WAYFIND-R: Delivering a high-quality real-world data (RWD) global registry of patients diagnosed with a solid tumor and profiled with next-generation sequencing (NGS)

4094

Christophe Le Tourneau, Allan Hackshaw, Jean-Yves Blay, Jan Geissler, Clare Turnbull, Camille Perret, Olga Skatkova, Martina von Meyenn, Rodrigo Dienstmann

Supplement: Key variables to be collected

Key variables to be collected for each site

Laboratory and NGS test details

- Number of NGS tests performed per month
- Laboratory accreditation type, if applicable
- Type of NGS test (e.g. hotspot, CGP, WES, WGS)
- NGS technique (e.g. amplicon, hybrid capture)
- If a panel was used, number and names of genes analyzed
- For commercial or lab-developed tests, information on analytic and clinical validation

Key variables to be collected for each participant

Medical history

- Prior cancer history (if any)
- Initial diagnosis details of current cancer (e.g. cancer diagnosis and date of diagnosis, stage)
- Prior cancer biomarker results
- Past outcome assessment for current cancer and criteria used (e.g. RECIST, if applicable)
- Details of past treatment and therapies
- Past major procedures

Medical information at baseline (collected within 3 months of NGS test order)

- Socio-demographic information
- Family history of cancer
- Comorbidities
- Current cancer details (e.g. stage)
- Clinical measures (e.g. performance status)
- NGS sample details and NGS test results
- Cancer biomarker results
- Current cancer treatments and therapies, including means of access
- Diagnostic procedures
- Physician and MTB decision-making

Medical information at follow-ups (collected at least every 6 months)

- Changes in information collected at baseline
- Additional NGS test results
- Additional cancer biomarker results
- New/ongoing cancer treatments, therapies, and major procedures, including means of access
- Reason(s) for treatment discontinuation, if applicable
- Date and means of assessing disease progression
- Date of death
- Rationale for additional NGS testing and decision-making
- Adverse events that have a direct clinical consequence on the administered therapies, patients, or occurrence of pregnancies

Abbreviations: CGP, comprehensive genomic profiling; MTB, molecular tumor board; NGS, next-generation sequencing; RECIST, Response Evaluation Criteria In Solid Tumours; WES, whole-exome sequencing; WGS, whole-genome sequencing.

WAYFIND-R: Delivering a high-quality real-world data (RWD) global registry of patients diagnosed with a solid tumor and profiled with next-generation sequencing (NGS)

4094

Christophe Le Tourneau, Allan Hackshaw, Jean-Yves Blay, Jan Geissler, Clare Turnbull, Camille Perret, Olga Skatkova, Martina von Meyenn, Rodrigo Dienstmann

Supplement: Conflicts of interest

CLT has received grants/contracts from MSD, has received travel expenses from MSD, BMS, and AstraZeneca, has received honoraria from and performed a consulting/advisory role for BMS, MSD, Merck Serono, Roche, Nanobiotix, GSK, Rakuten, Seattle Genetics, and AstraZeneca, and has been paid by Roche as an external consultant. **AH** has held stock in Illumina and ThermoFisher (sold in 2020), has received grants/contracts from Roche, MSD, Takeda, BMS, Boehringer Ingelheim, and Celgene (several pharmaceutical companies provided research funding for UCL-sponsored trials), has received honoraria from AbbVie, Boehringer Ingelheim, Takeda, AstraZeneca, Daiichi Sankyo, Merck Serono, Merck/MSD, UCB, and Roche for delivering general education/training in clinical trials, and has performed a consulting/advisory role for AbbVie, Roche, and GRAIL, Inc. **J-YB** has received grants/contracts from Novartis, Bayer, GSK, Pfizer, Deciphera, Roche, BMS, MSD, and AstraZeneca, has a leadership role with Innate Pharma, has received honoraria from Novartis, Bayer, Pfizer, Deciphera, and Roche, and has performed a consulting/advisory role for Novartis, Bayer, GSK, Pfizer, Deciphera, Roche, BMS, MSD, and AstraZeneca. **JG** has received grants/contracts from Novartis, Incyte, BMS, Pfizer, and Takeda, has received travel expenses from Amgen, Alnylam, BioMarin, Novartis, Pfizer, Roche, Servier, and UCB, and has performed a consulting/advisory role for Alnylam, Bayer, Boehringer Ingelheim, BioMarin, Daiichi Sankyo, Gilead, Janssen, Novartis, Pfizer, Roche, Servier, Sanofi, Sobi, and UCB. **CT** has received honoraria from AstraZeneca, Roche, and Pfizer, has performed a consulting/advisory role for AstraZeneca and Roche, and has attended a speaker’s bureau for AstraZeneca. **CP** is an employee of and holds stocks in F. Hoffmann-La Roche Ltd. **OS** is an employee of and holds stocks in F. Hoffmann-La Roche Ltd. **MvM** is an employee of and holds stocks in F. Hoffmann-La Roche Ltd. **RD** is an employee of Oncoclínicas Grupo, has business ownership in Trialing, has received a grant/contract from Merck, has performed a consulting/advisory role for Roche and Boehringer Ingelheim, and has attended speakers’ bureaus for Roche, Ipsen, Sanofi, MSD Oncology, Servier, Amgen, and Libbs.