Demographic Factors Associated with IPF Diagnostic Confidence

Mary Beth Scholand, MD[1]; Jeffrey Moore[2], MS; John L. Stauffer[3], MD; Karina Raimundo[3], MS; Wayne Pan[3], MD, PhD, MBA; Susan Limb[3], MD.

[1]University of Utah School of Medicine, Department of Internal Medicine, Pulmonary Division

[2] University of Michigan, Department of Biostatistics, Ann Arbor, MI.

[3]Genentech Inc., South San Francisco, CA.

Background

Per current guidelines, a multidisciplinary team (MDT) approach at an experienced specialist center is recommended in making the diagnosis of any interstitial lung disease (ILD).

The Pulmonary Fibrosis Foundation Patient Registry (PFF-PR) is a network of 42 PFF Care Centers in 27 US states (Fig.1) gathering data from well-characterized pulmonary fibrosis patients.

We assessed the association of demographic features with the confidence level of the PFF Care Center investigator in diagnosing idiopathic pulmonary fibrosis (IPF).

Methods

- We evaluated the association of 60 patient characteristics at enrollment and the confidence level of IPF diagnosis for 731 patients enrolled in the PFF-PR as of September 1, 2017. Not every patient was evaluated for each of the characteristics.
- Confidence in the IPF diagnosis was rated by the clinical site to be “low,” “medium,” or “high.”
- A X^2 test was used to examine associations between confidence level and categorical parameters, and a *t*-test for associations with continuous parameters.
- Location paramters are defined as distance from the patient’s home to the Care Center site in km, or categorized as *long* (>100 km;), *medium* (20-100 km) or *short distance* (<20 km).

Table 1. Summary of statistical analyses conducted on PFF-PR data.

Variable	Confidence Level for IPF Diagnosis						Total (N=771)		p-value
	High (N=589)		Medium (N=129)		Low (N=13)				
	N	Mean or Percent	N	Mean or Percent	N	Mean or Percent	N	Mean or Percent	
Age at Diagnosis									**0.0066**
< 50 years	10	1.70%	2	1.60%	2	15.40%	14	1.90%	
50-60 years	39	6.60%	13	10.10%	2	15.40%	54	7.40%	
60-70 years	192	32.60%	53	41.10%	4	30.80%	249	34.10%	
70-80 years	287	48.70%	49	38%	4	30.80%	340	46.50%	
> 80 years	61	10.40%	12	9.30%	1	7.70%	74	10.10%	
Region									
Midwest	**140**	**23.69%**	**19**	**14.73%**	**3**	**23.08%**	**162**	**22.10%**	**0.0342**
Northeast	145	24.53%	33	25.58%	4	30.77%	182	24.83%	0.6348
South	236	39.93%	59	45.74%	4	30.77%	299	40.79%	0.3455
West (REF)	70	11.84%	18	13.95%	2	15.38%	90	12.28%	
Insurance									
Medi-Medi	1	0.17%	2	1.55%	0	0%	3	0.41%	0.1608
Medicaid	4	0.68%	3	2.33%	2	15.38%	9	1.23%	0.3762
Medicare	**227**	**38.41%**	**40**	**31.01%**	**1**	**7.69%**	**268**	**36.56%**	**0.001**
Military/VA	3	0.51%	2	1.55%	0	0%	5	0.68%	0.695
None/Other/ Unknown	37	6.26%	10	7.75%	1	7.69%	48	6.55%	0.1688
Private (REF)	319	53.98%	72	55.81%	9	69.23%	400	54.57%	
Biopsy?									
Yes	**219**	**37.44%**	**26**	**20.31%**	**4**	**30.77%**	**249**	**34.30%**	**0.0003**
No	366	62.56%	102	79.69%	9	69.23%	477	65.70%	
HRCT?									
Yes	**486**	**86.79%**	**87**	**70.16%**	**10**	**83.33%**	**583**	**83.76%**	**<0.0001**
No	74	13.21%	37	29.84%	2	16.67%	113	16.24%	
Weight (lbs)	556	188.59 (38.08)	122	195.46 (43.56)	11	196.32 (43.59)	689	189.93 (39.22)	0.0678
SF-6D Total Score	**568**	**33.2 (4.17)**	**126**	**32.46 (4.14)**	**13**	**30.85 (4.49)**	**707**	**33.02 (4.18)**	**0.0328**

Results

- **Confidence level of IPF diagnosis was significantly associated with the region of the country of the clinical study site** (Midwest differing from the South, Northeast, and West), **insurance type** (Medicare vs. Private), **high resolution CT scan** performed, and **patient quality of life** as per the SF-6D Total Score.
- **Additional location parameters** urban/rural classification of the patient’s home, distance from the patient’s home to the Care Center site **did not show an association** with confidence level of IPF diagnosis.
- **Patient age was a significant predictor of IPF** when categorized as <50 years of age, 50-60, 60-70, 70-80 and 80+, with the “high” confidence group consisting of mostly older patients. The Midwest region had younger patients than the other 3 regions. The Medicare group consisted of slightly older patients than the private insurance group.

Discussion and Conclusion

Older age is associated with increased confidence of an IPF diagnosis. While US regional differences may be a factor, the distance from the PFF Care Center and urban/rural designation are not associated with the diagnostic certainty in patients evaluated at a PFF Care Center. **These data suggest a consistent diagnostic process at PFF Care Centers.**

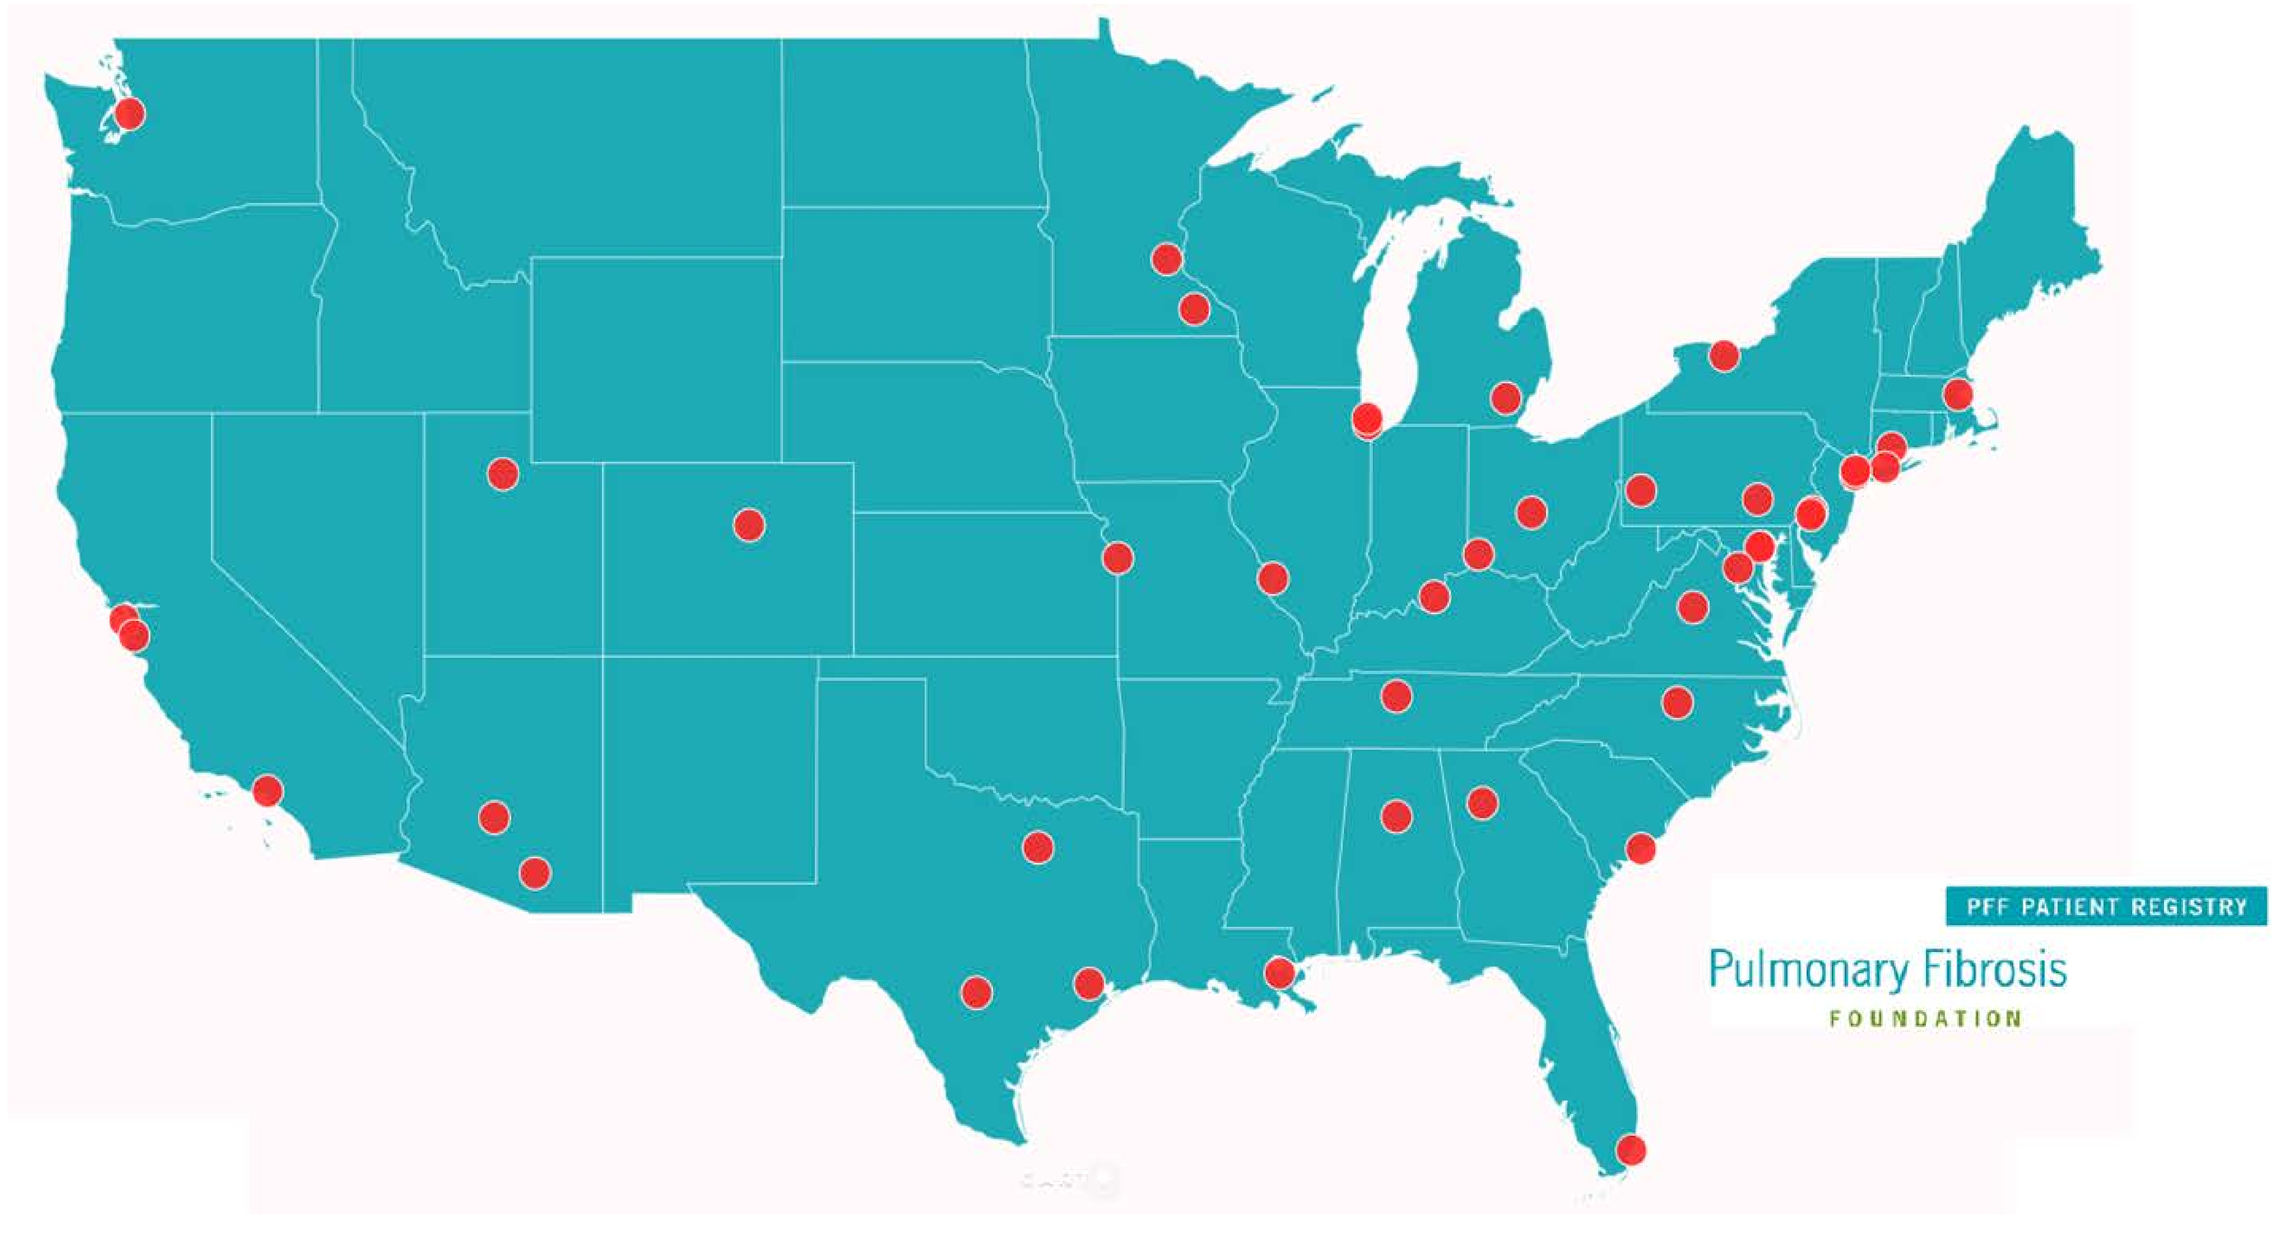

PFF PATIENT REGISTRY
Pulmonary Fibrosis
FOUNDATION

Figure 1: Map of the Pulmonary Fibrosis Foundation Registry sites.