A Phase 1b/2 study of the anti-myostatin adnectin RG6206 (BMS-986089) in ambulatory boys with Duchenne muscular dystrophy (DMD): A 72-week treatment update

Kathryn R. Wagner (1), Brenda L. Wong (2,3*), Barry J. Byrne (4), Cuixia Tian (3), Leslie K. Jacobsen (5*), Giridhar S. Tirucherai (5), Michael Rabbia (6), Heidemarie Kletzl (7), Rose Ong (7*), Karl Yen (7), Gautam Sajeev (8), James Signorovitch (8,9), Susan J. Ward (9), Clifford Bechtold (5*) and Michelle Krishnan (7)

(1) The Johns Hopkins School of Medicine, Baltimore, MD, USA; (2) UMass Memorial Children's Medical Center, Worcester, MA, USA; (3) Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA; (4) University of Florida, Gainesville, FL, USA; (5) Bristol-Myers Squibb, Princeton, NJ, USA; (6) Genentech, Inc, South San Francisco, CA, USA; (7) F. Hoffmann-La Roche, Basel, Switzerland; (8) Analysis Group, Inc., Boston, MA, USA; (9) Collaborative Trajectory Analysis Project (cTAP), Cambridge, MA, USA. *Institution at the time of study initiation.

Background

- Duchenne muscular dystrophy (DMD) is an X-linked, recessive neuromuscular disorder resulting in a loss of function of dystrophin protein and progressive muscle degeneration from early childhood.[1,2]
- Inhibition of myostatin, a negative regulator of muscle growth,[3] has been shown to increase skeletal muscle mass in several species, including humans.[3,4]
- RG6206 (BMS-986089) is a fully human anti-myostatin adnectin modified by the addition of an IgG-Fc tail to prolong half-life.[5]
 - — RG6206 inhibits myostatin activity by binding to serum-free myostatin or myostatin-ActRIIB complex and inhibiting downstream pSmad2/3 signalling via the inhibition of ALK4/5 (signaling receptor) recruitment.
 - — RG6206 is administered subcutaneously with weekly dosing at home.[5]
 - — In a Phase 1 healthy volunteer trial (NCT02145234)[6] RG6206 treatment reduced levels of free myostatin from baseline by a maximum of 96%.
 - This was associated with increases in thigh muscle volume and total LBM that were dependent on RG6206 dose and magnitude of free myostatin suppression.[7]

Design/Methods

Phase 1b/2 study

- Phase 1b/2 study of the novel anti-myostatin adnectin RG6206 in ambulatory boys with DMD, aged 5–10 years (NCT02515669).[8]
- Forty-three boys were randomized to receive weekly SC doses of RG6206 (4–50 mg) or placebo for 24 weeks (**Figure 1**).[8]
- All boys (including boys treated with placebo) received treatment with RG6206 for 48 weeks in the open-label phase (**Figure 1**).
- **Table 1** provides the demographics and baseline characteristics from the Phase 1b/2 study.

Cincinnati Children's Hospital Medical Center (CCHMC) cohort

- This cohort presents natural history data from a group of patients from the CCHMC.
- Changes in muscle mass were reported in the CCHMC cohort, analyses of which were conducted by the Collaborative Trajectory Analysis Project (cTAP) patient registry.
 - cTAP is a pre-competitive, multi-sponsor, multi-institution collaboration focused on using clinical data to improve drug development for DMD.

Figure 1. Phase 1b/2 study design in ambulatory boys with DMD aged 5–10 years[7,8]

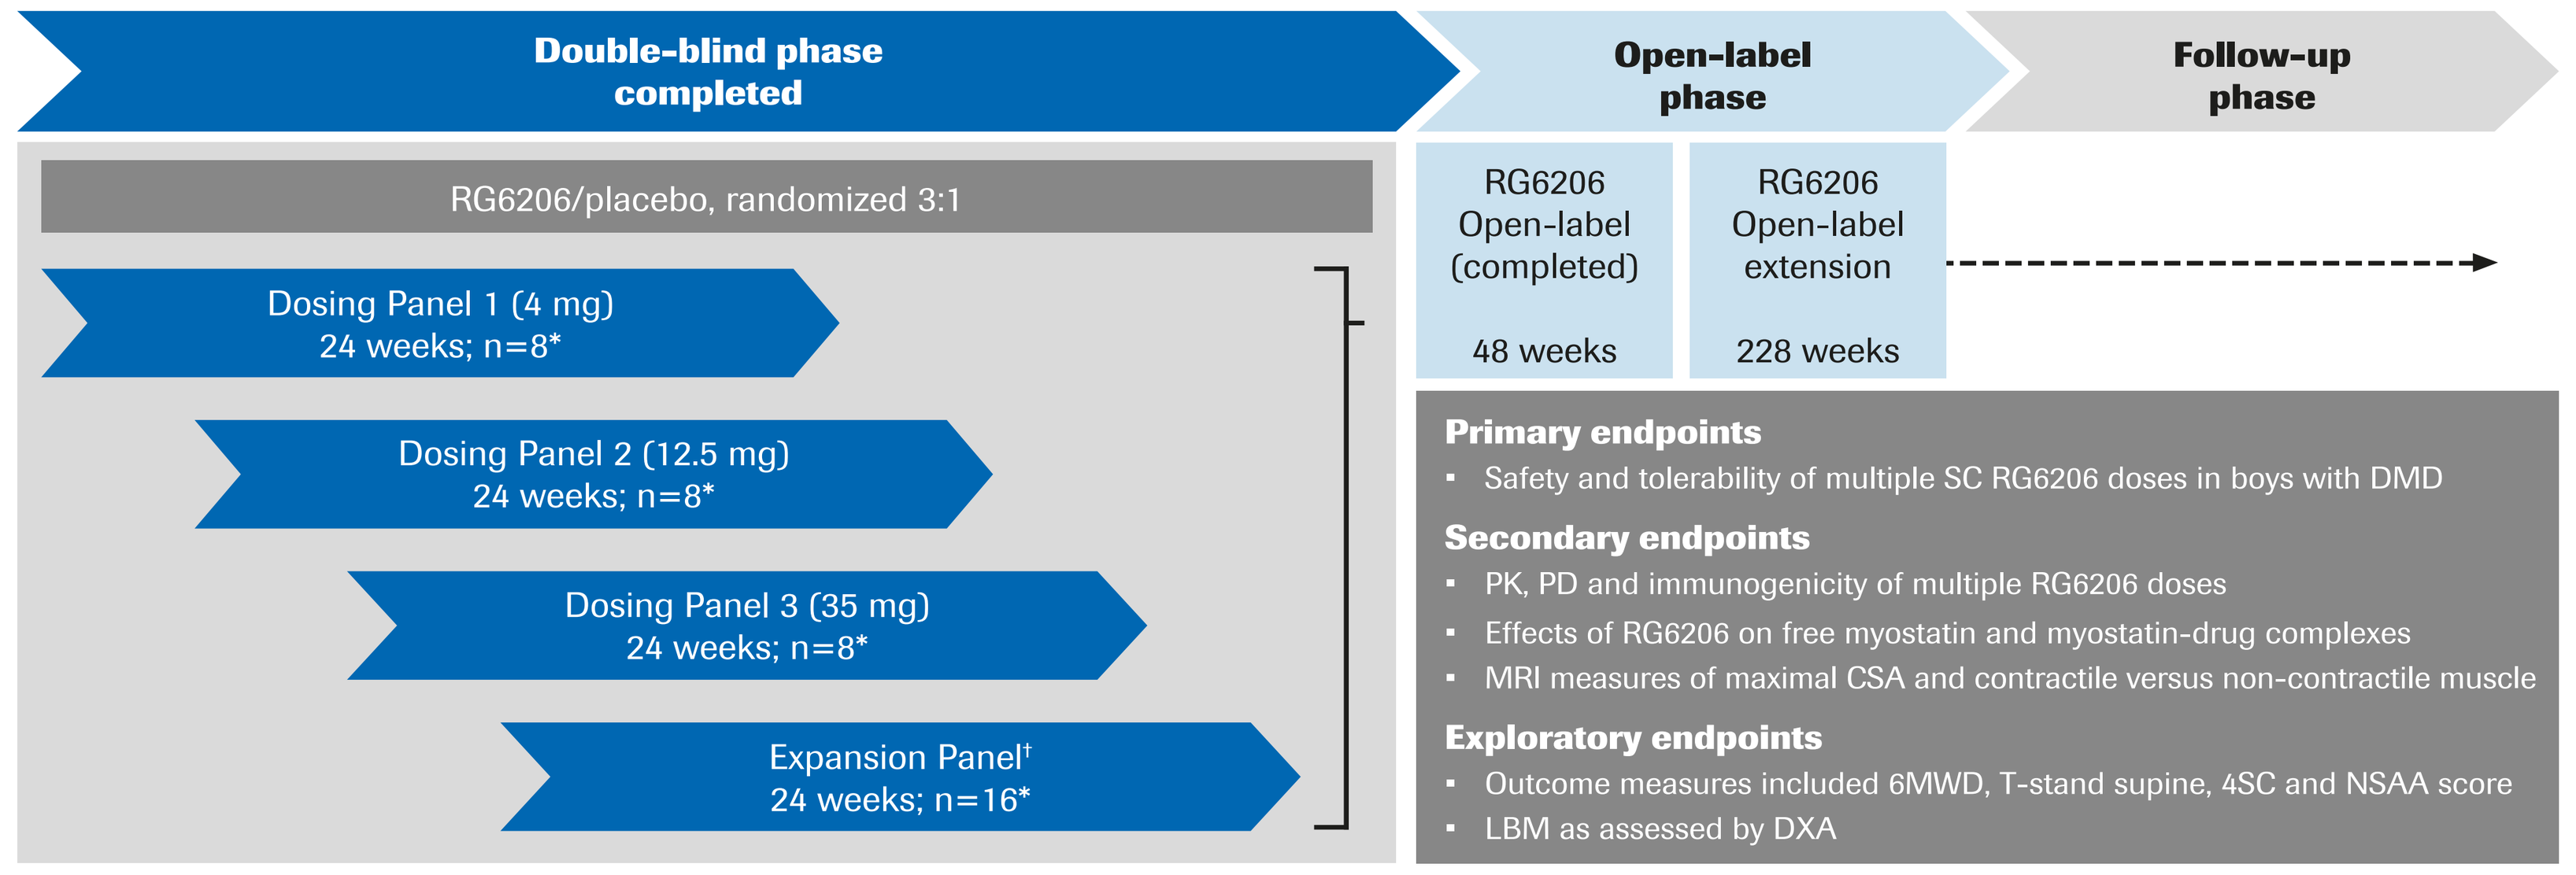
Double-blind phase completed
Open-label phase
Follow-up phase
RG6206/placebo, randomized 3:1
Dosing Panel 1 (4 mg) 24 weeks; n=8*
Dosing Panel 2 (12.5 mg) 24 weeks; n=8*
Dosing Panel 3 (35 mg) 24 weeks; n=8*
Expansion Panel† 24 weeks; n=16*
RG6206 Open-label (completed) 48 weeks
RG6206 Open-label extension 228 weeks
Primary endpoints
Safety and tolerability of multiple SC RG6206 doses in boys with DMD
Secondary endpoints
PK, PD and immunogenicity of multiple RG6206 doses
Effects of RG6206 on free myostatin and myostatin-drug complexes
MRI measures of maximal CSA and contractile versus non-contractile muscle
Exploratory endpoints
Outcome measures included 6MWD, T-stand supine, 4SC and NSAA score
LBM as assessed by DXA

*n, numbers per protocol; †Expansion panel utilizes the same dose as Dosing Panel 3.

Table 1: Phase 1b/2 study: Demographics and baseline characteristics*

	Placebo	Panel 1 RG6206	Panel 2 RG6206	Panel 3 RG6206	Expansion panel RG6206
n	11	7	6	6	13
RG6206 dose, mg **≥15 kg** **≥15 to ≤45 kg** **>45 kg**	N/A	4	12.5 20	35 50	35 50
Age, mean years (SD)	8.8 (1.3)	8.0 (2.2)	8.0 (1.8)	7.7 (2.3)	8.2 (1.6)
Age, n **5–6 years** **≥6–10 years**	1 10	3 4	1 5	3 3	3 10
Screening weight, mean kg (SD)	29.7 (7.6)	26.1 (6.4)	27.8 (6.6)	27.5 (6.7)	28.1 (9.0)
Screening weight, n **≤45 kg** **>45 kg**	10 1	7 0	6 0	6 0	12 1
Myostatin, median pg/mL (min–max)	1071 (500–2745)	835 (611–1006)	1114 (538–3552)	1447 (703–2143)	835 (492–1580)

Forty patients received daily doses of corticosteroids – three patients received intermittent doses.
*All treated subjects.

Please scan using your QR reader application to access this poster on your mobile device. NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details. Alternatively this can be accessed at: https://bit.ly/2KUwcFP

Results

Phase 1b/2 safety summary at Week 72

- At the time of the data cut (8th February 2018), no drug-related safety findings leading to withdrawal from the study were reported in any boys receiving RG6206 for 72 weeks.
- Most AEs were mild to moderate in intensity.
- The most common AEs considered to be related to study drug were injection site reactions, which were mild in intensity (except two moderate events: injection site erythema and injection site discomfort).
- Five SAEs were reported in four patients receiving RG6206 (influenza, rhabdomyolysis, femur fracture, conversion disorder and spinal compression fracture); all SAEs were considered unrelated to RG6206.
- At the time of the data cut (8th February 2018) there was no evidence of drug-related trends in safety lab parameters, vital signs, ECG parameters or echocardiogram parameters.

Phase 1b/2 PK and target engagement at Week 24

- At Week 24, RG6206 serum concentrations increased with dose and were accompanied by a dose-dependent reduction (77–97%) in free myostatin (**Figure 2**).
 - — Maximum myostatin suppression was obtained with the high (35 mg) dose.

Figure 2. Phase 1b/2 study: PK and target engagement at Week 24

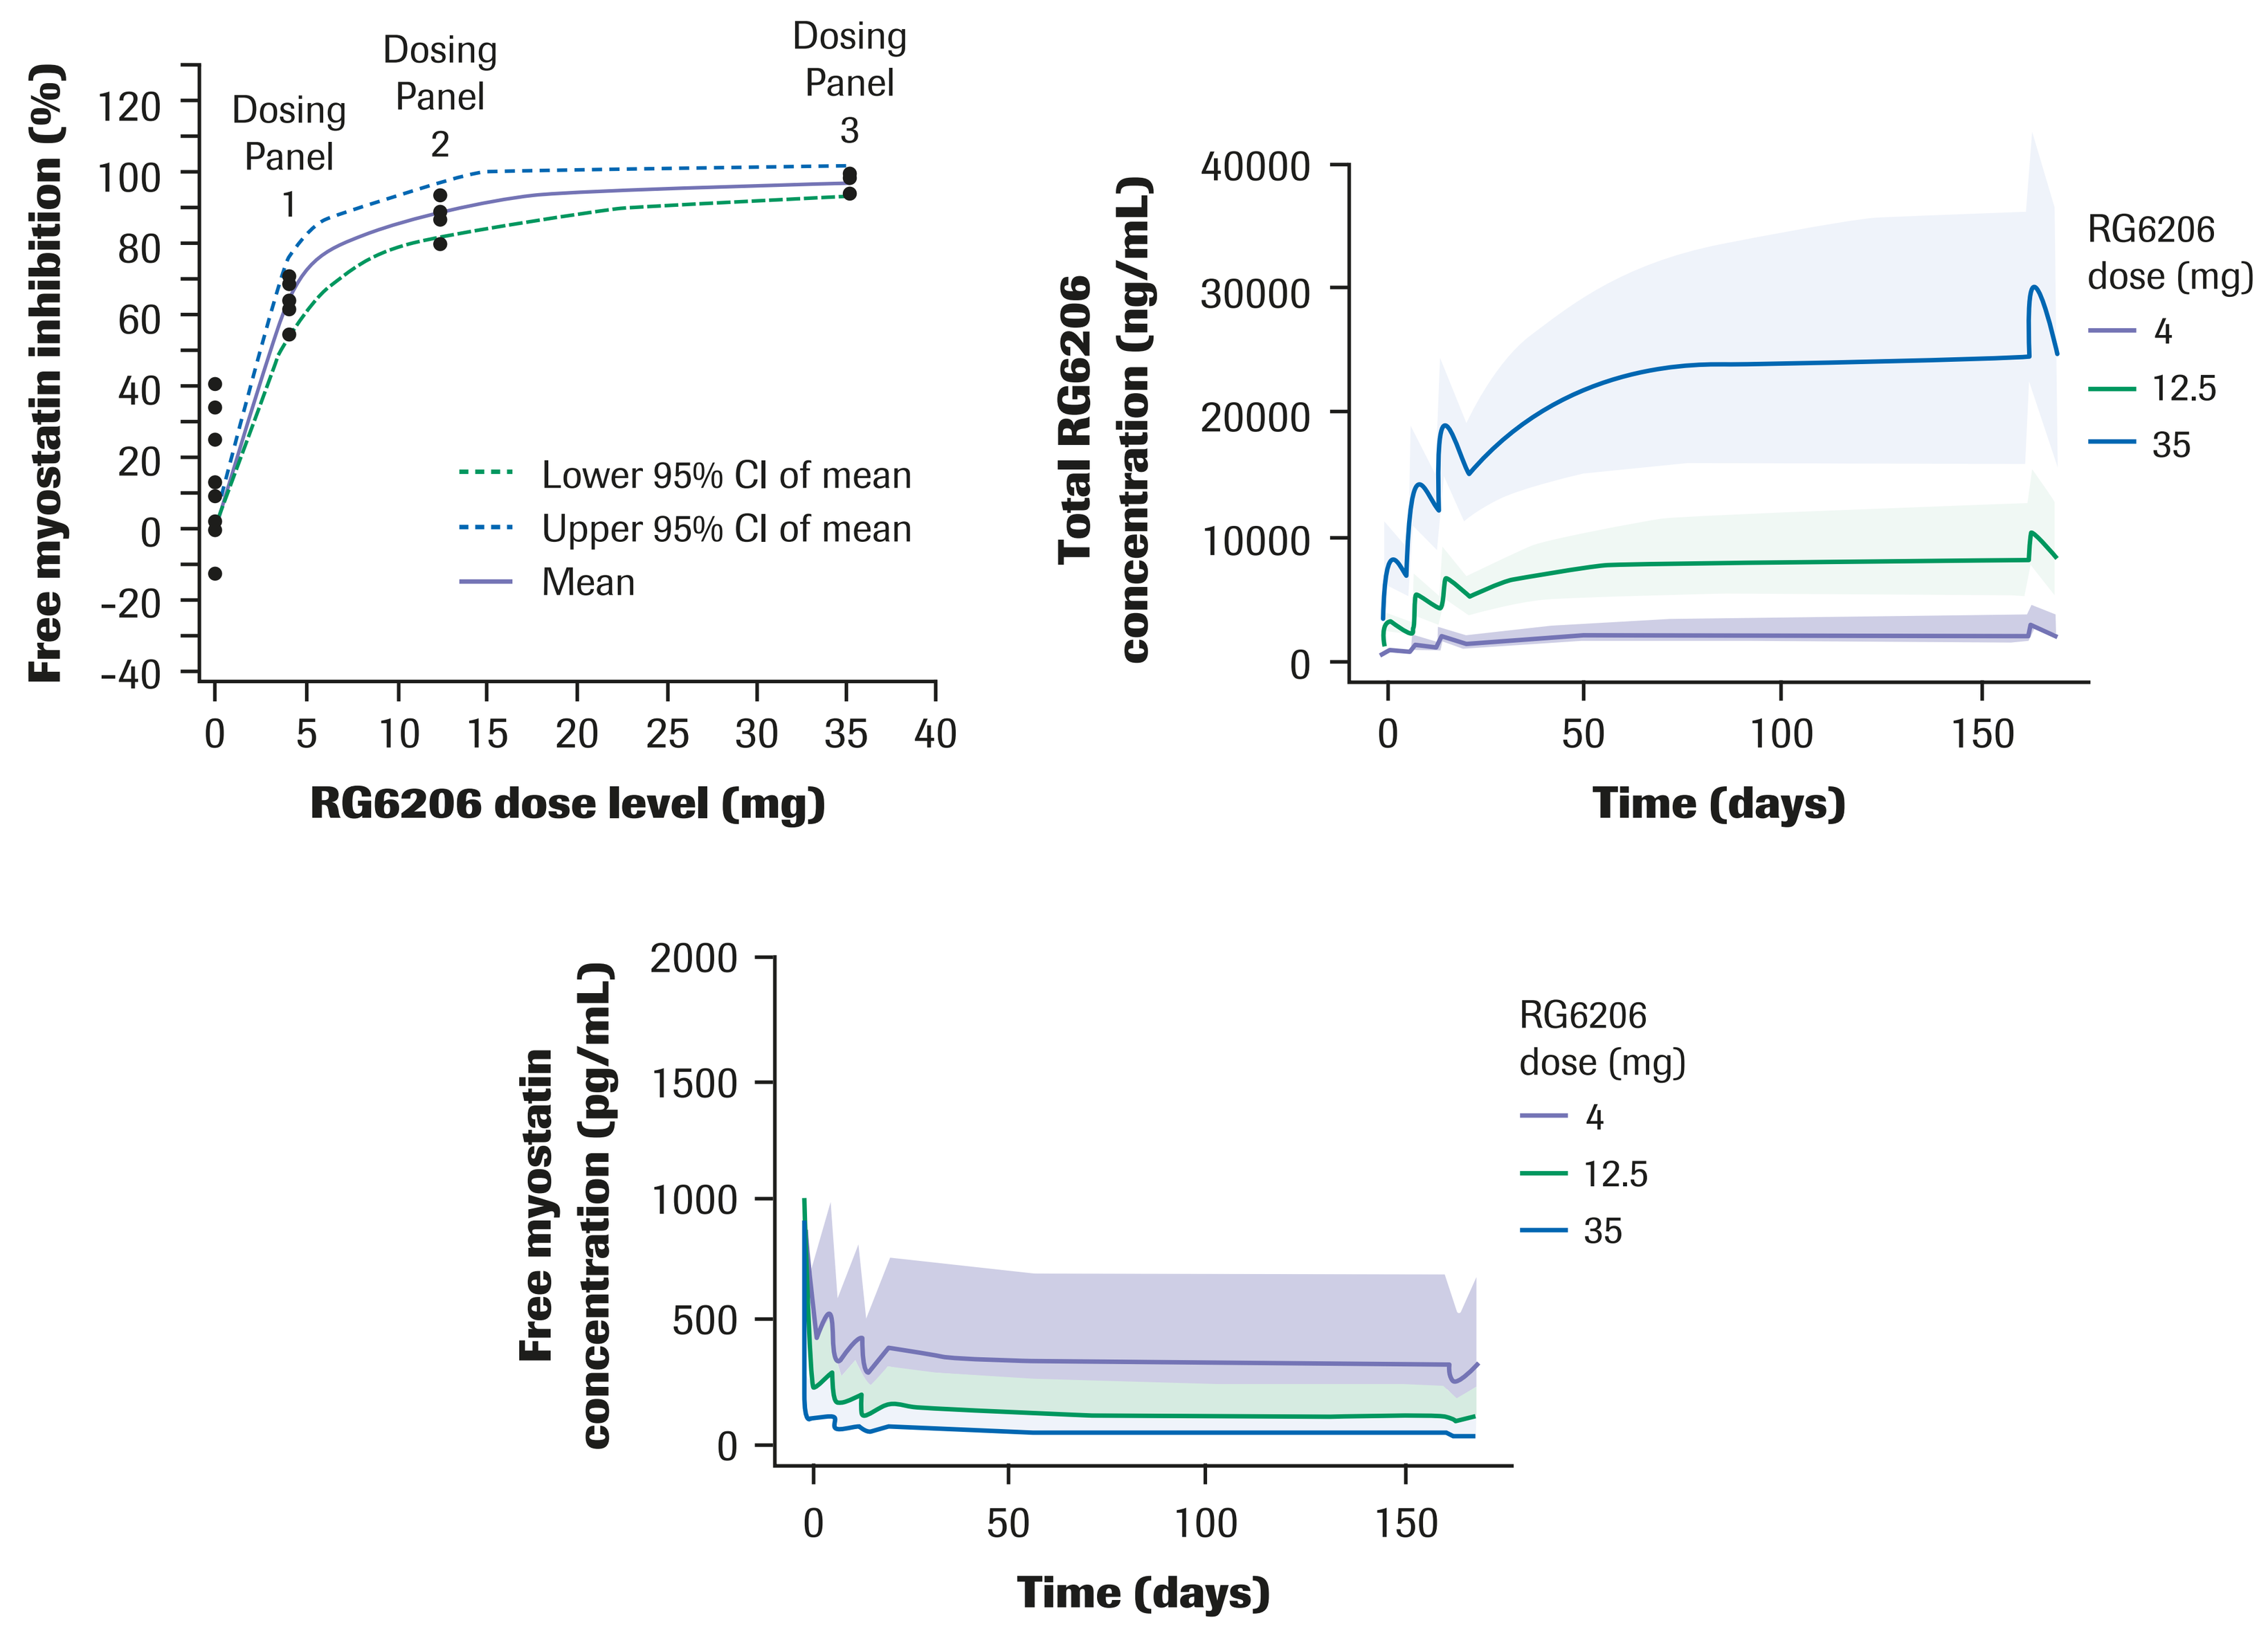
Dosing Panel 1
Dosing Panel 2
Dosing Panel 3
Free myostatin inhibition (%)
120
100
80
60
40
20
0
-20
-40
0 5 10 15 20 25 30 35 40
RG6206 dose level (mg)
Lower 95% CI of mean
Upper 95% CI of mean
Mean
Total RG6206 concentration (ng/mL)
40000
30000
20000
10000
0
0 50 100 150
Time (days)
RG6206 dose (mg)
4
12.5
35
Free myostatin concentration (pg/mL)
2000
1500
1000
500
0
0 50 100 150
Time (days)
RG6206 dose (mg)
4
12.5
35

Population PK analysis – lines are the population average and shaded areas represent the 90% prediction interval. Target myostatin suppression for Dosing Panels: 1 =≥50%, 2=≥ 85%, 3 and expansion =≥95%.

- DXA imaging showed that LBMi increased in boys receiving RG6206 for 72 weeks (**Table 2**).
- There was a decrease in LBMi in the CCHMC cohort, who had not received any treatment with RG6206 (**Table 2**).
- MRI results were consistent with the DXA data at Week 72 (data not shown).
- At Week 24, when all dose groups were pooled, boys on active treatment showed a 3.85% increase in LBMi (assessed using DXA) and a 5.49% increase in contractile CSA of right thigh (assessed using MRI) versus placebo (data not shown).[9]

Table 2. Absolute change in LBMi at Weeks 24 and 72 in boys treated with RG6206 and in boys with DMD in the CCHMC cohort measured by DXA

	RG6206 Phase 1b/2 study				CCHMC cohort	
	RG6206 (all doses pooled) n=32		Placebo n=11		Natural history n=63	
Time point (weeks)	**Model estimate (kg/m²)**	**SE**	**Model estimate (kg/m²)**	**SE**	**Model estimate (kg/m²)**	**SE**
Week 24	0.16	0.10	−0.13	0.17	−0.06	0.07
Week 72	0.26	0.15	-	-	−0.18	0.21

Phase 1b/2 Week 24 placebo results based on LSM estimates from ANCOVA including baseline value and age as covariates. Phase 1b/2 Week 24 and Week 72 active results based on LSM estimates from MMRM including baseline value and age as covariates. cTAP natural history results based on model estimates from RCRM model for which Week 24 estimate is the median of estimates calculated for the various models provided.

- **Table 3** provides the baseline characteristics in boys enrolled in the Phase 1b/2 study and boys in the CCHMC cohort.

Table 3. Baseline characteristics from boys enrolled in the Phase 1b/2 study and in boys with DMD in the CCHMC cohort

		RG6206 Phase 1b/2 study		CCHMC cohort
		RG6206 (all doses pooled) n=32	Placebo n=11	Natural history n=63
	Unique patients	32	11	56
Age (years)	**Mean±SD** **Median** **Range**	8.0±1.84 9.0 (6, 10)	8.8±1.33 9.0 (6, 10)	7.75±1.57 7.69 (5.28, 10.62)
Current steroid use	**Deflazacort** **Prednisone**	22 (69%) 10 (31%)	10 (90.9%) 1 (9.1%)	58 (92.06%) 9 (14.29%)
NSAA total score	**Mean±SD** **Median** **Range**	23.71±6.59 25.00 (8.00, 33.00)	25.36±6.04 26.00 (11.00, 33.00)	26.95±4.83 27.00 (12.00, 34.00)
4SC (stairs/sec)	**Mean±SD** **Median** **Range**	1.19±0.51 1.15 (0.46, 2.50)	1.44±0.57 1.29 (0.40, 2.41)	2.28±0.84 2.35 (0.59, 4.44)
LBMi	**Mean±SD** **Median** **Range**	10.09 9.89 (7.23, 12.28)	10.48 10.46 (7.83, 12.61)	13.42±1.32 13.18 (10.77, 16.70)

This was a modelling comparison using a small number of boys enrolled in the Phase 1b/2 study, further work is needed to refine the approach to be used with other trial and real-world data.

Conclusions

- At the time of the data cut (8th February 2018), no drug-related safety findings leading to withdrawal from the study were reported in any boys receiving RG6206 for 72 weeks.
- The PK profile of RG6206 was as expected and robust target engagement (myostatin suppression) was achieved.
- Imaging outcomes suggested that RG6206 treatment had a positive effect on muscle in boys with DMD.
- Forty-one boys from this study are now enrolled in a 228-week open-label extension.
- A Phase 2/3 RG6206 study is actively recruiting (NCT03039686)[10] worldwide.

Abbreviations

4SC, 4-stair climb (velocity); 6MWD, 6-Minute Walk Distance; ActRIIB, activin receptor Type IIB; AE, adverse event; ALK, anaplastic lymphoma kinase; ANCOVA, analysis of covariance; CCHMC, Cincinnati Children's Hospital Medical Center; CI, confidence interval; CSA, cross sectional area; cTAP, Collaborative Trajectory Analysis Project; DMD, Duchenne muscular dystrophy; DXA, dual-energy X-ray absorptiometry; ECG, electrocardiogram; Fc, fragment crystalizable; Ig, immunoglobulin; LBM, lean body mass; LBMi, lean body mass index; LSM, least-squares means; MMRM, mixed model for repeated measurements; MRI, magnetic resonance imaging; N/A, not applicable; NSAA, North-Star Ambulatory Assessment; PD, pharmacodynamics; PK, pharmacokinetics; RCRM, runoff coefficient routing model; SAE, serious adverse event; SC, subcutaneous; SD, standard deviation; SE, standard error; T-stand supine, time to standing from supine.

Acknowledgements

We thank all the patients and patient family members who participate in our studies. This study was funded by F. Hoffmann-La Roche. We acknowledge the contributions and support of our colleagues, and the study investigators. The authors thank Michelle Kim, PhD, of MediTech Media UK for providing editorial support for this poster, which was funded by F. Hoffmann-La Roche in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

References

1. Ryder S, et al. *Orphanet J Rare Dis*. 2017; 12:1–21;
2. Leung DG, et al. *Ann Neurol*. 2013; 74:404–411;
3. Sharma M, et al. *IUBMB Life*. 2015; 67:589–600;
4. Schuelke M, et al. *N Engl J Med*. 2004; 350:2682–2688;
5. Roche data on file;
6. Clinicaltrials.gov: NCT02145234 (Accessed April 2019);
7. Jacobsen LK, et al. Presented at 21st International World Muscle Society Congress. Granada, Spain; Oct 4–8, 2016;
8. Clinicaltrials.gov: NCT02515669 (Accessed April 2019);
9. Wagner K, et al. Presented at the Word Muscle Society Congress. Mendoza, Argentina; Oct 2–6, 2018;
10. Clinicaltrials.gov: NCT03039686 (Accessed April 2019).

Disclosures

- KW received honorarium from Sarepta, Wave, Fibrogen, Dynacure, Lion, PTC Therapeutics and research support from Pfizer, Roche, and Sarepta.
- BW received personal compensation from GSK and research support from Capricor Therapeutics.
- BB received personal compensation from Pfizer and licensing fees from the University of Florida.
- CT received research support from Pfizer, BMS, Roche, Fibrogen, PTC and Sarepta for clinical trials.
- LJ is an employee of Shire and holds stocks.
- GT is an employee of Bristol-Myers Squibb and hold stocks.
- MR and HK are employees of F. Hoffmann-La Roche and hold stocks.
- RO holds stocks for F. Hoffmann-La Roche.
- KY is an employee of F. Hoffmann-La Roche.
- JS and GS are employees of Analysis Group Inc., which received funding for this research from F. Hoffmann-La Roche as a member of cTAP.
- SJW receives funding from cTAP drug company members, including F. Hoffmann-La Roche, to manage the collaboration.
- CB received personal compensation from Bristol-Myers Squibb.
- MK received personal compensation from F. Hoffmann-La Roche and Biogen.

Presented at the 2019 American Academy of Neuroscience Annual Meeting, Philadelphia, PA, USA; May 4–10, 2019