JEWELFISH: Risdiplam (RG7916) increases SMN protein in non-naïve patients with SMA

CA Chiriboga[1*], E Mercuri[2], D Fischer[3], D Kraus[4], WY Yeung[5], H Kletzl[4], M Gerber[4], Y Cleary[4], K Gorni[4] and O Khwaja[4]

1. Department of Neurology, Columbia University Medical Center, New York, NY, USA; 2. Paediatric Neurology and Nemo Center, Catholic University and Policlinico Gemelli, Rome, Italy; 3. Division of Neuropediatrics, University Children's Hospital Basel, University of Basel, Basel, Switzerland; 4. Roche Pharmaceutical Research and Early Development, Roche Innovation Center Basel, Basel, Switzerland; 5. Roche Products Ltd, Welwyn Garden City, UK

*cac3@cumc.columbia.edu

Background

- Spinal muscular atrophy (SMA) is a severe, progressive, inherited neuromuscular disease leading to loss of motor function and reduced life expectancy.[1]
- SMA is caused by reduced levels of the survival of motor neuron (SMN) protein due to deletions and/or mutations of the *SMN1* gene.[1]
 - A second SMN gene, *SMN2*, produces only low levels of functional SMN protein.[2]
- Increasing preclinical evidence indicates that SMA is a multisystem disorder.[3]
 - Therapies that increase SMN protein levels systematically may have broader therapeutic benefit than those targeting motor neurons alone
- Risdiplam (RG7916; RO7034067) is an orally administered, centrally and peripherally distributed small molecule that modulates *SMN2* pre-mRNA splicing to increase SMN protein levels.[4]

Study design

- JEWELFISH (NCT03032172)[5] is a multicenter, open-label study primarily evaluating the safety and tolerability of once-daily oral administration of risdiplam in patients aged from 6 months to 60 years with SMA who have previously enrolled in Study BP29420 (MOONFISH)[6] with the splicing modifier RO6885247 or received previous treatment with nusinersen or olesoxime.*

JEWELFISH SMA, Non-naïve, aged 6 months to 60 years old (N=125†)	Primary endpoints	• Safety. • PK: Mean plasma concentration, C_{max}, AUC and C_{trough} of risidplam and metabolites.
	Secondary endpoints	• PK–PD relationship (PD investigations will include analyses of *SMN2* mRNA splice forms and SMN protein).

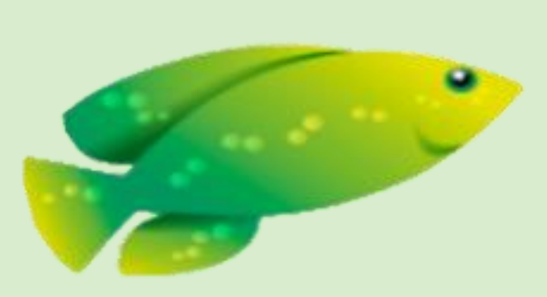

*Updated protocol is currently being submitted and approvals can be expected by Q4 2018–Q2 2019 at selected sites. Single dose level. †Target recruitment.

Patients in JEWELFISH show a diversity of copy number, SMA type and ambulatory status

	All patients (N=12)
Age at screening, years, median (range)	20 (13−52)
Gender, female/male, n (%)	3/9 (25.0/75.0)
Risdiplam treatment duration, median days (range)	303 (57–512)
SMA Type 2, n (%); Type 3, n (%)	6 (50); 6 (50)
SMN2 copy number, n (%)	
2	1 (8.3)
3	4 (33.3)
4	1 (8.3)
5	1 (8.3)
Unknown	5 (41.7)
Previous therapy, n (%)	
RO6885247 (MOONFISH)[6]	9 (75.0)
Nusinersen	3 (25.0)
Ambulatory, n (%)	3 (25.0)
Non-ambulatory, n (%)	9 (75.0)
MFM32 at baseline, median (range)	48.44 (2.1–84.4)

Data cut-off: 6 July 2018, intent-to-treat patients.

Risdiplam has been well tolerated in JEWELFISH

- To date, risdiplam has been well tolerated, and there have been no drug-related safety findings leading to withdrawal in any patient with SMA exposed to risdiplam.
- Safety data is available from 12 patients exposed to study drug from 57–512 days:
 - ophthalmological monitoring did not show any evidence of the retinal findings seen in preclinical monkey studies
 - 41 mild or moderate events were reported in 10 patients
 - the most frequent events were nasopharyngitis (5 events in 2 patients), pyrexia (3 events in 2 patients) and headache (3 events in 2 patients)
 - no SAEs were reported; no adverse trends were reported after a review of all available safety laboratory results, vital signs and ECG data
 - study treatment was not changed in response to any AEs

Data cut-off: 6 July 2018.

Supplementary information

Please scan using your QR reader application to access the graphs and data presented in this poster. NB: there may be associated costs for downloading data. These costs may be high if you are using your smartphone abroad. Please check your mobile data tariff or contact your service provider for more details. Alternatively this can be accessed at: http://bit.ly/2PJadyY

Risdiplam treatment leads to sustained increases in SMN protein levels in patients with Type 2/3 SMA

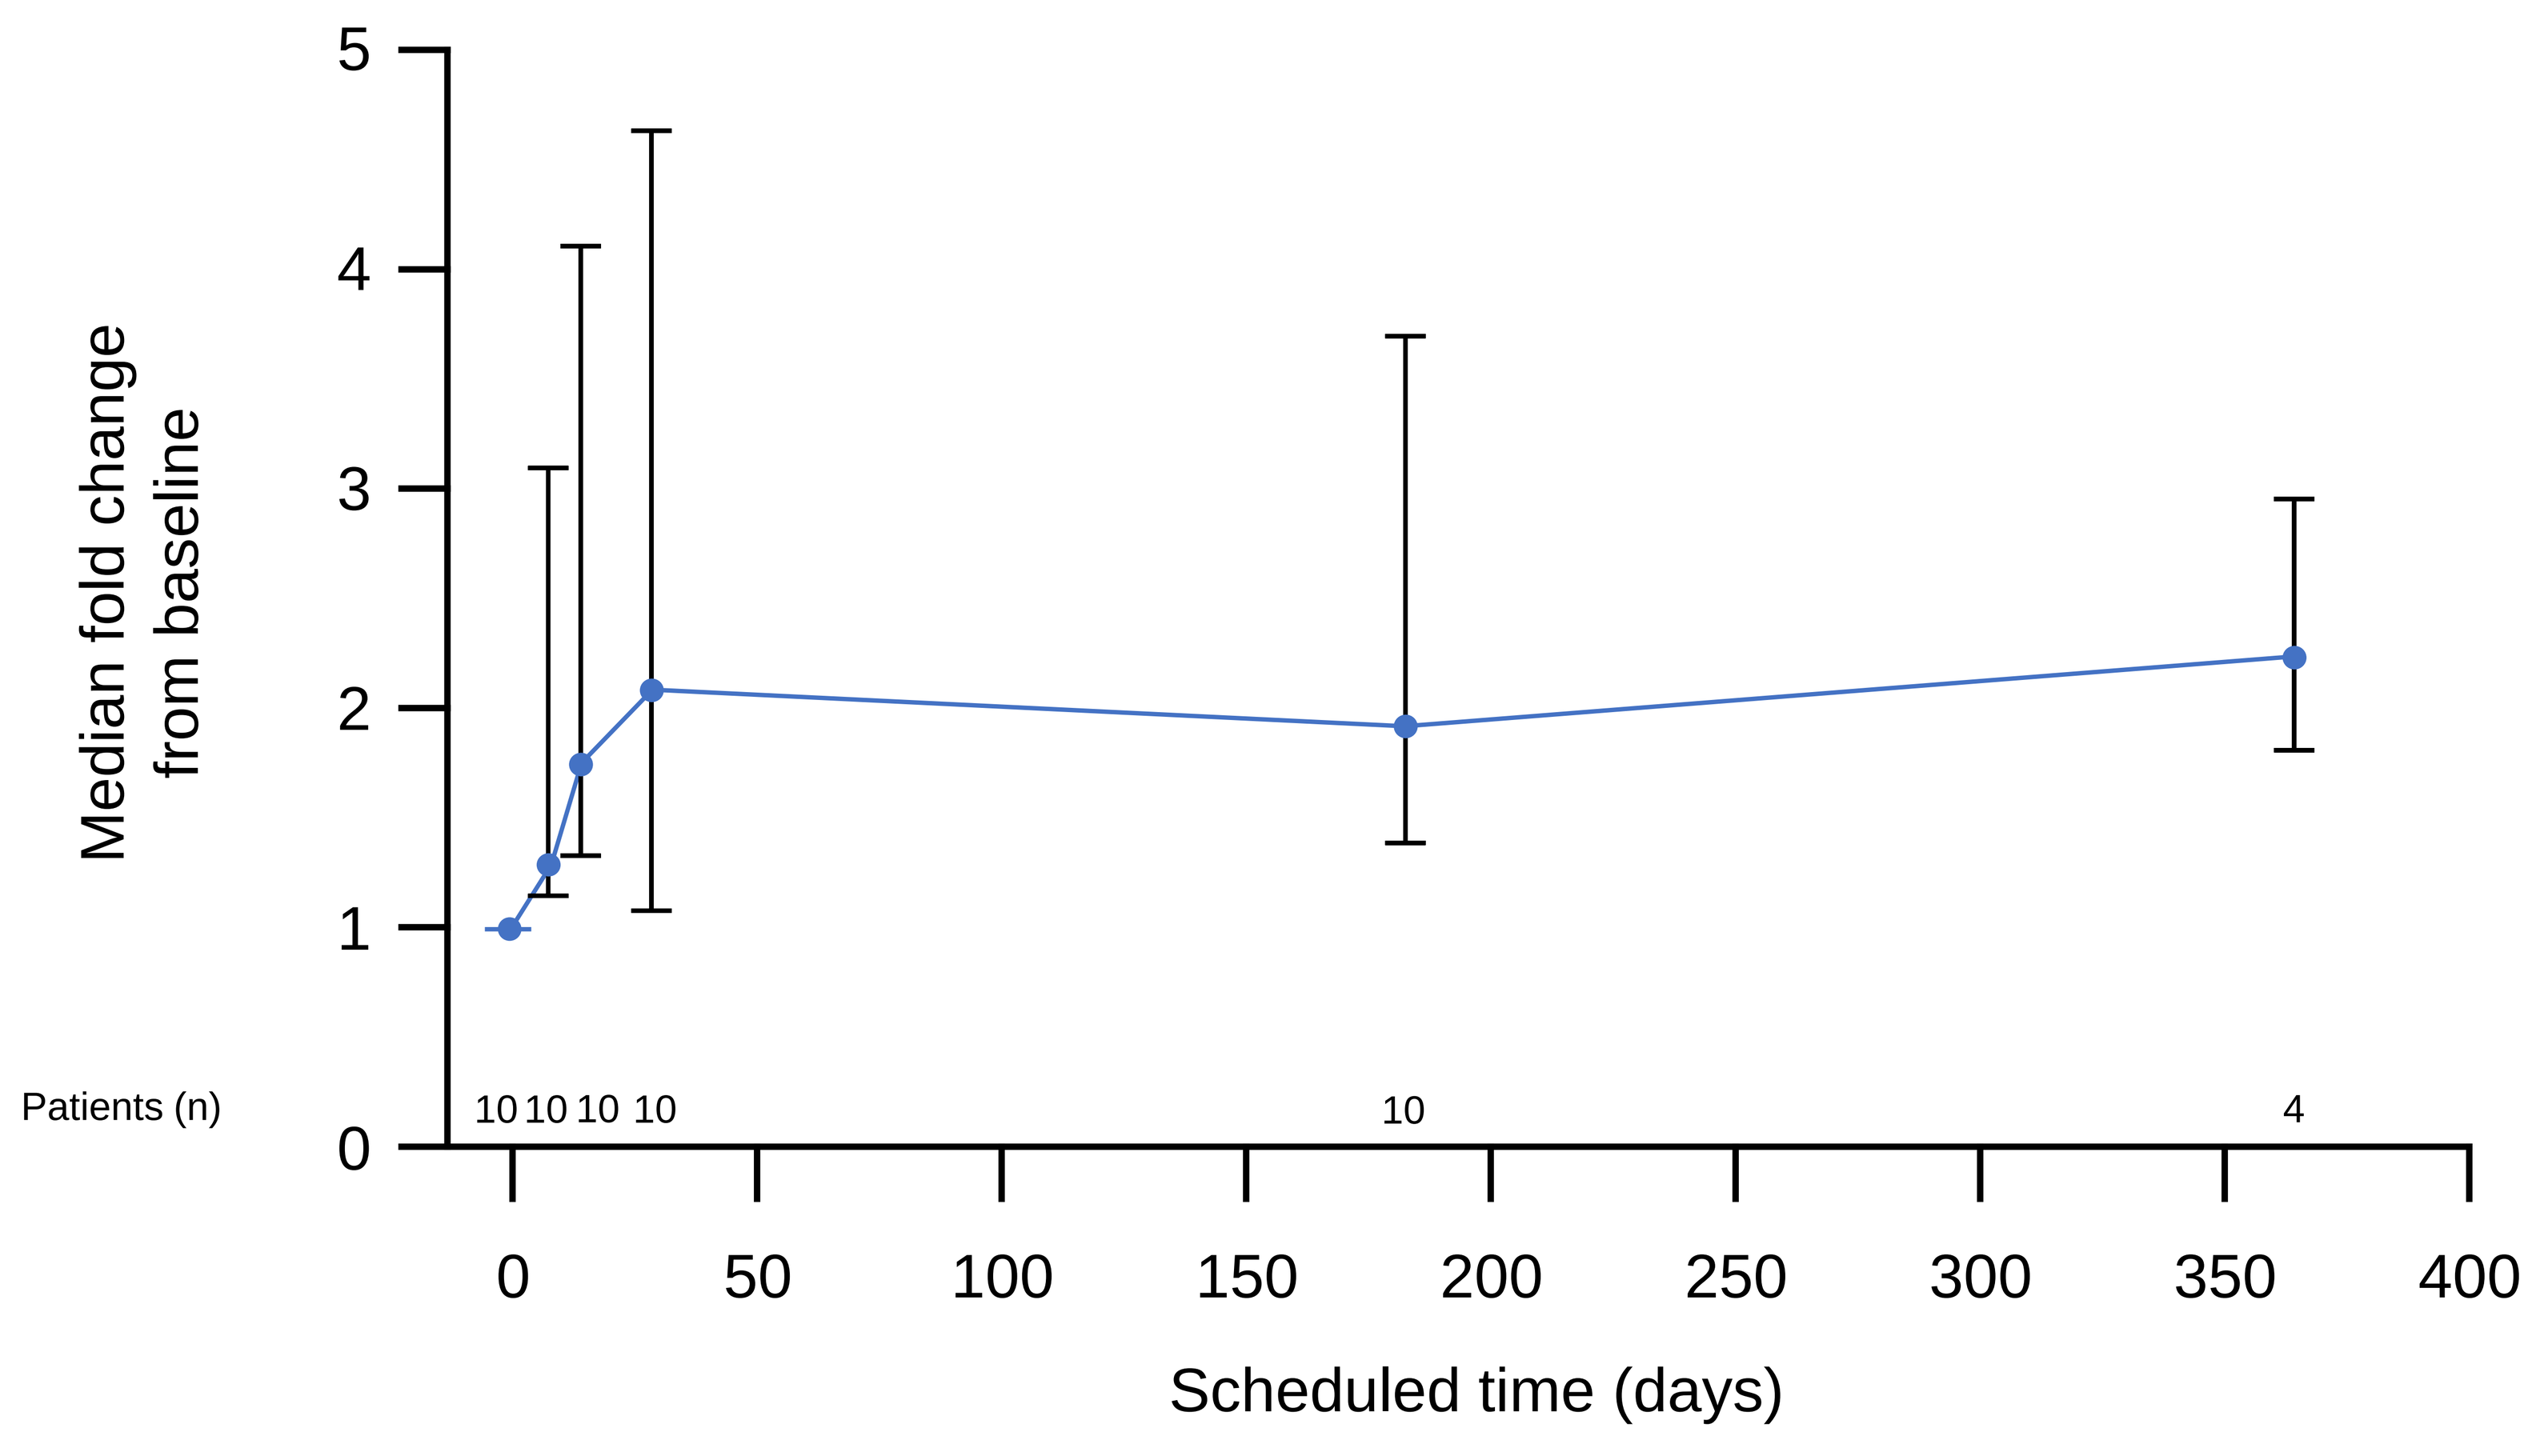
Median fold change from baseline
0
1
2
3
4
5
Patients (n)
10 10 10 10
10
4
0
50
100
150
200
250
300
350
400
Scheduled time (days)

Error bars represent minimum and maximum values. Data cut-off: 6 July 2018.

SMN protein levels in blood are similar in JEWELFISH and SUNFISH Part 1 following risdiplam exposure

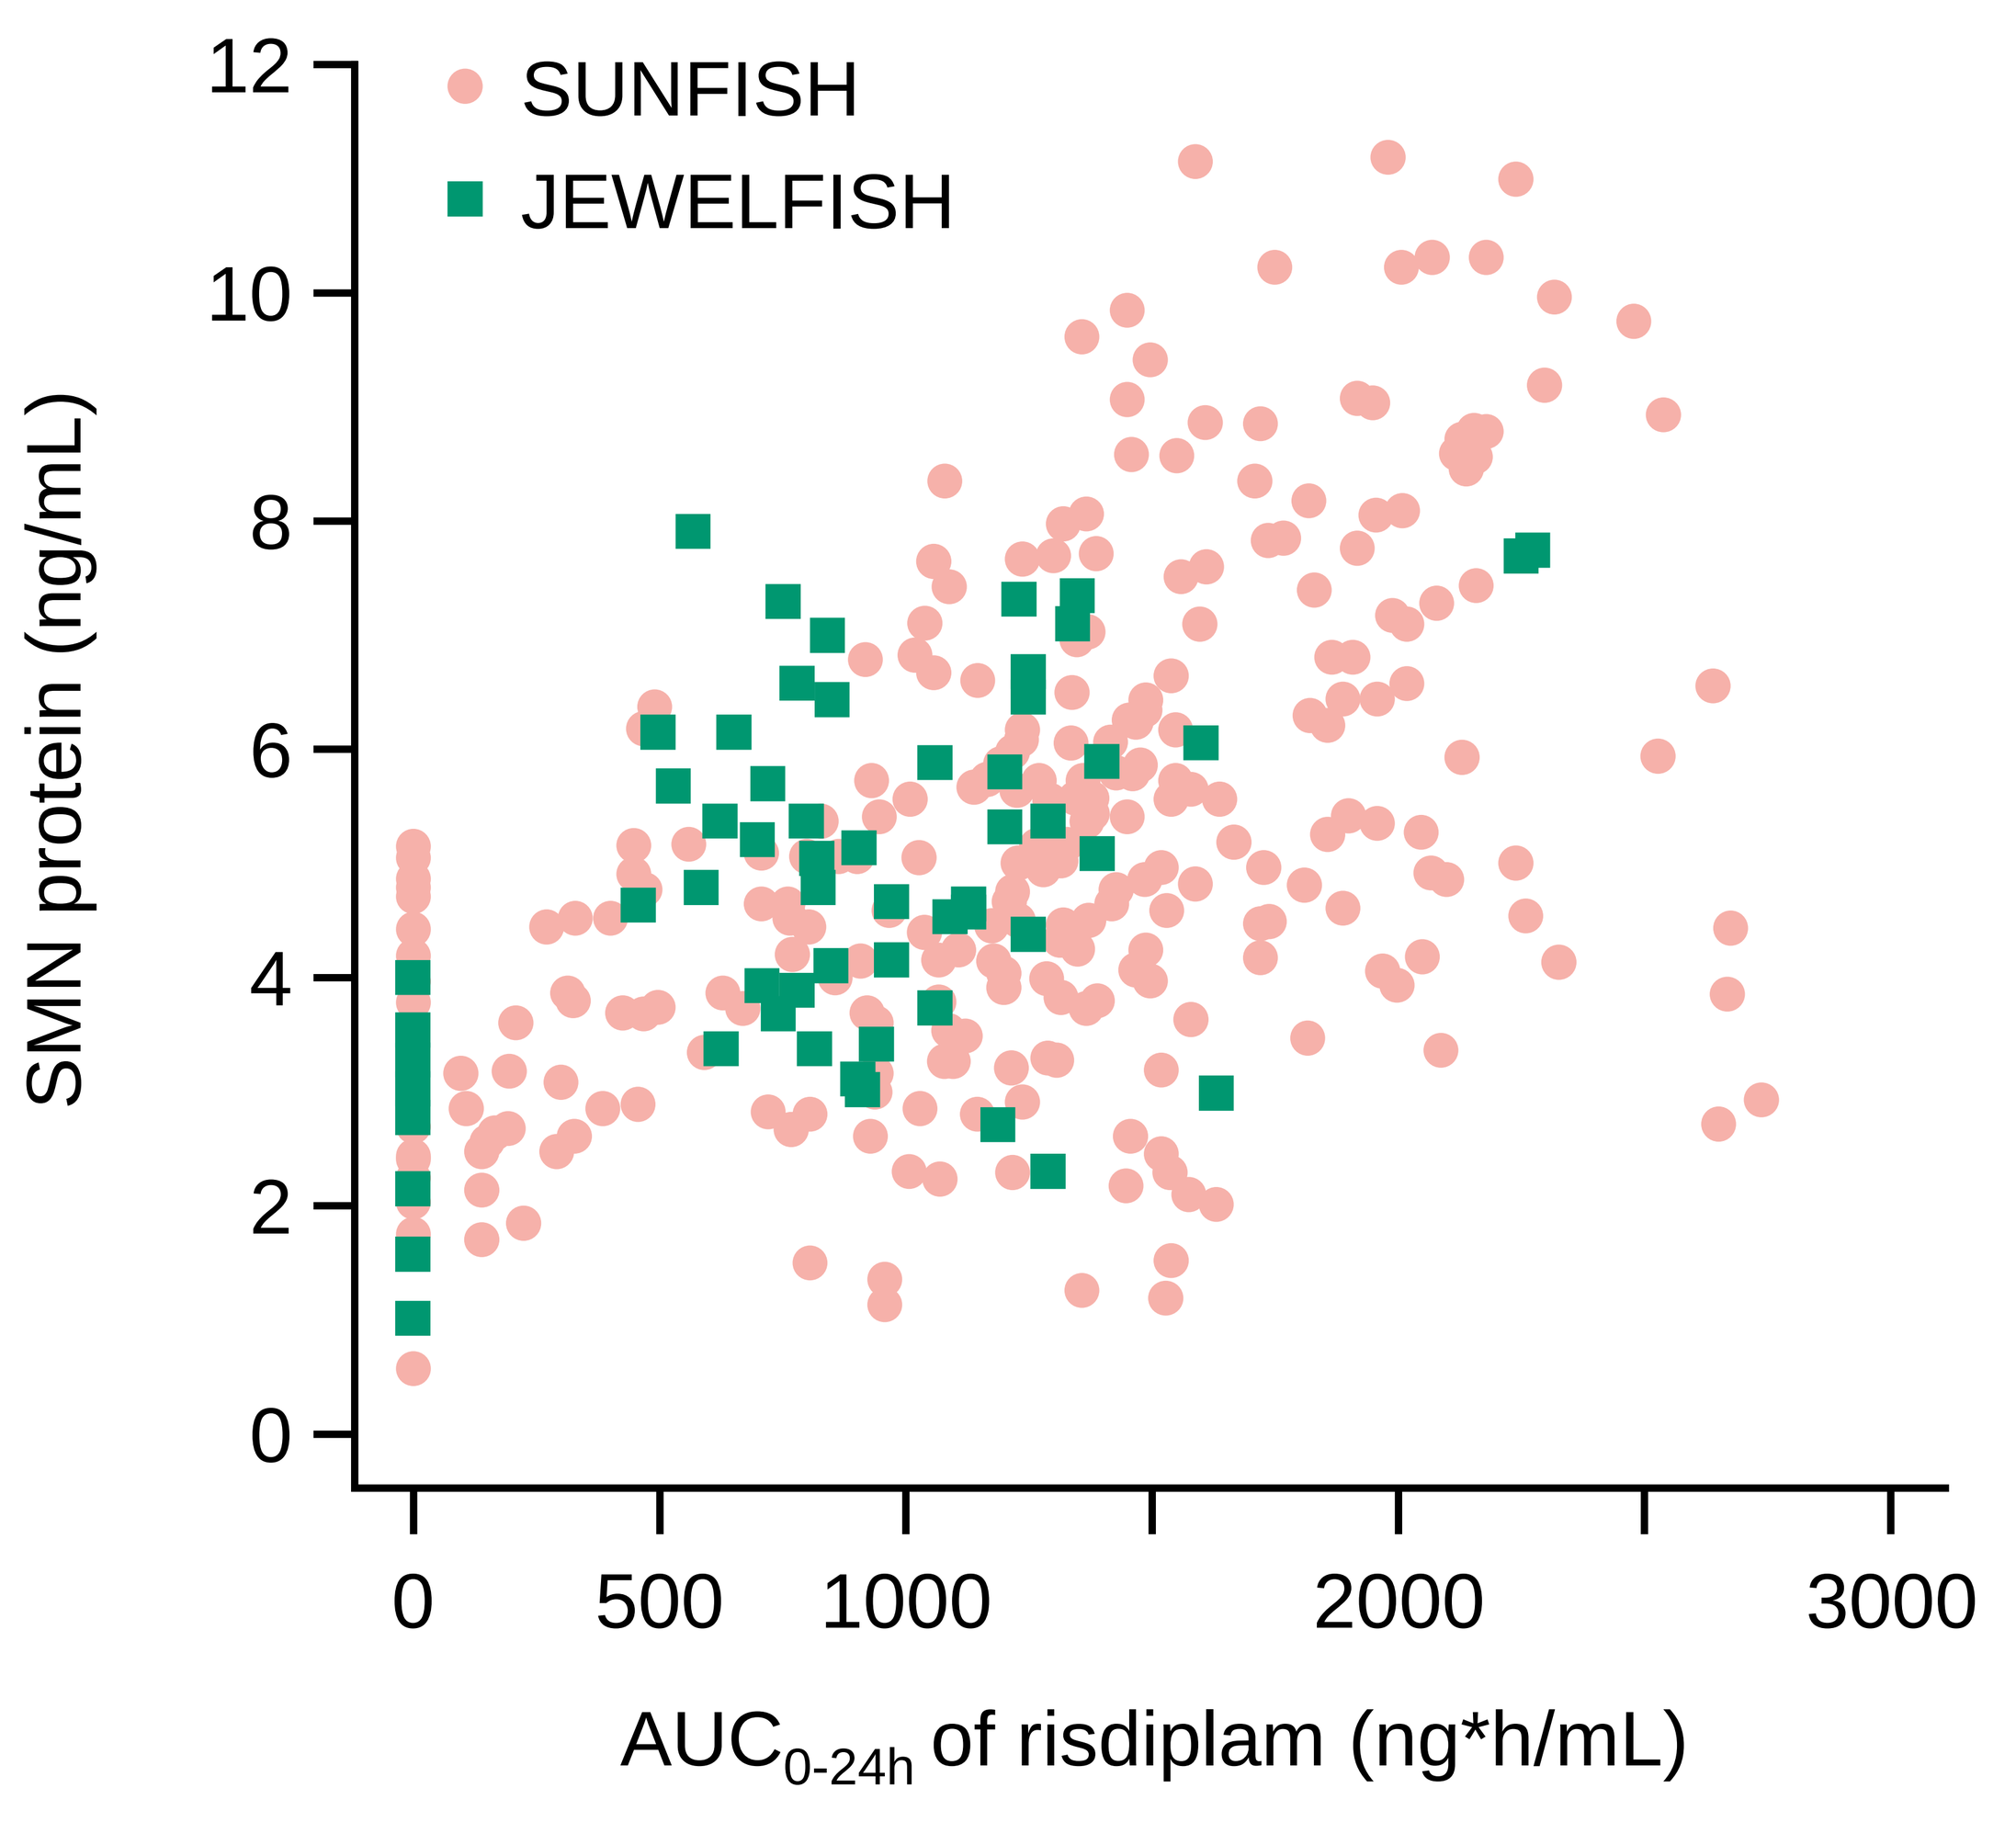
SUNFISH
JEWELFISH
SMN protein (ng/mL)
0
2
4
6
8
10
12
0
500
1000
2000
3000
AUC0-24h of risdiplam (ng*h/mL)

SUNFISH (NCT02908685) Part 1 is a study assessing the safety and efficacy of risdiplam in patients with Type 2 or 3 SMA aged 2–25 years.[7]

PD data showed a sustained >2-fold increase in median SMN protein versus baseline over 12 months of treatment

To date, there have been no drug-related AEs leading to withdrawal in any SMA patients exposed to risdiplam

JEWELFISH is currently recruiting in sites in Europe and the US.

Abbreviations

AEs, adverse events; $AUC_{0–24h}$, area under curve 0–24 hours; C_{max}, maximum observed plasma concentration; C_{trough}, trough plasma concentration; ECG, electrocardiogram; MFM32, motor function measure; mRNA, messenger ribonucleic acid; PD, pharmacodynamics; PK, pharmacokinetics; SAEs, serious adverse events; SMA, spinal muscular atrophy; SMN, survival of motor neuron.

Acknowledgments

We would like to thank the patients and their families for participation in these studies. This study is funded by F. Hoffmann La-Roche AG. The authors thank Paul Grimsey and Marianne Gerber of Roche Pharmaceutical Research and Early Development, Roche Innovation Center Basel, Basel, Switzerland for data analysis support. We would also like to thank our collaborators at PTC Therapeutics and the SMA Foundation. The authors thank Kayleigh Day, PhD, of MediTech Media, UK, for providing medical writing support, which was funded by F. Hoffmann La-Roche AG, Basel, Switzerland, in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

References

1. Mercuri E, et al. *Lancet Neurol*. 2012; 11:443–452;
2. Lorson CL, et al. *PNAS*. 1999; 96:6307–6311;
3. Hamilton G and Gillingwater TH. *Trends Mol Med.* 2013; 19:40–50;
4. Ratni H, et al. *J Med Chem*. 2018; 61:6501–6517;
5. ClinicalTrials.gov. NCT03032172 (Accessed Sep 2018);
6. ClinicalTrials.gov. NCT02240355 (Accessed Sep 2018);
7. ClinicalTrials.gov. NCT02908685 (Accessed Sep 2018).

Presented at the 23rd International Annual Congress of the World Muscle Society, Mendoza, Argentina, October 2–6, 2018

SUPPLEMENTARY INFORMATION – the contents below was not in the poster presented at WMS 2018 but is available via a QR code

JEWELFISH: Risdiplam (RG7916) increases SMN protein in non-naïve patients with SMA

CA Chiriboga[1*], E Mercuri[2], D Fischer[3], D Kraus[4], WY Yeung[5], H Kletzl[4], M Gerber[4], Y Cleary[4], K Gorni[4] and O Khwaja[4]

1. Department of Neurology, Columbia University Medical Center, New York, NY, USA; 2. Paediatric Neurology and Nemo Center, Catholic University and Policlinico Gemelli, Rome, Italy; 3. Division of Neuropediatrics, University Children's Hospital Basel, University of Basel, Basel, Switzerland; 4. Roche Pharmaceutical Research and Early Development, Roche Innovation Center Basel, Basel, Switzerland; 5. Roche Products Ltd, Welwyn Garden City, UK

*cac3@cumc.columbia.edu

Inclusion and exclusion criteria

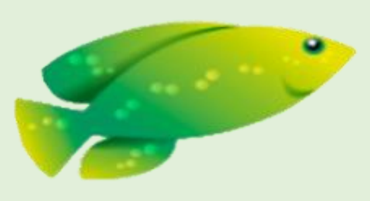 **JEWELFISH** SMA 6 months–60 years old	Key inclusion criteria	• Males and females 6 months to 60 years of age inclusive (at screening) • Confirmed diagnosis of 5q-autosomal recessive SMA, including: – genetic confirmation of homozygous deletion or heterozygosity predictive of loss of function of the *SMN1* gene – clinical history, signs, or symptoms attributable to SMA. • Previous enrollment in Study BP29420 (MOONFISH) with the splicing modifier RO6885247 or previous treatment with any of the following: – nusinersen (defined as having received ≥4 doses of nusinersen, provided that the last dose was received ≥90 days prior to screening) – olesoxime (provided that the last dose was received ≤12 months and ≥90 days prior to screening).
	Key exclusion criteria	• Concomitant participation in any investigational drug or device study • Participation in any investigational drug or device study, with the exception of studies with olesoxime or nusinersen, within 90 days of screening or five half-lives of the drug, whichever is longer • History of gene or cell therapy • Recently initiated treatment for SMA (<6 weeks prior to enrollment) with oral salbutamol or another beta 2-adrenergic agonist taken orally • Recent history (<1 year) of ophthalmologic disease
	Primary endpoints	• Safety • PK: – mean plasma concentration – C_{max} – AUC – C_{trough} of risdiplam and metabolites.
	Secondary endpoints	• PK–PD relationship (PD investigations will include analyses of *SMN2* mRNA splice forms and SMN protein)
	Exploratory endpoints	• Efficacy: – motor functions and milestones (MFM for patients 2–60 years, RULM for patients 2–60 years, 6MWT for patients 6–60 years, BSID-III for patients 6 months–2 years, HINE-2 for patients 6 months–2 years) – respiratory function – patient-reported (for patients 12–60 years) and caregiver-reported (for patients aged 2–60 years) SMAIS

Updated protocol is currently being submitted and approvals can be expected by Q4 2018–Q2 2019 at selected sites.

Abbreviations

6MWT, six-minute walk test; AUC, area under curve; BSID-III, Bayley Scales of Infant Development, Third Edition; C_{max}, maximum observed plasma concentration; C_{trough}, trough plasma concentration; HINE-2, Hammersmith Infant Neurological Examination Module 2; MFM, Motor Function Measure; PD, pharmacodynamics; PK, pharmacokinetics; RULM, Revised Upper Limb Module; SMA, spinal muscular atrophy; SMAIS, SMA independence Scale; SMN, survival of motor neuron.

Supplementary page

(This page provides additional online content and will not be included in the main poster. Attendees will be able to view and download this page by scanning the QR code on the main poster)