Herpes Zoster and Disseminated Zoster in Systemic Lupus Erythematosus and Lupus Nephritis: Incidence Rates in Real-World Claims Data

Katherine A. Belendiuk[1], Devika Chawla[1], Yingjie Ding[2], Matthew Cascino[1]

[1]Genentech Inc., South San Francisco, CA [2]Genesis Research, Hoboken, NJ

INTRODUCTION AND OBJECTIVES

- Systemic lupus erythematosus (SLE) is associated with an increased risk of varicella zoster reactivation (herpes zoster) with an estimated incidence rate of 1.2 cases per 100 patient years [1].
- Among patients with SLE, lupus nephritis (LN) has been identified as a risk factor for incident herpes zoster [2].
- Disseminated zoster is a highly morbid complication of herpes zoster that is typically associated with immunosuppression or immunodeficiency.
- Robust estimates of herpes zoster incidence rates in the SLE and LN populations have not previously been compared nor have rates of disseminated zoster in these populations been described.
- **The objective of this study was to estimate and compare incidence rates of herpes zoster and disseminated zoster in patients with SLE and LN in the United States using population-based claims data.**

METHODS

Study Dataset

- Truven Healthcare MarketScan® Commercial Claims and Encounters (Commercial) and Medicare Supplemental and Coordination of Benefits (COB) (Medicare) databases [~176 million patients]
- Patients receiving care between January 2000 and December 2014

SLE Inclusion Criteria (n = 76,354)

Validated algorithm [3]

- Inclusion criteria were based on a study using Swedish registry data to identify SLE

Algorithm modifications for the current project:

- Patients will have *at least two* medical claims with SLE (separated by at least 7 days) as one of the listed diagnoses (*International Classification of Diseases, Ninth Revision, Clinical Modification* [ICD-9-CM] code 710.0)
- At least 6 months continuous medical and drug coverage before and after index date
- Note that the SLE cohort *does not exclude* LN patients

LN Inclusion Criteria (n = 11,068)

Validated algorithm [4]

- Patients will have *more than two* medical claims with SLE as one of the listed diagnoses (ICD-9 code 710.0)
- Patients with greater than two claims for any combination of acute or chronic glomerulonephritis (including lupus glomerulonephritis), acute or chronic renal failure, nephritis or nephrotic syndrome (including due to lupus), renal failure or proteinuria (ICD-9 codes 580.00-586.00 and 791.0)

Algorithm modifications for the current project:

- Claims must be separated by at least 7 days
- At least one claim for SLE (validated algorithm bullet 1) and one claim for renal involvement (validated algorithm bullet 2) must occur within 365 days.
- At least 6 months continuous medical and drug coverage before and after index date

Outcome

- Incident herpes zoster cases were identified using ICD-9 codes 053.xxx
- Disseminated zoster cases were identified using ICD-9 codes 053.0, 053.10, 053.14, 053.2, 053.20, 053.21, 053.22, 053.29, 053.79, and 053.8

RESULTS

Table 1. Demographic characteristics of SLE and LN patients in Truven Marketscan from 2000-2014

Characteristic	SLE		LN	
	No. of patients	Percent	No. of patients	Percent
Total cohort	76,354	100.0%	11,068	100.0%
Gender				
Female	67,532	88.4%	9,323	84.2%
Male	8,822	11.6%	1,745	15.8%
Age at Index				
0-17	1,806	2.4%	435	3.9%
18-34	11,953	15.7%	1,905	17.2%
35-44	15,735	20.6%	1,933	17.5%
45-54	20,976	27.5%	2,488	22.5%
55-64	17,357	22.7%	2,532	22.9%
65+	8,527	11.2%	1,775	16.0%
Insurance Plan				
Comprehensive	7,879	10.3%	1,374	12.4%
EPO	857	1.1%	99	0.9%
HMO	12,684	16.6%	1,944	17.6%
POS	5,957	7.8%	898	8.1%
PPO	41,495	54.3%	5,789	52.3%
POS with capitation	1,214	1.6%	145	1.3%
CDHP	2,250	2.9%	298	2.7%
HDHP	1,096	1.4%	124	1.1%
Unknown	2,922	3.8%	397	3.6%
Region				
Northeast	12,097	15.8%	1,399	12.6%
North Central	16,990	22.3%	2,705	24.4%
South	30,915	40.5%	4,569	41.3%
West	15,348	20.1%	2,269	20.5%
Unknown	1,004	1.3%	126	1.1%
	Mean	**Median (IQR)**	**Mean**	**Median (IQR)**
Age at Index	48	49 (38, 58)	49	50 (37, 60)
Enrollment (years)[1]	6.3	5.2 (3.0, 8.7)	6.8	6.0 (3.5, 9.6)

[1] Enrollment in Marketscan database

Demographics

- SLE and LN cohorts included 76,354 and 11,068 patients, respectively.
- The majority of patients were female, privately insured, and between 35-64 years old.
- Mean enrollment duration was 6.3 (SLE) and 6.8 years (LN) (**Table 1**).

Herpes Zoster and Disseminated Zoster

- For patients with SLE, there were 4,284 incident cases of herpes zoster, of which 582 (14%) were considered disseminated.
- For patients with LN, there were 955 incident cases of herpes zoster, of which 157 (16%) were considered disseminated.
- Patients with LN had 1.6 (95% CI: 1.5-1.7) times the rate of herpes zoster as compared to patients with SLE.
- Patients with LN had 1.9 (1.6-2.3) times the rate of disseminated zoster as compared to patients with SLE.

Table 2. Incidence rates of herpes zoster and disseminated herpes zoster for SLE and LN patients

Diagnosis	Event	Person-years	Incidence[1]	Lower 95% CI	Higher 95% CI
SLE					
Herpes zoster	4,284	265,208	1.6	1.6	1.7
Disseminated herpes zoster	582	275,592	0.2	0.2	0.2
LN					
Herpes zoster	955	37,012	2.6	2.4	2.7
Disseminated herpes zoster	157	39,162	0.4	0.3	0.5
Incidence Rate Ratio: LN vs. SLE (unadjusted)					
			Incidence rate ratio	Lower 95% CI	Higher 95% CI
Herpes zoster			**1.6**	**1.5**	**1.7**
Disseminated herpes zoster			**1.9**	**1.6**	**2.3**

[1] Per 100 person-years

SUMMARY AND DISCUSSION

- **In this population-based analysis of claims data from the United States, the incidence rate of herpes zoster in SLE was consistent with a previous estimate [1], and the presence of LN was associated with a significant increase in the rate of herpes zoster.**
- A substantial minority (14-16%) of herpes zoster cases in this cohort were associated with dissemination.
- Limitations of this analysis include a lack of disease activity and treatment information and the use of administrative claims codes to identify SLE/LN and herpes zoster cases.
- Further characterization of the relative contributions of age, immunosuppressive therapies, and disease-related factors is warranted.

REFERENCES

1. Chakravarty EF et al. *Increased incidence of herpes zoster among patients with systemic lupus erythematosus*. Lupus, 2013. Mar;22(3):238-44.
2. Chen D et al. *Herpes zoster in patients with systemic lupus erythematosus: Clinical features, complications and risk factors.* Exp Ther Med, 2017. Dec;14(6):6222-6228.
3. Arkema EV et al. Case definitions in Swedish register data to identify systemic lupus erythematsus. BMJ Open, 2016. Jan 4;6(1):e007769.
4. Chibnik LB et al. *Identification and validation of lupus nephritis cases using administrative claims data.* Lupus, 2010. May;19(6):741-3.

Presented at the American College of Rheumatology Annual Meeting; October 19-24, 2018; Chicago, IL

This study was funded by F. Hoffmann- La Roche Ltd..