Autoimmune Testing Patterns Associated with IPF Diagnostic Confidence

Mary Beth Scholand[1], MD; Jeffrey Moore[2], MS; John L. Stauffer[3], MD; Wayne Pan[3], MD, PhD, MBA; Susan Limb[3], MD.
[1]University of Utah School of Medicine, Department of Internal Medicine, Pulmonary Division, Salt Lake City, UT
[2] University of Michigan, Department of Biostatistics, Ann Arbor, MI
[3] Genentech Inc., South San Francisco, CA

Background

Per current guidelines, an autoimmune evaluation should be done on every interstitial lung disease (ILD) patient prior to making a diagnosis of idiopathic pulmonary fibrosis (IPF).[1] The recommended minimal serologies include rheumatoid factor (**RF**), anti-cyclic citrullinated peptide (**anti-CCP**), and anti-nuclear antibody (**ANA**) titer and pattern.[2]

The Pulmonary Fibrosis Foundation Patient Registry (PFF-PR) is a network of 42 PFF Care Centers in 27 US states (Fig. 1) gathering data from pulmonary fibrosis patients.

Using data from the PFF-PR, we assessed the association of various autoimmune measures with the confidence level of the PFF Care Center investigator in diagnosing IPF.

Materials and Methods

- We evaluated the association of autoantibodies at enrollment and the confidence level of IPF diagnosis for 731 patients enrolled in the PFF-PR as of September 1, 2017. Not every patient was evaluated for each of the autoantibodies.
- In each case, autoantibodies are assessed to be either “normal” or “abnormal”.
- Confidence in the IPF diagnosis was rated by the clinical site to be “low,” “medium,” or “high.”
- Fisher’s Exact and X^2 Tests were used to examine associations between autoantibodies and confidence level of diagnosis.

Table 1. Association of autoimmune measures with confidence in PF diagnosis

	Confidence Level for IPF Diagnosis						Total (N=771)		*p*-value
	High (N=589)		Medium (N=129)		Low (N=13)				
Parameters	N	Percent	N	Percent	N	Percent	N	Percent	
ANA	344		91			7	442		**0.0228**
Abnormal	76	22.09%	30	32.97%	3	42.86%	109	24.66%	
Normal	268	77.91%	61	67.03%	4	57.14%	333	75.34%	
RF	336		91		10		437		**0.0283**
Abnormal	28	8.09%	14	15.38%	2	20%	44	9.84%	
Normal	318	91.91%	77	84.62%	8	80%	403	90.16%	
Anti-CCP	240		74		9		366		
Abnormal	14	5.83%	5	6.80%	0	0%	19	5.88%	0.8799
Normal	226	94.17%	68	93.15%	10	100%	304	94.12%	
Skin Cancer									**0.0255**
Yes	29	4.91%	13	10.08%	1	7.69%	43	5.87%	
No	562	95.09%	116	89.92%	12	92.31%	690	94.13%	
Diabetes									0.0961
Yes	129	21.83%	20	15.50%	2	15.38%	151	20.60%	
No	462	78.17%	109	84.50%	11	84.62%	582	79.40%	
Raynaud’s Disease									**0.0009**
Yes	13	2.20%	11	8.53%	0	0%	24	3.27%	
No	578	97.80%	118	91.47%	13	100%	709	96.73%	

Results

- As shown in Table 1, confidence level of **IPF diagnosis was significantly associated with ANA and RF**. The anti-CCP did not impact diagnostic confidence.
- Excluding the relatively small number of patient for which diagnosis confidence was “low”, **the pattern of significance remains for the association of “Medium” or “High”** using a Chi-Square test for the first two, and a Fisher’s Exact Test for third.
- Additionally, **clinical comorbidities that were also associated with confidence level include** having a history of **skin cancer** or **Raynaud’s disease.**

Summary and Future Directions

- **Positive ANA and RF titers are associated with lower confidence that the patient has IPF.**
- The low predictive value of these particular serologies for autoimmune disease may contribute to decreased diagnostic confidence for IPF.
- Further study is necessary to understand the significance of these positive serologies in the identification of interstitial pneumonias with autoimmune features.

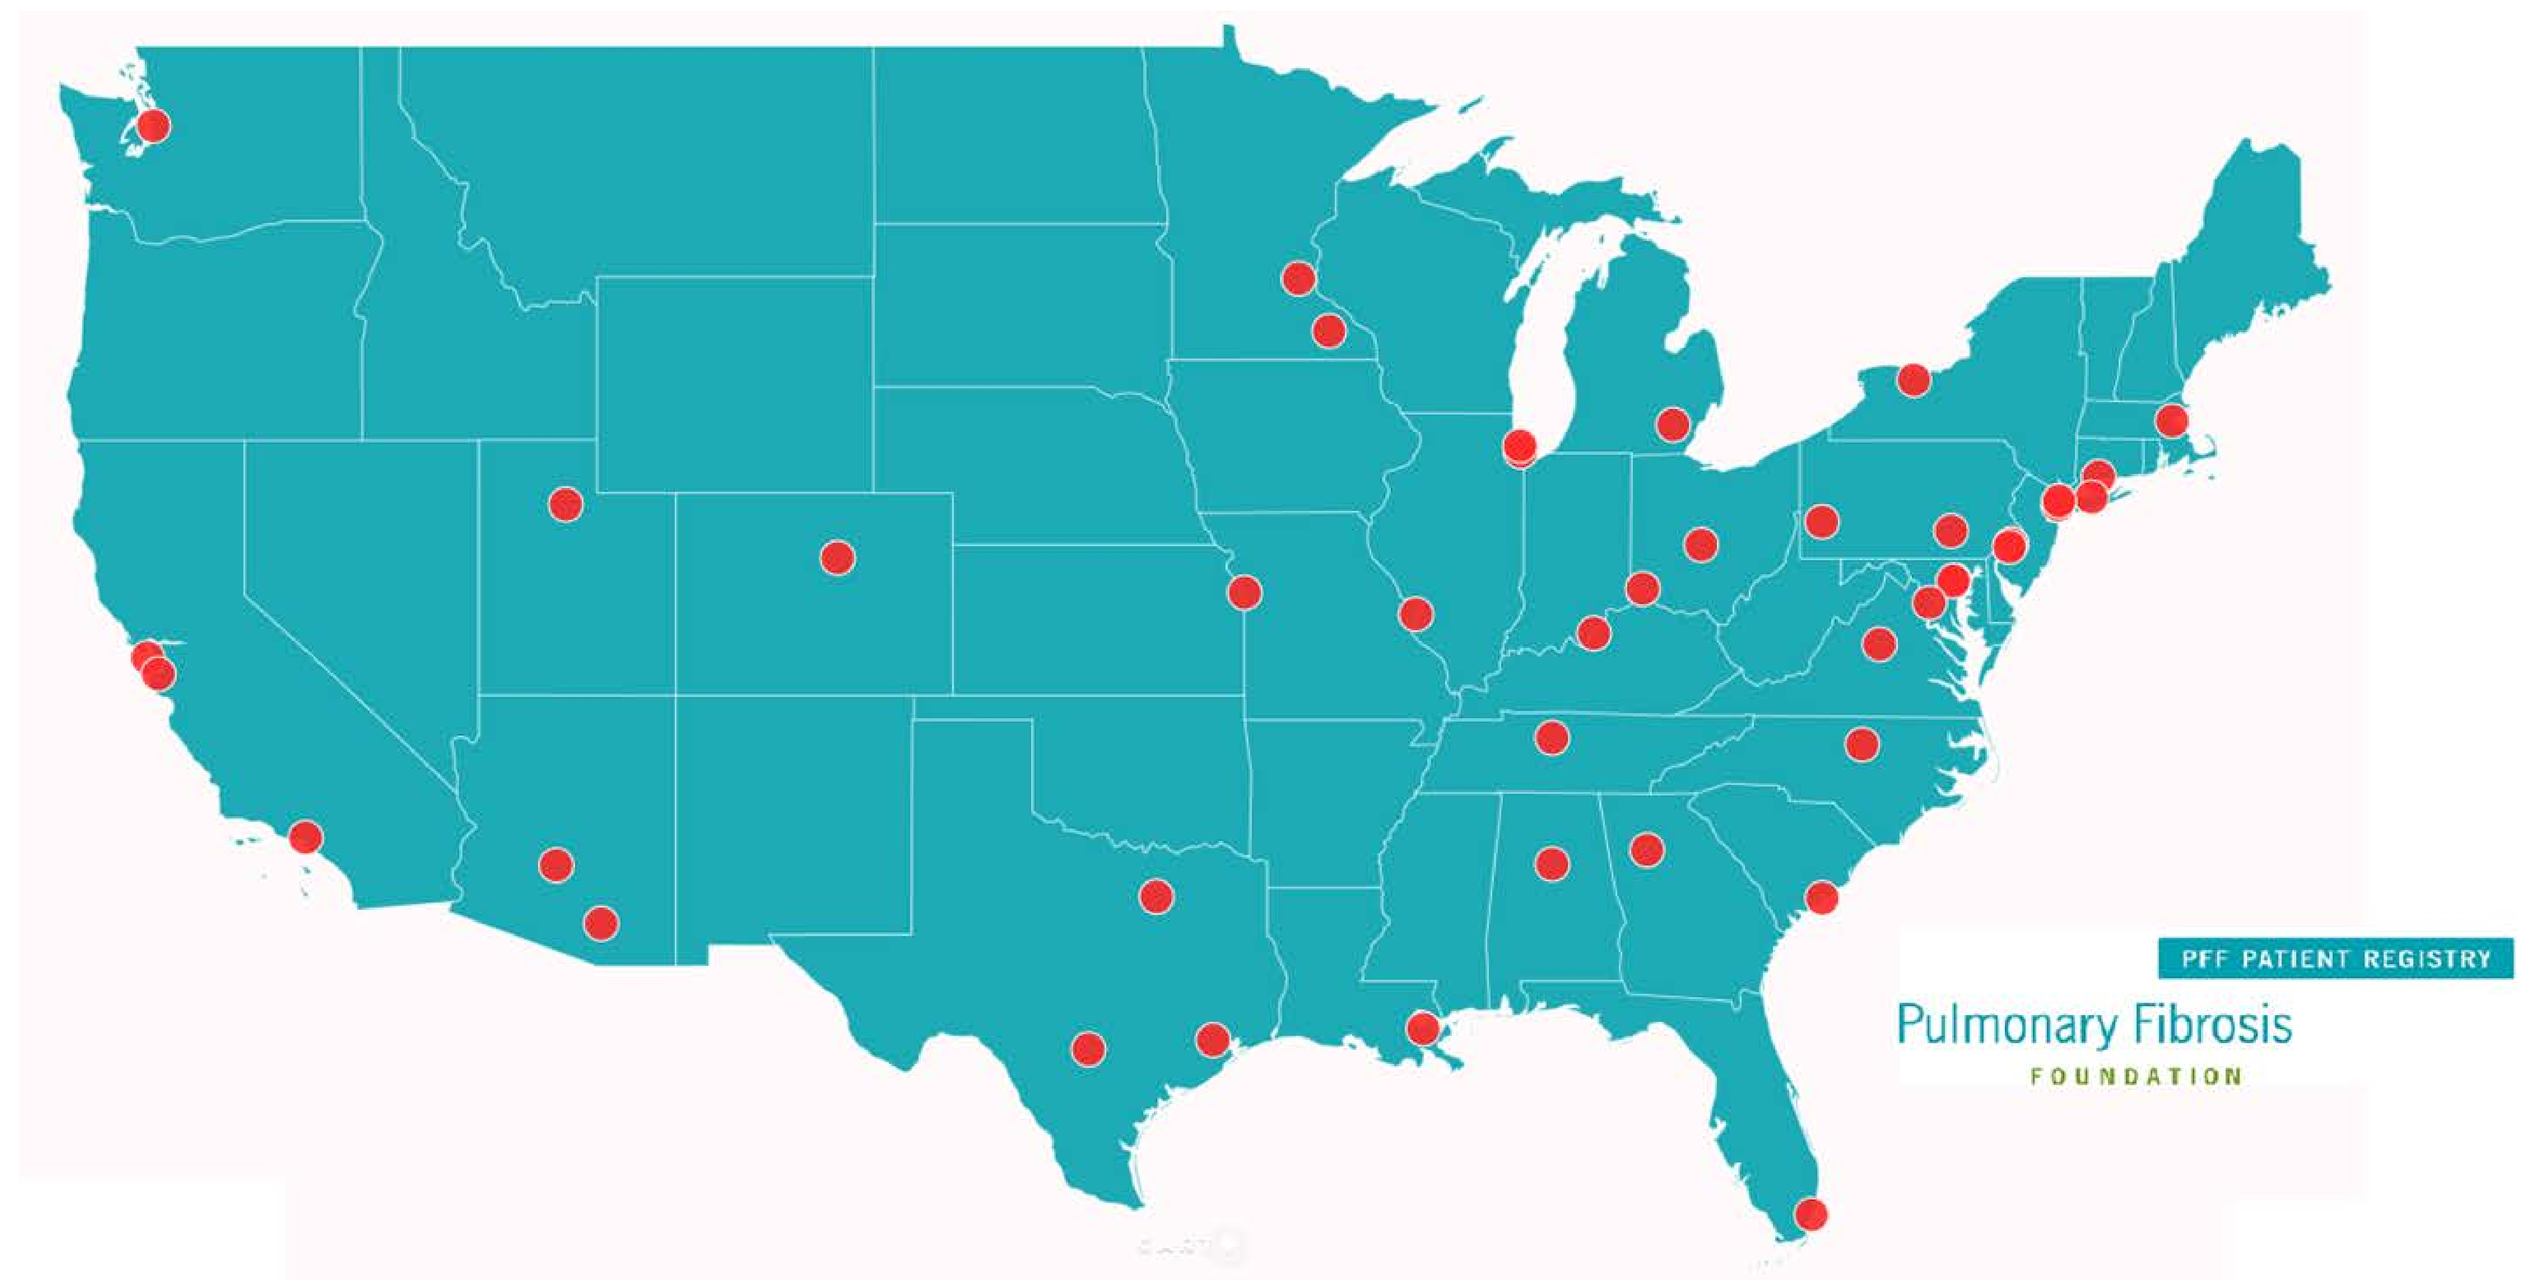
PFF PATIENT REGISTRY
Pulmonary Fibrosis
FOUNDATION

Figure 1. Map of PFF-PR sites.

References

1. Raghu *et al.* Am J Respir Crit Care Med. 2011. Mar 15; 183(6):788-824.
2. Fischer *et al*. Eur Respir J 2015; Oct; 46(4):976-87.