Defining clinically meaningful change on the composite Unified Huntington's Disease Rating Scale (cUHDRS)

Dylan Trundell (1), Giuseppe Palermo (2), Scott Schobel (2), Jeffrey D. Long (3,4), Blair R. Leavitt (5), Sarah J. Tabrizi (6)

(1) Roche Products Ltd, Welwyn Garden City, UK; (2) F. Hoffmann-La Roche Ltd, Basel, Switzerland; (3) Department of Psychiatry, Carver College of Medicine, University of Iowa, Iowa City, Iowa, USA;
(4) Department of Biostatistics, College of Public Health, University of Iowa, Iowa City, Iowa, USA; (5) Centre for Molecular Medicine and Therapeutics, Department of Medical Genetics, University of British Columbia, Vancouver, British Columbia, Canada;
(6) Huntington's Disease Research Centre, UCL Institute of Neurology, London, UK.

Background

- Huntington's disease (HD) is a genetic, neurodegenerative and ultimately fatal disease that has a devastating impact on families across generations. HD is characterised by a triad of cognitive, behavioural and motor symptoms, which result in functional disability and progressive loss of independence.[1]
- The composite Unified Huntington's Disease Rating Scale (cUHDRS) has been developed to assess disease progression in early-to-moderate manifest HD. It combines four assessments: Total Functional Capacity (TFC), Total Motor Score (TMS), Symbol Digit Modalities Test (SDMT) and Stroop Word Reading (SWR).[2]
- Composite endpoints consist of multiple measures; qualitative and quantitative evidence have been generated to aid interpretation of the cUHDRS by linking the score to established measures of meaningful daily function and independence.

Objective

To enhance interpretation of cUHDRS score and clinically meaningful change on the cUHDRS

Design/Methods

Qualitative evidence

- To assess the conceptual relevance of the cUHDRS to patients and caregivers, the following research was conducted.
 — A literature review, comprising a targeted search of qualitative studies using PubMed, supplemented by additional articles (Huntington's Disease Society of America [HDSA] Survey,[3] FDA Voice of the Patient[4]).
 — Patient/caregiver interviews, comprising one-to-one telephone interviews with members of HDSA following a semi-structured interview guide.
- Key themes (symptoms and impacts) emerging from the qualitative research were summarised to better understand the conceptual relevance of the cUHDRS.

Quantitative measures

Population/data sources

- Data from Enroll-HD (periodic dataset 3, December 2016) were used. Patients similar to those in GENERATION HD1 (an ongoing randomised, double-blind, placebo-controlled Phase III study evaluating the efficacy and safety of RG6042 in manifest HD) were included (manifest HD, 25–65 years, Independence Scale [IS] ≥70, CAG–age product score [CAP] ≥400), except for 'association with function' where all manifest HD patients were included to provide a broader reflection of the relationship between these measures.
- To estimate meaningful within-patient cUHDRS score change, data from Enroll-HD (population as above) and data from REGISTRY (manifest HD, 25–65 years, TFC 5–13, CAP ≥400) were used.

Outcome measures

- The measures used in this study are summarised in Table 1.

Table 1. Measures used in the study

CGI-S, Clinical Global Impression-Severity; FA, Functional Assessment; IS, Independence Scale; SDMT, Symbol Digit Modalities Test; SWR, Stroop Word Reading; TFC, Total Functional Capacity; TMS, Total Motor Score.

Function/Independence			Motor Function	Cognitive Function		Global Severity
TFC	IS	FA	TMS	SDMT	SWR	CGI-S

- The cUHDRS is scored using the following formula:

$$cUHDRS = \frac{TFC - 8.8}{2.8} - \frac{TMS - 34.4}{17.4} + \frac{SDMT - 25.2}{12.4} + \frac{SWR - 58.0}{21.2} + 10$$

Relation to functional ability

- Manifest HD patients were grouped according to cUHDRS score and the proportion of patients achieving each item on the Unified Huntington's Disease Rating Scale Functional Assessment (FA) was calculated (Enroll-HD).

Meaningful change estimation

- Anchor-based analyses (linear regression) were carried out to estimate meaningful, within-patient change.
 — 1-category worsening on the, Clinical Global Impression-Severity (CGI-S) scale over 12 months (REGISTRY) for constituent scales.
 — 1-category worsening on CGI-S (REGISTRY) and a decline of 10 points on IS (Enroll-HD) for cUHDRS.
 — A distribution-based analysis, standard error of measurement (SEM) was calculated as a supportive analysis (SEM=Standard Deviation $[SD]_{Baseline} * \sqrt{[1\text{-test-retest reliability}]}$).
- Meaningful change estimates were compared to the ratio of the SD used in z-score calculations.

Please scan using your QR reader application to access this poster on your mobile device. NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details. Alternatively this can be accessed at: https://bit.ly/2Zxav1A

Results

Qualitative evidence

Qualitative evidence captures the significance of disease-related cognitive impairment, motor impairment and loss of daily functional ability to HD patients and their families **(Figure 1)**.

Cognition

- Cognitive impairment is commonly reported in individuals with HD.[3,4]
 — >60% of patients (N=215) and >70% of caregivers (N=729) reported the presence of dysexecutive syndrome, and declines in cognitive processing speed and attention.[3]
 — Executive function/cognitive decline was selected as the most impactful symptom by 22% of patients (N=189) and by 24.3% of caregivers (N=701) second only to chorea (30.1%).[3]
 — Cognitive impairment was selected as the HD symptom with the most significant impact on daily life by participants of the FDA Voice of the Patient meeting for HD.[4]
- Examples of cognitive impairment in HD include slowed processing speed and reduced capacity for decision-making, problem-solving, planning/organising/sequencing, perseveration and multitasking.[5]

Motor

- Motor dysfunction also greatly impacts the lives of HD patients and their families.
- Patients and caregivers described the impact of motor impairment on faculties including balance, uncontrolled movements and speech.[4]
- Chorea movement/balance was considered the most impactful symptom by caregivers and the third most impactful by patients.[3]

Functional capacity

- Concept elicitation interviews identified that the concepts assessed by TFC items (e.g. managing finances and activities of daily living) are important to patients/caregivers.[6]
- Cognitive debriefing of TFC demonstrated that changes are meaningful across the scale.[6]

Figure 1: Qualitative evidence supporting the composition of the cUHDRS

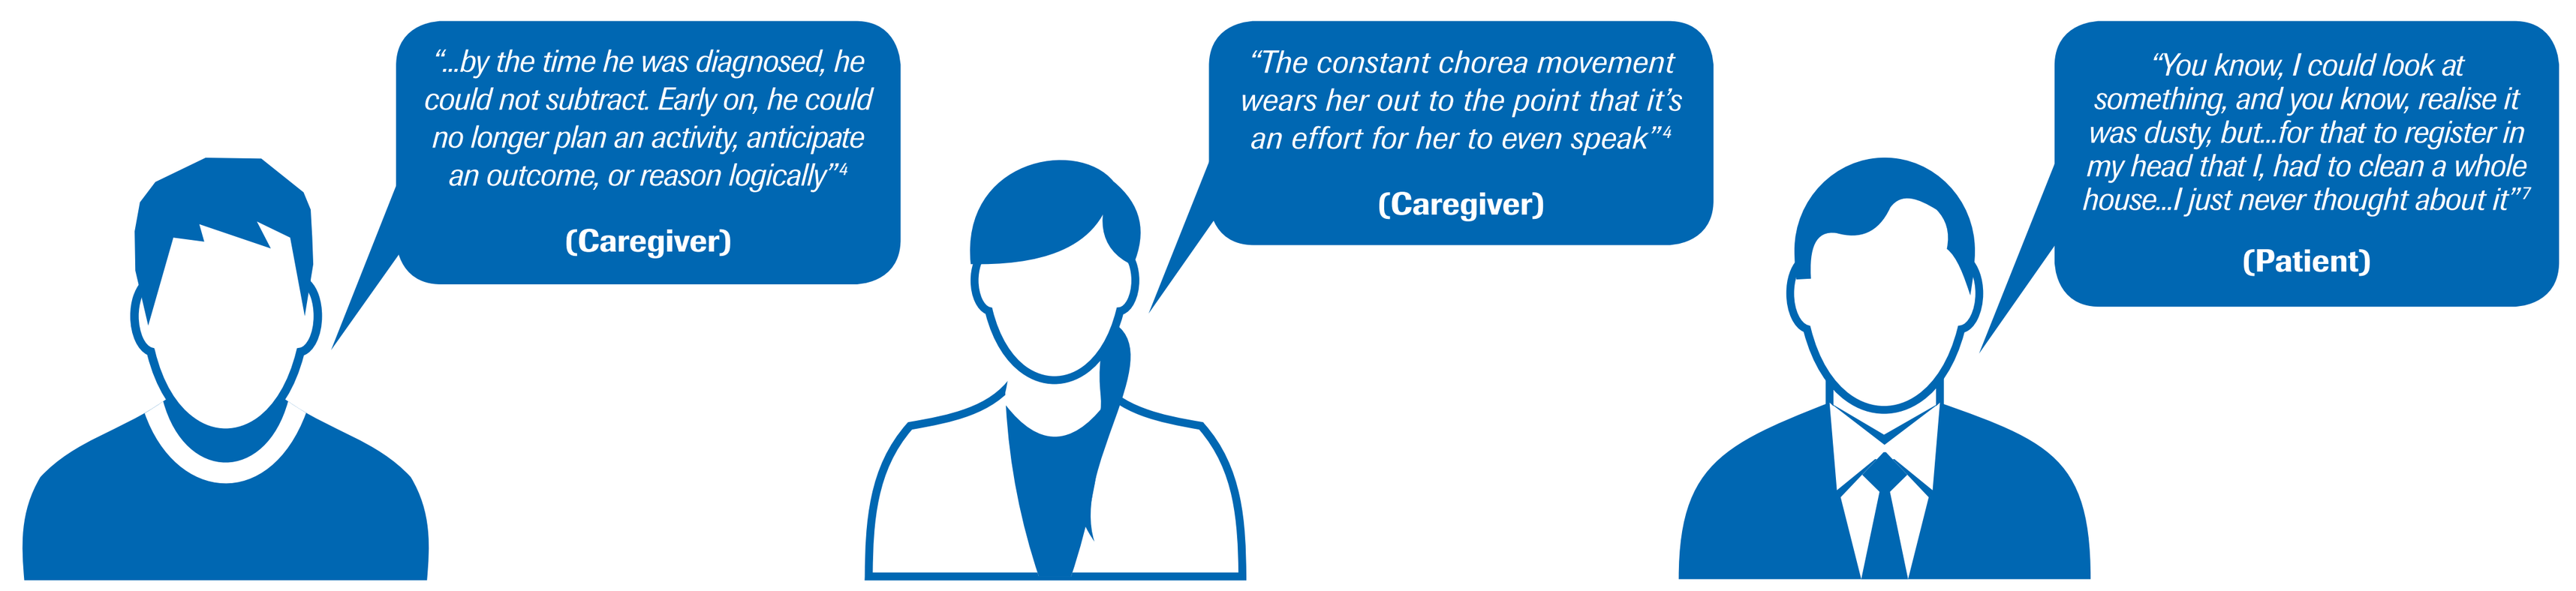
"...by the time he was diagnosed, he could not subtract. Early on, he could no longer plan an activity, anticipate an outcome, or reason logically"[4]
(Caregiver)
"The constant chorea movement wears her out to the point that it's an effort for her to even speak"[4]
(Caregiver)
"You know, I could look at something, and you know, realise it was dusty, but...for that to register in my head that I, had to clean a whole house...I just never thought about it"[7]
(Patient)

Quantitative evidence

Relationship between cUHDRS and functional ability

- As cUHDRS score declines, the proportion of patients achieving each item on the FA also declines, demonstrating an association between cUHDRS score and functional ability **(Figure 2)**.

Figure 2: Proportion of patients achieving each FA item by cUHDRS score

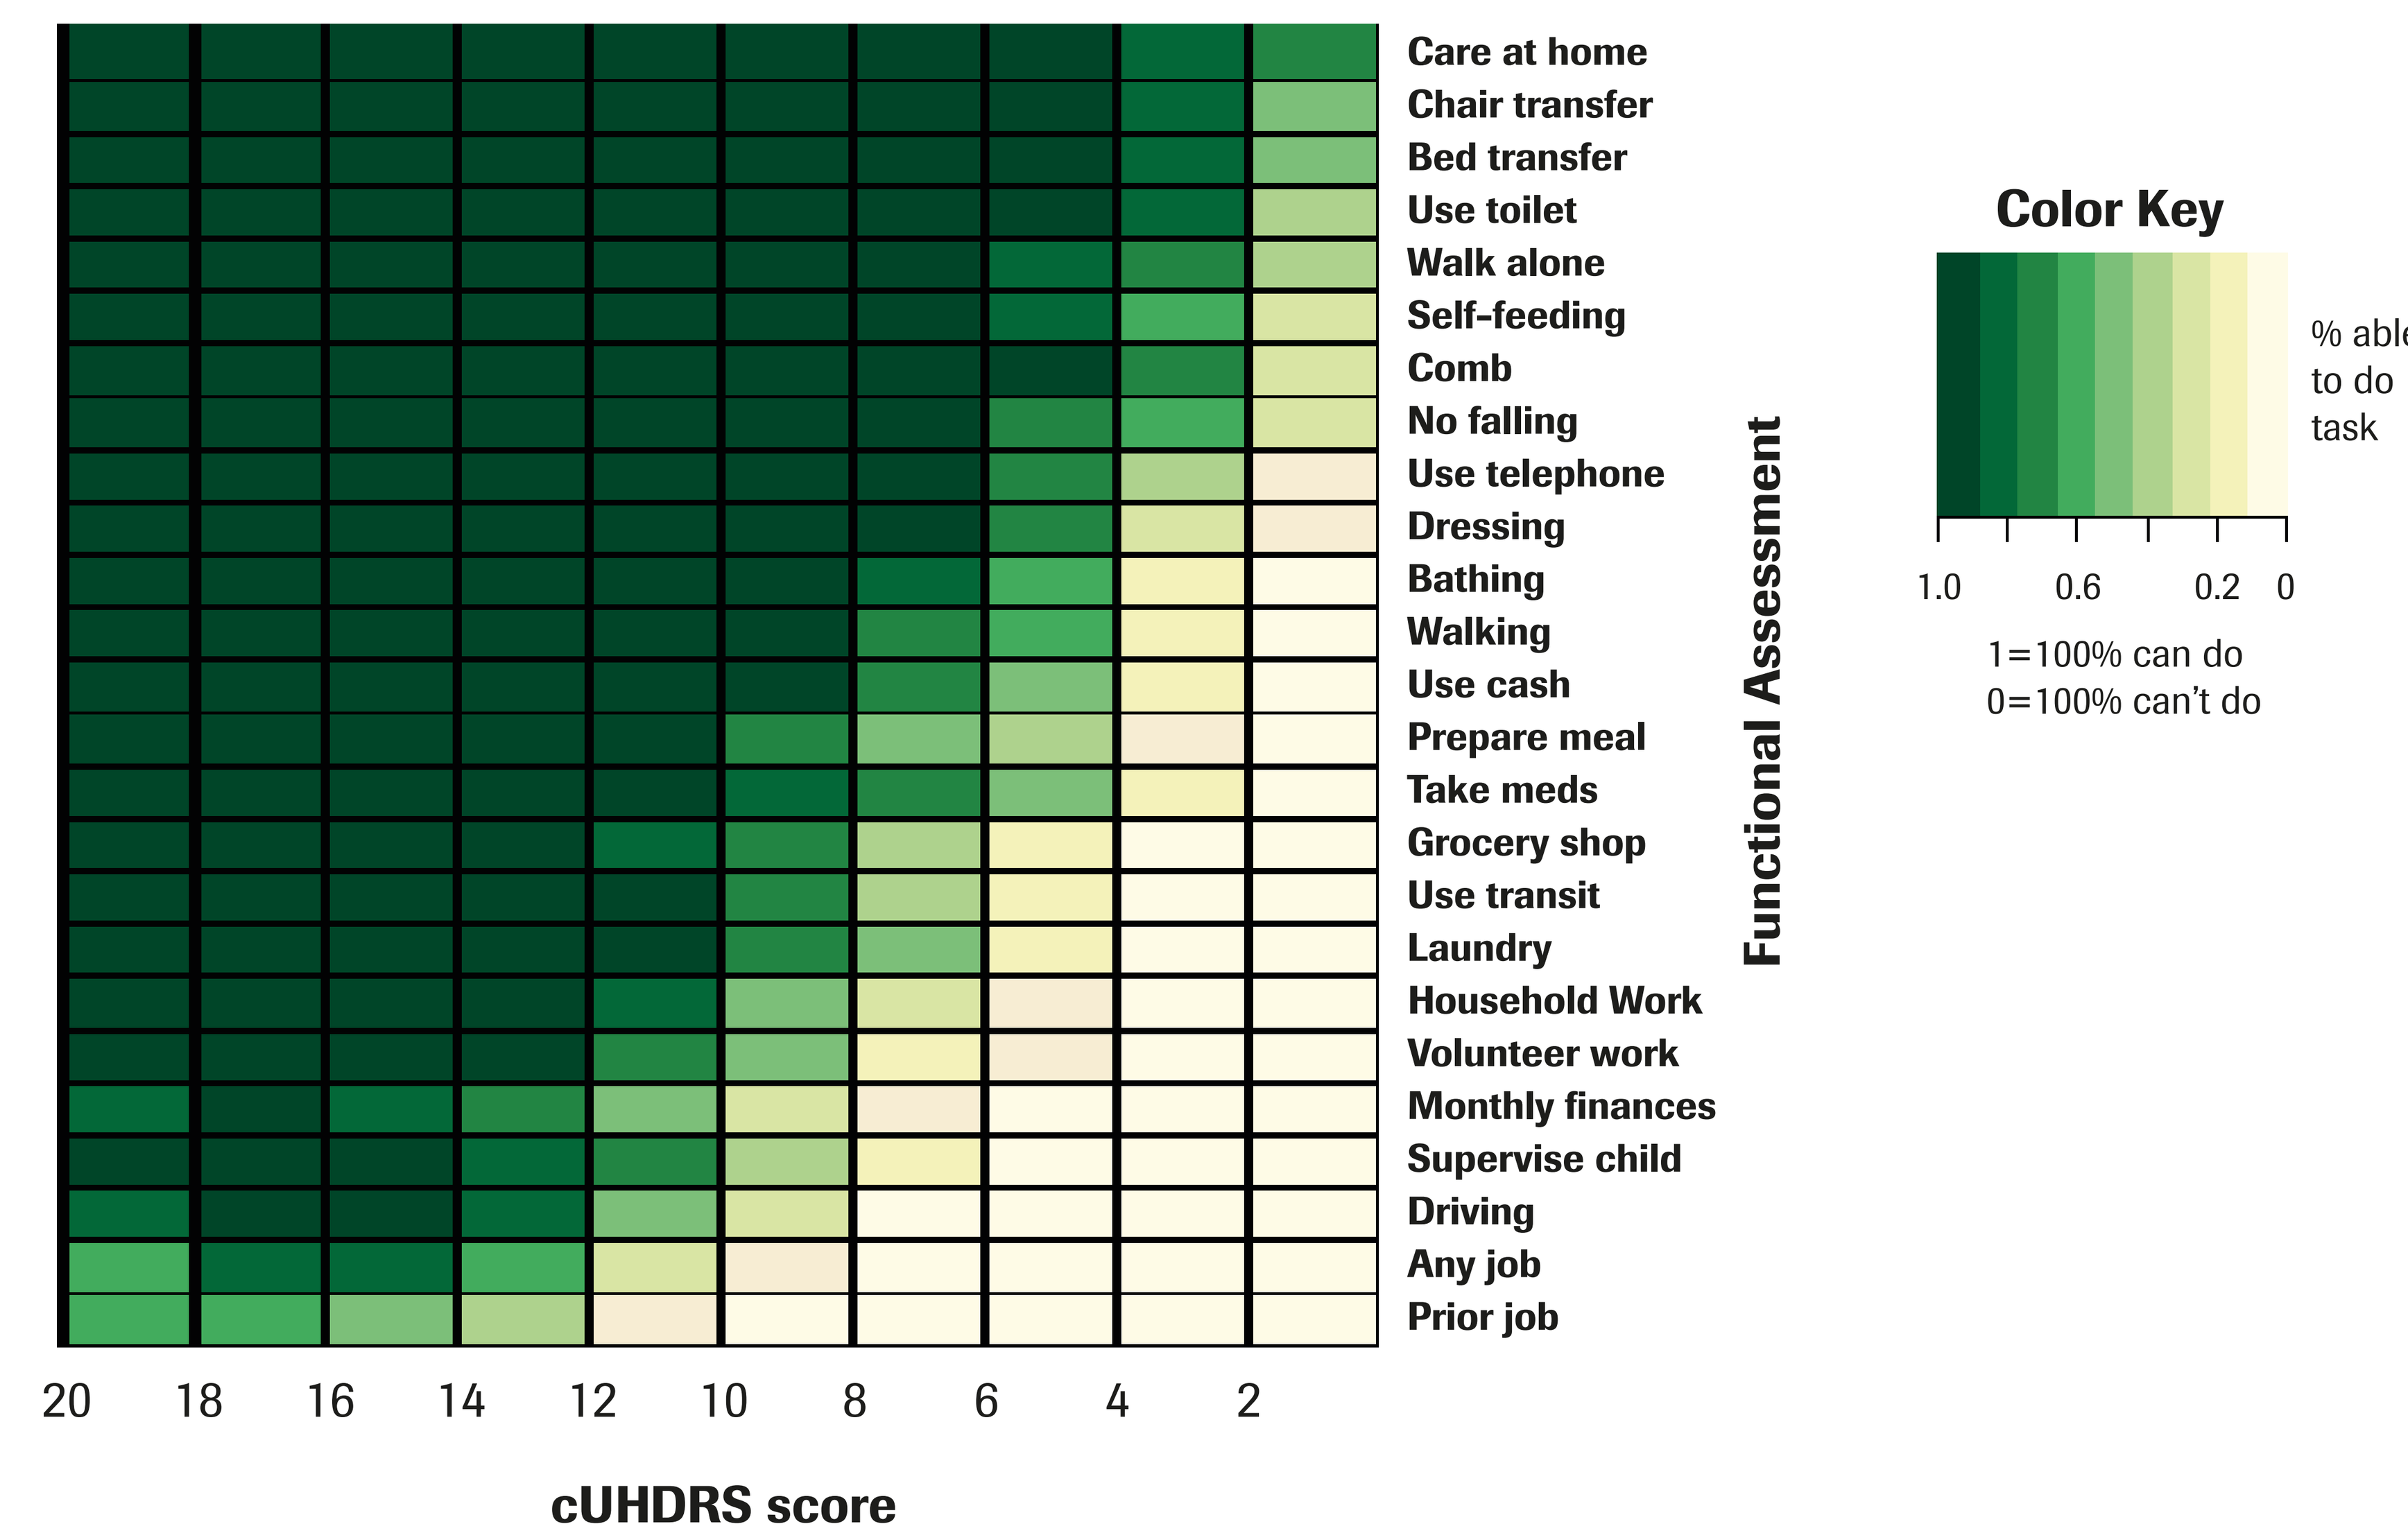
Care at home
Chair transfer
Bed transfer
Use toilet
Walk alone
Self-feeding
Comb
No falling
Use telephone
Dressing
Bathing
Walking
Use cash
Prepare meal
Take meds
Grocery shop
Use transit
Laundry
Household Work
Volunteer work
Monthly finances
Supervise child
Driving
Any job
Prior job
Functional Assessment
20 18 16 14 12 10 8 6 4 2
cUHDRS score
Color Key
% able to do task
1.0 0.6 0.2 0
1=100% can do
0=100% can't do

Meaningful change estimates for the individual scales and composite score

- Anchor-based estimates for cUHDRS are 1.22 (CGI-S) and 1.14 (IS); SEM=0.80.
- Anchor-based estimates for the constituent scales of the cUHDRS are shown in **(Table 2)**.
- A similar ratio to the denominators used to create z-scores was observed **(Table 2)**.
 — A meaningful change on each constituent scale will contribute a similar overall score change to the cUHDRS score (0.24–0.46 points).
 — If the threshold for the TFC is met but the thresholds for the constituent scales are missed by a single point each, the resultant cUHDRS score change is 1.16.
- A decline of 1.2 points on cUHDRS is recommended as an estimate of meaningful within-patient decline for use in a responder analysis endpoint in clinical trials in patients with early-to-moderate manifest HD.
- This estimate is:
 — approximately equal to anchor-based estimates and larger than SEM
 — larger than the cUHDRS score if only TFC (or, if all constituent scales but TFC) meets the threshold for worsening (this provides confirmation of clinical worsening through multiple scales; TFC is highlighted as it provides the greatest cUHDRS contribution per constituent scale point change).

Table 2: Anchor-based estimates of meaningful within-patient change and denominators used in creating z-scores of the constituent scales of the cUHDRS

	TFC	TMS	SDMT	SWR
Anchor-based estimates	1	8	3	6
Denominators	2.8	17.4	12.4	21.2
Ratio of denominators	1	6.2	4.4	7.6

Conclusions

- Qualitative research provides supportive evidence of the conceptual relevance of the constituent scales of the cUHDRS.
- The cUHDRS score has a clear association with functional ability.
- Meaningful change estimates have been calculated for the four constituent scales and the composite score.
- A worsening of 1.2 points on cUHDRS can be considered clinically meaningful for early-to-moderate manifest HD.

Acknowledgements

The HDSA is a non-profit organisation dedicated to improving the lives of the HD community.

Enroll-HD is a clinical research platform and longitudinal observational study for HD families intended to accelerate progress towards therapeutics. Enroll-HD incorporates data from REGISTRY; an observational study of the European Huntington's Disease Network (EHDN). The EHDN is a non-profit research network committed to advancing research and improving clinical care in HD. Enroll-HD and the EHDN are sponsored by the CHDI Foundation, a non-profit biomedical research organisation exclusively dedicated to collaboratively developing therapeutics for HD. These observational studies would not be possible without the vital contribution of the research participants and their families.

We thank all the patients who participate in our studies and their families. This study is funded by F. Hoffmann-La Roche. The authors thank Craig O'Hare, of MediTech Media UK for providing editorial support for this poster, which was funded by F. Hoffmann-La Roche in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

Abbreviations

CAP, CAG–age product score; CGI-S, Clinical Global Impression-Severity; cUHDRS; composite Unified Huntington's Disease Rating Scale; EHDN, European Huntington's Disease Network; FA, Functional Assessment; HD, Huntington's disease; HDSA, Huntington's Disease Society of America; IS, Independence Scale; SD, standard deviation; SDMT, Symbol Digit Modalities Test; SEM, standard error of the mean; SWR, Stroop Word Reading; TFC, Total Functional Capacity; TMS, Total Motor Score.

References

1. Bates GP, et al. *Nat Rev Dis Primers*. 2015; 1:15005;
2. Schobel S, et al. *Neurology*. 2017; 89:2495–2502;
3. Simpson JS, et al. *J Huntingtons Dis*. 2016; 5:395–403;
4. FDA: The Voice of the Patient Report – Huntington's Disease (March, 2016) https://www.fda.gov/downloads/ForIndustry/UserFees/PrescriptionDrugUserFee/UCM491603.pdf (Accessed April 2019);
5. Carlozzi NE, et al. *J Health Psychol*. 2013; 18: 212–225;
6. Roche: data on file;
7. Williams J, et al. *PLoS Curr*. 2011; 3:RRN1254.

Disclosures

- DT, GP and SS are employees of F. Hoffmann-La Roche.
- JL has received personal compensation from Wave Life Sciences USA Inc, Vaccinex Inc., Azevan Pharmaceuticals Inc., F. Hoffmann-La Roche, HSG (for uniQure biopharma B.V.) and Mitoconix Bio Limited.
- ST has undertaken consultancy services for F. Hoffmann-La Roche.
- BL has received personal compensation from F. Hoffmann-La Roche, Teva, uniQure, Nuredis, PTC therapeutics, sRNAlytics and Ionis Pharmaceuticals, personal compensation as Editor-in-Chief of the Journal of Huntington's Disease, and funding grants from Teva Pharmaceuticals, uniQure and Nuredis Pharmaceuticals.

Presented at the 2019 American Academy of Neuroscience Annual Meeting, Philadelphia, PA, USA; May 4–10, 2019