial J84.1 code
 - In order to refine further for patients who had a diagnosis of IPF, more detailed clinical data were extracted from electronic medical records (EMRs) obtained from primary care and four respiratory clinics in Stockholm and Uppsala county councils
 - The IPF subcohort comprised of patients from the pulmonary fibrosis cohort with an ICD-10 code of J84.1, registered between November 2004 and March 2017, and a diagnosis of IPF recorded in EMRs
- Data extracted from the Swedish national health registers for patients in this analysis included survival (death from pulmonary fibrosis and death from any cause), rates of lung transplant, oxygen use and hospitalisation, and comorbid diagnoses for both cohorts

RESULTS

Study Population

- Overall, 18 750 patients with an ICD-10 code of J84.1 recorded in the Swedish National Patient Register between 2001 and 2015 were identified, of which 17 247 (92.0%) met the eligibility criteria (pulmonary fibrosis cohort; **Figure 1**)
 - It was not possible to confirm if these patients had a formal diagnosis of IPF; therefore, these patients are described hereafter as patients with pulmonary fibrosis
- A total of 1837 patients from Stockholm and Uppsala within the pulmonary fibrosis cohort with an ICD-10 code of J84.1 recorded in EMRs between November 2004 and March 2017 were identified
 - In this subcohort, 291 patients were identified who had a diagnosis of IPF according to their EMR (IPF subcohort; **Figure 1**)
- In the pulmonary fibrosis cohort and the IPF subcohort, mean (standard deviation) age at diagnosis was 74.6 (10.5) and 70.6 (9.0) years, respectively; 62.5% and 70.1% of patients, respectively, were male

Survival

- For death from any cause and from pulmonary fibrosis, respectively, 5-year survival was 30% and 70% in the pulmonary fibrosis cohort and 60% and 78% in the IPF subcohort
- Death from any cause or from pulmonary fibrosis within 1 year of pulmonary fibrosis diagnosis was associated with male sex and higher age at diagnosis in both cohorts ($P < 0.001$ for the pulmonary fibrosis cohort; $P \leqslant 0.03$ for the IPF subcohort; **Table 1**)

Figure 1. Construction of the Study Cohorts

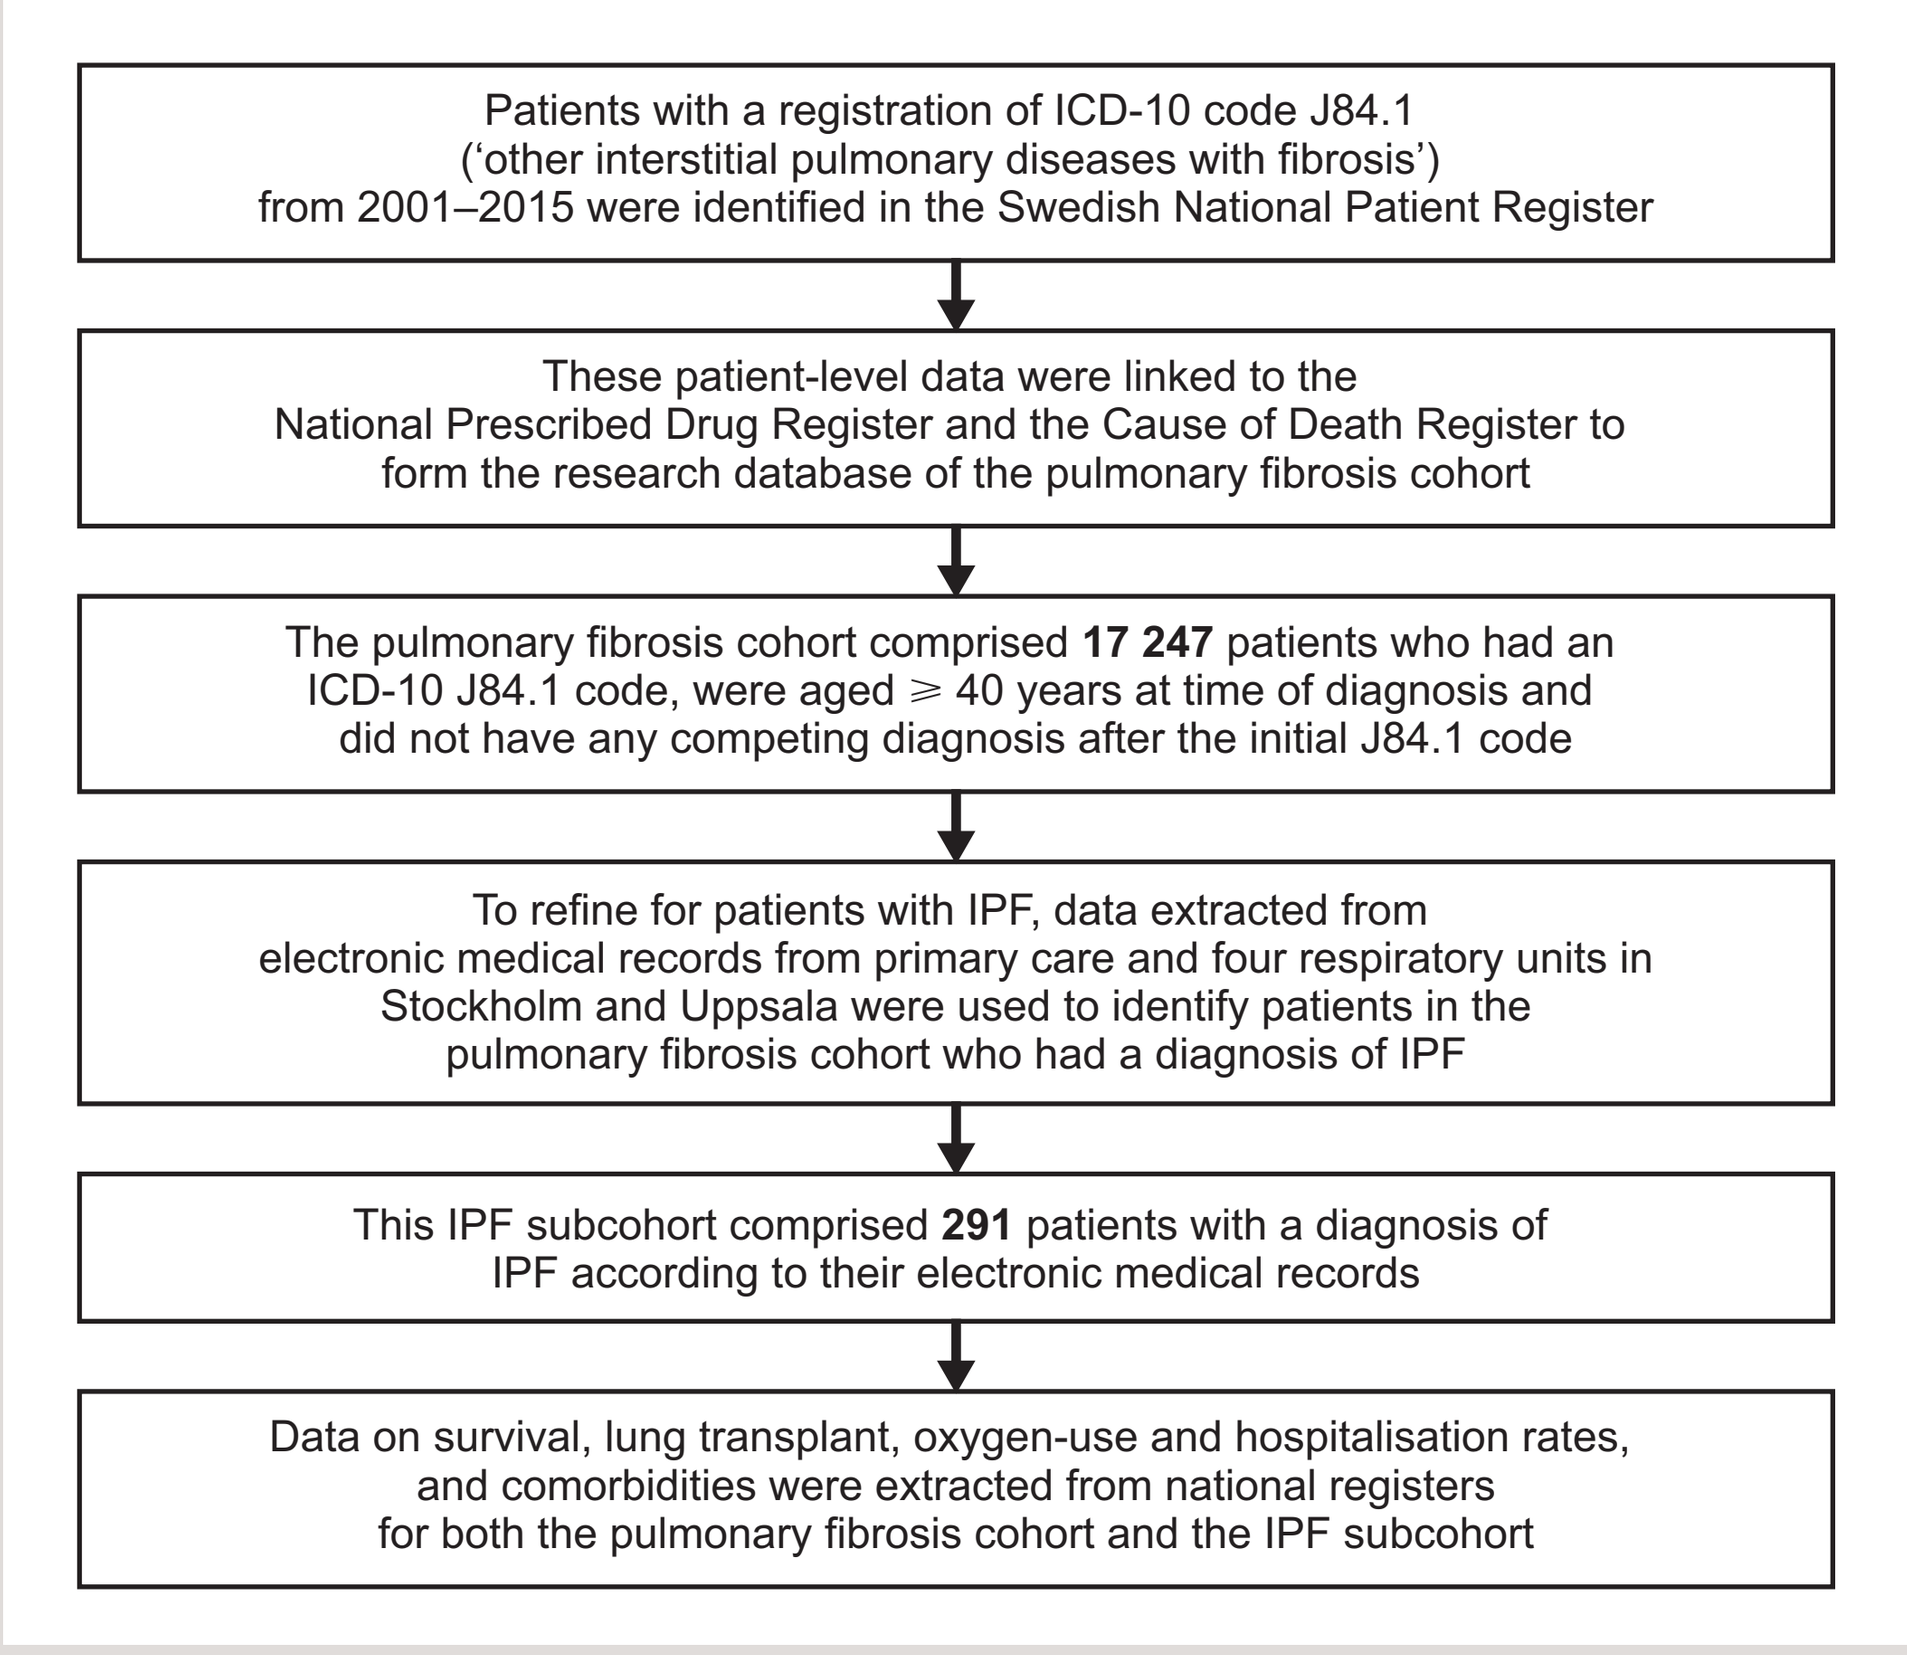
Patients with a registration of ICD-10 code J84.1 ('other interstitial pulmonary diseases with fibrosis') from 2001–2015 were identified in the Swedish National Patient Register
These patient-level data were linked to the National Prescribed Drug Register and the Cause of Death Register to form the research database of the pulmonary fibrosis cohort
The pulmonary fibrosis cohort comprised 17 247 patients who had an ICD-10 J84.1 code, were aged ⩾ 40 years at time of diagnosis and did not have any competing diagnosis after the initial J84.1 code
To refine for patients with IPF, data extracted from electronic medical records from primary care and four respiratory units in Stockholm and Uppsala were used to identify patients in the pulmonary fibrosis cohort who had a diagnosis of IPF
This IPF subcohort comprised 291 patients with a diagnosis of IPF according to their electronic medical records
Data on survival, lung transplant, oxygen-use and hospitalisation rates, and comorbidities were extracted from national registers for both the pulmonary fibrosis cohort and the IPF subcohort

ICD-10, International Statistical Classification of Diseases and Related Health Problems, 10th Revision; IPF, idiopathic pulmonary fibrosis.

Table 1. Associations Between Sex/Age at Diagnosis and Death From Any Cause/From Pulmonary Fibrosis Within 1 Year of Pulmonary Fibrosis Diagnosis in Swedish Patients in 2001–2015*

Variable	Events, n	Median survival, years†	Patients alive at 1 year after diagnosis, % (95% CI)	Unadjusted HR (95% CI)	*P* value
Pulmonary fibrosis cohort (n = 17 247)					
Death from any cause					
Sex					$<$ 0.001
Male (n = 10 771)	7484	2.3	66.6 (65.7, 67.5)	1 (reference)	
Female (n = 6476)	4179	3.1	72.0 (70.9, 73.1)	0.82 (0.79, 0.85)	
Age at diagnosis (n = 17 247)	11 663	2.6	68.6 (67.9, 69.3)	1.06 (1.06, 1.06)	$<$ 0.001
Death from pulmonary fibrosis					
Sex					$<$ 0.001
Male (n = 10 771)	2602	13.9	86.2 (85.5, 86.9)	1 (reference)	
Female (n = 6476)	1379	–	89.1 (88.3, 89.9)	0.79 (0.74, 0.84)	
Age at diagnosis (n = 17 247)	3981	–	87.3 (86.8, 87.9)	1.05 (1.04, 1.05)	$<$ 0.001
IPF cohort (n = 291)					
Death from any cause					
Sex					0.001
Male (n = 204)	91	5.6	95.0 (91.9, 98.0)	1 (reference)	
Female (n = 87)	30	9.2	96.5 (92.7, 100.0)	0.51 (0.34, 0.78)	
Age at diagnosis (n = 291)	121	7.4	95.4 (93.0, 97.8)	1.05 (1.02, 1.07)	$<$ 0.001
Death from pulmonary fibrosis					
Sex					0.03
Male (n = 204)	50	9.6	96.9 (94.5, 99.3)	1 (reference)	
Female (n = 87)	17	14.3	97.7 (94.5, 100.0)	0.54 (0.31, 0.94)	
Age at diagnosis (n = 291)	67	12.7	97.1 (95.2, 99.1)	1.05 (1.02, 1.08)	0.003

CI, confidence interval; HR, hazard ratio; IPF, idiopathic pulmonary fibrosis.
* Associations were assessed using Cox proportionate hazards models.
† Not observable if $<$ 50% of the population at risk by the end of the study period had not experienced the event.

Non-Pharmacological Treatment Patterns

- Lung transplant and oxygen-use rates, respectively, were 0.4% and 11.0% in the pulmonary fibrosis cohort and 0.7% and 61.9% in the IPF subcohort (**Figure 2**)
 - The majority of the patients in each cohort who had lung transplants and/or used oxygen did so $>$ 12 months post-diagnosis

Figure 2. Non-Pharmacological Treatment Patterns Before and After Diagnosis of Pulmonary Fibrosis in Swedish Patients in 2001–2015 in (a) the Pulmonary Fibrosis Cohort and (b) the IPF Cohort

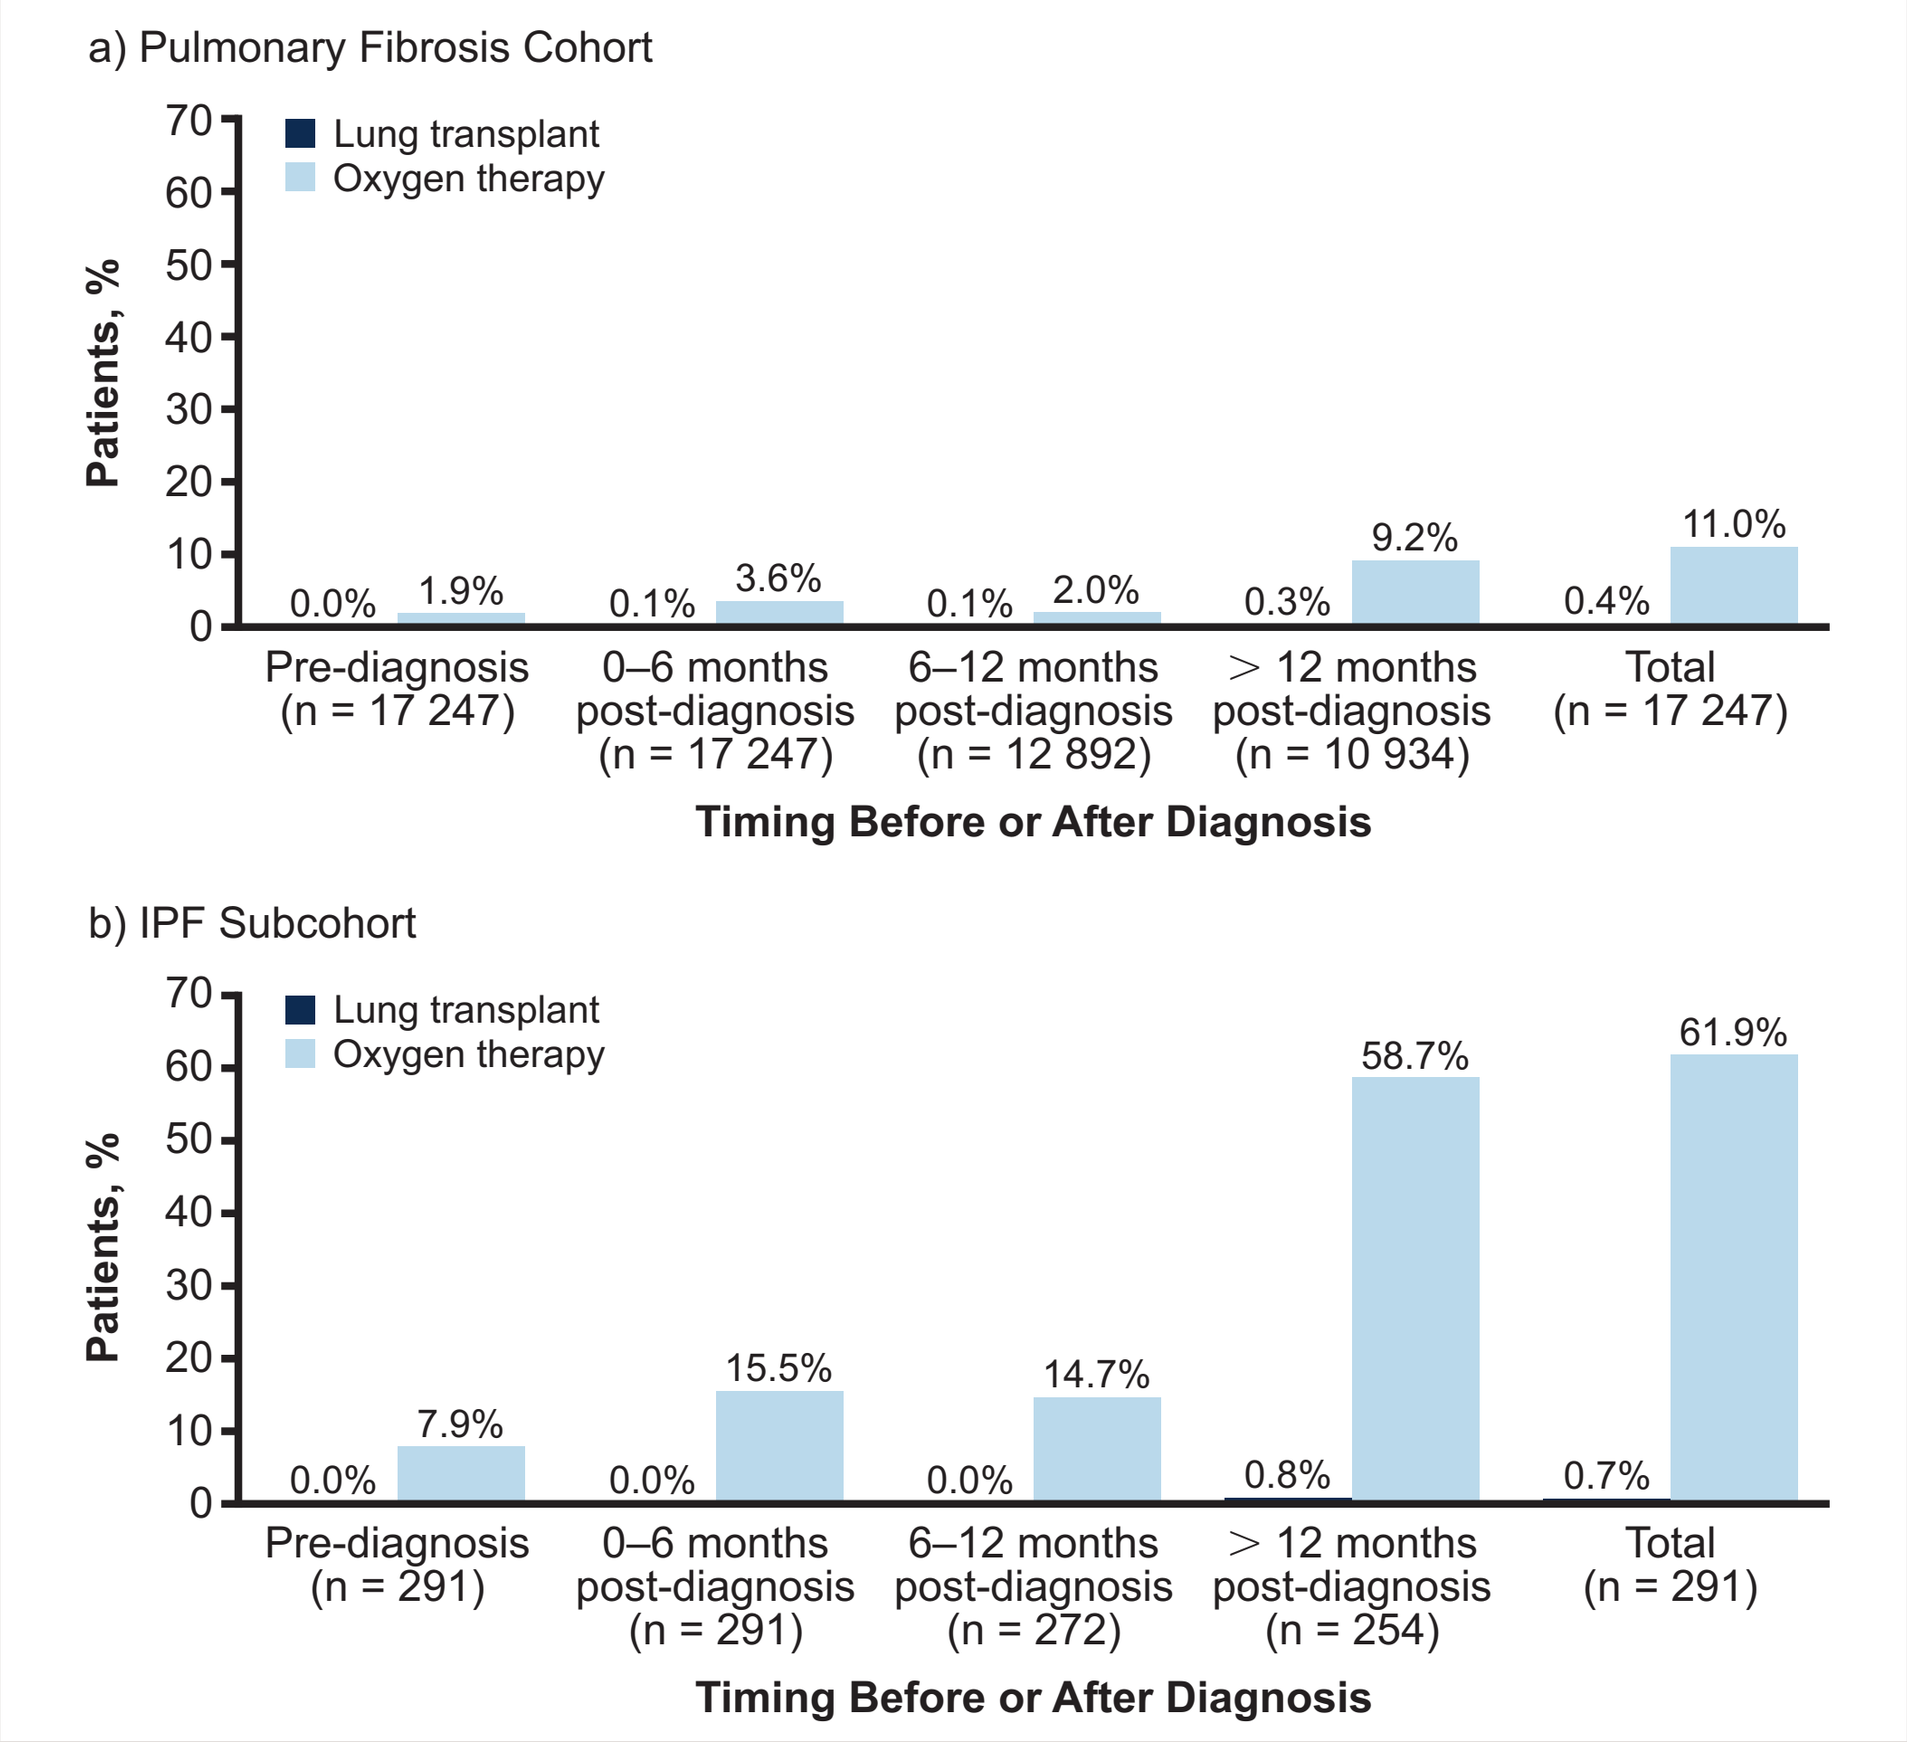
a) Pulmonary Fibrosis Cohort
Lung transplant
Oxygen therapy
Patients, %
0.0% 1.9%
0.1% 3.6%
0.1% 2.0%
0.3% 9.2%
0.4% 11.0%
Pre-diagnosis (n = 17 247)
0–6 months post-diagnosis (n = 17 247)
6–12 months post-diagnosis (n = 12 892)
> 12 months post-diagnosis (n = 10 934)
Total (n = 17 247)
Timing Before or After Diagnosis
b) IPF Subcohort
Lung transplant
Oxygen therapy
Patients, %
0.0% 7.9%
0.0% 15.5%
0.0% 14.7%
0.8% 58.7%
0.7% 61.9%
Pre-diagnosis (n = 291)
0–6 months post-diagnosis (n = 291)
6–12 months post-diagnosis (n = 272)
> 12 months post-diagnosis (n = 254)
Total (n = 291)
Timing Before or After Diagnosis

IPF, idiopathic pulmonary fibrosis.

Hospitalisations

- Total rate of hospitalisation with pulmonary fibrosis as the main diagnosis was 47.3% in the pulmonary fibrosis cohort and 61.9% in the IPF subcohort post-diagnosis (**Figure 3**)
 - For most of these hospitalisations, patients were admitted to non-pulmonary wards (36.7%) rather than pulmonary wards (10.7%) in the pulmonary fibrosis cohort; corresponding rates were more similar in the IPF subcohort (35.7% and 26.1% for non-pulmonary and pulmonary wards, respectively)
 - The majority of these hospitalisations occurred in the first year after diagnosis, with rates of 35.2% (28.2% in pulmonary wards; 7.1% in other wards) in the pulmonary fibrosis cohort and 31.6% (13.1% in pulmonary wards; 18.6% in other wards) in the IPF subcohort

Figure 3. Pulmonary Fibrosis-Related Hospitalisation Rates After Diagnosis of Pulmonary Fibrosis in Swedish Patients in 2001–2015 in (a) the Pulmonary Fibrosis Cohort and (b) the IPF Cohort

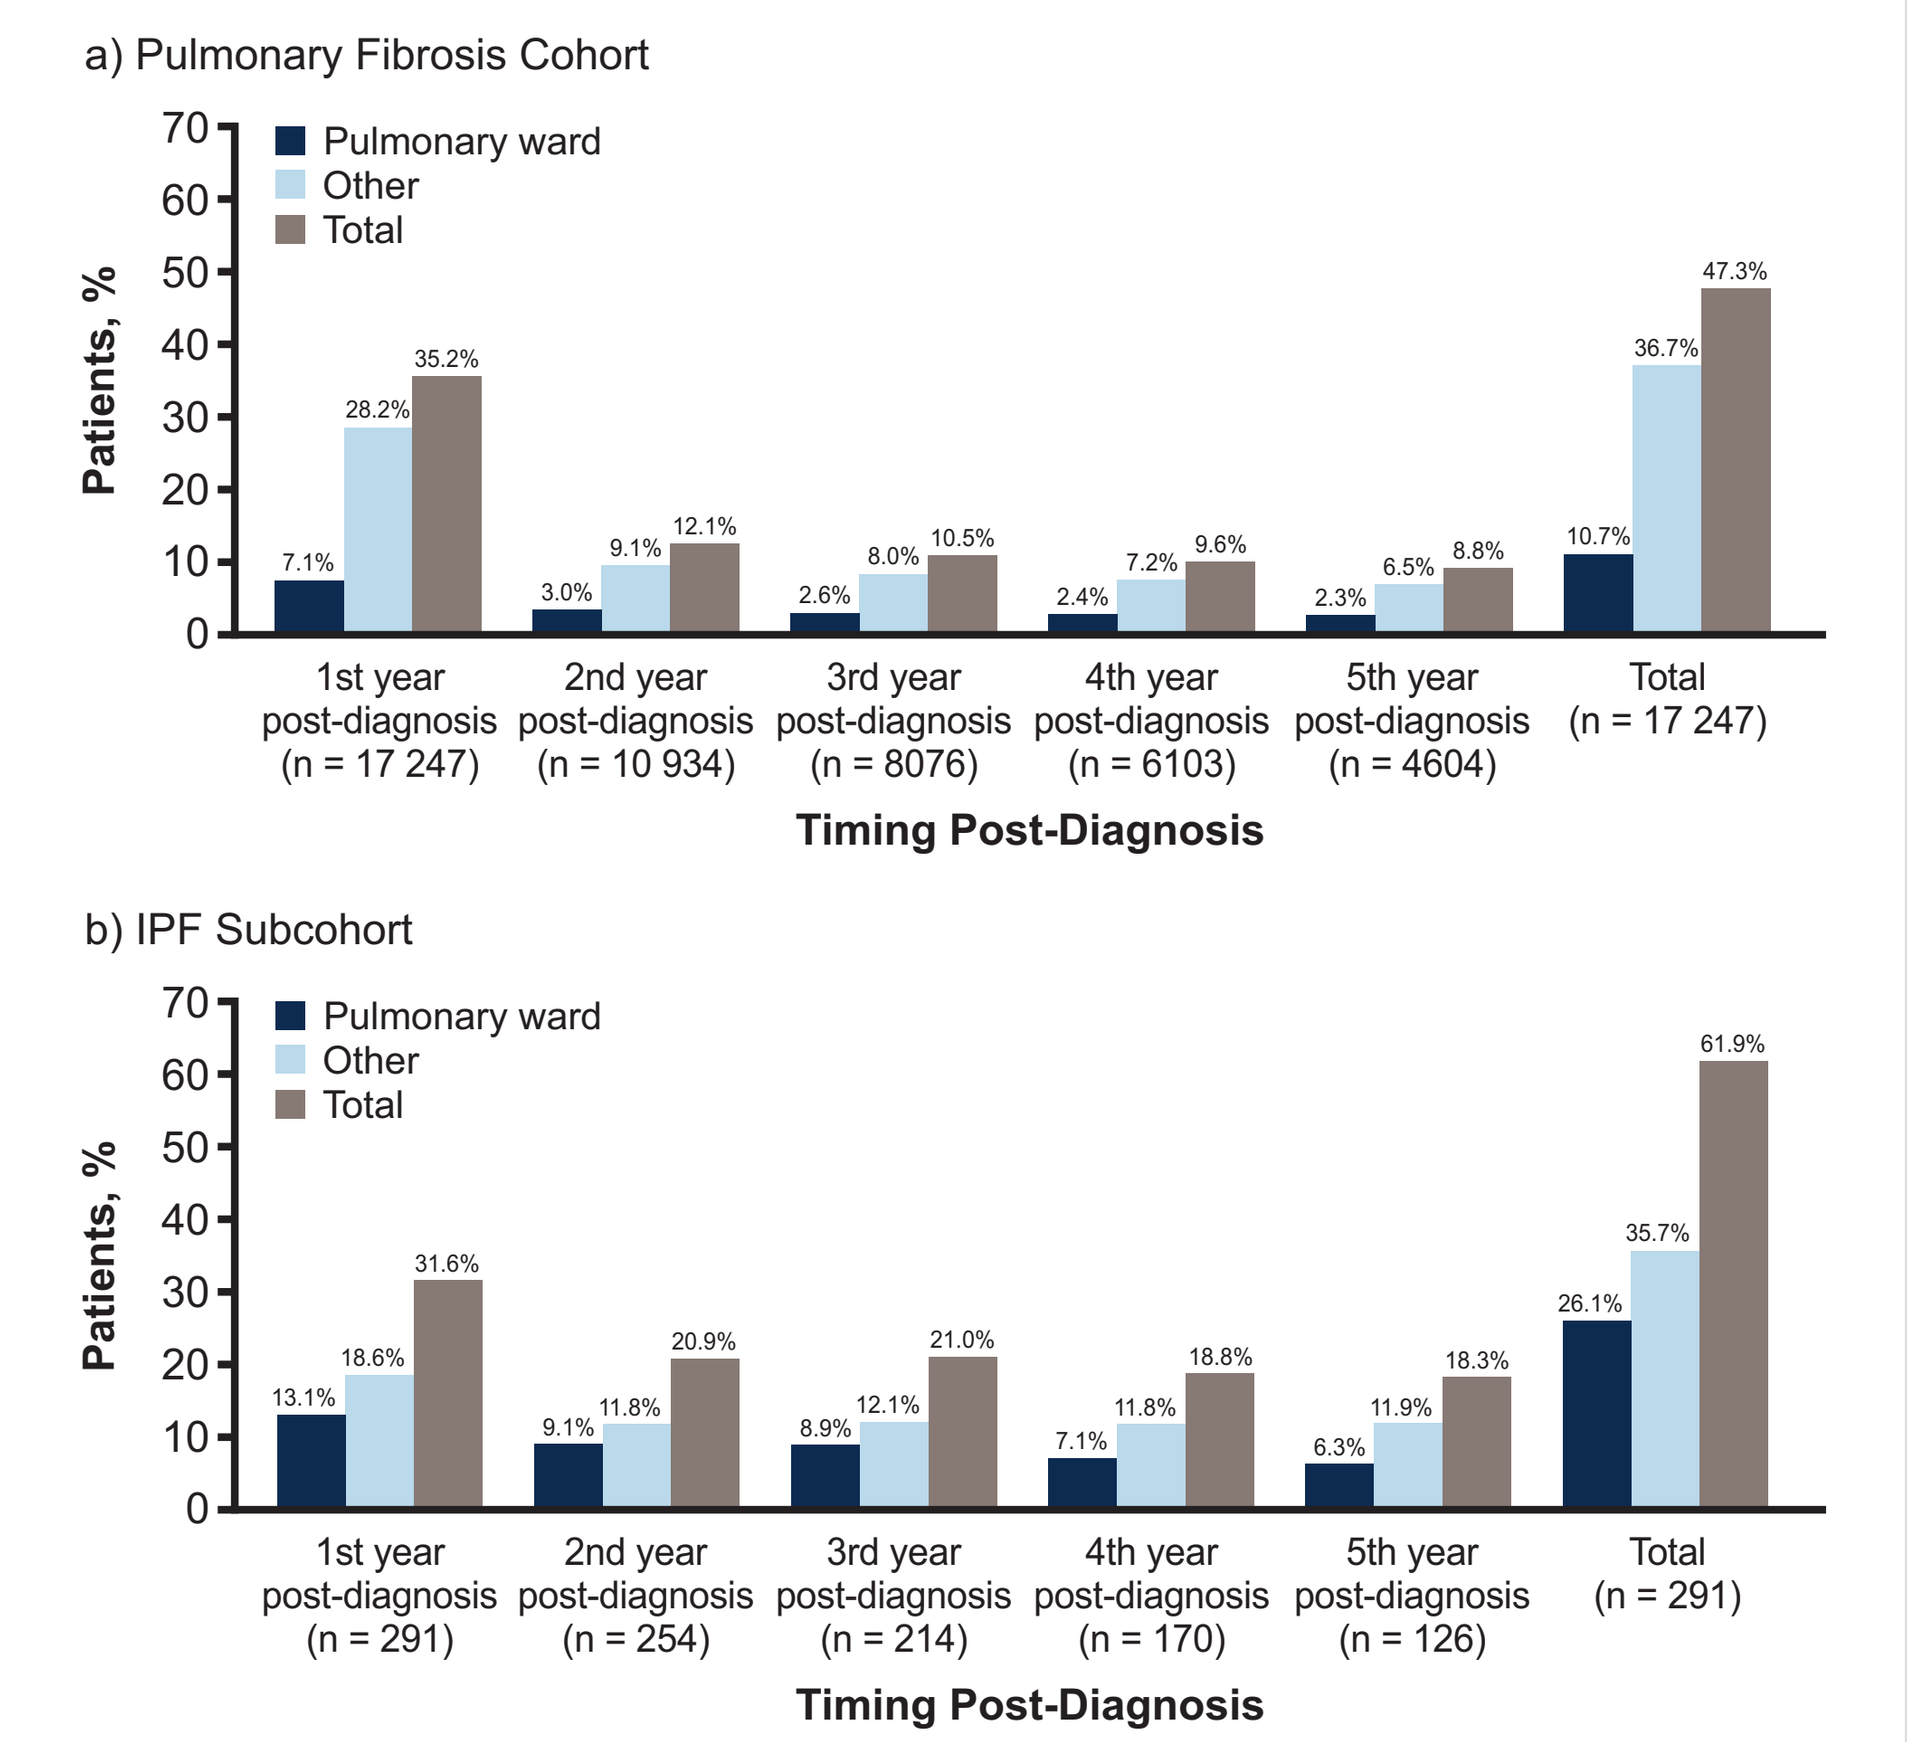
a) Pulmonary Fibrosis Cohort
Pulmonary ward
Other
Total
Patients, %
7.1% 28.2% 35.2%
3.0% 9.1% 12.1%
2.6% 8.0% 10.5%
2.4% 7.2% 9.6%
2.3% 6.5% 8.8%
10.7% 36.7% 47.3%
1st year post-diagnosis (n = 17 247)
2nd year post-diagnosis (n = 10 934)
3rd year post-diagnosis (n = 8076)
4th year post-diagnosis (n = 6103)
5th year post-diagnosis (n = 4604)
Total (n = 17 247)
Timing Post-Diagnosis
b) IPF Subcohort
Pulmonary ward
Other
Total
Patients, %
13.1% 18.6% 31.6%
9.1% 11.8% 20.9%
8.9% 12.1% 21.0%
7.1% 11.8% 18.8%
6.3% 11.9% 18.3%
26.1% 35.7% 61.9%
1st year post-diagnosis (n = 291)
2nd year post-diagnosis (n = 254)
3rd year post-diagnosis (n = 214)
4th year post-diagnosis (n = 170)
5th year post-diagnosis (n = 126)
Total (n = 291)
Timing Post-Diagnosis

IPF, idiopathic pulmonary fibrosis.

- Rate of pulmonary fibrosis-related hospitalisations per year was associated with male sex and higher age at diagnosis in the pulmonary fibrosis cohort ($P < 0.001$) but not the IPF subcohort (not significant; **Table 2**)

Comorbid Diagnoses

- The most common comorbid diagnoses were other infections of airways (besides bacterial pneumonia), ischaemic heart disease and dyspnoea in the pulmonary fibrosis cohort, and other infections of airway (besides bacterial pneumonia), dyspnoea and musculoskeletal pain in the IPF subcohort at any time (**Figure 4**)
 - Within 1 year of pulmonary fibrosis diagnosis, the most common comorbid diagnoses were ischaemic heart disease, congestive heart failure and dyspnoea in the pulmonary fibrosis cohort. The most common comorbid diagnoses in the IPF subcohort were cough, dyspnoea and ischaemic heart disease within 1 year of pulmonary fibrosis diagnosis

Table 2. Associations Between Sex/Age at Diagnosis and Pulmonary Fibrosis-Related Hospitalisations Per Year After Pulmonary Fibrosis Diagnosis in Swedish Patients in 2001–2015*

Parameter	Co-efficient (95% CI)	*P* value
Pulmonary fibrosis cohort (n = 17 247)		
Intercept	–2.26 (–2.69, –1.83)	$<$ 0.001
Age at diagnosis	0.02 (0.01, 0.03)	$<$ 0.001
Sex (female)	–0.29 (–0.41, –0.16)	$<$ 0.001
IPF subcohort (n = 291)		
Intercept	0.12 (–3.27, 3.52)	0.94
Age at diagnosis	–0.01 (–0.06, 0.03)	0.55
Sex (female)	–0.78 (–1.73, 0.18)	0.11

CI, confidence interval; IPF, idiopathic pulmonary fibrosis.
Parameter estimates are given on the log-linear scale.
* Associations were assessed using generalised linear modelling with negative binomial distribution.

Figure 4. Summary of Common Comorbid Diagnoses ($\geqslant$ 10% of Patients at Any Time) in Patients With Pulmonary Fibrosis in Sweden in 2001–2015 in (a) the Pulmonary Fibrosis Cohort and (b) the IPF Cohort

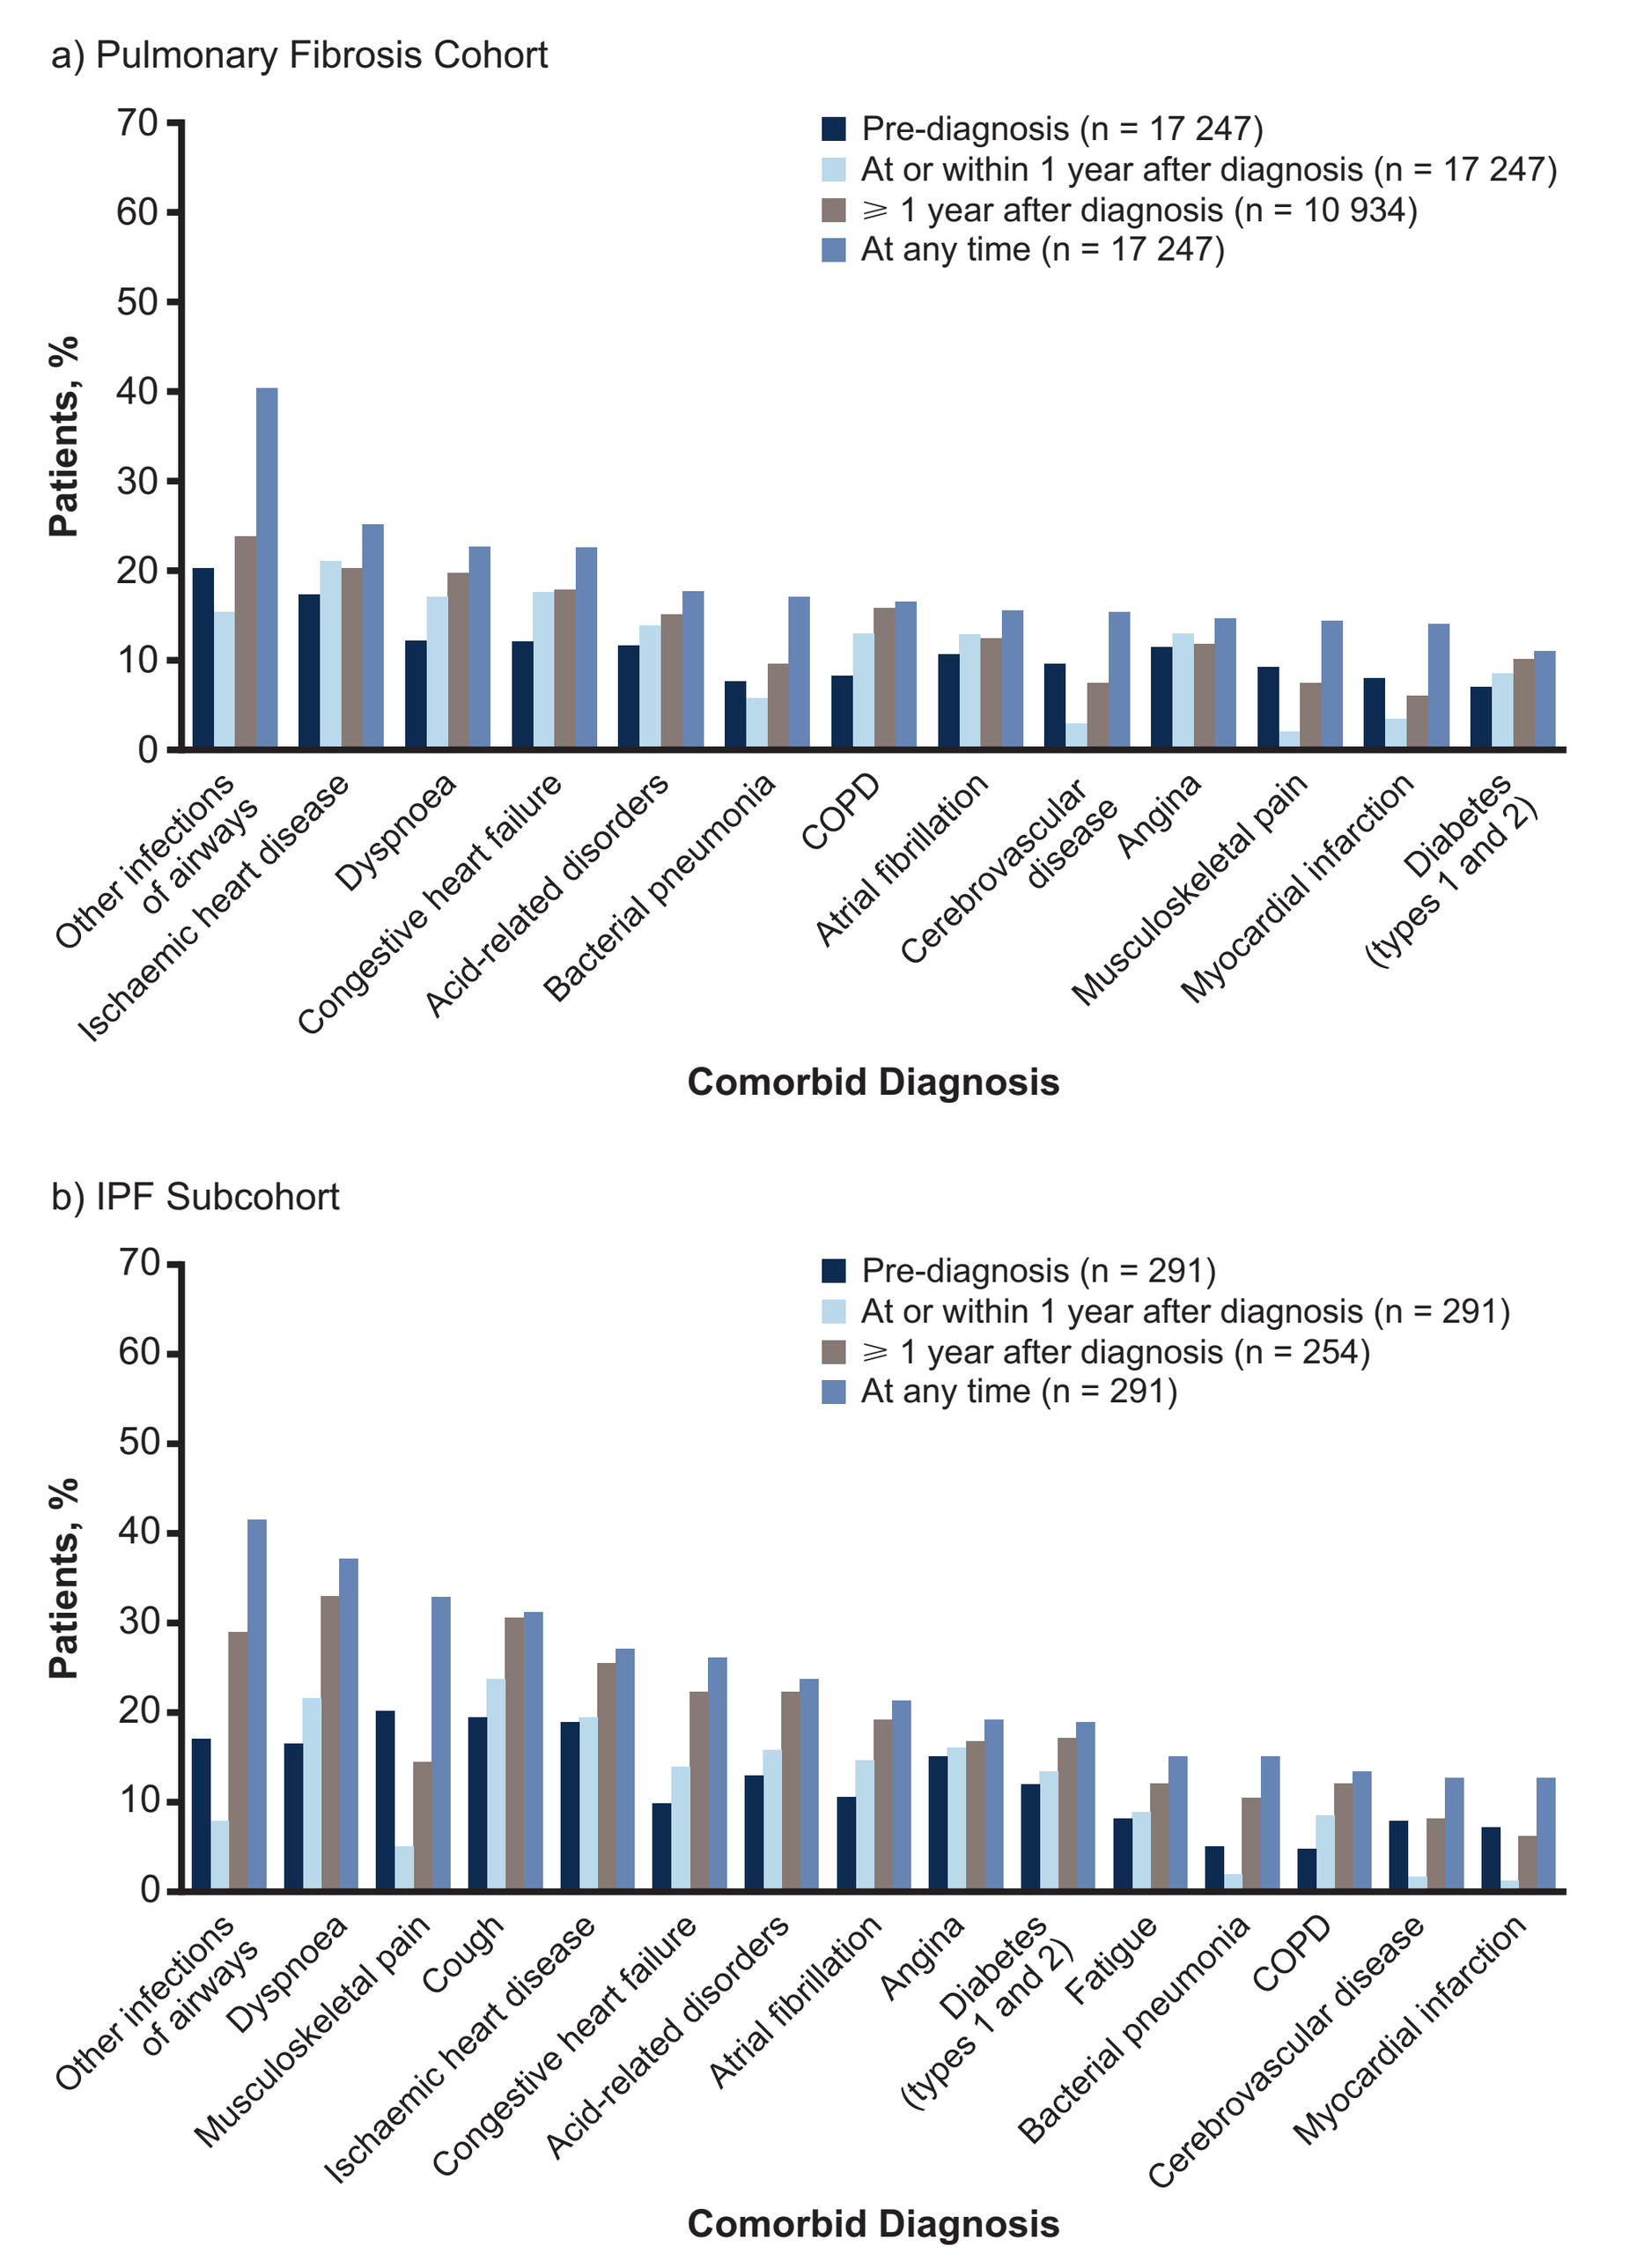
a) Pulmonary Fibrosis Cohort
Pre-diagnosis (n = 17 247)
At or within 1 year after diagnosis (n = 17 247)
⩾ 1 year after diagnosis (n = 10 934)
At any time (n = 17 247)
Patients, %
Other infections of airways
Ischaemic heart disease
Dyspnoea
Congestive heart failure
Acid-related disorders
Bacterial pneumonia
COPD
Atrial fibrillation
Cerebrovascular disease
Angina
Musculoskeletal pain
Myocardial infarction
Diabetes (types 1 and 2)
Comorbid Diagnosis
b) IPF Subcohort
Pre-diagnosis (n = 291)
At or within 1 year after diagnosis (n = 291)
⩾ 1 year after diagnosis (n = 254)
At any time (n = 291)
Patients, %
Other infections of airways
Dyspnoea
Musculoskeletal pain
Cough
Ischaemic heart disease
Congestive heart failure
Acid-related disorders
Atrial fibrillation
Angina
Diabetes (types 1 and 2)
Fatigue
Bacterial pneumonia
COPD
Cerebrovascular disease
Myocardial infarction
Comorbid Diagnosis

COPD, chronic obstructive pulmonary disease; IPF, idiopathic pulmonary fibrosis.

CONCLUSIONS

- Prognosis was poor and comorbidity burden was substantial in this cohort of patients with pulmonary fibrosis and subcohort with IPF
- Comorbid conditions in both cohorts were generally consistent with previously studied populations of patients with IPF[6–8]
- Male sex and higher age at diagnosis were associated with increased death by any cause or due to pulmonary fibrosis in both cohorts, and with increased pulmonary fibrosis-related hospitalisations in the pulmonary fibrosis cohort
 - The higher mortality in older patients highlights the importance of early case detection and diagnosis and improved management of the disease
- Overall, this study underscores the importance of pulmonary fibrosis as a cause of respiratory-related morbidity and mortality in the Swedish population

DISCLOSURES

Data within the poster are directly derived from research sponsored by F. Hoffmann-La Roche, Ltd./Genentech, Inc. The authors thank Dr Ragnar Linder, who was involved in the design of the study and interpretation of study results and contributed to the development of the abstract for this poster

C. Magnus Sköld has received research grants from Boehringer Ingelheim, Roche and Sandoz, and consultancy or speaker fees from AstraZeneca, Boehringer Ingelheim, GlaxoSmithKline, Meda, Novartis, Mundipharma, Sandoz, Chiesi, Almirall, InterMune and Roche

Giovanni Ferrara has received honoraria for lectures from Roche and Boehringer Ingelheim

REFERENCES

1. Ley B, et al. *Am J Respir Crit Care Med.* 2011;183:431–440.
2. Nathan SD, et al. *Chest.* 2011;140:221–229.
3. American Cancer Society. Cancer Facts & Figures 2018. Available at: https://www.cancer.org/content/dam/cancer-org/research/cancer-facts-and-statistics/annual-cancer-facts-and-figures/2018/cancer-facts-and-figures-2018.pdf. Last accessed: 8 August 2018.
4. Hubbard RB, et al. *Am J Respir Crit Care Med.* 2008;178:1257–1261.
5. Collard HR, et al. *J Med Econ.* 2012;15:829–835.
6. Raghu G, et al. *Eur Respir J.* 2015;46:1113–1130.
7. Behr J, et al. *Eur Respir J.* 2015;46:186–196.
8. Jo HE, et al. *Eur Respir J.* 2017;49:1601592.

http://bit.ly/2wu8Gpi

Presented at the European Respiratory Society International Congress; 15–19 September 2018; Paris, France.

This poster was sponsored by F. Hoffmann-La Roche, Ltd.
Third-party writing assistance was furnished by CMC AFFINITY, a division of Complete Medical Communications, Ltd.