Abstract #4585
Poster #193

Clinical Value of Atezolizumab + Bevacizumab for First-Line Unresectable Hepatocellular Carcinoma (HCC): A Network Meta-Analysis

Arndt Vogel,[1] Lorenza Rimassa,[2,3] Huichuan Sun,[4] Ghassan K. Abou-Alfa,[5,6] Anthony El-Khoueiry,[7] David James Pinato,[8] Monica Daigl,[9] Javier Sanchez Alvarez,[9] Panos Orfanos,[9] Michael Leibfried,[10] Marie-Hélène Blanchet Zumofen,[9] Vincent Gaillard,[9] Philippe Merle[11]

[1]Hannover Medical School, Hannover, Germany; [2]Department of Biomedical Sciences, Humanitas University, Pieve Emanuele (Milan), Italy; [3]Medical Oncology and Hematology Unit, Humanitas Cancer Center, Humanitas Clinical and Research Center-IRCCS, Rozzano (Milan), Italy; [4]Zhongshan Hospital, Fudan University, Shanghai, China; [5]Memorial Sloan Kettering Cancer Center, New York, NY, US; [6]Weill Medical College at Cornell University, New York, NY, US; [7]USC Norris Comprehensive Cancer Center, USC, Los Angeles, CA, US; [8]Department of Surgery and Cancer, Imperial College London, London, UK; [9]F. Hoffmann-La Roche Ltd., Basel, Switzerland; [10]Genentech Inc., South San Francisco, CA, US; [11]Hospital De La Croix-Rousse, Lyon, France

BACKGROUND

- Sorafenib and lenvatinib are the standards of care for patients with advanced HCC[1,2]
- Single-agent anti–programmed death-1 (anti–PD-1) checkpoint inhibitor nivolumab failed to show a superior overall survival (OS) benefit vs sorafenib in the CheckMate459 trial[3]
- Combination atezolizumab + bevacizumab showed statistically significant and clinically meaningful OS and progression-free survival (PFS) benefits compared with sorafenib in the IMbrave150 trial[4]
- Bevacizumab was evaluated as a single agent in an HCC population not screened for varices.[5] The synergy of atezolizumab + bevacizumab is based on their complementary effects as immuno-modulators and mutual reinforcements at different steps in the cancer immunity cycle[6,7]
- In the absence of head-to-head clinical trials, we conducted a network meta-analysis (NMA) to compare the efficacy and safety of atezolizumab + bevacizumab, lenvatinib and nivolumab using sorafenib as a common comparator

METHODS

Systematic Literature Review

- A systematic literature review identified randomized controlled trials in adults (≥ 18 years) with locally advanced or metastatic HCC and no prior systemic HCC therapy
 - Primary sources included MEDLINE, Embase and the Cochrane Library searched through May 28, 2019
 - Secondary sources included presentations at relevant scientific congresses (2016-2019), references in screened publications, health technology assessment organization reports and the International Clinical Trials Registry Platform
- Studies of therapies approved for any line of HCC treatment with data reported for first-line treatment since sorafenib approval in 2007 were eligible for inclusion
- A total of 8783 records were screened, 55 trials were reviewed and 9 trials were included in the overall evidence network; 3 studies of atezolizumab + bevacizumab, lenvatinib or nivolumab vs sorafenib were included in this NMA (Figure 1)

Figure 1. Evidence Network Based on Included Studies[2-4]

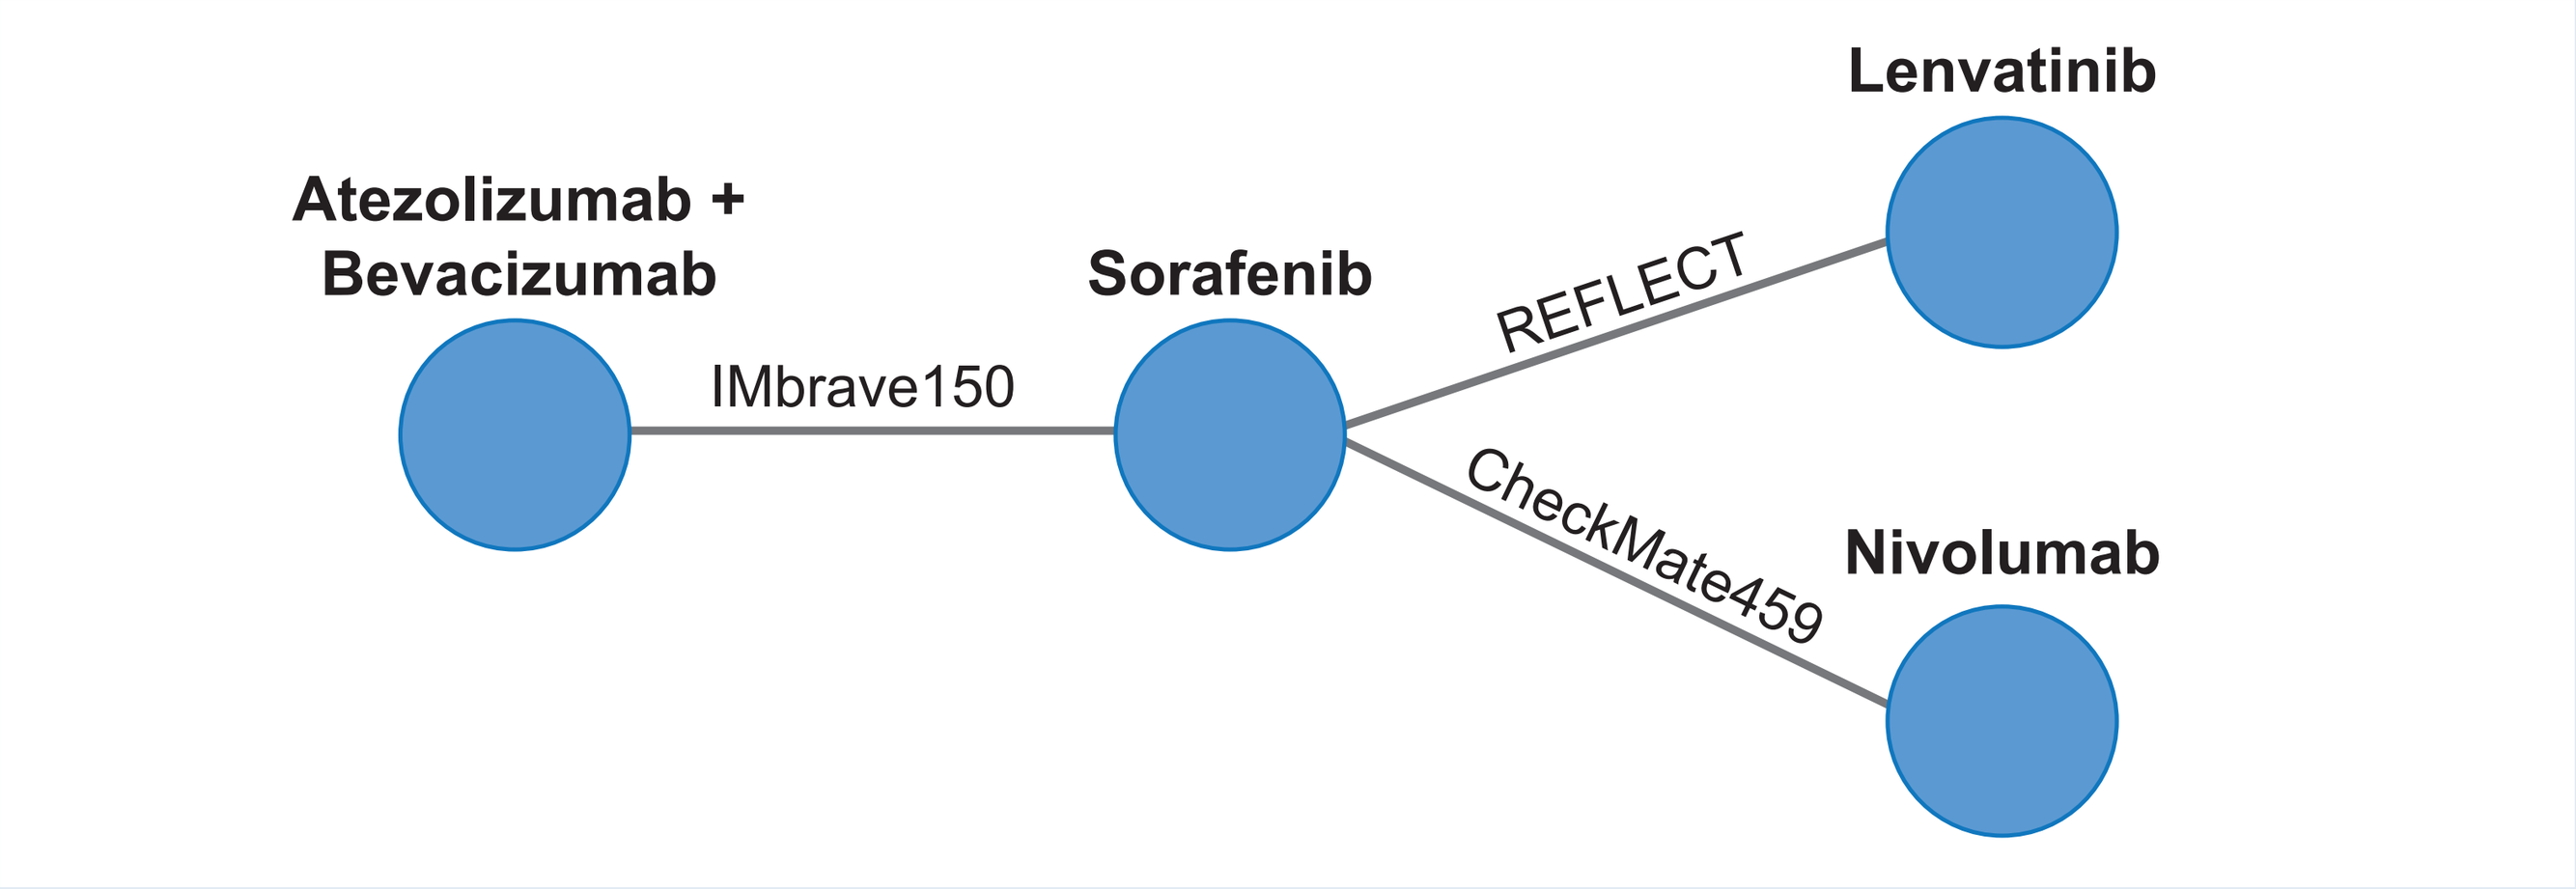
Atezolizumab + Bevacizumab
Sorafenib
Lenvatinib
Nivolumab
IMbrave150
REFLECT
CheckMate459

Statistical Analysis

- The base case NMA compared the relative efficacy of atezolizumab + bevacizumab vs sorafenib observed in the IMbrave150 study with that of lenvatinib vs sorafenib in REFLECT and of nivolumab vs sorafenib in CheckMate-459
- Reported hazard ratios (HRs) for OS and PFS were extracted from published studies
- A generalized linear model with random effects was used to estimate indirect treatment effects
 - Informative priors for the heterogeneity of treatment effects across trials were adopted given the limited number of trials to inform each pairwise comparison[8]
 - HRs with 95% credible intervals (CrIs) and Bayesian posterior probability of atezolizumab + bevacizumab being superior to other treatments were calculated for each treatment comparison
- Analyses were performed to assess subgroups based on macrovascular invasion (MVI) and extrahepatic spread (EHS), etiology, and geography (Asia-Pacific region vs rest of world); results from all-comers were compared with those of the subgroups to confirm the findings were robust and insensitive to subgroup specifications
- Descriptive statistics summarized safety outcomes reported from IMbrave150 and REFLECT because CheckMate-459 did not report comparable statistics for adverse event (AEs)

RESULTS

Study Populations

- The study populations of IMbrave150, REFLECT and CheckMate-459 were considered sufficiently similar to be compared in the quantitative analysis (Table 1)
- Age was generally similar across trials, with a slightly greater proportion of patients aged ≥ 65 years in IMbrave150 than in REFLECT
- The REFLECT trial included a larger proportion of patients from the Asia-Pacific region
- CheckMate-459 included a larger proportion of patients with non-viral etiology
- A greater proportion of patients in IMbrave150 had MVI compared with REFLECT (not reported from CheckMate459); however, the proportions of patients with MVI and/or EHS were similar across trials
- Of note, REFLECT excluded patients with main portal vein trunk or bile duct invasion and those with > 50% liver involvement; these patients comprised approximately 20% of the IMbrave150 population

Table 1. Key Patient Characteristics From IMbrave150, REFLECT and CheckMate-459[2-4]

	IMbrave150[4]		REFLECT[2]		CheckMate-459[3]	
	Atezo + Bev	**Sorafenib**	**Lenvatinib**	**Sorafenib**	**Nivolumab**	**Sorafenib**
Patients, n	336	165	478	476	371	372
Male, %	82	83	85	84	85	85
Median age, y	64	66	63	62	65	65
Age ≥ 65 y, %	**48**	**55**	44	41	Not reported	
White, %	37	32	28	30	Not reported	
Asian, %	56	58	**70**	**68**	Not reported	
Asia-Pacific region, %	40[a]	41[a]	**67**	**67**	40	40
ECOG PS 1, %	38	38	36	37	27	30
AFP ≥ 200 ng/mL, %	43	45	46	39	39	43
Hepatitis B, %	49	46	53	48	31	31
Hepatitis C, %	21	22	19	26	23	23
Non-viral etiology, %	30	32	28	26	**45**	**45**
MVI and/or EHS	77	73	69	71	75	70
MVI, %	**38**	**43**	**23**	**19**	Not reported	
EHS, %	63	56	61	62	Not reported	
Prior radiotherapy, %	10	10	10	13	12	11
Prior local therapy, %	52	48	Not reported		51	56

AFP, alpha-fetoprotein; ECOG PS, Eastern Cooperative Oncology Group performance status; EHS, extrahepatic spread; MVI, macrovascular invasion.
[a] Does not include patients from Japan.

Overall Survival

- Base case analysis suggested improved OS benefit with atezolizumab + bevacizumab and > 90% probability of atezolizumab + bevacizumab offering superior OS benefit vs lenvatinib, nivolumab or sorafenib (Table 2)
- Sensitivity analyses were generally consistent with the primary findings

Table 2. Indirect OS Comparisons

Comparison	Hazard Ratio (95% CrI)	Probability of Atezo + Bev Superiority[a]
Atezo + bev vs **lenvatinib**	0.63 (0.32, 1.25)	93.7%
Atezo + bev vs **nivolumab**	0.68 (0.35, 1.38)	90.3%
Atezo + bev vs **sorafenib**[b]	0.58 (0.35, 0.99)	97.6%

[a] Based on Bayesian posterior probability.
[b] Based on the results of this NMA. These are not the results of direct comparison in the clinical trial.

Progression-Free Survival

- Base case analysis suggested improved PFS benefit with atezolizumab + bevacizumab and > 85% probability of atezolizumab + bevacizumab offering superior PFS benefit vs nivolumab or sorafenib (Table 3)
- Sensitivity analyses were not feasible because subgroup results for PFS according to RECIST 1.1 were not reported for REFLECT or CheckMate-459

Table 3. Indirect PFS Comparisons

Comparison	Hazard Ratio (95% CrI)	Probability of Atezo + Bev Superiority[a]
Atezo + bev vs **lenvatinib**	0.91 (0.23, 3.65)	61.5%
Atezo + bev vs **nivolumab**	0.63 (0.16, 2.59)	85.5%
Atezo + bev vs **sorafenib**[b]	0.59 (0.23, 1.58)	92.3%

[a] Based on Bayesian posterior probability.
[b] Based on the results of this NMA. These are not the results of direct comparison in the clinical trial.

Safety

- Nearly all patients reported an AE in both REFLECT and IMbrave150
- In REFLECT, a greater proportion of patients receiving lenvatinib experienced Grade ≥ 3 AEs than those receiving sorafenib
- In IMbrave150, the proportions of patients with Grade ≥ 3 AEs in the atezolizumab + bevacizumab and sorafenib groups were similar (Table 4)
- AEs among patients receiving sorafenib appeared to be similar across trials

Table 4. Adverse Events Reported in IMbrave1504 and REFLECT[2]

	IMbrave150[4]		REFLECT[2]	
Patients, n (%)	**Atezo + Bev (n = 329)**	**Sorafenib (n = 156)**	**Lenvatinib (n = 476)**	**Sorafenib (n = 475)**
Any AE	323 (98.2)	154 (98.7)	470 (98.7)	472 (99.4)
Serious AE	125 (38.0)	48 (30.8)	205 (43.1)	144 (30.3)
AE of Grade ≥ 3	201 (61.1)	95 (60.9)	357 (75.0)	316 (66.5)
AE leading to discontinuation of any treatment component[a]	51 (15.5)[b]	16 (10.3)	63 (13.2)	43 (9.1)

[a] Data from IMbrave150 included discontinuations related to atezolizumab or bevacizumab or both.
[b] Includes AE leading to discontinuation from any treatment component; 23 of 329 patients (7%) experienced an AE leading to discontinuation from both components.

STRENGTHS AND LIMITATIONS

- This NMA included comparable study populations in a common evidence network for indirect comparison of relative efficacy data from clinical trials
- Although NMAs are insensitive to differences in prognostic factors across the trials that constitute the network, any differences in predictive factors (i.e., factors that impact the efficacy of the compounds being compared) can impact the findings; imbalances in any unknown predictive factors should be considered in the interpretation of this NMA
- As opposed to naively comparing individual arms from different trials, the NMA approach respects the randomization of the underlying trials; however, the NMA itself is not a randomized study and, therefore, the results are exploratory in nature

SUMMARY

- This NMA suggested improved OS and PFS benefits with first-line combination atezolizumab + bevacizumab vs lenvatinib, nivolumab or sorafenib
- The reported safety profiles were generally similar for atezolizumab + bevacizumab and lenvatinib; however, types and classification of AEs, impact on the patient experience and differences in follow-up time and treatment exposure were not considered in this descriptive safety summary
- In the absence of head-to-head clinical trials between atezolizumab + bevacizumab and lenvatinib or nivolumab, these indirect comparisons may help clinicians and population health managers consider the relative efficacy and safety of these first-line treatment options for patients with unresectable HCC
- Atezolizumab and bevacizumab should be considered a new standard of care in patients with unresectable HCC[9]

REFERENCES

1. Bouattour M, et al. *Liver Cancer.* 2019;8:341-358.
2. Kudo M, et al. *Lancet*. 2018;391(10126):1163-1173.
3. Yau T, et al. *Ann Oncol*. 2019;30(suppl 5):v851-v934.
4. Finn RS, et al. *N Engl J Med.* 2020;382(20):1894-1905.
5. Chen DS, Mellman I. *Immunity*. 2013;39(1):1-10.
6. Chen DS, Hurwitz H. *Cancer J*. 2018;24(4):193-204.
7. Siegel AB, et al. *J Clin Oncol*. 2008;26:2992-2998.
8. Turner RM, et al. *Stat Med*. 2015;34(6):984-998.
9. National Comprehensive Cancer Network. Hepatobiliary Cancers. Version 2.2020.

CONTACT INFORMATION

- For questions, please email Dr. Vogel at Vogel.Arndt@mh-hannover.de

ACKNOWLEDGMENTS

- This study is sponsored by F. Hoffmann-La Roche, Ltd
- Medical writing support for this poster was provided by Jeff Frimpter, MPH, of Health Interactions and funded by F. Hoffmann-La Roche, Ltd

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission from ASCO® and the author of this poster

2020 American Society of Clinical Oncology (ASCO) Annual Meeting – 29 May-31 May, 2020, Virtual Scientific Program