San Antonio Breast Cancer Symposium® – December 5–9, 2023

Circulating tumor DNA dynamics in acelERA Breast Cancer: a Phase II study of giredestrant for estrogen receptor-positive, HER2-negative, previously treated advanced breast cancer

PO1-05-07

Ann Collier[1], Aditya Bardia[2], Joohyuk Sohn[3], Elgene Lim[4], Mariana Chavez-MacGregor[5], Miguel Martín[6], Jorge Martinalbo[7], Pablo D. Pérez-Moreno[1], Heather M. Moore[1]

[1]Genentech, Inc., South San Francisco, CA; [2]Massachusetts General Hospital, Harvard Medical School, Boston, MA; [3]Yonsei University College of Medicine, Seoul, Republic of Korea; [4]Garvan Institute of Medical Research, St Vincent's Clinical School, University of New South Wales, Darlinghurst, NSW, Australia; [5]The University of Texas MD Anderson Cancer Center, Houston, TX; [6]Hospital Gregorio Marañón, Universidad Complutense, GEICAM, CIBERONC, Madrid, Spain; [7]F. Hoffmann-La Roche Ltd, Basel, Switzerland

BACKGROUND

- A common mechanism of resistance to endocrine therapy is the presence of an activating mutation in the ligand-binding domain of the *ESR1* gene.[1]
- Giredestrant, a highly potent, nonsteroidal, oral, selective estrogen receptor antagonist and degrader (SERD), has a similar mechanism of action to fulvestrant and other oral SERDs, but is superior to them in terms of preclinical potency.[2–4] As well as being well tolerated in the treatment of advanced breast cancer (either as a single agent or combined with the cyclin-dependent kinase 4/6 inhibitor palbociclib), giredestrant has shown encouraging antitumor activity, including in patients with *ESR1*-mutated (*ESR1*m) tumors and in patients who have previously received fulvestrant.[5–8]
- acelERA Breast Cancer (NCT04576455) was a randomized, Phase II study that compared giredestrant with physician's choice of endocrine therapy (PCET) in patients with ER-positive, HER2-negative, previously treated advanced breast cancer.[9] The primary endpoint was investigator-assessed progression-free survival (PFS), and although acelERA Breast Cancer did not reach statistical significance for this, a numeric improvement was shown for giredestrant vs. PCET (hazard ratio [HR] 0.81; 95% confidence interval [CI] = 0.60, 1.10; p = 0.18).[9] There was also a consistent treatment effect across most key subgroups, which was more pronounced in patients with *ESR1*m tumors (HR 0.60; 95% CI = 0.35, 1.03).[9] Clinical benefit was also observed across common *ESR1* mutations.[10]
- We present an exploratory biomarker analysis of circulating tumor (ct)DNA dynamics.

METHODS

- Clinical cutoff was July 22, 2022, with N = 303 total patients and a median follow-up of 12.9 months.
- Cycle 1, Day 1 (C1D1, n = 229), Cycle 2, Day 1 (C2D1, n = 220), and end of treatment (EOT, n = 155) ctDNA was evaluated by FoundationOne® Liquid CDx (Foundation Medicine, Inc., Cambridge, MA).
- Tumor fraction (TF) is a measure of ctDNA level. TF calculation was based on a composite algorithm incorporating multiple factors including aneuploidy, variant allele frequency, and canonic alterations.

Figure 1: Summary of patient numbers and available ctDNA datasets from acelERA BC

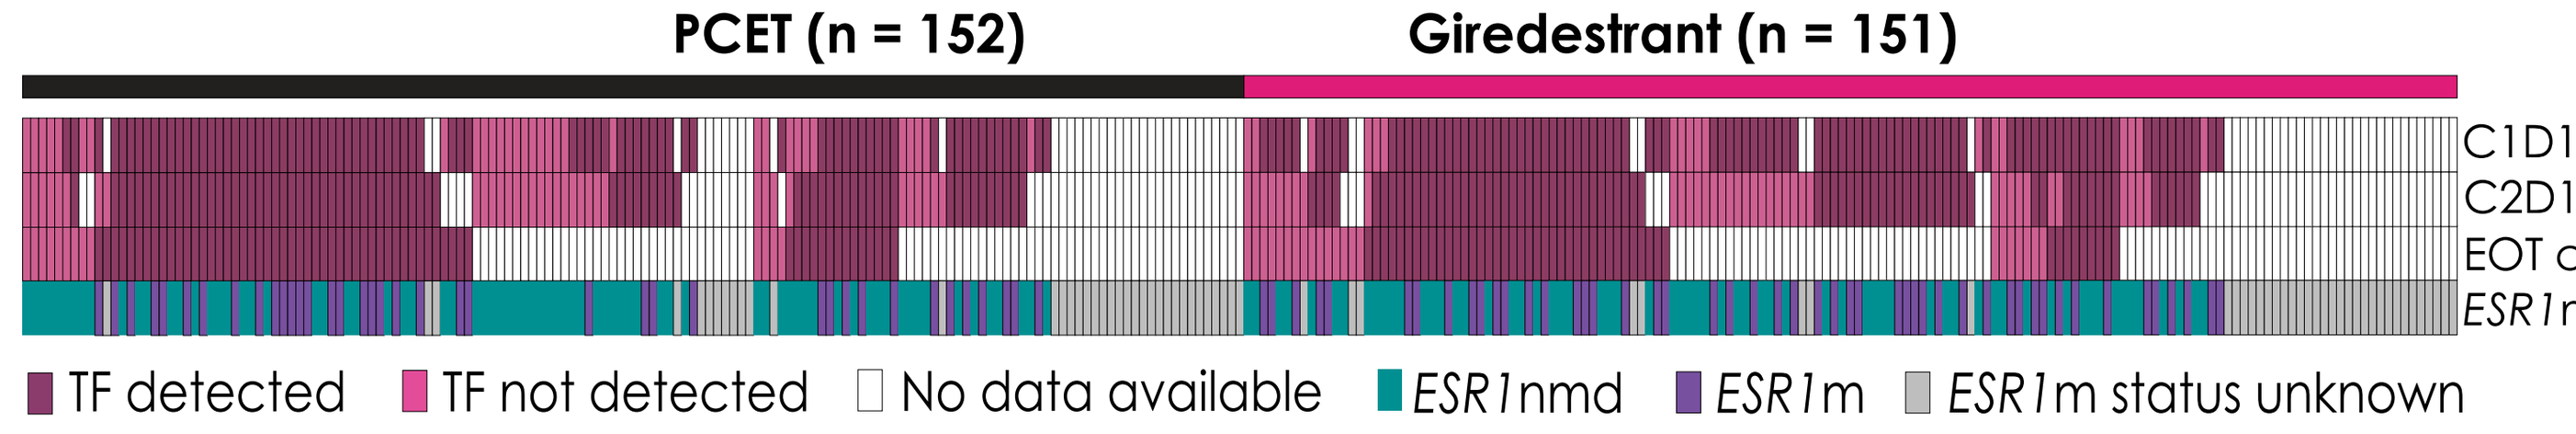
PCET (n = 152)
Giredestrant (n = 151)
C1D1 ctDNA
C2D1 ctDNA
EOT ctDNA
ESR1m status
TF detected
TF not detected
No data available
ESR1nmd
ESR1m
ESR1m status unknown

Number of patients at C1D1		ctDNA detected *ESR1*m	ctDNA detected *ESR1*nmd	ctDNA not detected	Missing
Giredestrant		49	47	18	37
PCET	Aromatase inhibitor	10	11	9	8
	Fulvestrant	33	29	23	29

*ESR1*nmd, no *ESR1* mutation detected.

RESULTS

Figure 2: Longitudinal changes in ctDNA short variants with endocrine therapy

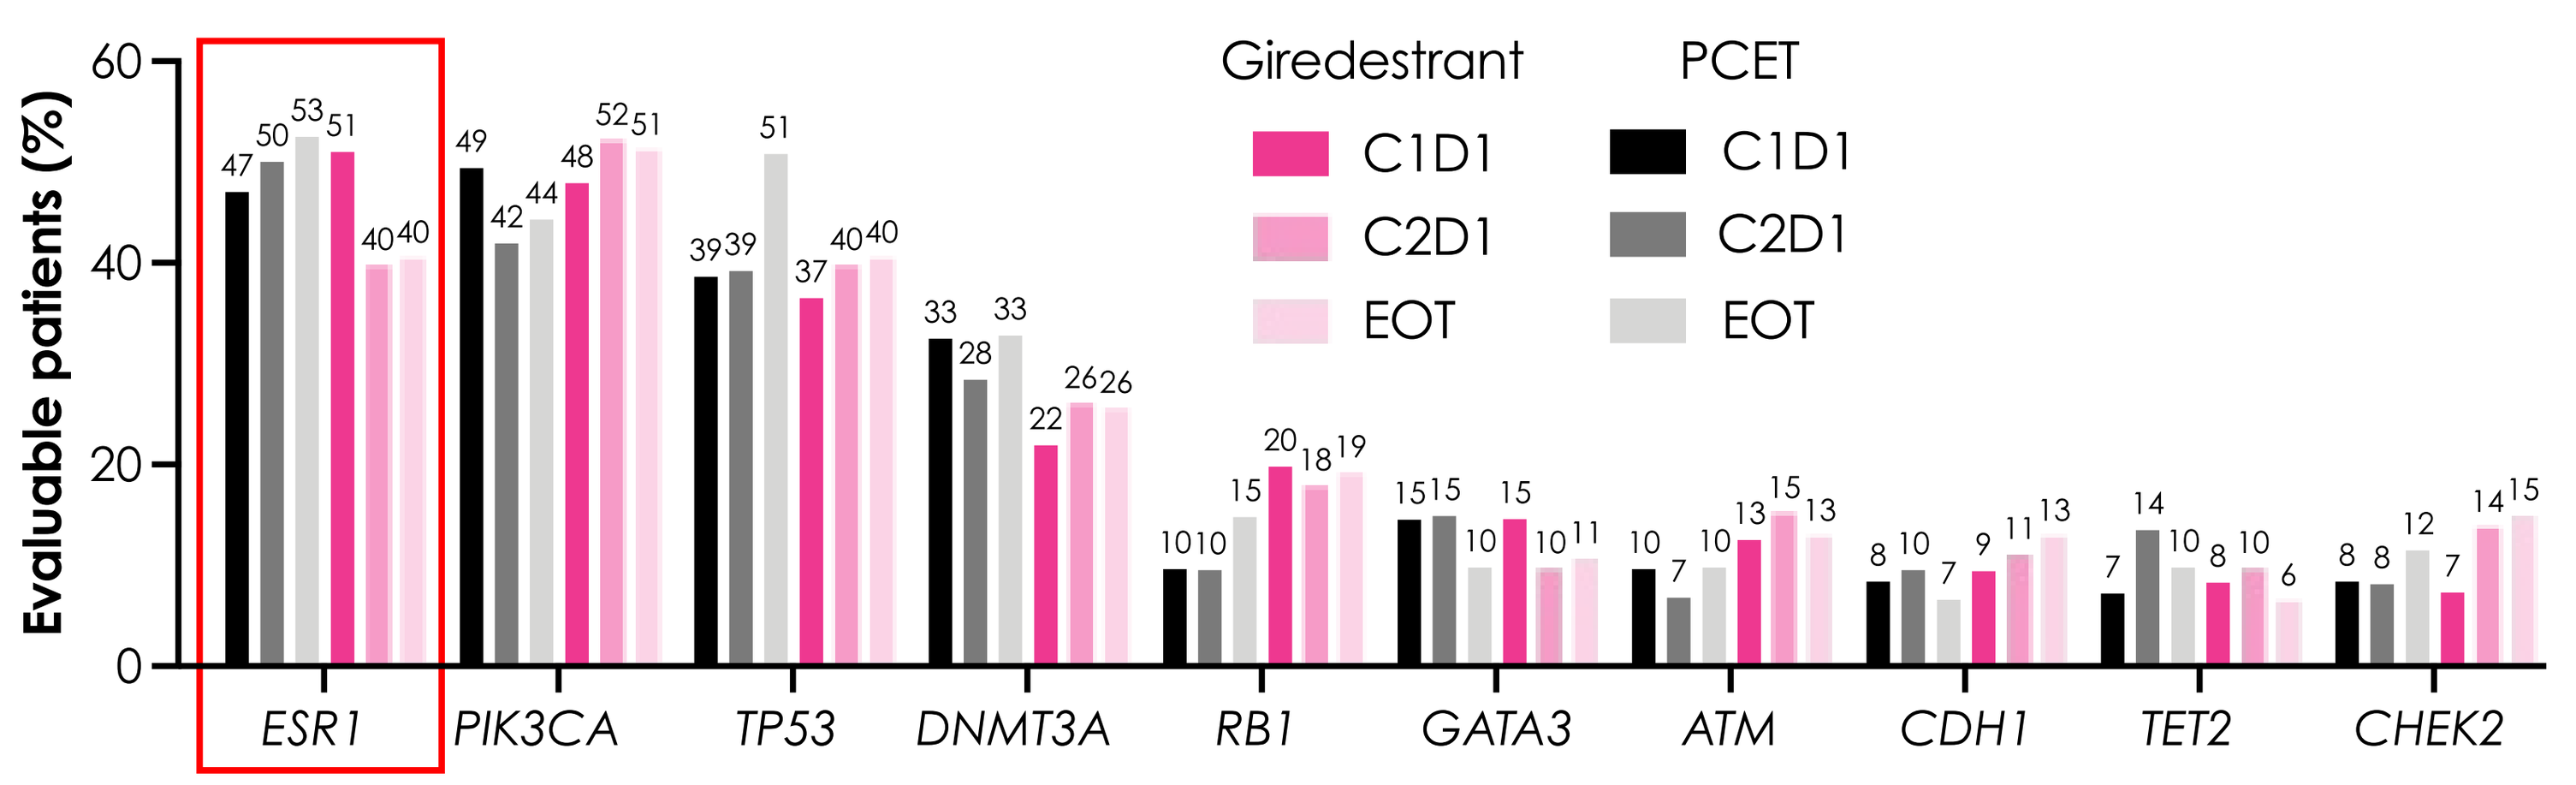
Evaluable patients (%)
Giredestrant
PCET
C1D1
C2D1
EOT
ESR1
PIK3CA
TP53
DNMT3A
RB1
GATA3
ATM
CDH1
TET2
CHEK2

- The prevalence of *ESR1*m ctDNA decreased longitudinally with giredestrant, and increased slightly with PCET.
- The prevalence of other gene mutations largely remained relatively stable on-treatment in both treatment arms.
- Patients without detectable TF were excluded from this analysis.
- Giredestrant: C1D1 n = 96; C2D1 n = 73; EOT n = 47; PCET: C1D1 n = 83; C2D1 n = 74; EOT n = 61.

RESULTS

Figure 3: ctDNA levels and dynamics were associated with response to endocrine therapy

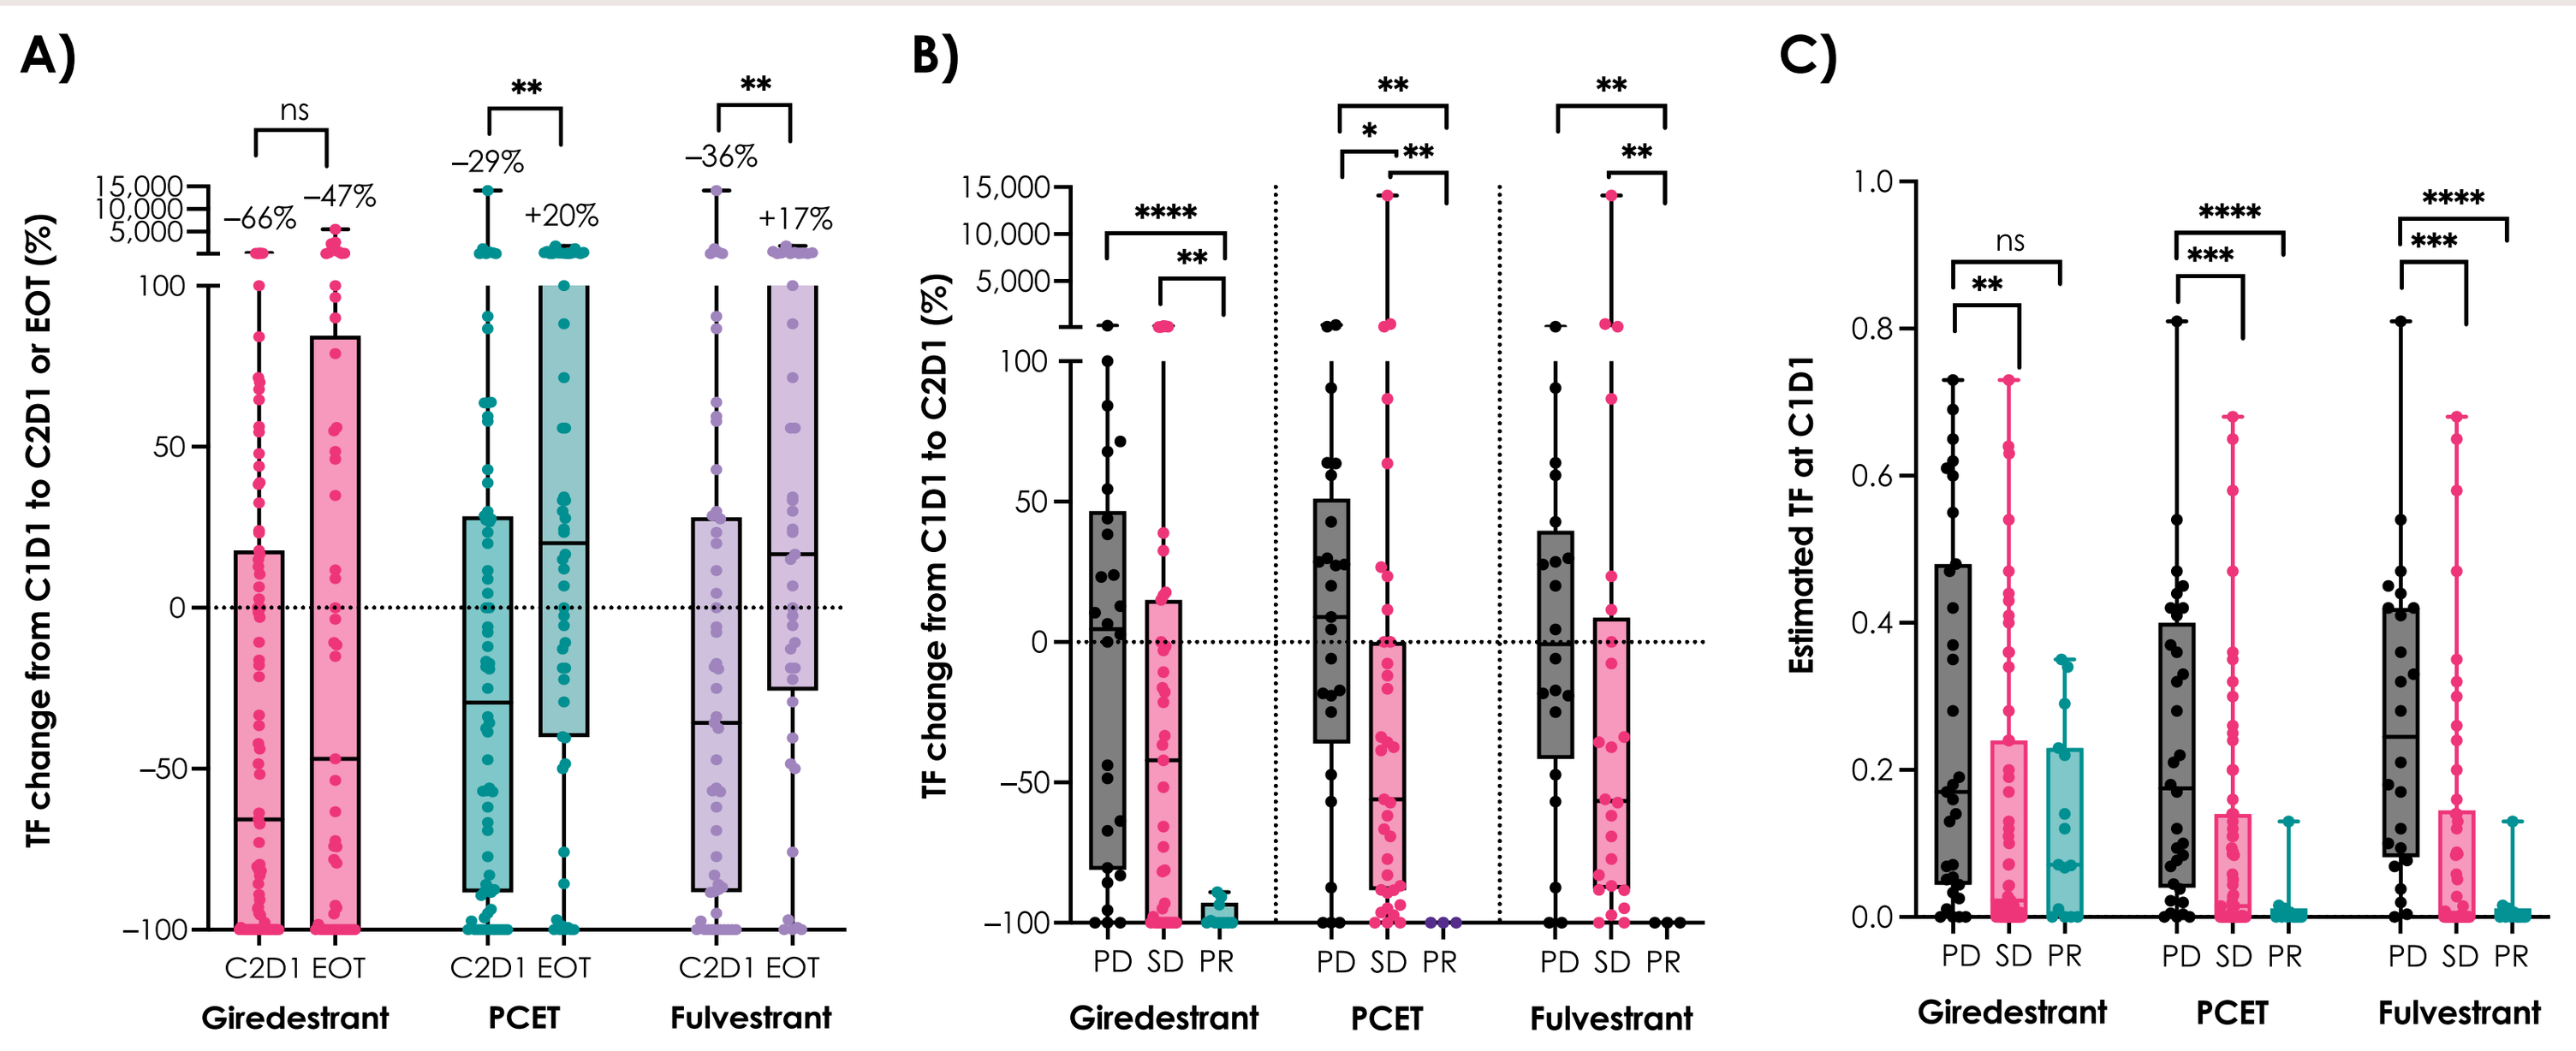
A)
TF change from C1D1 to C2D1 or EOT (%)
–66%
–47%
–29%
+20%
–36%
+17%
C2D1 EOT
Giredestrant
PCET
Fulvestrant
B)
TF change from C1D1 to C2D1 (%)
PD SD PR
C)
Estimated TF at C1D1

- Median ctDNA levels were not significantly different at baseline between arms: giredestrant 0.100 (95% CI = 0.044, 0.160); PCET 0.073 (0.020, 0.120); fulvestrant 0.085 (0.016, 0.140).
- ctDNA levels decreased in 58/89 (65%) of giredestrant-treated patients, 44/75 (59%) of PCET-treated patients, and 34/56 (61%) of fulvestrant-treated patients with detectable/estimable TF at baseline.
- A) The median change from C1D1 to C2D1 was greater with giredestrant (–66%) compared with PCET (–29%) or fulvestrant (–36%). At EOT, PCET-treated patients saw a significant increase in ctDNA levels relative to C1D1. This effect was not seen with giredestrant, which still showed a median of –47% ctDNA reduction.
- B) and C) ctDNA reduction was significantly higher and baseline ctDNA levels were significantly lower in patients with PR or SD compared with those with PD (best investigator response), regardless of the treatment arm.

Median values are listed above the respective bars. * p < 0.05; ** p < 0.01; *** p < 0.001; **** p < 0.0001 (Mann–Whitney U test). ns, not significant; PD, progressive disease; PR, partial response; SD, stable disease.

Figure 4: ctDNA reduction at C2D1 with giredestrant was improved in patients with *ESR1*m tumors

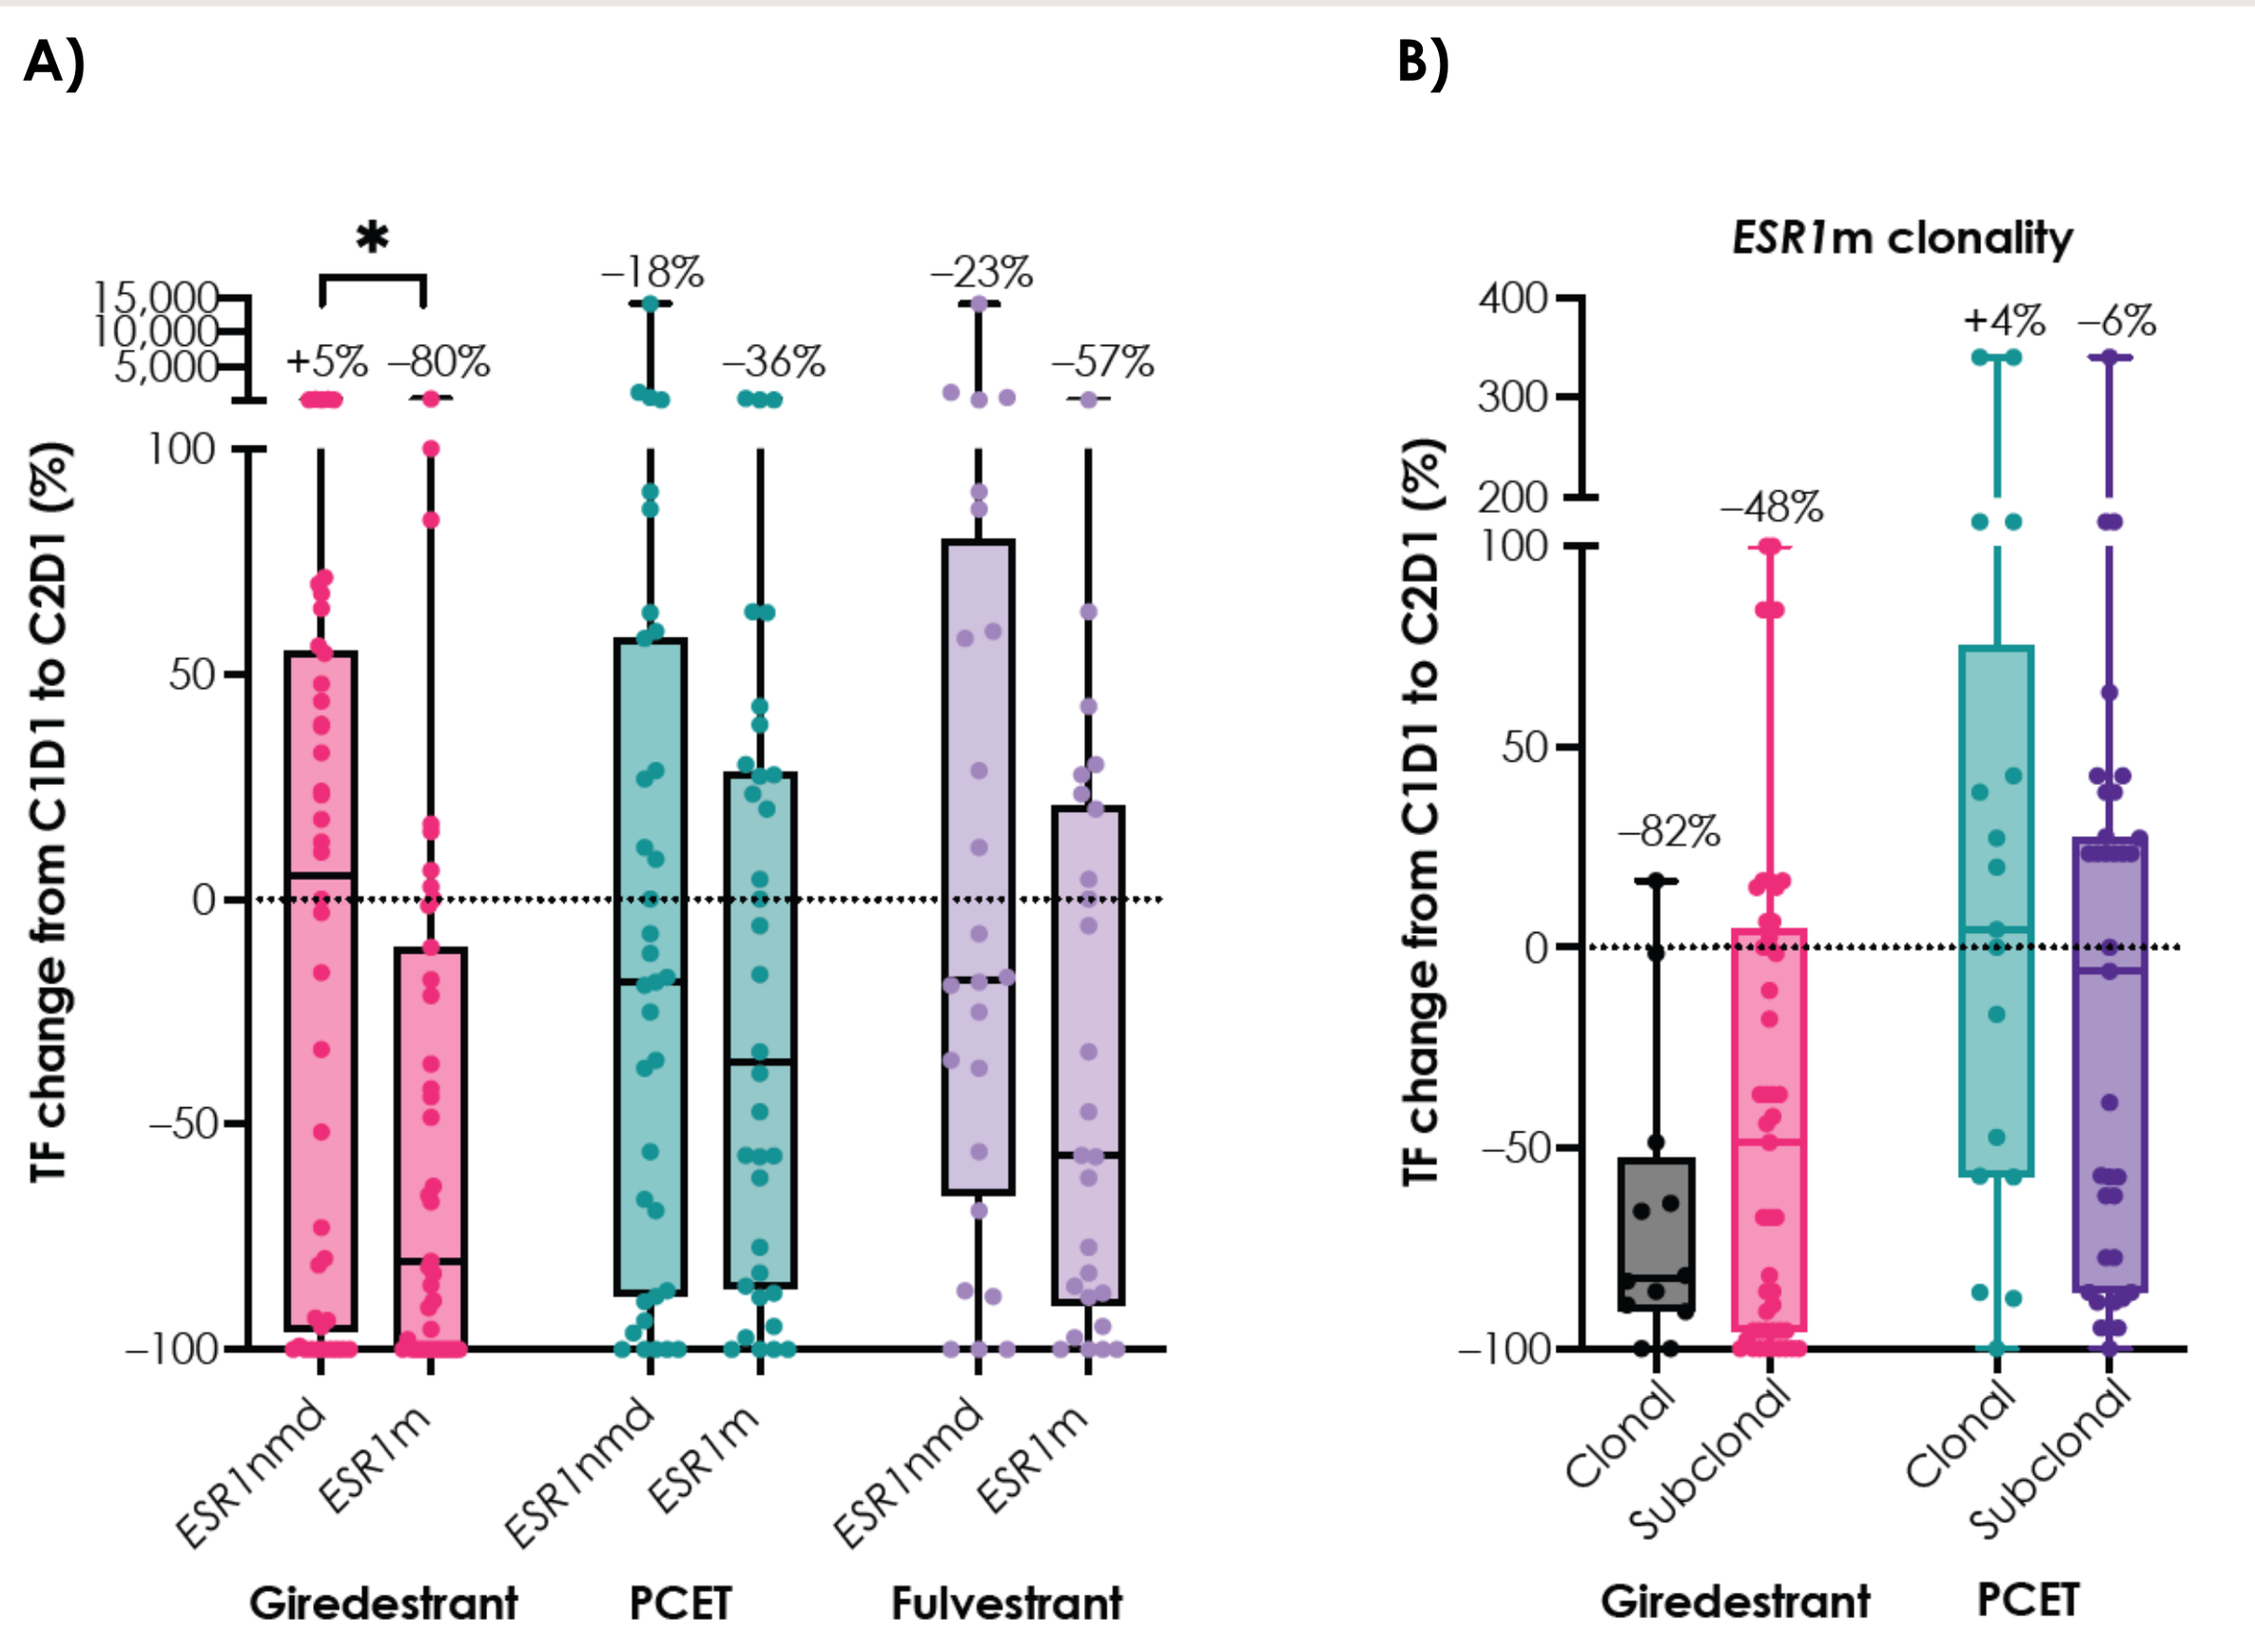
A)
TF change from C1D1 to C2D1 (%)
+5%
–80%
–18%
–36%
–23%
–57%
ESR1nmd
ESR1m
Giredestrant
PCET
Fulvestrant
B)
ESR1m clonality
–82%
–48%
+4%
–6%
Clonal
Subclonal

- A) Specifically in response to giredestrant, the level of ctDNA reduction was greater in patients with *ESR1*m tumors (–80%) vs. those with *ESR1*nmd (+5%).
- B) Patients with clonal *ESR1*m tumors experienced greater ctDNA reduction in response to giredestrant.

Median values are listed above the respective bars. * p = 0.0165 (Mann–Whitney U test).

Figure 5: *ESR1* MAF reduction was significantly improved with giredestrant over PCET, particularly in patients with D538G and Y537X variants

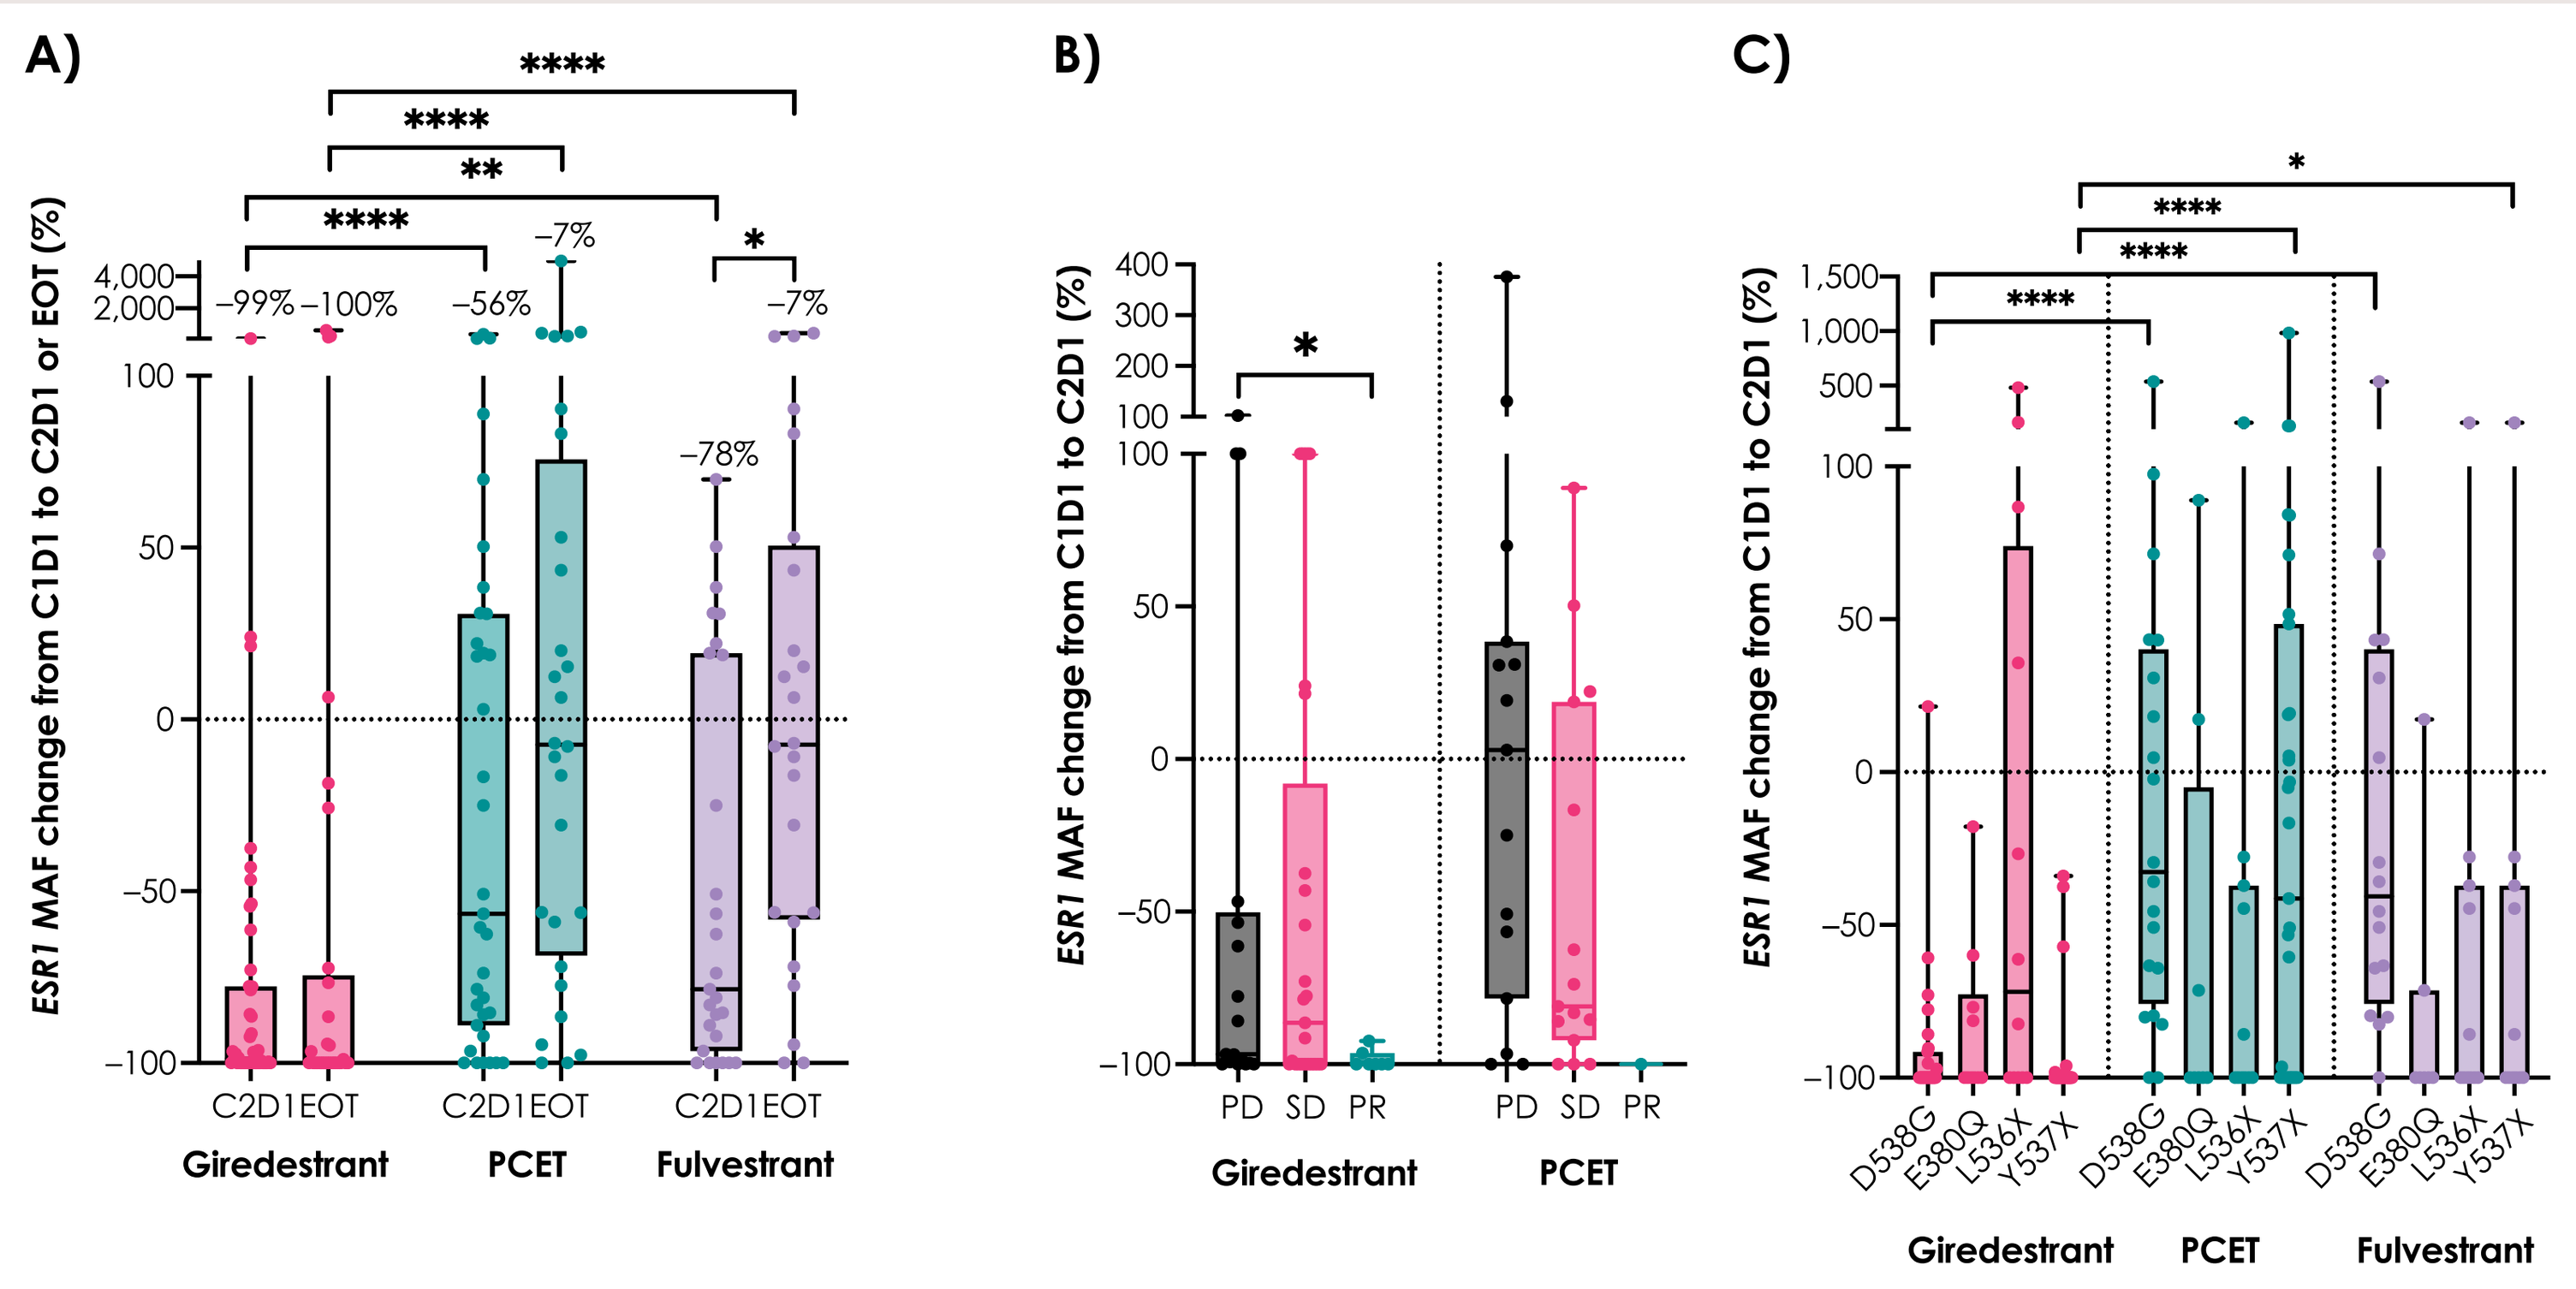
A)
ESR1 MAF change from C1D1 to C2D1 or EOT (%)
–99%
–100%
–56%
–7%
–78%
–7%
C2D1 EOT
Giredestrant
PCET
Fulvestrant
B)
ESR1 MAF change from C1D1 to C2D1 (%)
PD SD PR
C)
D538G
E380Q
L536X
Y537X

- Median *ESR1* MAF levels were not significantly different at baseline between arms: giredestrant 1.6 (95% CI = 0.56, 3.03); PCET 2.8 (0.62, 7.70); fulvestrant 2.29 (0.62, 7.70).
- A) *ESR1* MAF reduction on-treatment was significantly greater in response to giredestrant (–99%) vs. PCET (–56%) or fulvestrant (–78%). This observation was consistent in EOT samples.
- B) A trend for greater on-treatment changes in *ESR1* MAF was observed in patients with PR, compared with those with PD or SD, regardless of treatment arm.
- C) *ESR1* MAF reduction was significantly greater for D538G and Y537X variants in response to giredestrant (–100%) vs. PCET (–33% and –42%) or fulvestrant (–41% and –100%).

Median values are listed above the respective bars. In patients with more than one *ESR1* mutation, the MAF is represented as a sum. * p < 0.05; ** p = 0.0063; *** p < 0.001; **** p < 0.0001 (Mann–Whitney U test). MAF, mutant allele frequency.

Figure 6: ctDNA detection was a prognostic biomarker for endocrine therapy both at baseline and on-treatment

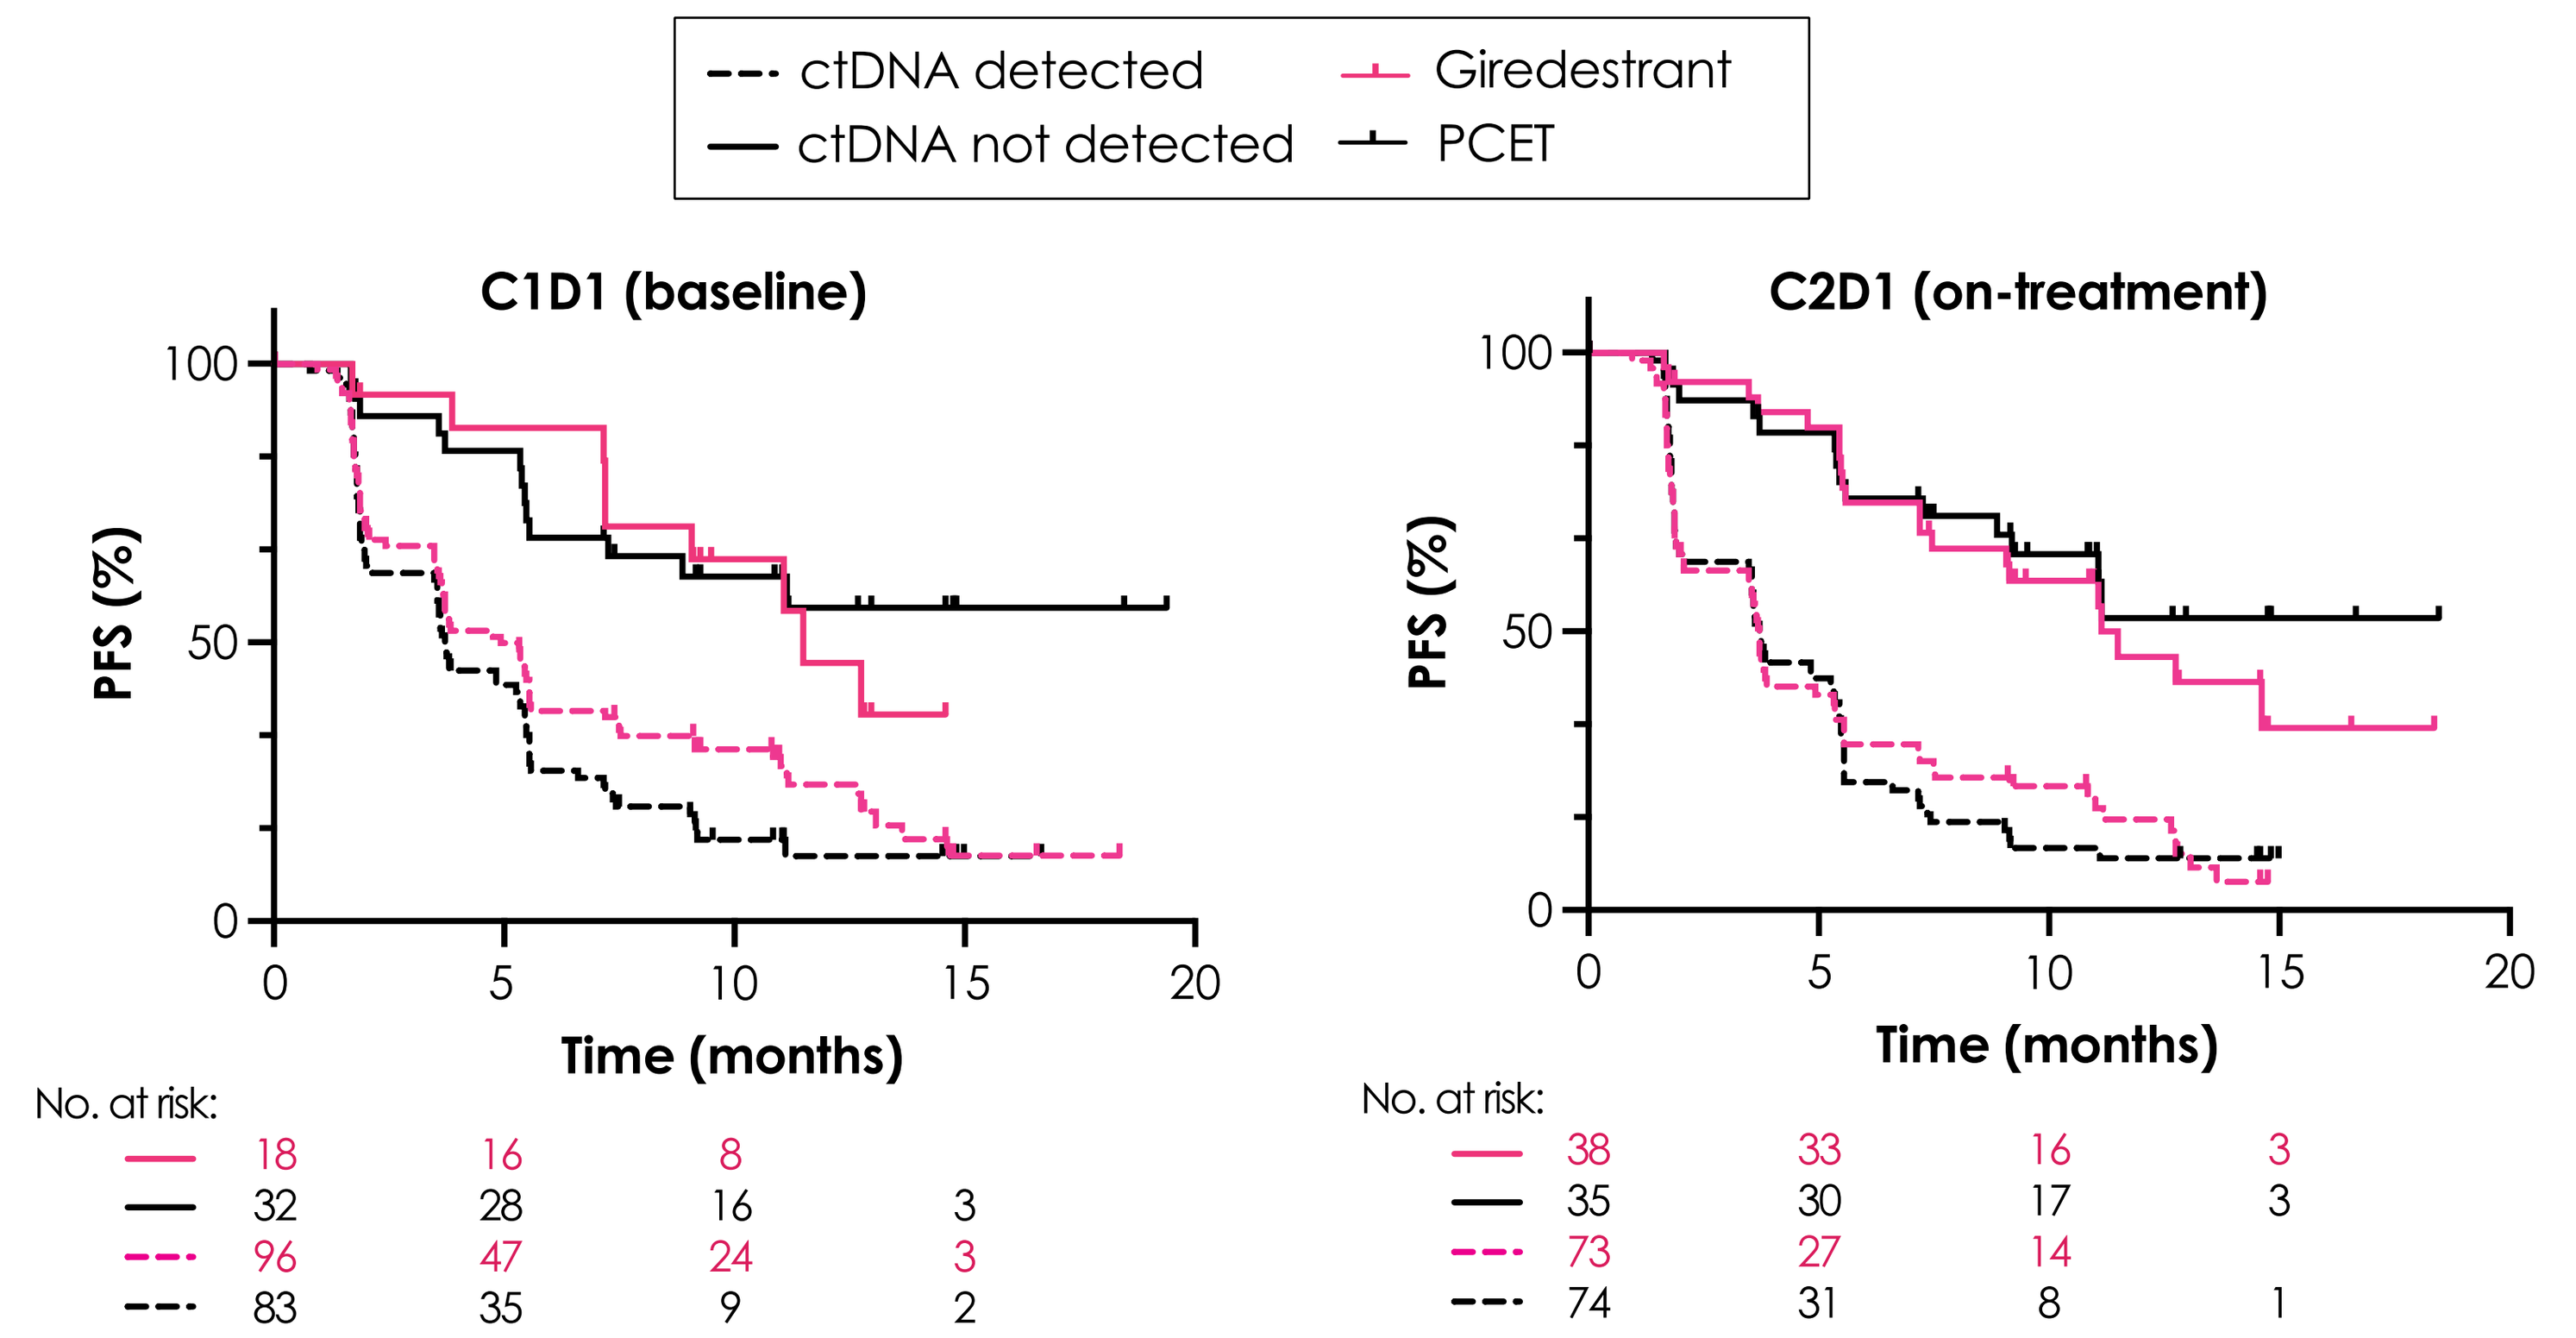
ctDNA detected
ctDNA not detected
Giredestrant
PCET
C1D1 (baseline)
C2D1 (on-treatment)
PFS (%)
Time (months)
No. at risk:
18 16 8
32 28 16 3
96 47 24 3
83 35 9 2
38 33 16 3
35 30 17 3
73 27 14
74 31 8 1

- Better PFS was observed, regardless of treatment arm, in patients with no detectable ctDNA at both C1D1 and C2D1.

Figure 7: ctDNA clearance on-treatment was a prognostic biomarker for endocrine therapy

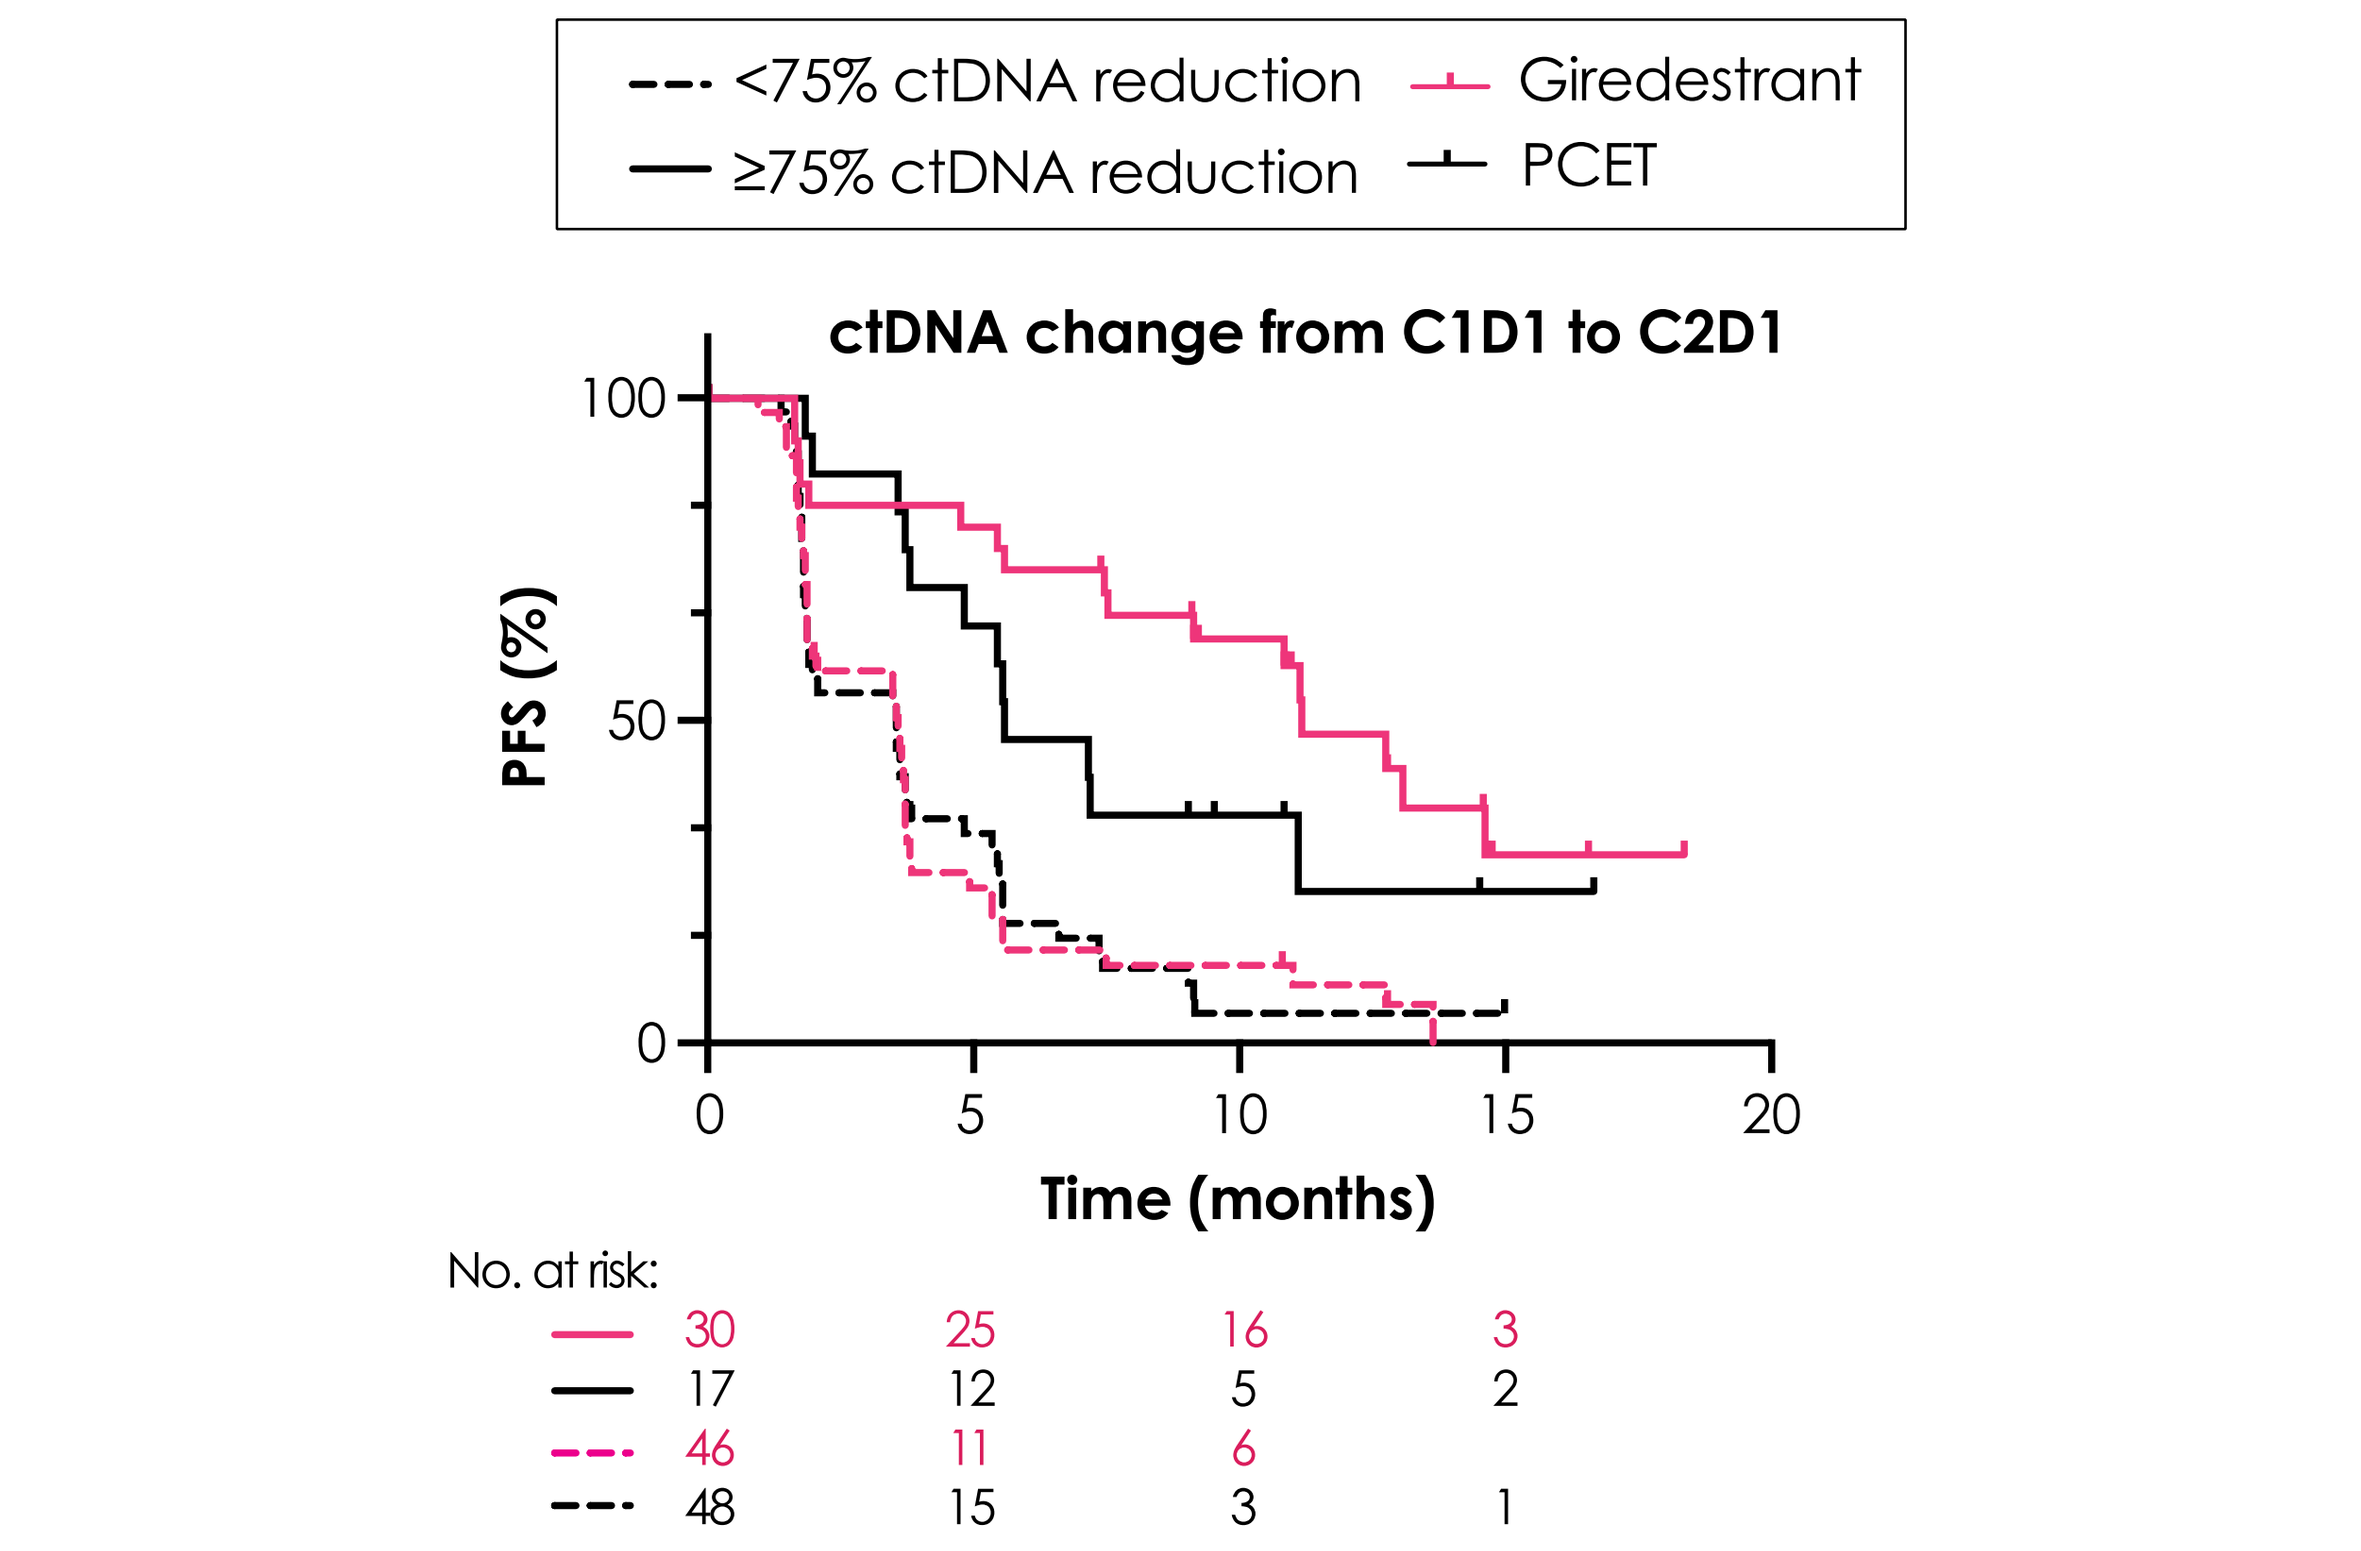
<75% ctDNA reduction
≥75% ctDNA reduction
Giredestrant
PCET
ctDNA change from C1D1 to C2D1
PFS (%)
Time (months)
No. at risk:
30 25 16 3
17 12 5 2
46 11 6
48 15 3 1

- Better PFS was observed, regardless of treatment arm, in patients who exhibited ctDNA reduction of ≥75% on-treatment.
- This effect was greater with giredestrant compared with PCET.

CONCLUSIONS

- **ctDNA levels decreased on-treatment in the majority of patients treated with either giredestrant or PCET; giredestrant elicited a greater magnitude of ctDNA reduction compared with PCET.**
- **Specifically in response to giredestrant, ctDNA reduction was greater in patients with *ESR1*m tumors, particularly those with clonal mutations.**
- **Reduction of ctDNA and *ESR1* MAF was associated with better response, regardless of treatment arm; PFS was improved in patients with high ctDNA reduction in both treatment arms, but to a greater degree with giredestrant.**
- ***ESR1* MAF reduction was greater with giredestrant (–99%) vs. PCET (–56%) or fulvestrant (–78%).**
- **ctDNA detection both at baseline and on-treatment was associated with poor prognosis.**

CONFLICTS OF INTEREST

AC reports employment by Genentech, Inc. and stocks/shares in F. Hoffmann-La Roche Ltd.
Please refer to the abstract for all author conflicts of interest.
All authors have received research support in the form of third-party writing assistance for this poster from F. Hoffmann-La Roche Ltd. This study is sponsored by F. Hoffmann-La Roche Ltd.

ACKNOWLEDGMENTS

We thank the patients who participated in this study, their families, and the staff, research coordinators, and investigators at each participating institution.
Research support in the form of third-party writing assistance for this poster, furnished by Daniel Clyde, PhD, of Health Interactions, was provided by F. Hoffmann-La Roche Ltd.

REFERENCES

1. Jeselsohn R, *et al*. *Nat Rev Clin Oncol* 2015; **12:**573–583; 2. Metcalfe C, *et al*. SABCS 2018; P5-04-07;
3. Liang J, *et al*. *J Med Chem* 2021; **64:**11841–11856; 4. Chen YC, *et al*. *Expert Opin Investig Drugs* 2022; **31:**515–529;
5. Moore HM, *et al*. ASCO 2021; 577; 6. Oliveria M, *et al*. ASCO 2023; 1061;
7. Hurvitz SA, *et al*. *Lancet Oncol* 2023; **24:**1029–1041;
8. Jhaveri KL, *et al*. *Clin Cancer Res* 2023; ePub ahead of print;
9. Martin Jimenez M, *et al*. ESMO 2022; 211MO; 10. Lim E, *et al*. SABCS 2022; PD13-04.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the authors of this poster.

This presentation is the intellectual property of the authors. Contact collier.annie@gene.com for permission to reprint and/or distribute.