Poster number : P2419

Pharmacokinetics and safety in ICU Patients and Healthy Participants Following Single Dose Administration of Zosurabalpin (RG6006), a Novel Pathogen-Specific Antibiotic for the Treatment of Serious *Acinetobacter* Infections

Emmanuelle Cottreel[1], Bruno François[2], Akram Khan[3], Andreas Günther[4], Amanda Messer[5], Laurie Millar[5], Katie Patel[5], Lu Yuan[6], Thomas Lodise[7], Michael Lobritz[1]

[1] Roche Pharma Research and Early Development, Cardiovascular and Metabolism, Immunology, Infectious Diseases and Ophthalmology, Roche Innovation Center Basel, F. Hoffmann La Roche Ltd, Grenzacherstrasse 124, 4070 Basel, Switzerland; [2] Medical-surgical ICU and Inserm CIC-1435 & UMR 1092, CHU Dupuytren, Limoges, France; [3] Oregon Health and Science University, Portland, Oregon, USA; [4] Roche Pharma Research and Early Development, Translational PKPD and Clinical Pharmacology, Pharmaceutical Sciences, Roche Innovation Center Basel, Switzerland. Grenzacherstrasse 124, 4070 Basel, Switzerland; [5] Roche Innovation Center Welwyn, Roche Products Ltd, Hexagon Place, 6 Falcon Way, Welwyn Garden City, Hertfordshire, AL7 1TW, United Kingdom; [6] China Innovation Center of Roche, Roche R&D Center (China) Ltd, 720 Cai Lun Road, Building 5, Shanghai, 201203, Pudong, China; [7] Albany College of Pharmacy and Health Sciences, Albany, New York, USA

Background

- Zosurabalpin is a first-in-class novel tethered macrocyclic peptide antibiotic in Phase 1 clinical development (Zampaloni 2024).
- It is active against *Acinetobacter* spp., including carbapenem-resistant *Acinetobacter baumannii-calcoaceticus* complex organisms, exerting its antimicrobial activity through inhibition of the lipopolysaccharide transporter (LptB2FGC) (Pahil 2024).
- It is being developed for the treatment of severe infections caused by *Acinetobacter*, such as hospital-acquired bacterial pneumonia (HABP), ventilator-associated bacterial pneumonia (VABP) and bacteremia.
- An understanding of drug exposure in ICU patients is an important consideration for this patient population. Pharmacokinetic (PK) studies in ICU patients have identified augmented exposures related to covariates of critical illness, potentially affecting drug efficacy (Lodise 2021). We used data from two clinical studies to describe the plasma exposure of zosurabalpin in ICU patients in comparison with healthy participants (HPs).

Methods

- NCT05614895[(1)] is a multicenter, single dose, uncontrolled, open label, pharmacokinetic study. In the first three study cohorts, 27 ICU patients with HABP, VABP or bacteremia received 600 mg IV zosurabalpin.
- Part 1 of NCT04605718 was an investigator/participant-blind, placebo-controlled, randomized, single ascending dose study, in which HPs were randomized 3:1 to receive IV zosurabalpin or placebo in sequential single ascending dose cohorts. In one of the cohorts, 6 HPs received 600 mg IV zosurabalpin.
- Study drug was administered as a 100 mL solution (normal saline) and administered IV over the course of one hour.
- Plasma PK parameters were estimated by non-compartmental analysis.
- Safety was monitored throughout the studies: until 72 h ± 4 h post-dose in NCT05614895, and until 14 (± 2) days post-dose in Part 1 of NCT04605718.

(1) NCT05614895 is ongoing; data from this study is preliminary.

Results – Pharmacokinetics (I)

- In all participants, C_{max} was observed shortly after the end of infusion.
- The observed variability of PK data in ICU patients was larger than in HPs but within the limit to maintain 80% power for estimation of the 95% confidence interval for the geometric mean estimate for each PK parameter within the range of 60% to 140%.
- On average, in ICU patients, mean C_{max} was lower than in HPs.
- On average, the clearance of zosurabalpin was reduced and the AUC_{inf} was increased in patients compared to HPs (Table 3).

Table 3: Plasma PK parameters following single 1-hr IV dose administration of 600 mg zosurabalpin

Population	T_{max}	C_{max} (µg/mL)	Free C_{max} (µg/mL)	AUC_{Inf} (h•µg/mL)	Free AUC_{Inf} (h•µg/mL)	$T_{1/2}$ (h)	V_{ss} (L)	CL (L/h)
ICU patients (N=27)	1.08 (1.08 - 2.08)	23.5 (28.2%)	11.5 (27.4%)	164 (49.6%)	80.7 (49.1%) (2)	9.73 (38.6%)	39.3 (23.4%)	3.67 (49.6%)
Healthy participants (N=6)	1.03 (0.92 - 1.08)	31.1 (7.4%)	11.9 (7.4%)(1)	97.3 (15.6%)	37.4 (15.6%)(1)	6.32 (15.2%)	30.0 (6.9%)	6.17 (15.6%)

Table displays geometric mean and geometric mean CV%, except for T_{max}: median and minimum - maximum
T_{max}: time to maximum plasma concentration; C_{max}: maximum observed zosurabalpin concentration in plasma; AUC_{inf}: area under the zosurabalpin plasma concentration–time curve from time 0 extrapolated to infinity; $T_{1/2}$: apparent terminal elimination half life; V_{ss}: volume of distribution at steady state; CL: total body clearance.
(1) Estimated as total zosurabalpin × average unbound fraction. Unbound fraction was determined *in vitro* and average value was 38.4%.
(2) Based on the individual time-averaged unbound fraction.

Results – Demographics and baseline characteristics

Table 1: Demographics and baseline characteristics of ICU patients and healthy participants

Demographics and baseline characteristics	ICU patients N=27	Healthy participants N=6
Age, years, mean ± SD	60.1 ± 14.8	45.3 ± 12.6
Gender, n (%)		
Male	19 (70.4%)	6 (100.0%)
Female	8 (29.6%)	0
BMI, kg/m2, mean ± SD	31.9 ± 8.0	26.5 ± 1.1
Median (min-max)	31.0 (19.5 – 49.4)	26.3 (25.2 - 27.9)
Infection (1)		
HABP	8 (29.6%)	0
VABP	7 (25.9%)	0
Bacteremia	16 (59.3%)	0
Primary site of infection (bacteremic patients)		
Intra-abdominal infection	1 (3.7%)	NA
Meningitis	1 (3.7%)	NA
Pneumonia	7 (25.9%)	NA
Skin and soft tissue infection	2 (7.4%)	NA
Urinary tract infection	5 (18.5%)	NA
APACHE II, median (min-max)	14 (5-28)	NA
Serum creatinine (umol/L), mean ± SD	151.4 ± 157.8	94.3 ± 18.3
Median (min-max)	75.16 (38.9 - 685.3)	92.8 (70.7 - 115)
Renal replacement therapy, n (%)	2 (7.4%)	0
Baseline albumin, g/L, mean ± SD	27.3 ± 5.4	41.0 ± 2.2
Median (min-max)	27.0 (19.0 - 39.0)	41.0 (37.0 - 43.0)
Diabetes mellitus, n (%)	4 (14.8%)	0

(1) Some patients had bacteremia in addition to HABP or VABP.
APACHE II: Acute Physiology and Chronic Health Disease Classification System II; BMI: body mass index; HABP: hospital-acquired bacterial pneumonia; NA: not applicable SD: standard deviation; VABP: ventilator-associated bacterial pneumonia.

Results – Pharmacokinetics (II)

Figure 1: Median Plasma Zosurabalpin Concentration vs. Time Profiles following single 1-hr IV dose administration of 600mg zosurabalpin in ICU patients and healthy participants

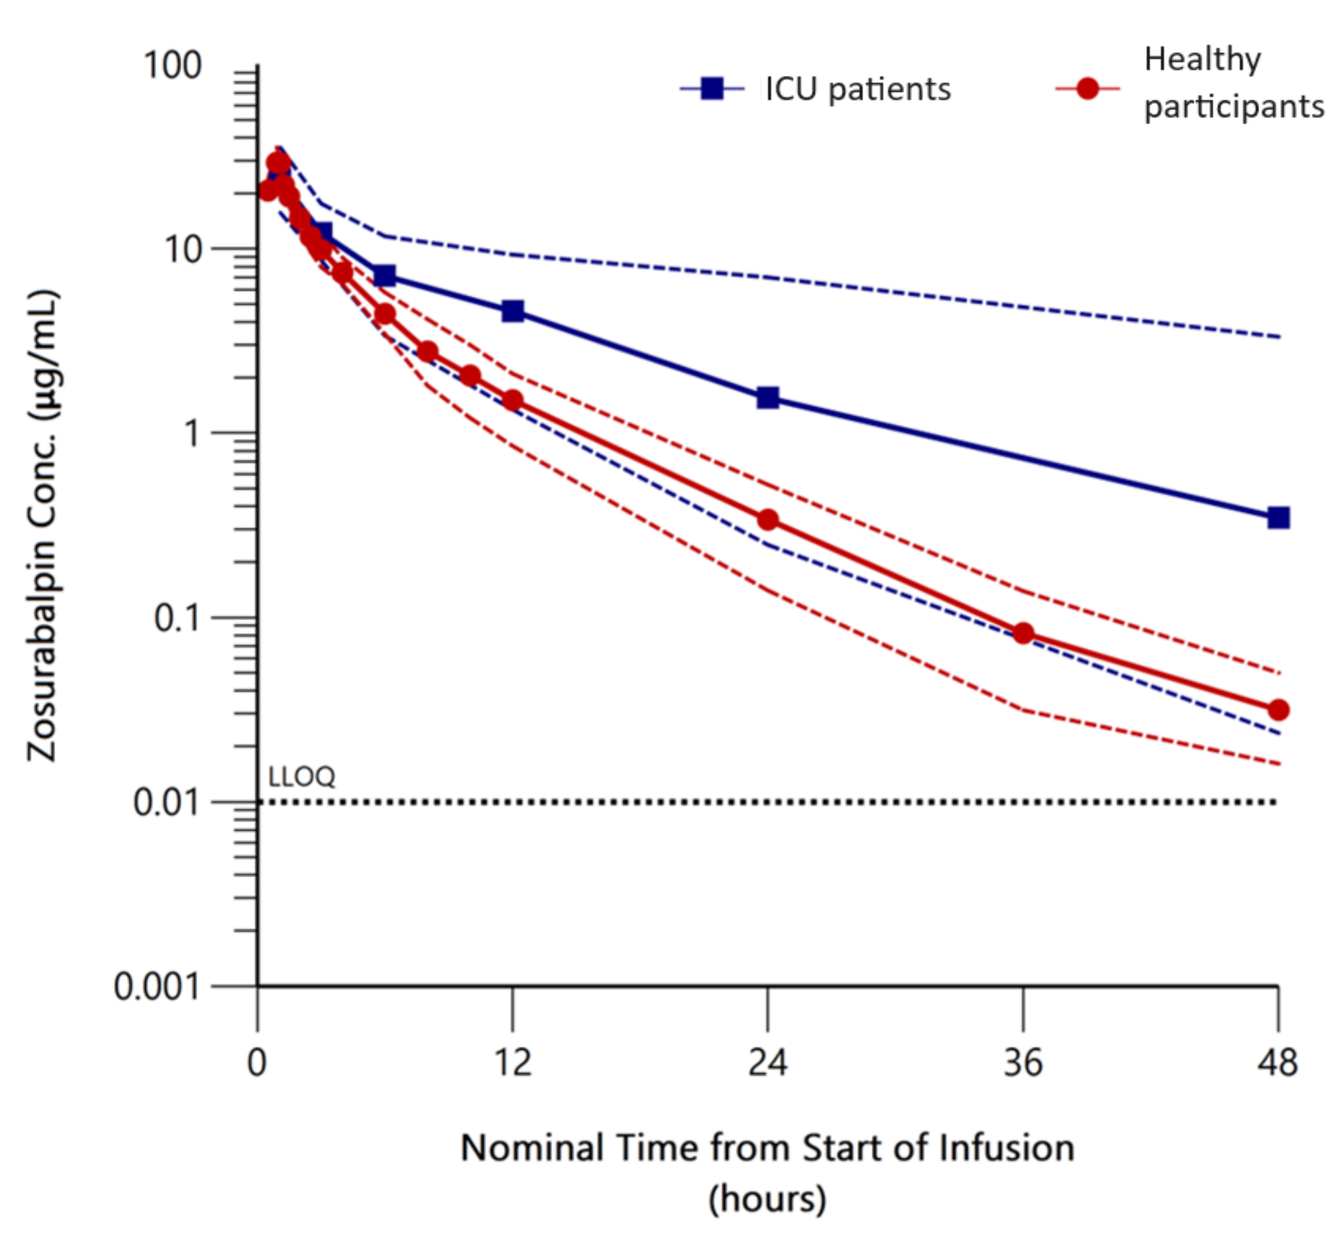

ICU patients
Healthy participants
Zosurabalpin Conc. (µg/mL)
100
10
1
0.1
0.01
0.001
LLOQ
0
12
24
36
48
Nominal Time from Start of Infusion (hours)

The solid blue and red lines are the observed medians. The dashed blue and red lines are the observed 10th and 90th percentiles.

Figure 2: Individual Zosurabalpin Concentrations vs. Time Profiles Following a single 1-hr IV dose administration of 600mg zosurabalpin in ICU patients[(1)]

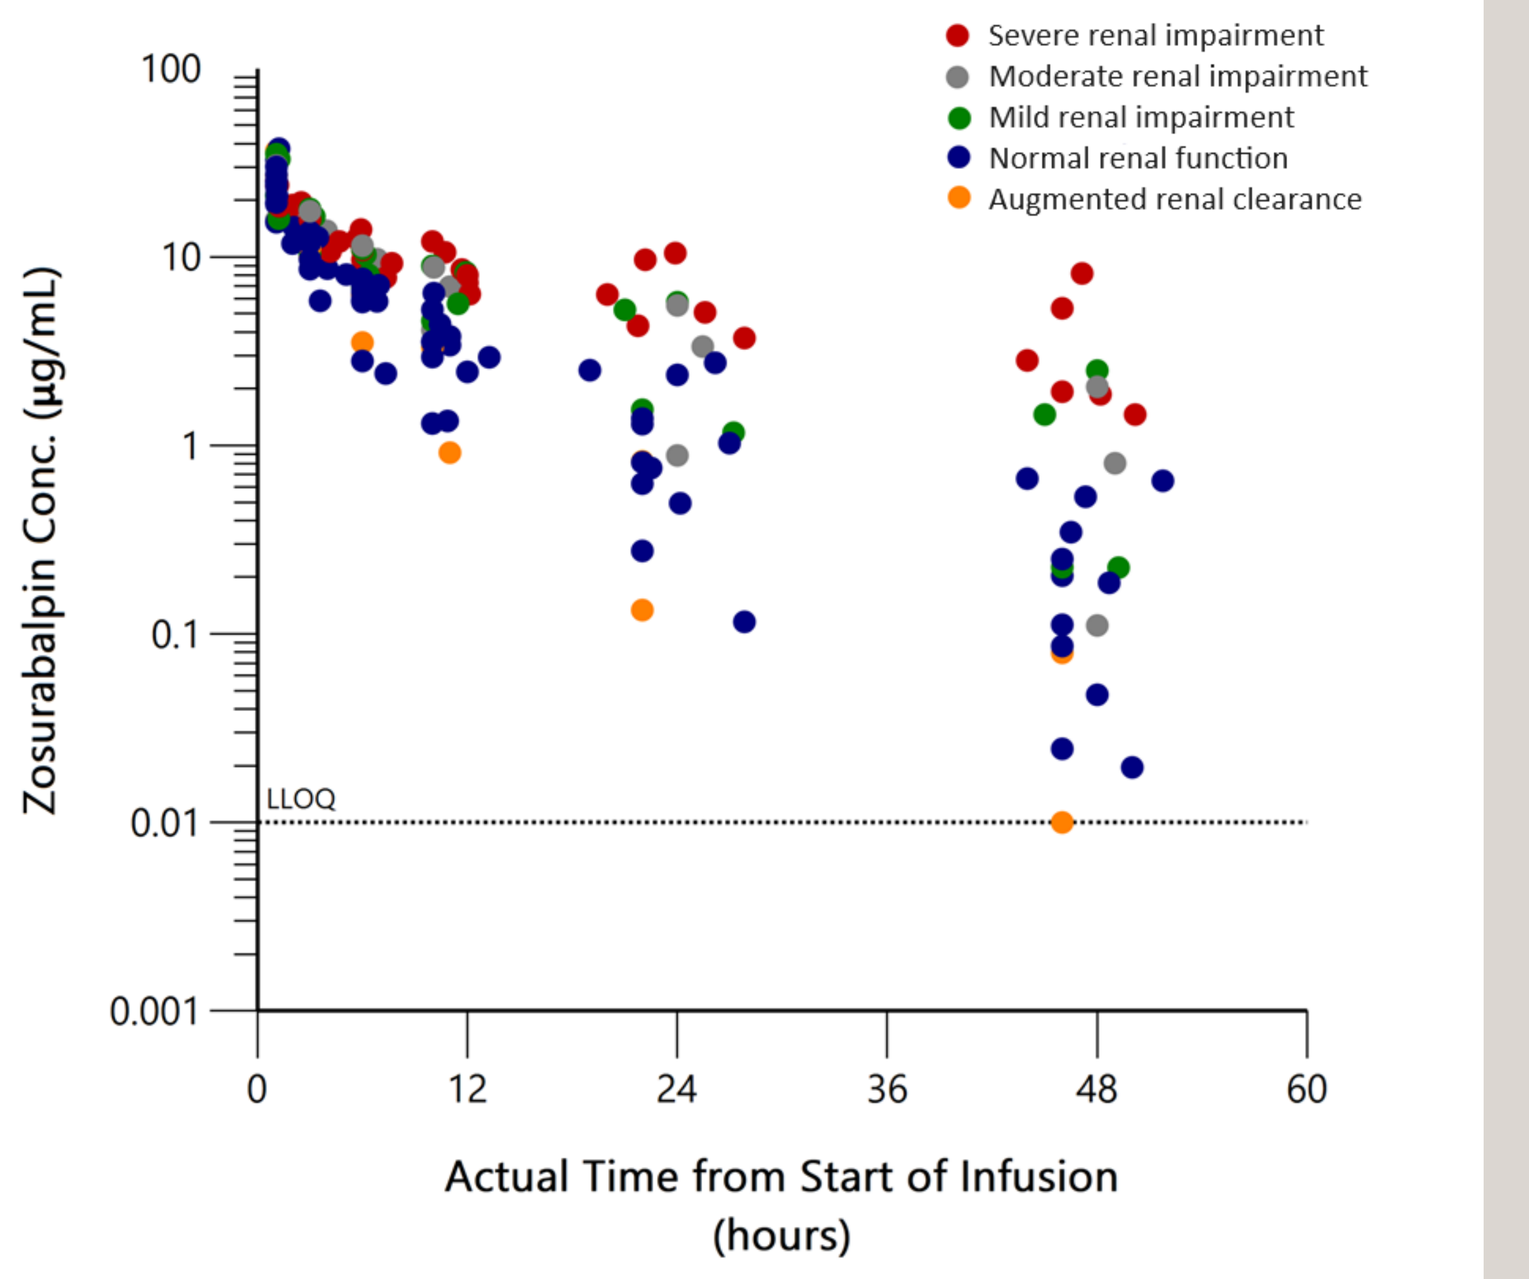

Severe renal impairment
Moderate renal impairment
Mild renal impairment
Normal renal function
Augmented renal clearance
Zosurabalpin Conc. (µg/mL)
100
10
1
0.1
0.01
0.001
LLOQ
0
12
24
36
48
60
Actual Time from Start of Infusion (hours)

(1) CKD-EPI creatinine equation (2021)
Severe renal impairment: estimated glomerular filtration rate (eGFR) < 30 mL/min/1.73m²; moderate renal impairment: eGFR ≥ 30 mL/min /1.73m² and ≤ 59 mL/min /1.73m²; mild renal impairment: eGFR ≥ 60 mL/min /1.73m² and ≤ 89 mL/min /1.73m²; normal renal function: eGFR > 89 mL/min/1.73m² and < 130mL/min /1.73m²; augmented renal clearance: eGFR ≥ 130 mL/min/1.73m².

Results – Safety

- In ICU patients, 8 adverse events (AEs) were reported. All AEs were non-serious and 2 were considered as related to the study treatment:
 - Grade 1 hepatic cytolysis was detected on the day after dosing. Maximum ALT and AST were observed 2 days after dosing (respectively 2.3 x ULN and 2.5 x ULN). Bilirubin stayed within normal range. The AE resolved without treatment after 13 days.
 - Grade 1 infusion related reaction manifested as hyperemia of face and neck, 2 hours after the end of infusion. Levocetirizine and montelukast were administered. The AE resolved after 4 hours.
- In HPs, 3 AEs were reported. All were grade 1, non-serious and considered as unrelated to the study treatment.
- All participants completed the studies as planned.

Table 2: Adverse events following a single 1-hr IV dose administration of 600mg zosurabalpin

Adverse event (coded term) (1)	Most extreme NCI CTCAE (2) grade	Seriousness	Relationship to study treatment	Outcome (3)
ICU patients (N=27)				
Renal failure	1	No	No	Recovered/Resolved
Urinary tract infection	3	No	No	Recovered/Resolved
Hepatic cytolysis	3	No	No	Recovered/Resolved
Hepatic cytolysis	1	No	Yes	Recovered/Resolved
Deep vein thrombosis	2	No	No	Recovered/Resolved
Abscess oral	1	No	No	Recovered/Resolved
Pulmonary oedema	2	No	No	Recovered/Resolved
Infusion related reaction	1	No	Yes	Recovered/Resolved
Healthy participants (N=6)				
Diarrhoea	1	No	No	Recovered/Resolved
Abdominal discomfort	1	No	No	Recovered/Resolved
Nodule	1	No	No	Recovered/Resolved

(1) Investigator text for AEs encoded using MedDRA version 26.0.
(2) NCI CTCAE: Common Terminology Criteria for Adverse Events v6.0
(3) In NCT05614895, safety follow-up for 72 h ± 4 h post-dose; in Part 1 of NCT04605718, safety follow-up for 14 (± 2) days post-dose.

Conclusions

- A single IV dose of 600 mg zosurabalpin was safe and well tolerated in ICU patients and HPs.
- The PK of zosurabalpin was different in ICU patients than in HPs.
- Characterization of the PK of zosurabalpin in ICU patients is of paramount importance to inform appropriate dose selection in future therapeutic trials ensuring that patients with serious *Acinetobacter* infections achieve adequately putative efficacious exposures.

References

Lodise TP. Antimicrob Agents Chemother. 2021 Feb 17;65(3):e01809-20.
Pahil KS. Nature. 2024 Jan;625(7995):572-577.
Zampaloni C. Nature. 2024 Jan;625(7995):566-571.

Acknowledgements
The authors thank the participants and their families for their trust and participation, and the personnel at study sites for their commitment to the study.

Acknowledgement of funding
This project has been supported in whole or in part with federal funds from the Department of Health and Human Services; Administration for Strategic Preparedness and Response; Biomedical Advanced Research and Development Authority, under contract number HHSO100201600038C.