269

Adverse Event Reporting Rates and Placebo/Standard-of-Care-Arm ACR Responses Vary by Region in Rheumatoid Arthritis Trials

K. Tuckwell, E. Teng, J. Chia, D. Lee, J. Galanter, J. Peake, D. Keebler

Genentech, South San Francisco, CA, USA

INTRODUCTION

- Clinical trials are becoming increasingly globalized; more are being conducted in Asia, Latin America (LatAm), and the Russian Federation and Eastern Europe (RFEE)[1]
- Less stringent regulatory barriers to study start-up in RFEE mean that early enrollment in clinical trials, and therefore interim analyses, might be skewed toward RFEE
- Adverse event (AE) reporting rates and placebo (or standard-of-care arm) responses can differ by geographic region and level of economic development[2-4]
- Efficacy and safety might not be generalizable across regions. This potential heterogeneity and its implications for rheumatoid arthritis (RA) trials have not been studied in detail
- Confidence that interim analysis populations are representative of final analysis populations is necessary. Therefore, we examined existing trial data from the TransCelerate initiative[5] to better understand geographic differences and potential implications
 - TransCelerate is a placebo/standard-of-care data-sharing initiative among 19 biopharmaceutical companies (AbbVie, Allergan, Amgen, Astellas, AstraZeneca, Boehringer Ingelheim, Bristol-Myers Squibb, EMD Serono/Merck, GlaxoSmithKline, Johnson & Johnson, Eli Lilly, Merck, Novartis Pharmaceuticals, Novo Nordisk, Pfizer, Roche, Sanofi, Shionogi, and UCB) that includes nontreatment arm data for multiple non-oncology indications/diseases

OBJECTIVE

- To assess potential regional differences in rates of American College of Rheumatology (ACR) response and AE reporting using patient-level data from RA trials in the TransCelerate initiative

METHODS

- Patient-level data were obtained for 17 RA trials available through TransCelerate as of August 2017, and data were analyzed from 7 trials for which country-specific information was available (ClinicalTrial.gov IDs: NCT01198002, NCT01202760, NCT01202773, NCT01404585, NCT00605735, NCT00048581, NCT00647270)
 - Included were joint counts, other domains needed for ACR scoring, demographics, and AEs
- Patients were grouped by region (Asia, LatAm, RFEE, USA), and differences in demographics, AE reporting rates, and ACR responses were evaluated
- After a significant omnibus chi-squared test result, pairwise comparisons were made between regions using the Fisher exact test, with false discovery rate (FDR) correction for multiple comparisons
- All patients were included in analyses of AE reporting. Only patients with data sufficient to calculate ACR scores were included in ACR response analyses

Patients

- Data were available for 913 patients from the placebo/standard-of-care arms of the 7 trials analyzed
- Data for ACR scoring were available for 667 patients
- Demographics for patients by region are shown in **Table 1**

Table 1. Patient Demographics by Region

Region	AE Analysis, n	ACR Response Analysis, n	Female, %	Median Age, y (SD; range)
USA	369	268	81	55 (11; 21-78)
RFEE	213	160	81	51 (12; 25-79)
LatAm	167	132	89	51 (12; 22-76)
Asia	164	107	88	52 (12; 18-83)

ACR, American College of Rheumatology; AE, adverse event; LatAm, Latin America; RFEE, Russian Federation and Eastern Europe; SD, standard deviation.

RESULTS

ACR20/50 Response

- The highest rate of ACR20 response was seen in Asia, whereas the lowest rate of ACR50 response was seen in RFEE (**Figure 1**)
- After FDR correction, ACR20 rates in Asia remained significantly higher than in RFEE and the USA
- For ACR50, only the response difference between RFEE and LatAm persisted after FDR correction

Figure 1. Percentages of patients who attained ACR20/50 response 12 weeks after baseline by region.

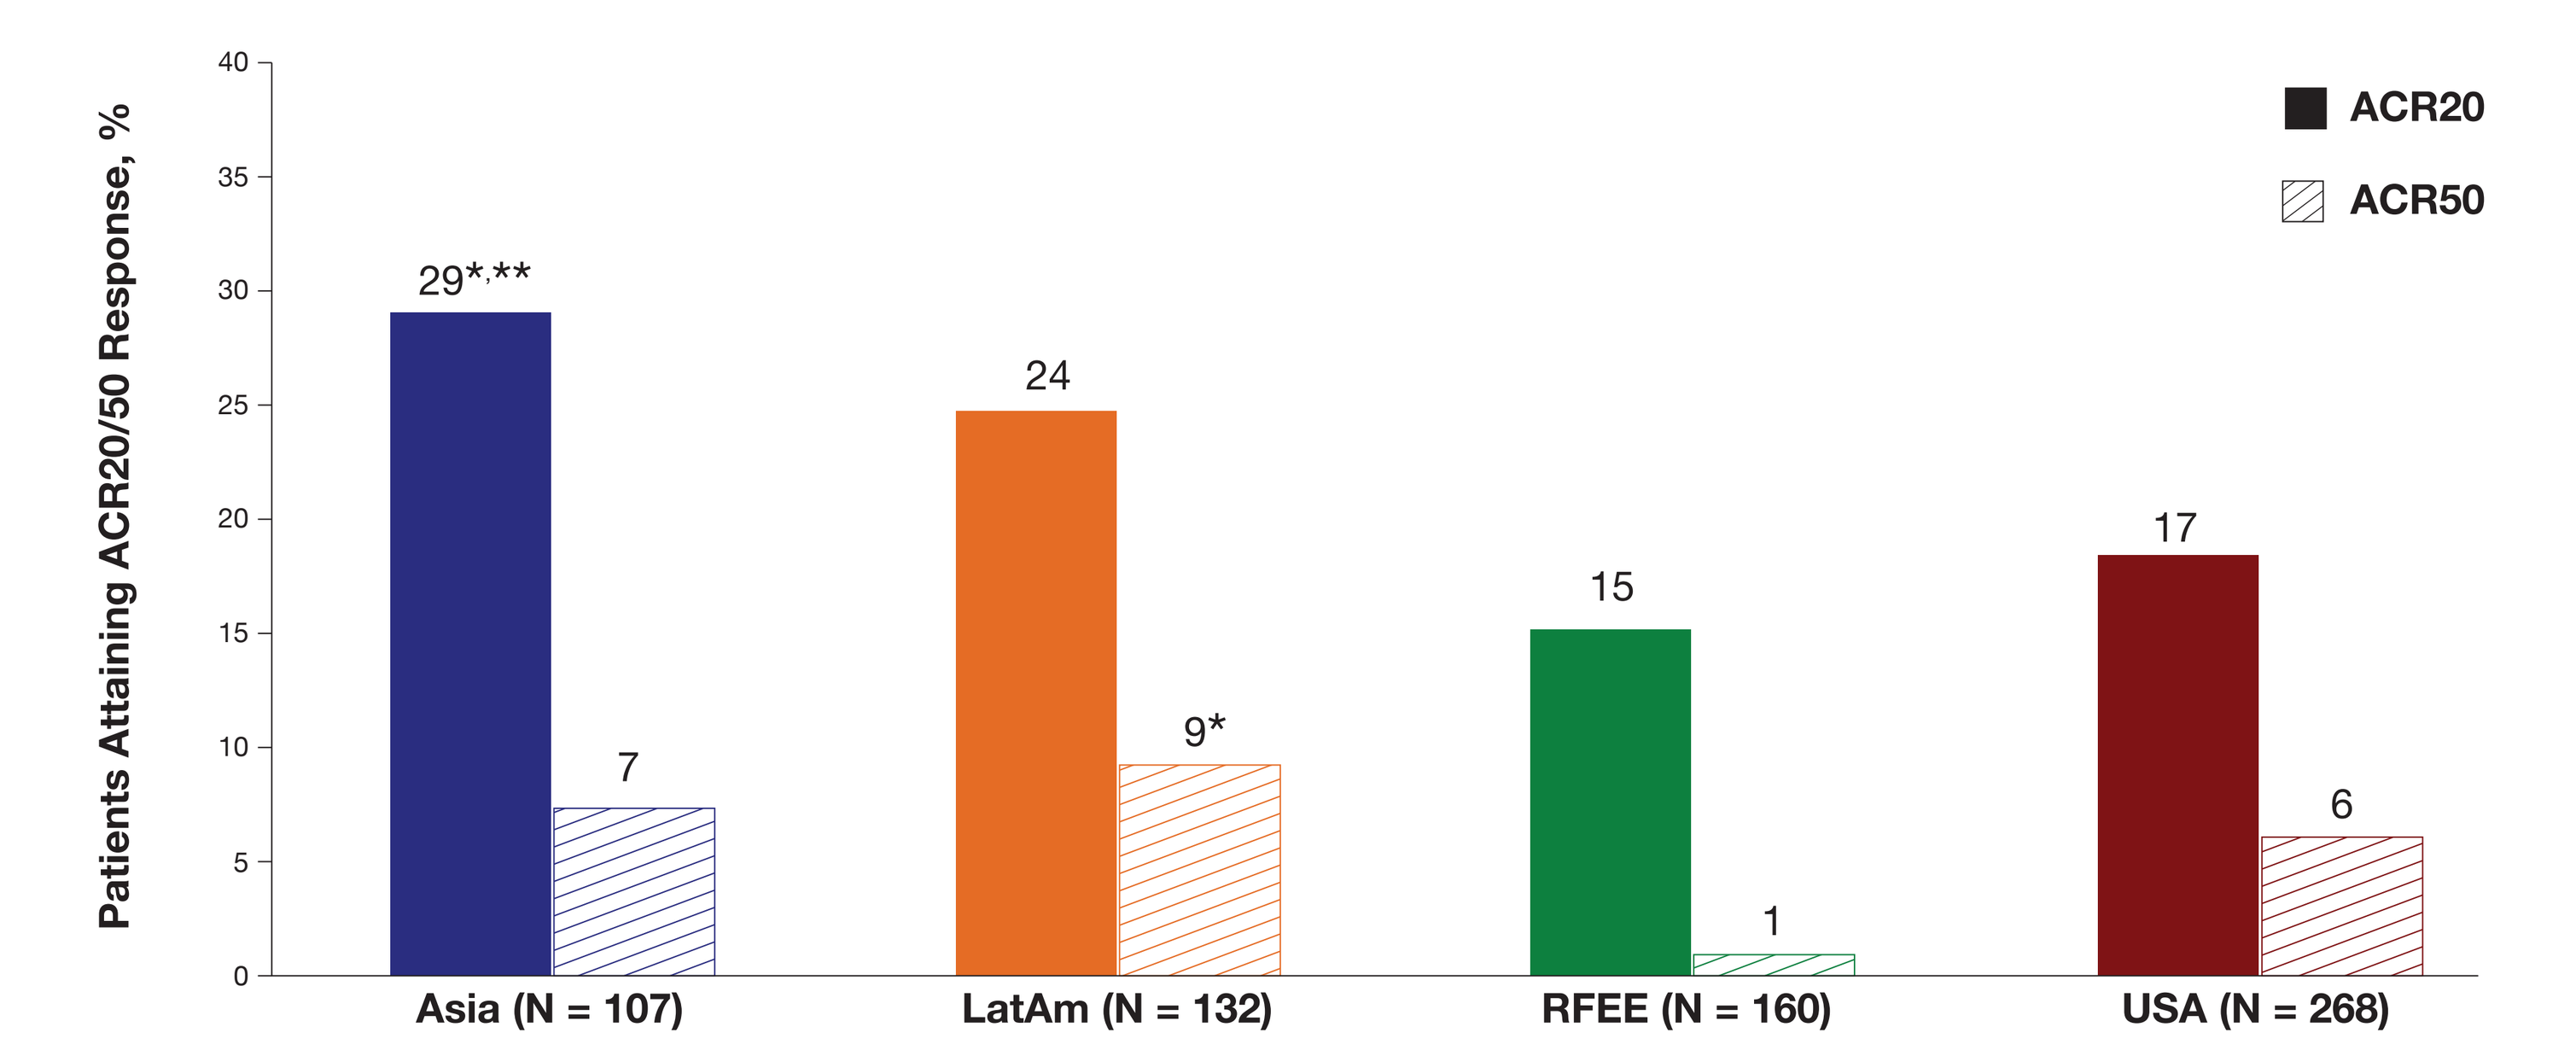
ACR20
ACR50
Patients Attaining ACR20/50 Response, %
29*,**
7
24
9*
15
1
17
6
Asia (N = 107)
LatAm (N = 132)
RFEE (N = 160)
USA (N = 268)

ACR, American College of Rheumatology; LatAm, Latin America; RFEE, Russian Federation and Eastern Europe.
*$p < 0.05$ versus RFEE.
**$p < 0.05$ versus USA.
All p values corrected for multiple comparisons.

Adverse Event Reporting Rates

- The lowest AE reporting rates were seen in RFEE (**Figure 2**)
- After FDR correction, significantly lower 12-week and 52-week AE reporting rates were seen in RFEE than in Asia, LatAm, and the USA

Figure 2. Percentages of patients by region who reported ≥1 AE 12 and 52 weeks after baseline.

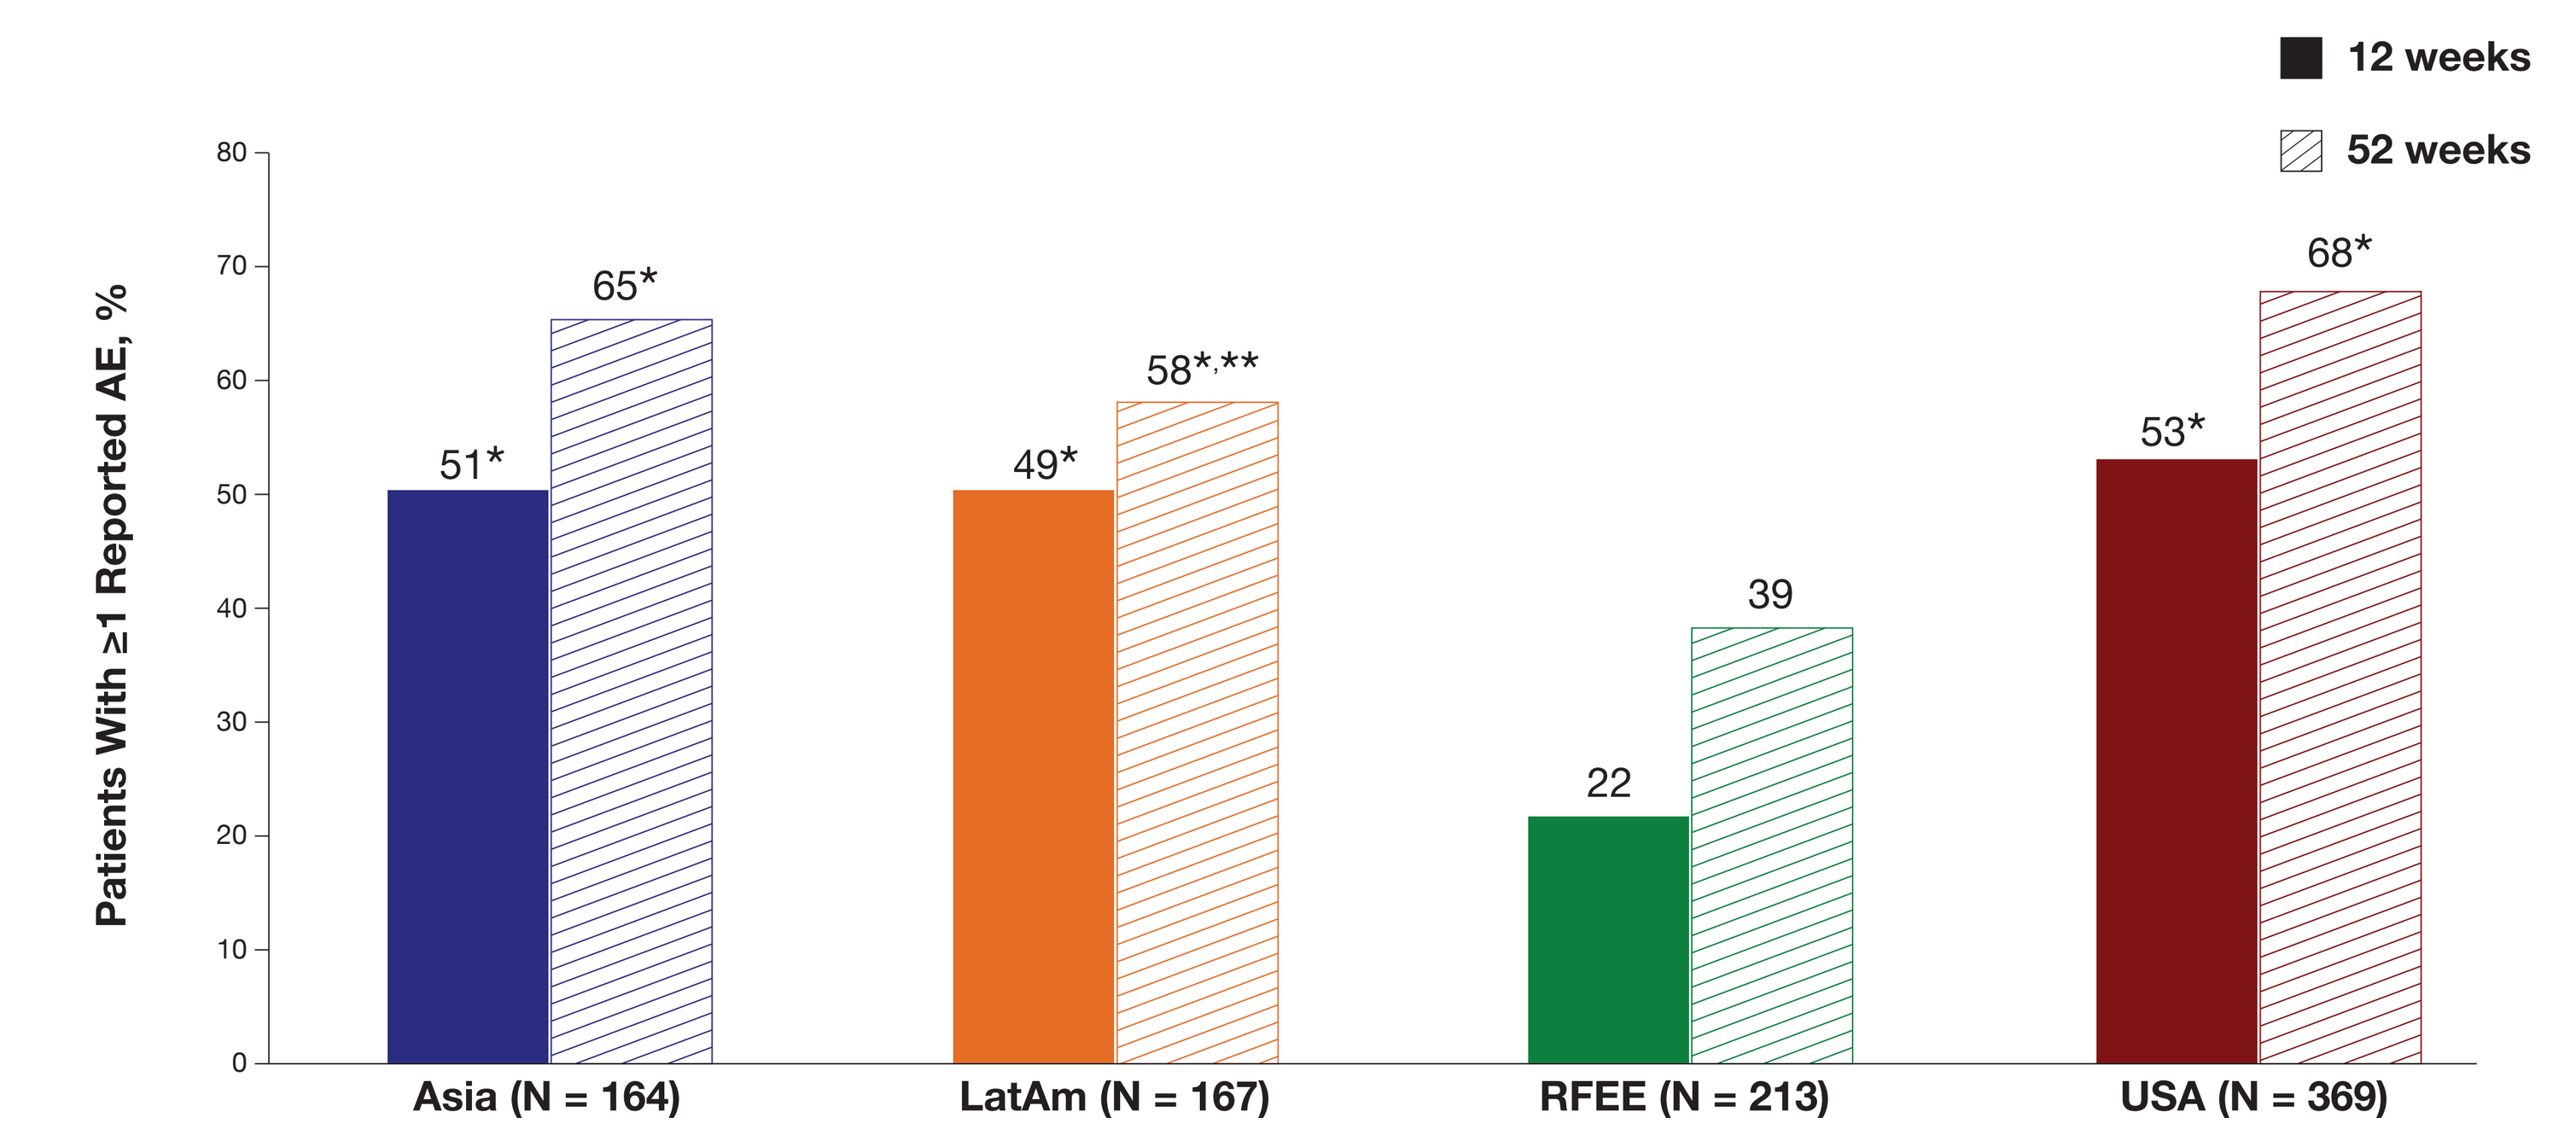
12 weeks
52 weeks
Patients With ≥1 Reported AE, %
51*
65*
49*
58*,**
22
39
53*
68*
Asia (N = 164)
LatAm (N = 167)
RFEE (N = 213)
USA (N = 369)

AE, adverse event; LatAm, Latin America; RFEE, Russian Federation and Eastern Europe.
*$p < 0.05$ versus RFEE.
**$p < 0.05$ versus USA.
All p values corrected for multiple comparisons.

CONCLUSIONS

- There were significant regional differences in ACR50 response and AE reporting rates; differences were especially notable among LatAm, RFEE, and Asia
 - Future patient populations from these regions may show distinct efficacy/safety profiles regardless of treatment
- Findings are consistent with, but do not prove, the hypothesis of lower placebo/standard-of-care arm ACR50 response and lower AE reporting rates in RFEE
- Caveat: Only 7 trials were included; generalizability may be limited
- Further contribution of industry-sponsored trial data into the TransCelerate registry could provide greater power for future analyses

REFERENCES

1. Drain PK et al. *Nat Rev Drug Discov*. 2014;13:166-167.
2. Contopoulos-Ioannidis D et al. *J Clin Epidemiol*. 2016;78(suppl C):10-21.
3. Yusuf S, Wittes J. *N Engl J Med*. 2016;375:2263-2271.
4. Xu X et al. *Arthritis Rheumatol*. 2016;68(suppl 10):abstr 2594.
5. Gill D. *Nat Rev Drug Discov*. 2014;13:787-788.

DISCLOSURES

D. Keebler, E. Teng, J. Chia, J. Galanter, and K. Tuckwell are shareholders and employees of Genentech.

D. Lee and J. Peake are employees of Genentech.

To receive a PDF of this poster on your phone:

- Go to getscanlife.com from your mobile browser to download the free barcode reader application
- Scan the code and get access to content

Presented at the 2018 American College of Rheumatology/Association of Rheumatology Health Professionals Annual Meeting (ACR/ARHP); October 19–24, 2018; Chicago, Illinois

This study was funded by F. Hoffmann-La Roche Ltd.
Support for third-party writing assistance for this poster was provided by F. Hoffmann-La Roche Ltd.